Abstract No. 1782-P

Phase 1b Study Results of AMPK/Mitochondrial Activator ATX-304 in Prediabetic Obese Participants

Ruth I Thieroff-Ekerdt*(1), David Jacobs(2), James A Hall(2), Erik J Schneider(1), Jessica Ainscough(1), James G Peyer(1), Tim Heise(3)
*corresponding author

Cambrian Bio (1); Betagenon AB (2); Profil Institut für Stoffwechselforschung GmbH (3)

AMPLIFIER OWNED BY Cambrian

BACKGROUND 1

Obesity is a highly prevalent condition and an independent risk factor for cardiometabolic diseases[1]

- ATX-304 is a clinical stage orally administered, peripherally acting small molecule in development for weight management with a novel mode of action
 - ATX-304 is a dual AMPK and mitochondrial activator
 - In vitro and preclinical data suggest that the combined action appears to avoid the safety issue of AMPK activators (glycogen accumulation) and mitochondrial uncoupling (increase in heart rate and hyperthermia)

- In high fat diet-induced and genetic animal models of obesity and diabetes, ATX-304 showed pleiotropic effects
 - At lower doses (≤0.8 mg/g food), ATX-304 improved glucose and lipid metabolism, reduced liver fat and hepatic de novo lipogenesis, and increased energy expenditure[2,3]
 - At higher doses (2 mg/g food in rodents[2], up to 300 mg/kg in dogs[4], at an exposure of 4500-5500 µg/mL·h), ATX-304 reduced body weight without decrease in food consumption[2]

ATX-304 is an investigational drug agent that is not available for human use outside a clinical trial setting.

The effects of ATX-304 were assessed in a Phase 1b translational study in patients with prediabetes and obesity.

METHODS 2

- **Study Design and Sample Size:** Randomized, double-blind, placebo-controlled, optional open label extension (OLE); N=23, 2:1 randomization (15 participants on ATX-304, 8 on placebo)

- **Study Population:**
 - Prediabetic (HbA1c 5.7-6.4%) and impaired fasting plasma glucose or impaired glucose tolerance
 - Obese (BMI 30-39 kg/m²)

Fig 1: Treatment and Endpoints

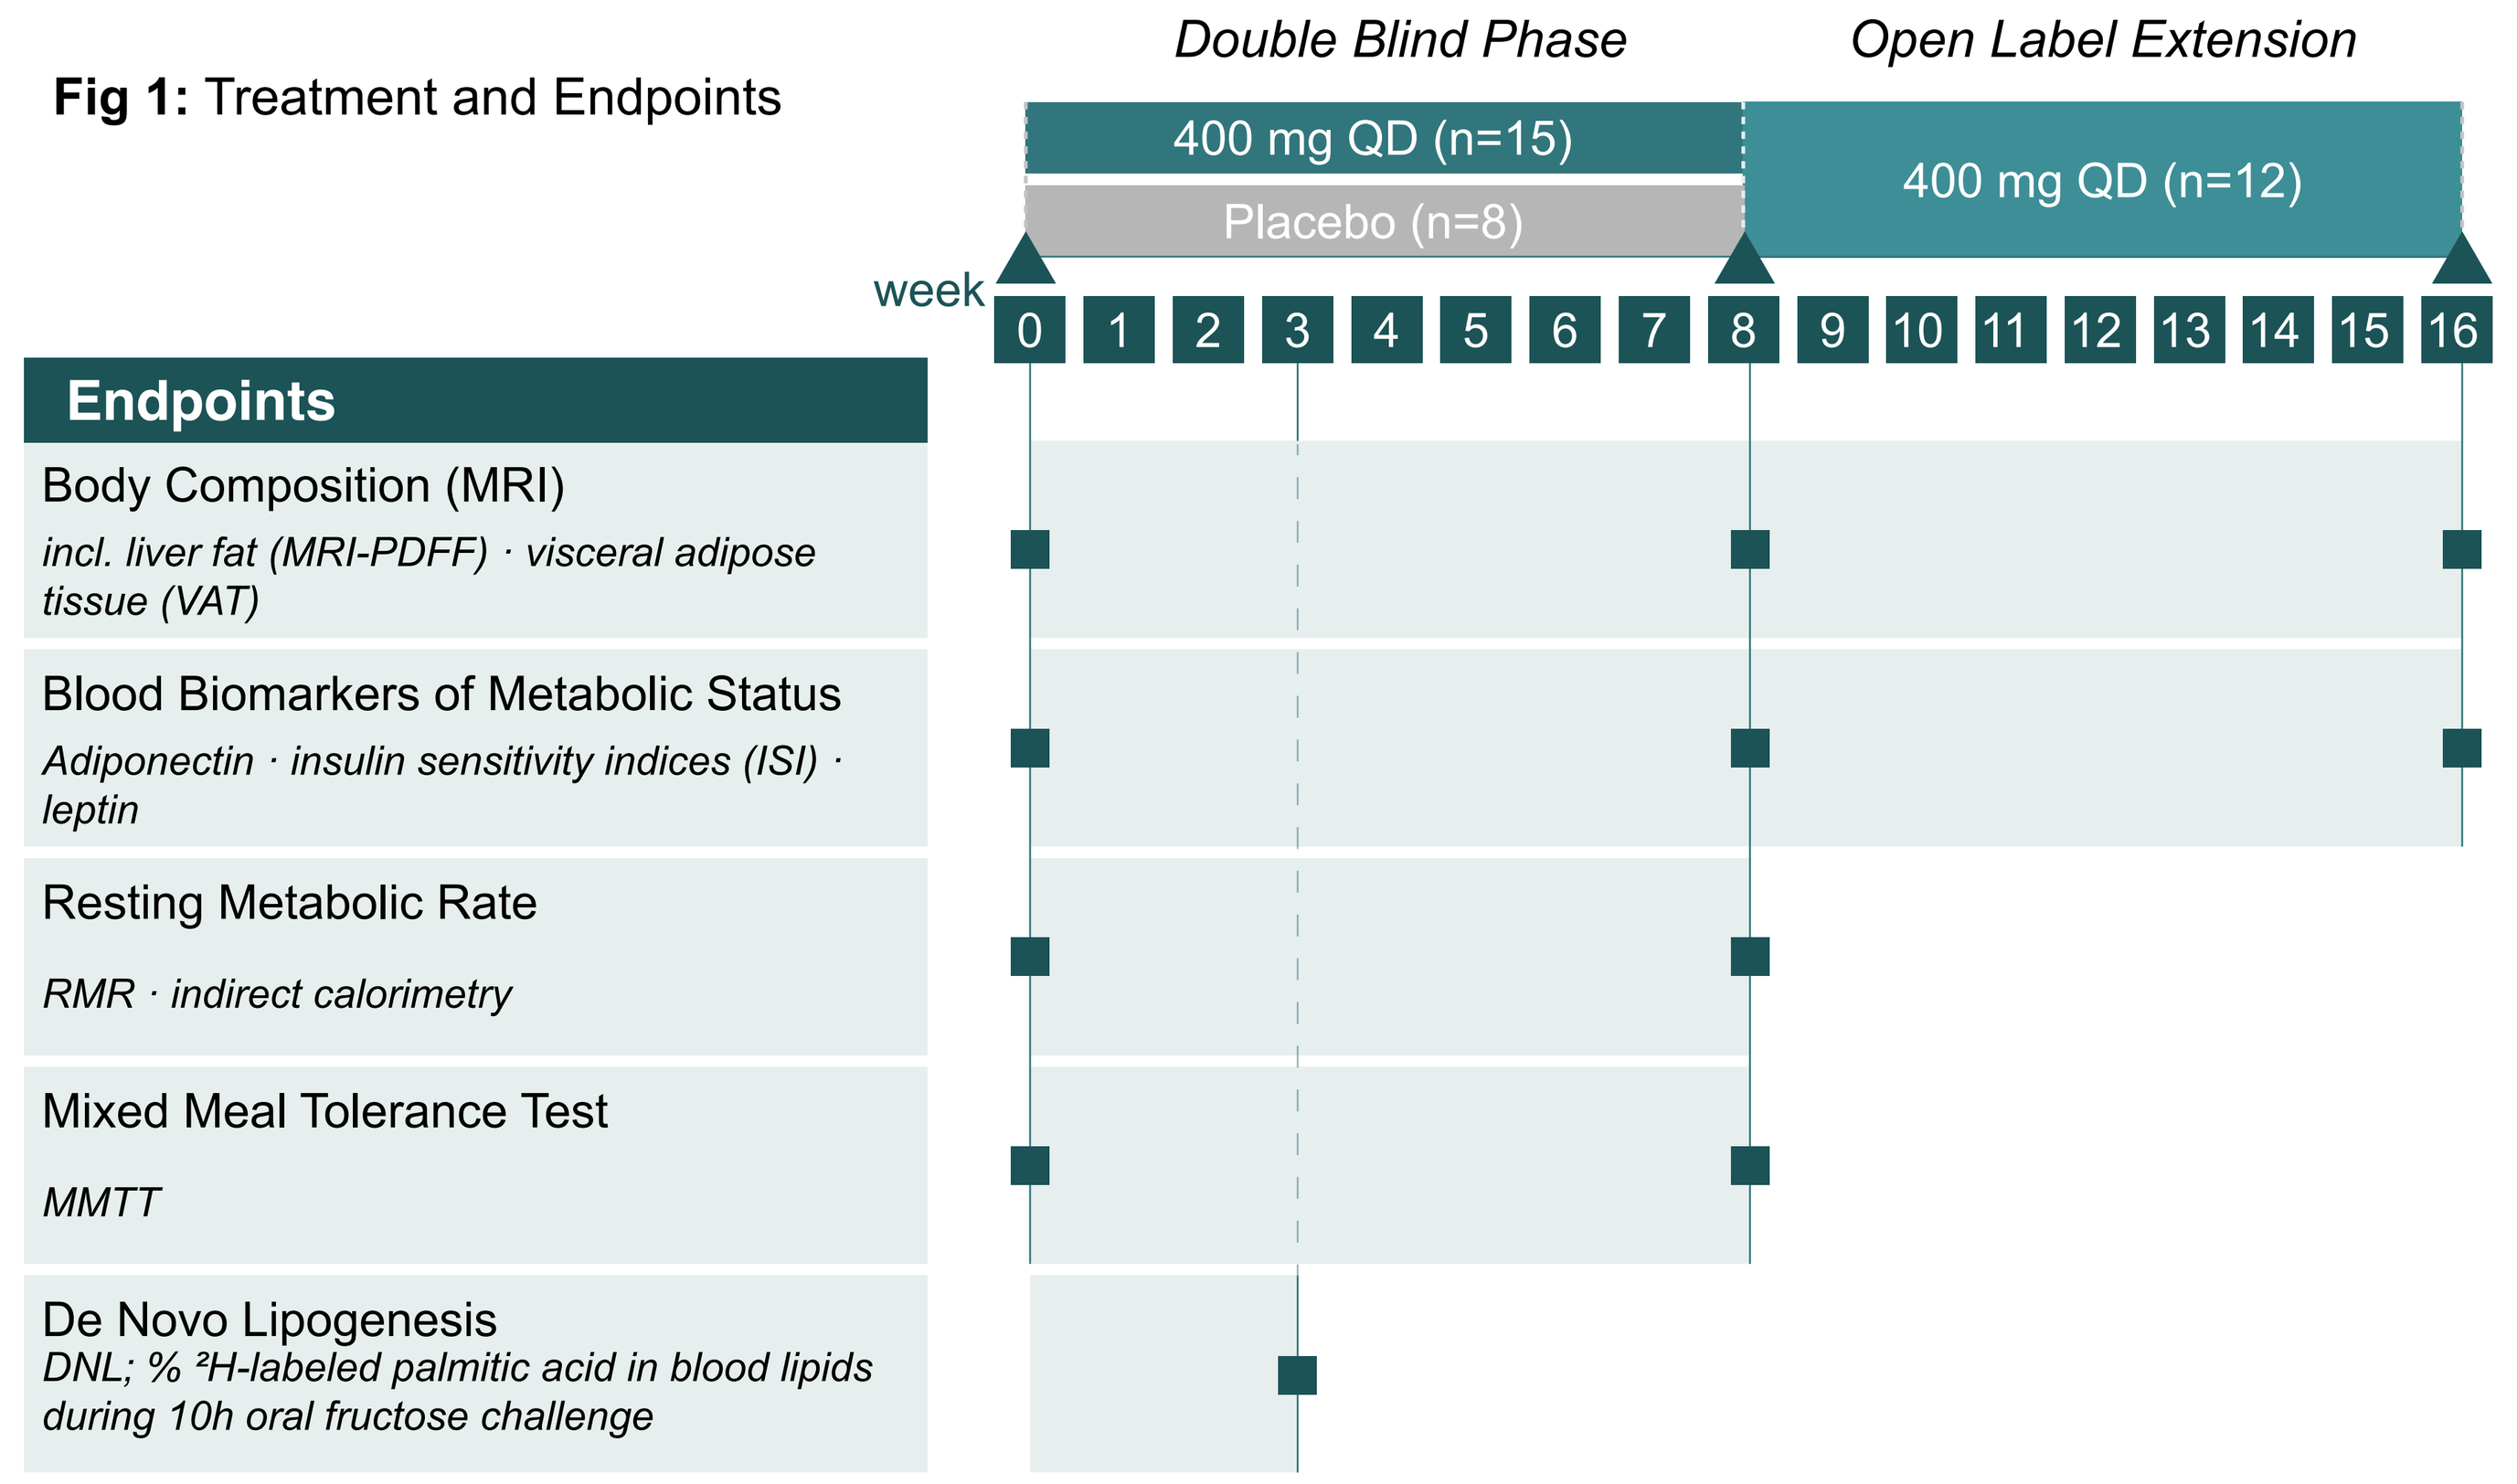

Double Blind Phase
Open Label Extension
400 mg QD (n=15)
Placebo (n=8)
400 mg QD (n=12)
week
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16
Endpoints
Body Composition (MRI)
incl. liver fat (MRI-PDFF) · visceral adipose tissue (VAT)
Blood Biomarkers of Metabolic Status
Adiponectin · insulin sensitivity indices (ISI) · leptin
Resting Metabolic Rate
RMR · indirect calorimetry
Mixed Meal Tolerance Test
MMTT
De Novo Lipogenesis
DNL; % ²H-labeled palmitic acid in blood lipids during 10h oral fructose challenge

RESULTS 3

Study Disposition, Demographics, and Safety

DISPOSITION:
- N=23 (15 ATX-304, 8 placebo) enrolled and completed placebo-controlled 8-week treatment period
- N=12 (8 continued ATX-304, 4 switched from placebo) enrolled and completed 8-week OLE period

DEMOGRAPHICS
- White (95.7%), Non-Hispanic (95.7%), Male (73.9%), mean age 54.2 yrs, BMI 34.3 kg/m², mean HbA1c 5.88%
- Higher baseline liver fat and fasted insulin in ATX-304 group, otherwise balanced baseline condition

SAFETY
- Predominantly mild treatment-emergent adverse events (TEAE), similar frequency to placebo
- No adverse events indicative of mitochondrial uncoupling, particularly no increase in continuously monitored core body temperature and 24-hour heart rate
- No new safety signals in OLE

Table 1: All TEAEs in more than 1 Participant

Preferred Term	ATX-304 (N=15) N (%)/Events/Related Events	Placebo (N=8) N (%)/ Events/Related Events
Diarrhea*	8 (53.3) / 8 / **4**	2 (25) / 2 / **2**
Dizziness	4 (26.7) / 4 / **3**	1 (12.5) / 1 / **0**
Vomiting*	4 (26.7) / 4 / **2**	0
Decreased Appetite+	3 (20) / 3 / **3**	1 (12.5) / 1 / **0**
Nausea*	2 (13.3) / 2 / **1**	2 (25) / 3 / **3**
Headache	2 (13/3) / 2 / **2**	1 (12.5) / 1 / **1**
Dyspepsia	1 (6.7) / 1 / **1**	2 (25) / 2 / **2**
Flatulence	1 (6.7) / 1 / **1**++	1 (12.5) / 1 / **1**
Fatigue	0	2 (25) / 2 / **2**

* A cluster of gastrointestinal AEs events was observed during fructose challenge for measurement of DNL
\+ Events on single days
++ Event of mild flatulence 8 h after last treatment (D57), considered related by investigator, serious due to overnight hospitalization, resolved within 6.25 hours

PK and PD

PHARMACOKINETICS
- Mean C_{max} (SD) 98.8 µg/mL (32.7); estimated AUC_{0-24} 2200-2400 h*µg/mL
 - Lower in this study with obese participants than observed in normal weight participants in previous study with same dose (138.4 µg/mL)
- Mean plasma $t_{1/2}$ (SD) 98.8h (21.8); steady state reached after appr. 21 days
 - Minimal fluctuation of plasma concentration over dosing interval

PHARMACODYNAMICS
- Significant changes in lipid metabolism, body composition, and energy expenditure
 - Time dependent increase in adiponectin (Fig 2), decrease in plasma triglycerides (Fig 3), liver fat (Fig 4) and VAT (Fig 5), and increase in RMR (Fig 6)
 - Minimal weight loss, as expected at this exposure (Fig 5)
 - Reduction in hepatic DNL in high exposure subgroup (n=6, C_{max}>100µg/ml) (data not shown)
 - No effect on glucose metabolism in MMTT, ISI, and plasma leptin levels (data not shown)

ATX-304 increased fasting adiponectin and decreased triglycerides

Fig 2: Mean % Change from Baseline in Plasma Adiponectin (+/- SEM)

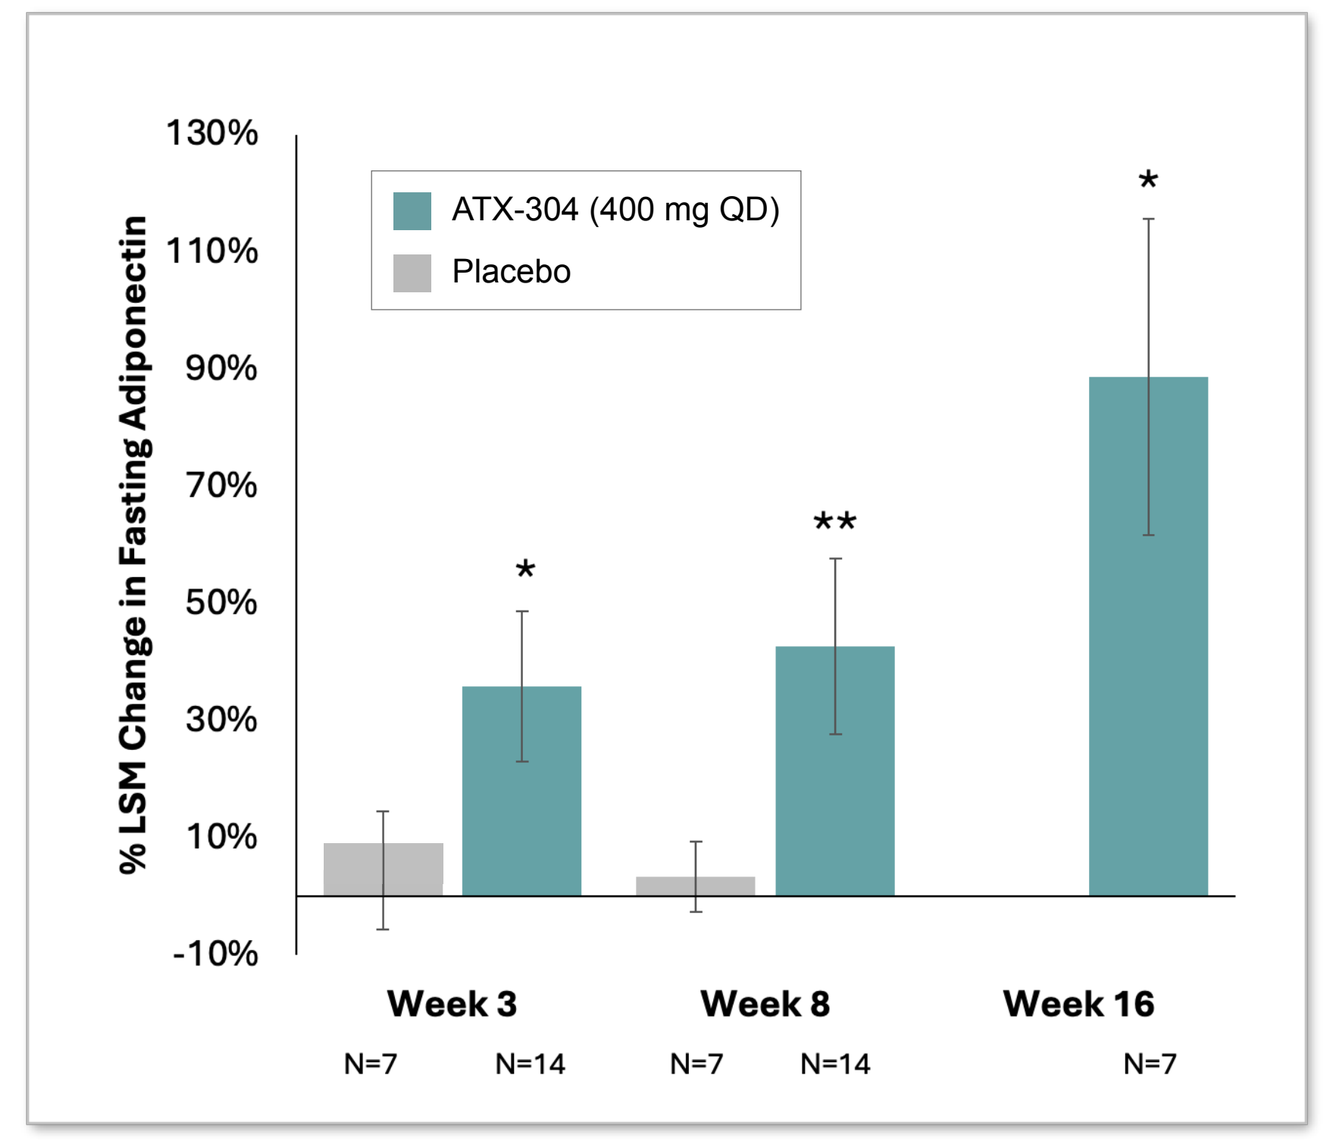

% LSM Change in Fasting Adiponectin
130%
110%
90%
70%
50%
30%
10%
-10%
ATX-304 (400 mg QD)
Placebo
*
**
*
Week 3
Week 8
Week 16
N=7 N=14 N=7 N=14 N=7

*P-value < 0.05; **P-value < 0.01 vs. baseline, ANCOVA based on Least Squares Mean. Week 16 data from a post-hoc analysis in subjects treated with ATX-304 for 16 weeks

Fig 3: Mean % Change from Baseline in Plasma Triglycerides (+/- SEM)

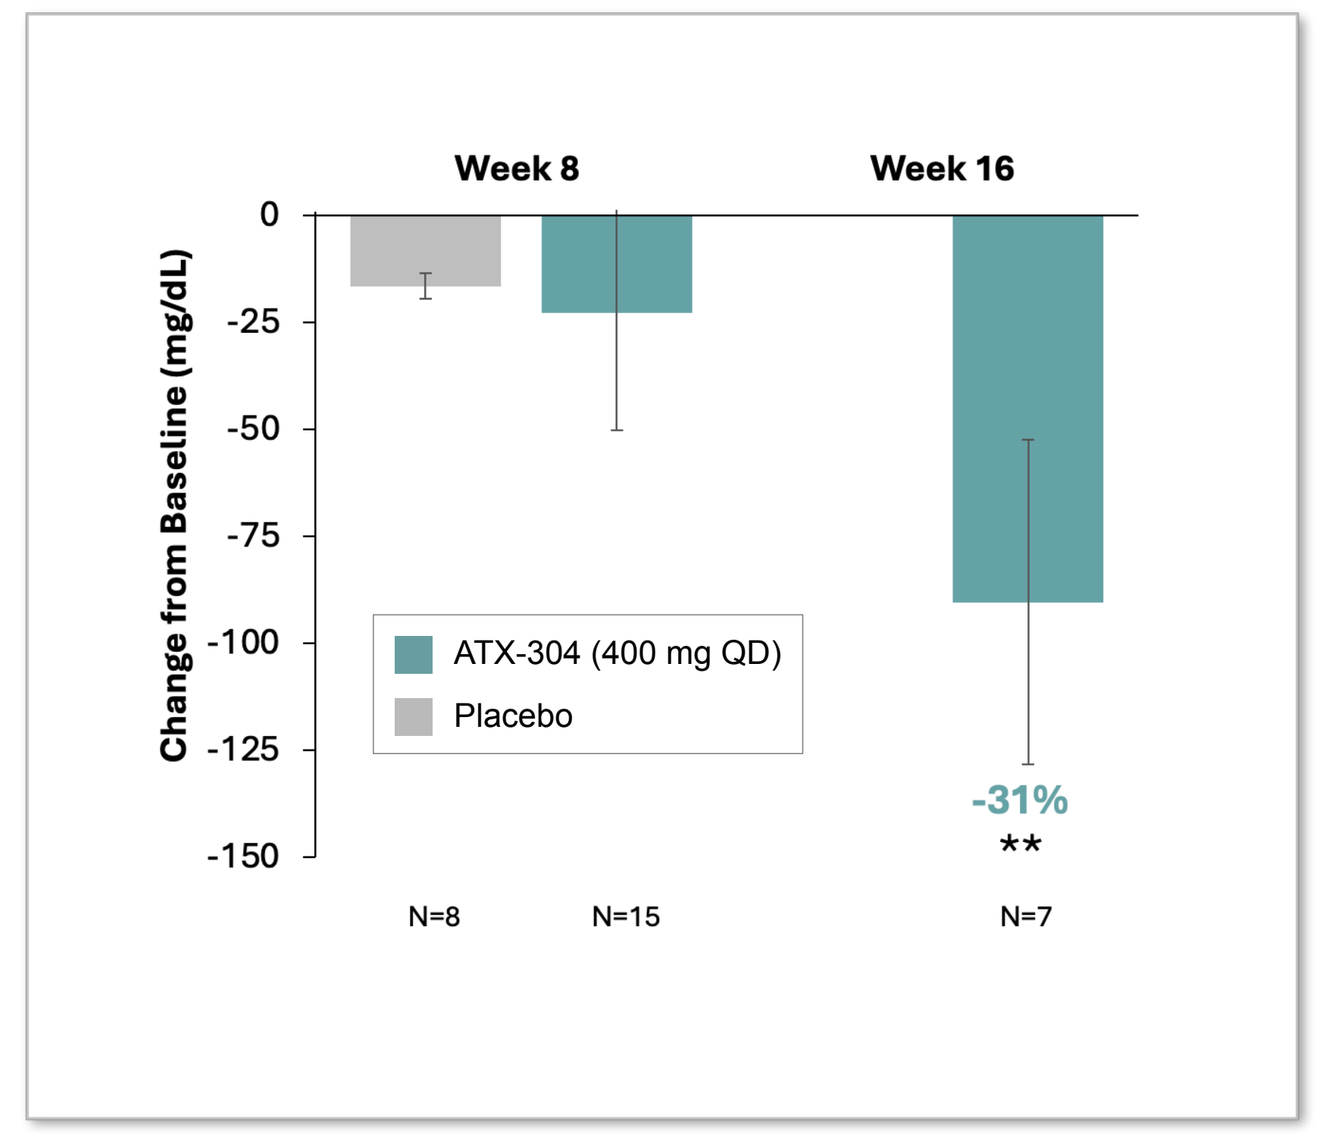

Change from Baseline (mg/dL)
Week 8
Week 16
0
-25
-50
-75
-100
-125
-150
ATX-304 (400 mg QD)
Placebo
-31%
**
N=8 N=15 N=7

**P-value < 0.01 vs. baseline, ANCOVA based on Least Squares Mean. Week 16 data from a post-hoc analysis in subjects treated with ATX-304 for 16 weeks

ATX-304 decreased liver fat and visceral adipose tissue (VAT)

Fig 4: Mean % Change From Baseline in Liver Fat (MRI-PDFF) (+/- SEM)

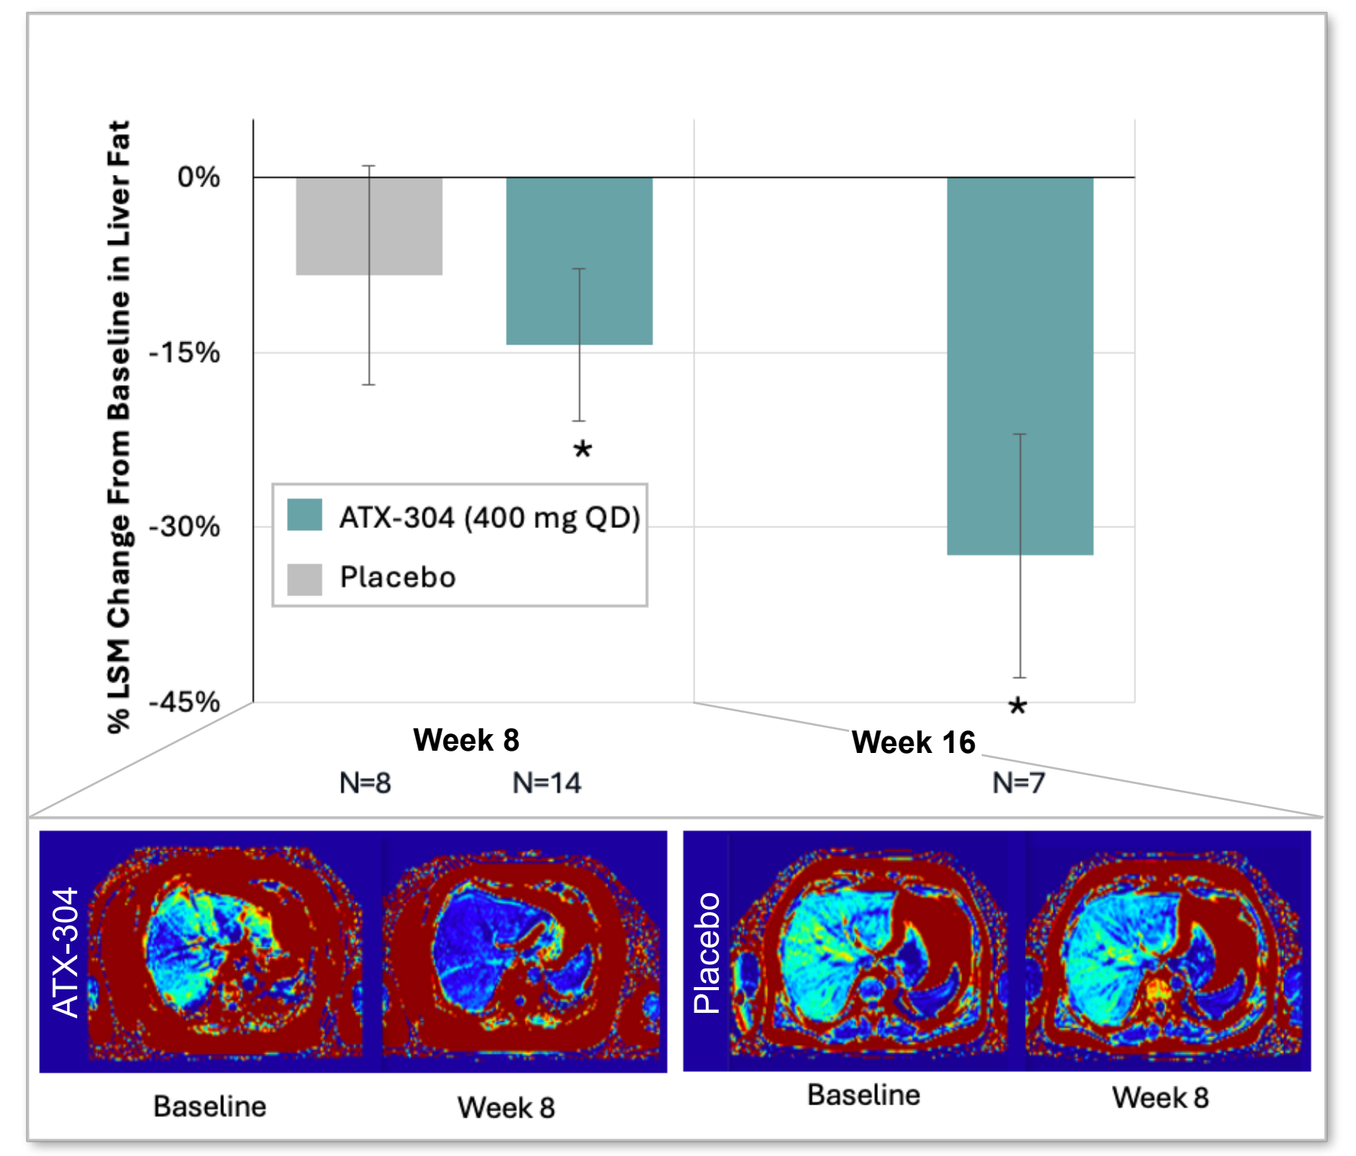

% LSM Change From Baseline in Liver Fat
0%
-15%
-30%
-45%
ATX-304 (400 mg QD)
Placebo
*
*
Week 8
Week 16
N=8 N=14 N=7
ATX-304
Placebo
Baseline
Week 8
Baseline
Week 8

P-value < 0.05; ANCOVA based on Least Squares Mean. MRI-PDFF: Magnetic Resonance Imaging-Proton Density Fat Fraction. MRI–PDFF images are scans from 2 subjects with similar baseline liver fat (appr 10%). Week 16 data from a post-hoc analysis in subjects treated with ATX-304 for 16-weeks.

Fig 5: Mean % Change From Baseline in VAT and Body Weight (+/- SEM)

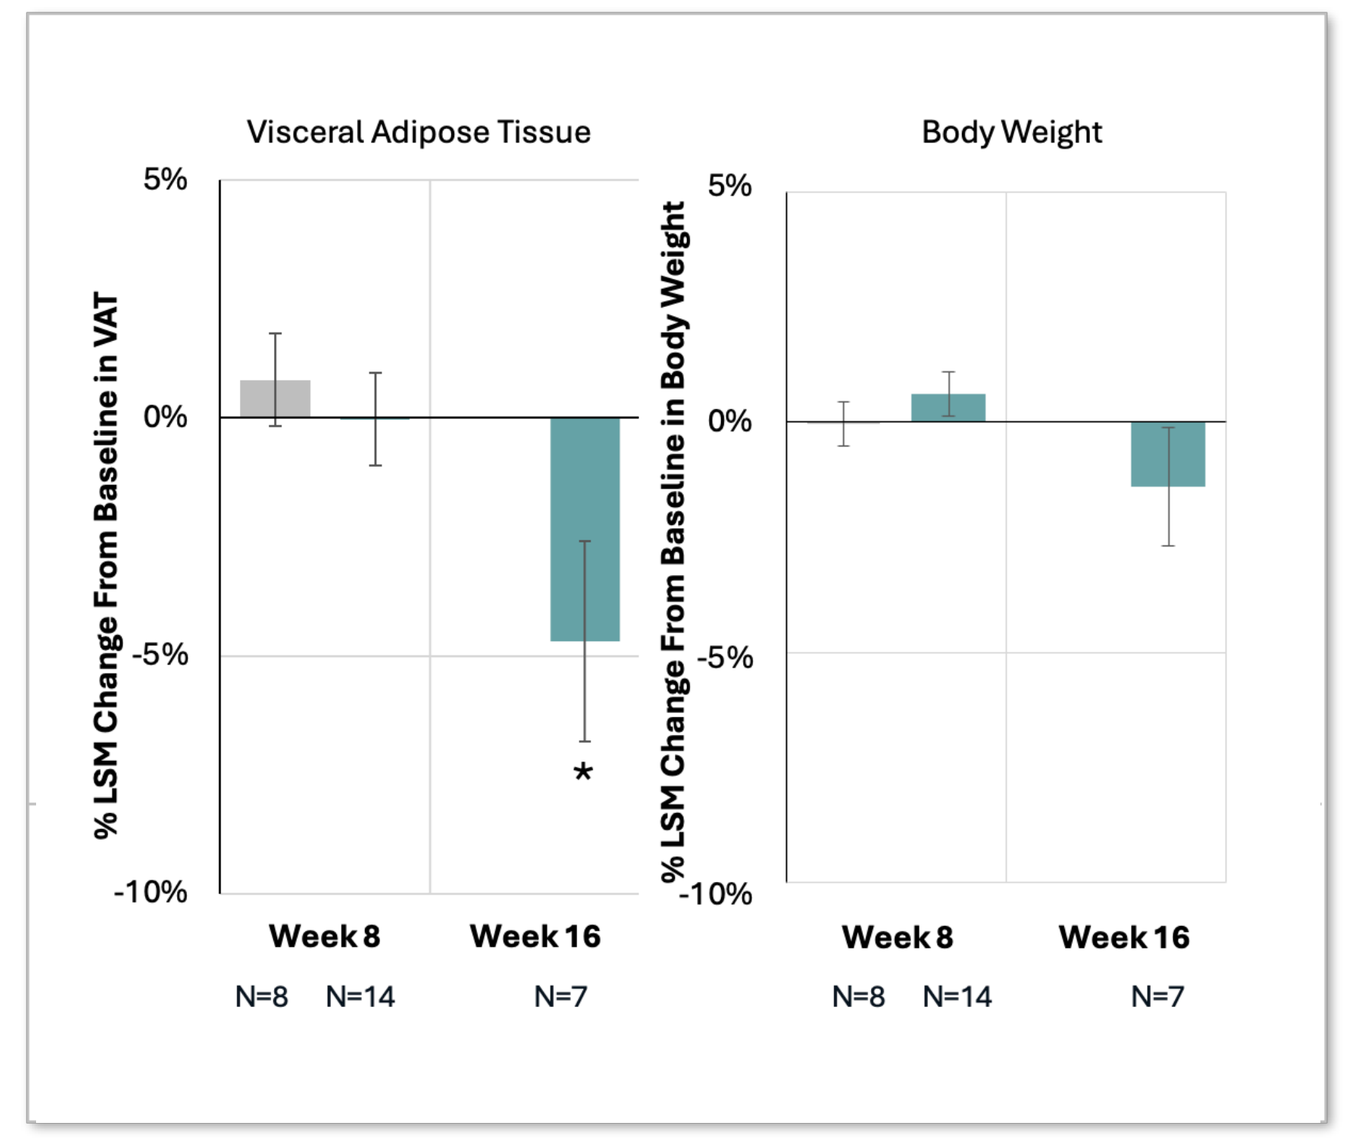

Visceral Adipose Tissue
Body Weight
% LSM Change From Baseline in VAT
% LSM Change From Baseline in Body Weight
5%
0%
-5%
-10%
*
Week 8
Week 16
N=8 N=14 N=7

P-value < 0.05; ANCOVA based on Least Squares Mean. Week 16 data from a post-hoc analysis in subjects treated with ATX-304 for 16-weeks.

ATX-304 increased resting metabolic rate in the Phase 1b Study

Fig 6: % LSM Change from Baseline in RMR, Week 8 (+/- SEM)

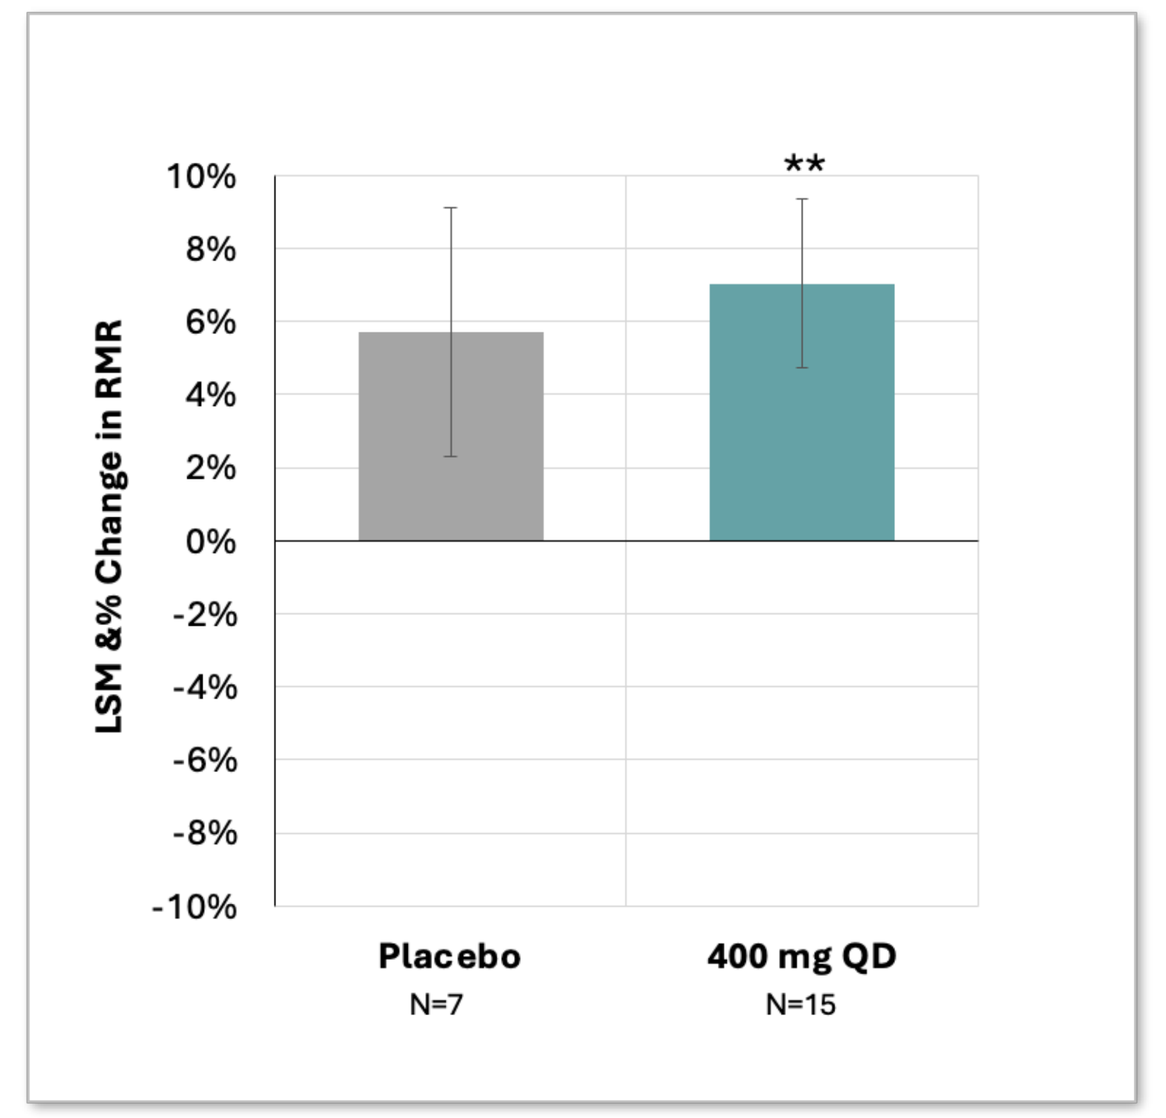

LSM &% Change in RMR
10%
8%
6%
4%
2%
0%
-2%
-4%
-6%
-8%
-10%
**
Placebo
N=7
400 mg QD
N=15

**P-value < 0.01 vs. baseline. ANCOVA. LSM: Least Squares Mean; RMR: Resting Metabolic Rate

Patient Level Data

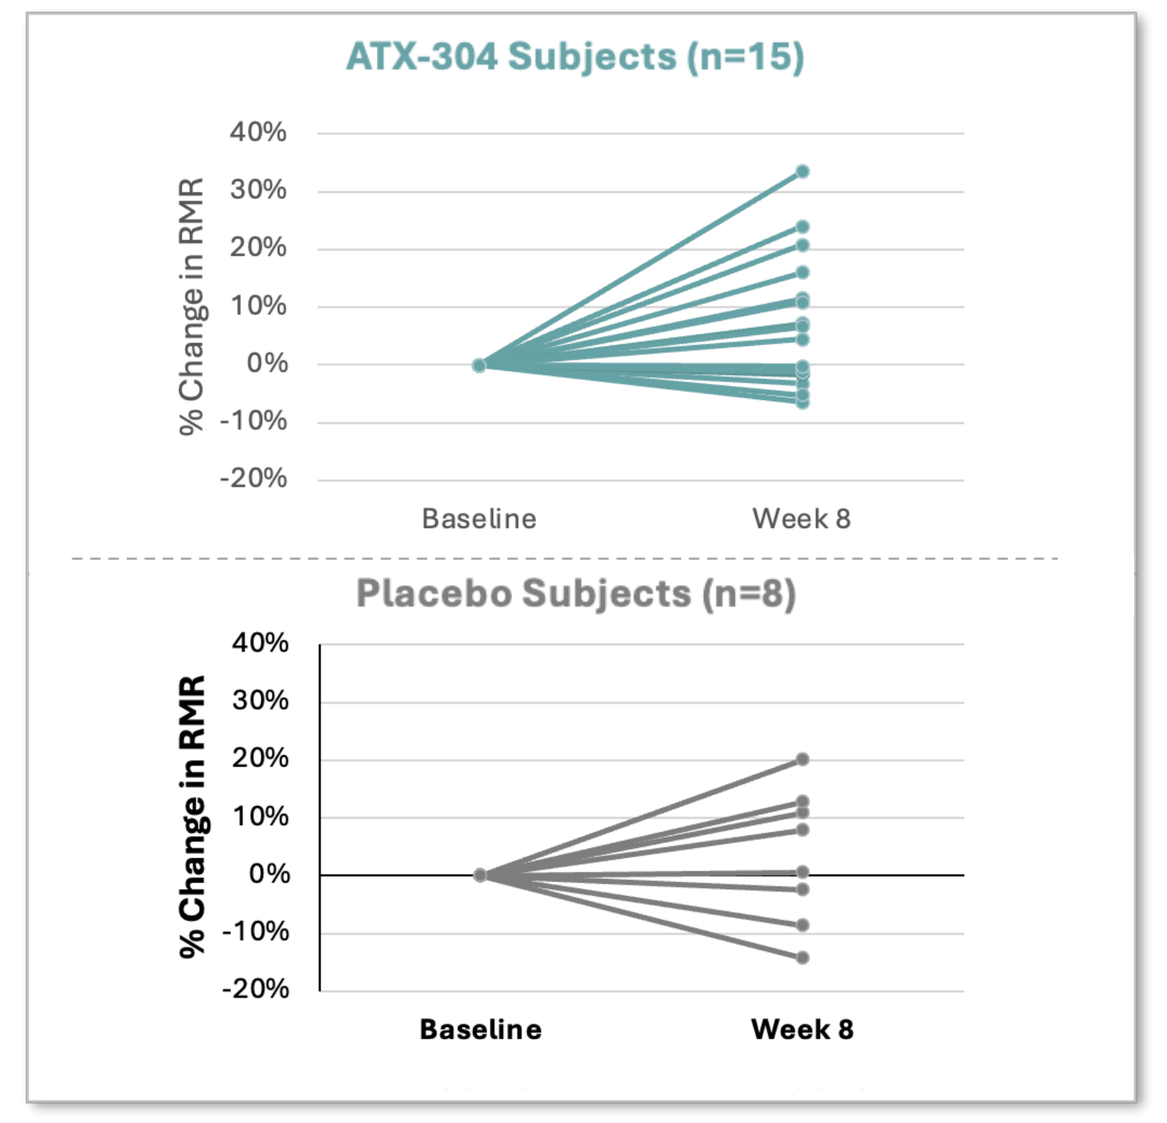

ATX-304 Subjects (n=15)
% Change in RMR
40%
30%
20%
10%
0%
-10%
-20%
Baseline
Week 8
Placebo Subjects (n=8)
% Change in RMR
40%
30%
20%
10%
0%
-10%
-20%
Baseline
Week 8

CONCLUSIONS 4

- ATX-304 is an investigational oral small molecule and the first dual AMPK and mitochondrial activator
- Exposure reached the threshold for translating metabolic effects predicted from rodent models
- ATX-304 showed significant favorable effects on lipid metabolism and body composition
- ATX-304 increased resting metabolic rate significantly without change in appetite and food intake
- Treatment-emergent adverse events were mostly mild and appeared in similar frequency as placebo

Phase 2 studies at higher exposures are planned to assess the utility of ATX-304 to reduce body weight by mobilizing lipids in adipose and ectopic fat tissue and increasing energy expenditure

References

1. Rubino F, et al. Definition and diagnostic criteria of clinical obesity. *Lancet Diabetes Endocrinol.* 2025;13(3):221-262. DOI: 10.1016/S2213-8587(24)00316-4.
2. Steneberg P et al. PAN-AMPK activator O304 improves glucose homeostasis and microvascular perfusion in mice and type 2 diabetes patients. *JCI Insight*. 2018 Jun 21;3(12). DOI: 10.1172/jci.insight.99114.
3. Norlin S, et al. O304 ameliorates hyperglycemia in mice by dually promoting muscle glucose effectiveness and preserving β-cell function. *Commun Biol*. 2023 Aug 25;6(1):877. DOI: 10.1038/s42003-023-05255-6
4. Data on file, Betagenon AB

Disclosures

(1) Ruth I Thieroff-Ekerdt, Erik J Schneider, and Jessica Ainscough are employees and shareholders of Cambrian Bio and work on behalf of the sponsor of the study, Betagenon AB
(1) James G Peyer is the founder and a shareholder of Cambrian Bio
(2) David Jacobs was an employee and is a shareholder of Cambrian Bio at the time of conduct of the study
(2) James A Hall is an employee and shareholder of Betagenon AB
(3) Tim Heise is an employee and shareholder of Profil Institut für Stoffwechselforschung GmbH, the contract research organisation executing the study

Get In Touch

Scan the QR code to visit the contact form on our website, or visit CambrianBio.Com

American Diabetes Association's 86th Scientific Sessions, June 5-8, 2026, New Orleans, LA