Abstract No. 1788-P

AMPK Activation by ATX-304 Is Not Secondary to Changes in ATP Levels

Erik J Schneider(1), Ruth I Thieroff-Ekerdt(1), James G Peyer(1), Anders Gudiksen(3), Henriette Pilegaard(3), Naomi X. Y. Ling(4)(5), Jonathan S Oakhill(4)(5), James A Hall*(2)
*corresponding author

Cambrian Bio(1); Betagenon AB(2); University of Copenhagen(3); St. Vincent's Institute of Medical Research(4); University of Melbourne(5)

AMPLIFIER | OWNED BY Cambrian

INTRODUCTION 1

- ATX-304 is a novel, clinical-stage, small molecule with dual action:
 - ATX-304 increases AMP-activated protein kinase (AMPK) activity
 - ATX-304 increases mitochondrial respiration via increased proton leak
- Increasing AMPK activity, therefore increasing substrate supply for mitochondrial respiration, may change the Mitochondrial Membrane Potential (MMP) and ATP response to increased proton leak[1,2]
- To understand the effect of dual AMPK and mitochondrial activation, ATX-304 effects on MMP, ATP levels, and AMPK activation were investigated up to a solubility limit of 20 μM

EXPERIMENTAL METHODS 2

Experiments in Isolated Mitochondria & Permeabilized Muscle Fibers

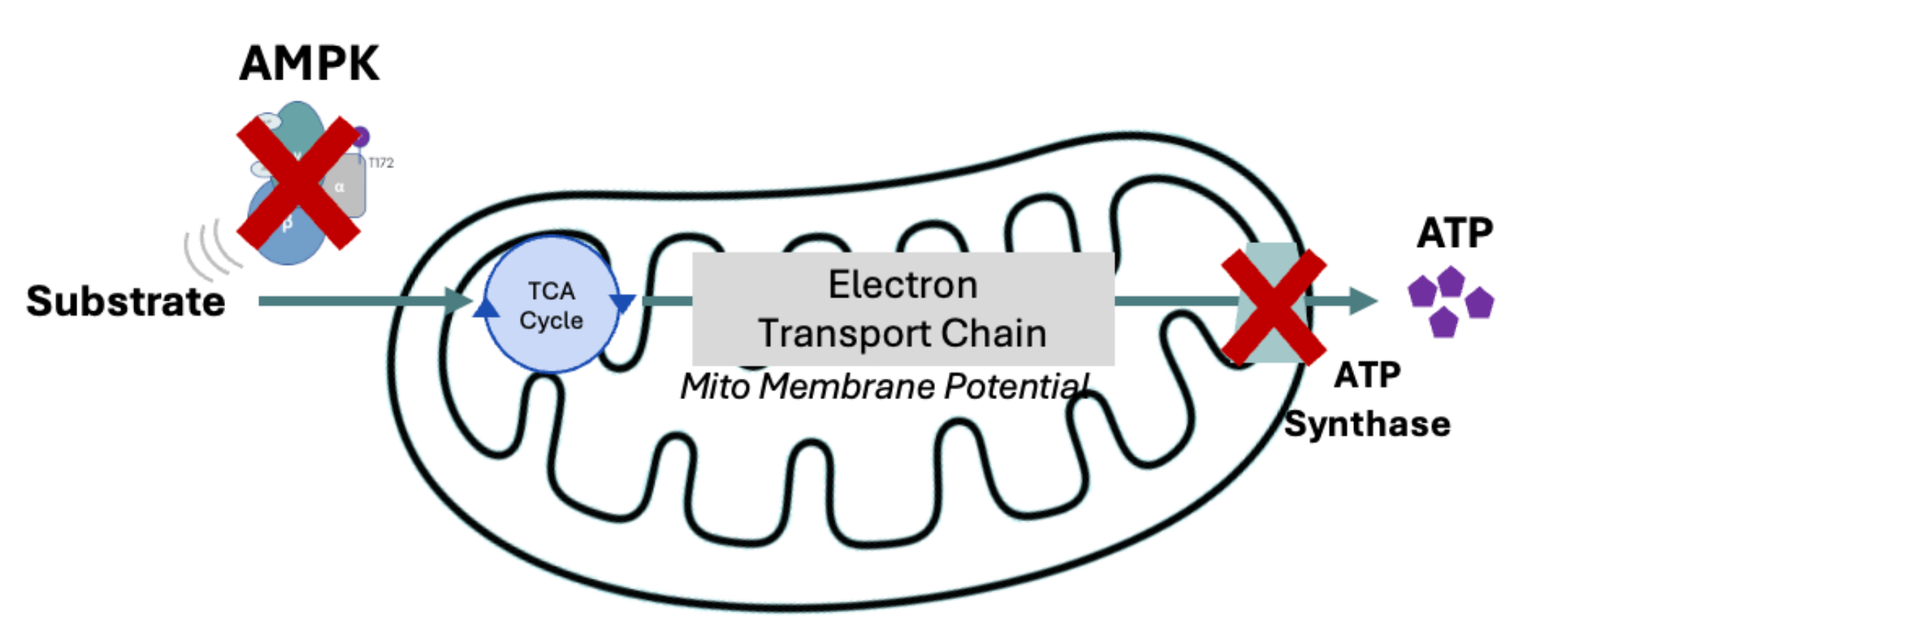
AMPK
Substrate
TCA Cycle
Electron Transport Chain
Mito Membrane Potential
ATP
ATP Synthase

- This test system allows study of ATX-304 solely on mitochondrial activity
 - Mitochondrial function assessed with high resolution respirometry (Oroboros Bioenergetics Analyzer)
- An uncoupled state is created to assess the effect of mitochondrial uncouplers
 - Fixed substrates are provided and ATP synthase is inhibited
- **Readouts:**
 - O_2 consumption
 - Mitochondrial Membrane Potential
 - Reactive Oxygen Species (H_2O_2 emission)

Experiments in Intact Cells

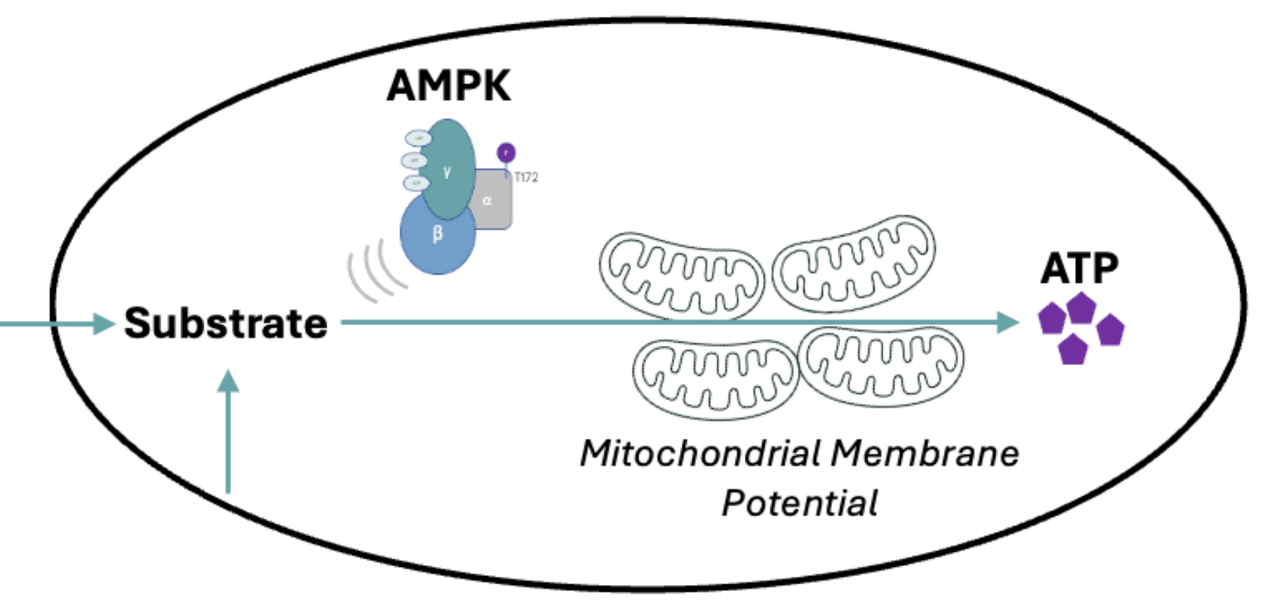
AMPK
Substrate
ATP
Mitochondrial Membrane Potential

- This test system allows study of mitochondrial activity and AMPK activity together (including AMPK action on substrate uptake and mobilization)
 - ATP synthase and ATP production are fully active
- ATX-304 activity reflects a net balance of both AMPK and mitochondrial action
- **Readouts:**
 - Mitochondrial Membrane Potential
 - Reactive Oxygen Species (H_2O_2 emission)
 - Cellular ATP levels
 - Activation of mutant AMP-insensitive AMPK

RESULTS 3

ATX-304 in Isolated Mitochondria

Experimental Set-Up

- Test system assesses maximal uncoupled respiration in isolated mitochondria from mouse muscle fibers
- Fixed substrate supply (Pyruvate, Malate, Glutamate) + Succinate provided for maximal electron transport chain function
- ATP synthesis is inhibited by withholding ADP
- O_2 consumption measured with Oroboros Bioenergetics Analyzer; measurement of proton gradient (Mitochondrial Membrane Potential; MMP) via fluorescence with TMRM dye (tetramethylrhodamine methyl ester)

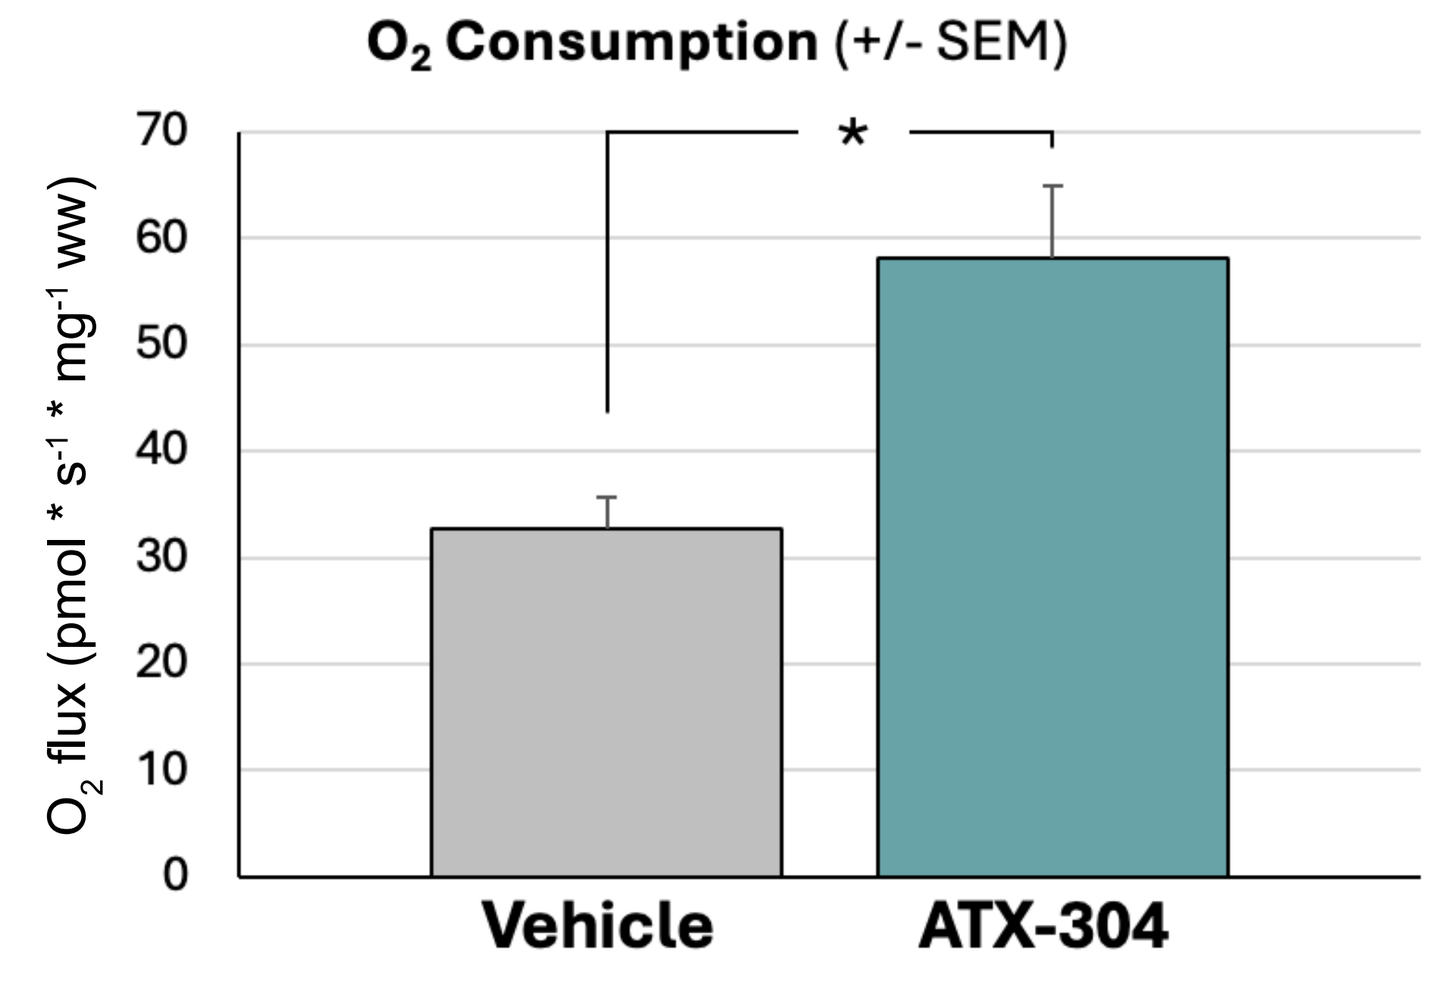
O2 Consumption (+/- SEM)
O2 flux (pmol * s-1 * mg-1 ww)
*
Vehicle
ATX-304

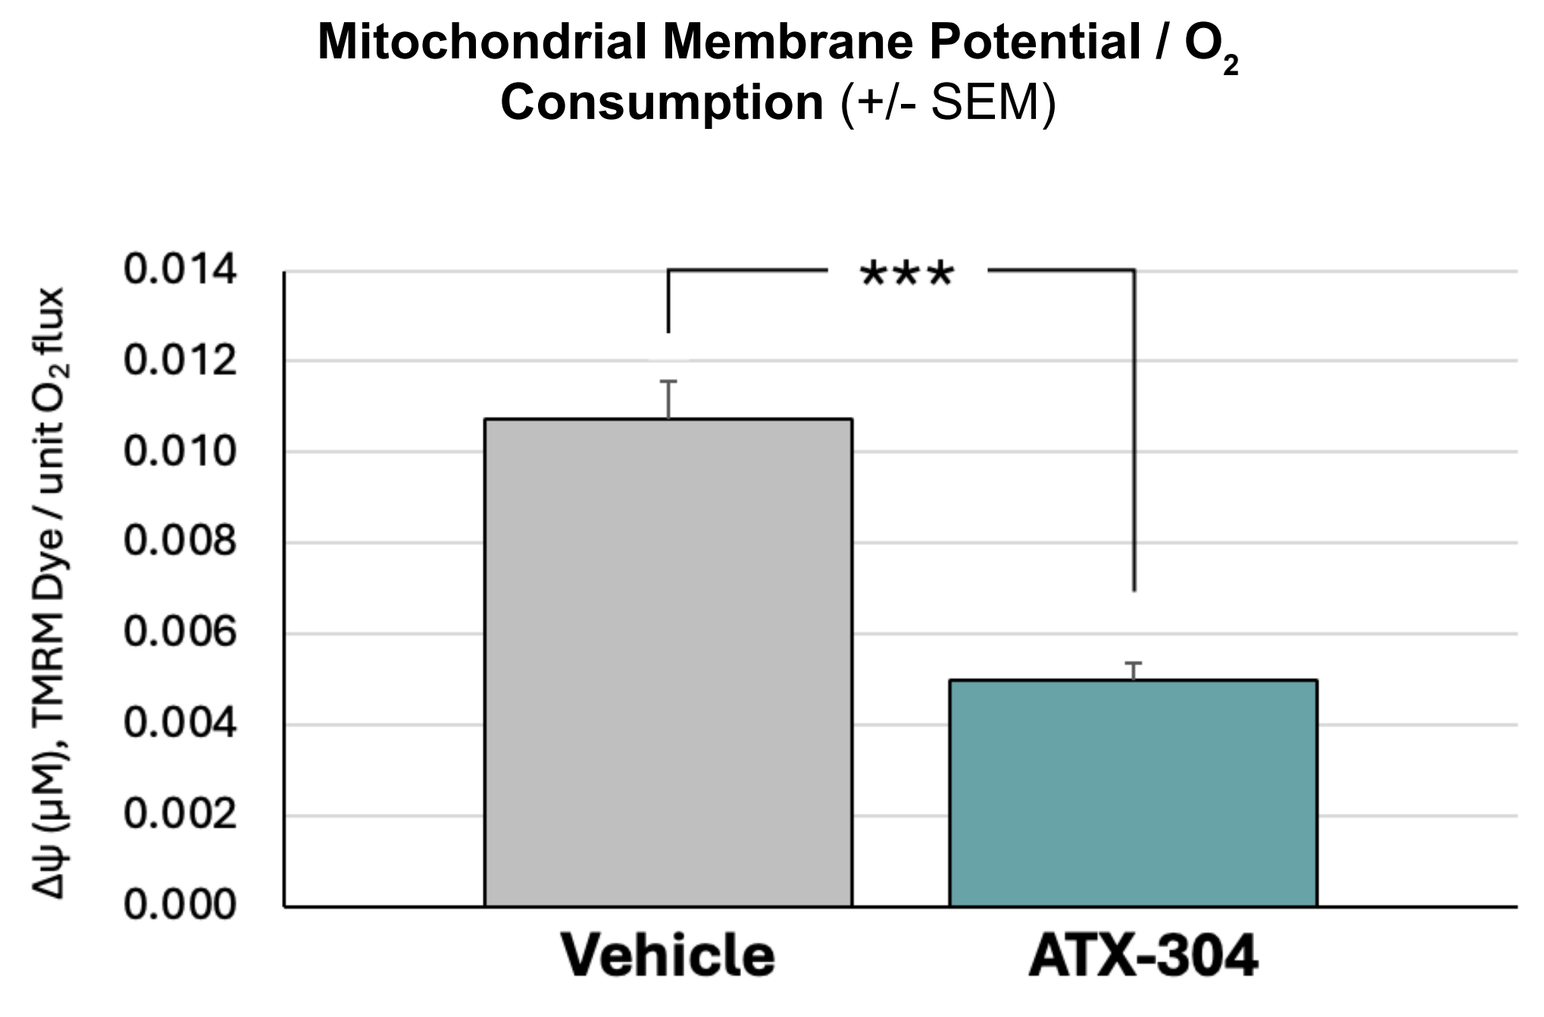
Mitochondrial Membrane Potential / O2 Consumption (+/- SEM)
Δψ (μM), TMRM Dye / unit O2 flux

Vehicle
ATX-304

* P-Value < 0.05; ***P-Value < 0.001 Mann-Whitney U test (between group)

Key Takeaway:

- In isolated mitochondria in an uncoupled state, ATX-304 increases O_2 consumption and decreases MMP, indicative of increased proton leak through the mitochondrial membrane

ATX-304 in Permeabilized Mouse Muscle Fibers

Experimental Set-Up

- Test system assesses uncoupled respiration in permeabilized (via saponin) mouse muscle fibers
- ATP synthesis is inhibited by withholding ADP and adding oligomycin, an ATP synthase inhibitor
- Fixed substrate supply (Pyruvate, Malate, Glutamate) + Succinate provided for maximal ETC function
- CATR (Carboxyatractyloside; ANT1 inhibitor) added to assess ANT1 participation in ATX-304 action on proton leak
- O_2 consumption measured with Oroboros Bioenergetics Analyzer; measurement of Reactive Oxygen Species (ROS; H_2O_2) emission via fluorescence with Amplex Red dye

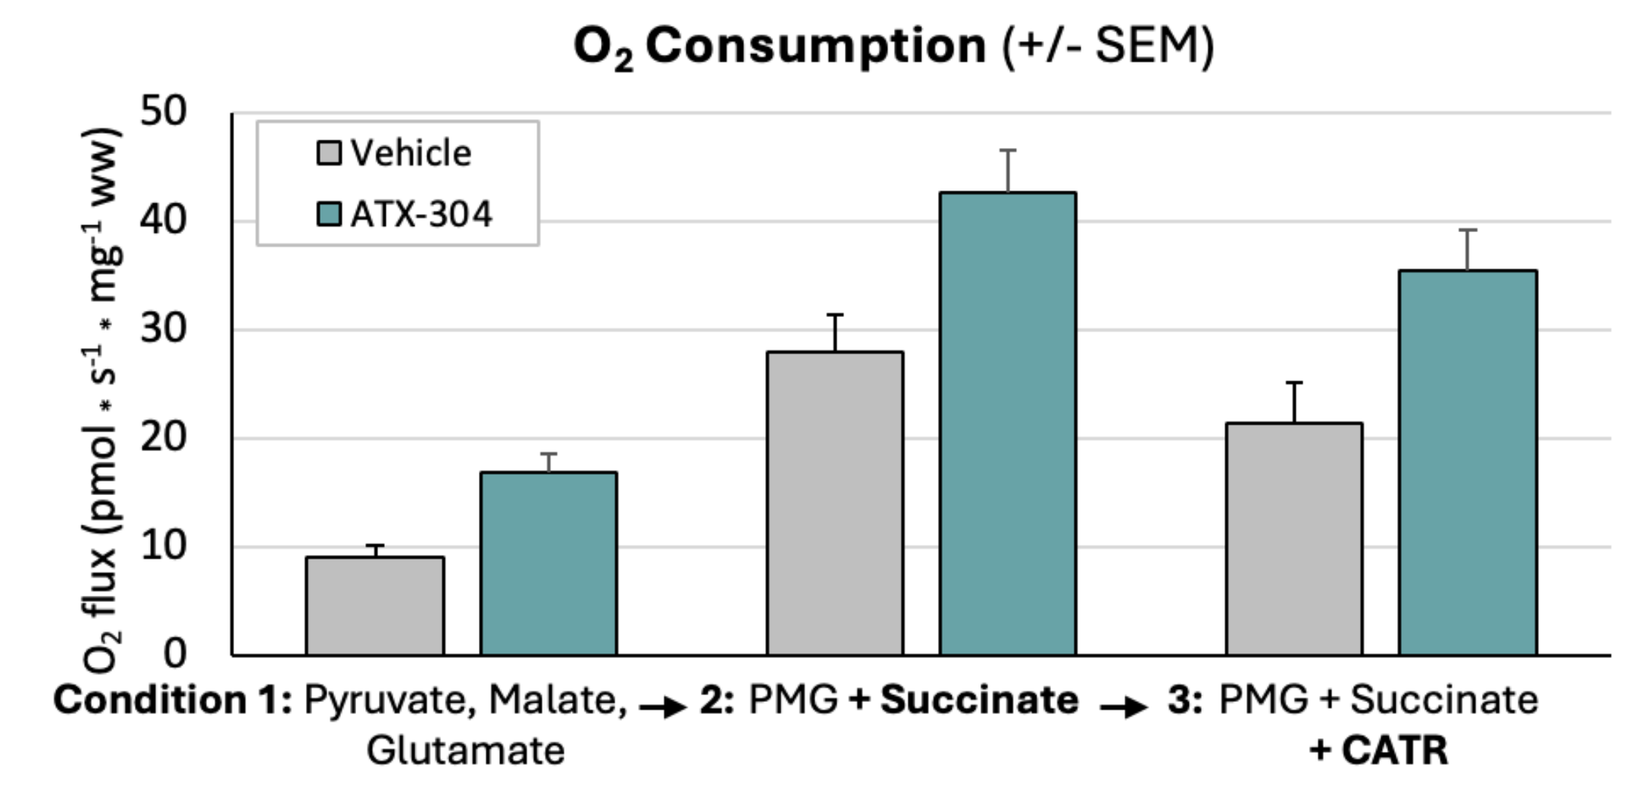
O2 Consumption (+/- SEM)
O2 flux (pmol • s-1 • mg-1 ww)
Vehicle
ATX-304
Condition 1: Pyruvate, Malate, Glutamate → 2: PMG + Succinate → 3: PMG + Succinate + CATR

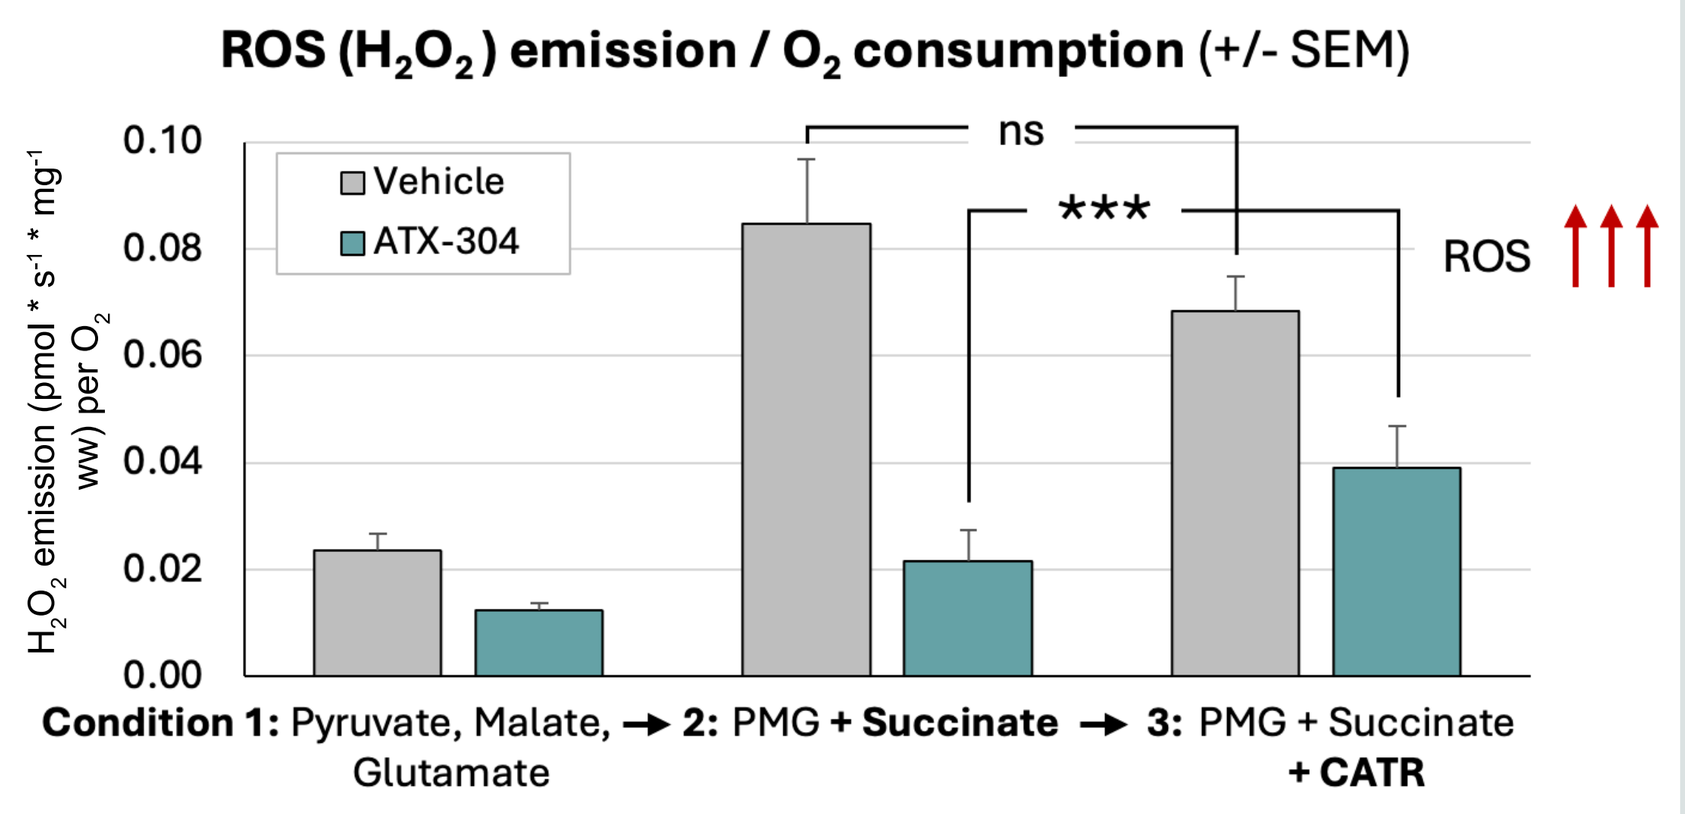
ROS (H2O2) emission / O2 consumption (+/- SEM)
H2O2 emission (pmol * s-1 * mg-1 ww) per O2
Vehicle
ATX-304
ns

ROS
Condition 1: Pyruvate, Malate, Glutamate → 2: PMG + Succinate → 3: PMG + Succinate + CATR

*** P-Value <0.001 Wilcoxon signed-rank test (within group)

ROS is produced if the ETC is active but there is no consumption of the proton gradient for ATP synthesis. Physiological uncoupling via proton leak reduces the proton gradient and minimizes ROS production during periods of low ATP demand

Key Takeaway:

- **Conditions 1 & 2:**
 - In control fibers, as ETC activity is maximized by adding succinate (increasing O_2 consumption) ROS production is increased
 - In ATX-304 treated fibers, ROS production is decreased due to increased proton leak
- **Condition 3:** An ATX-304-driven decrease in ROS is reduced by CATR, indicative of ANT1 participation in proton leak

ATX-304 in Intact Cells

Experimental Set-Up

- Test system assesses MMP and ROS emission in intact cells (murine embryonic fibroblasts)
- MMP assessed via fluorescence with TMRE dye (tetramethylrhodamine ethyl ester)
- ROS (H_2O_2) emission measured via fluorescence with DCFDA dye (dichlorodihydrofluorescein diacetate)
- FCCP, an ionophore, positive control for MMP reduction; TBHP, ROS inducer, positive control for ROS emission increase

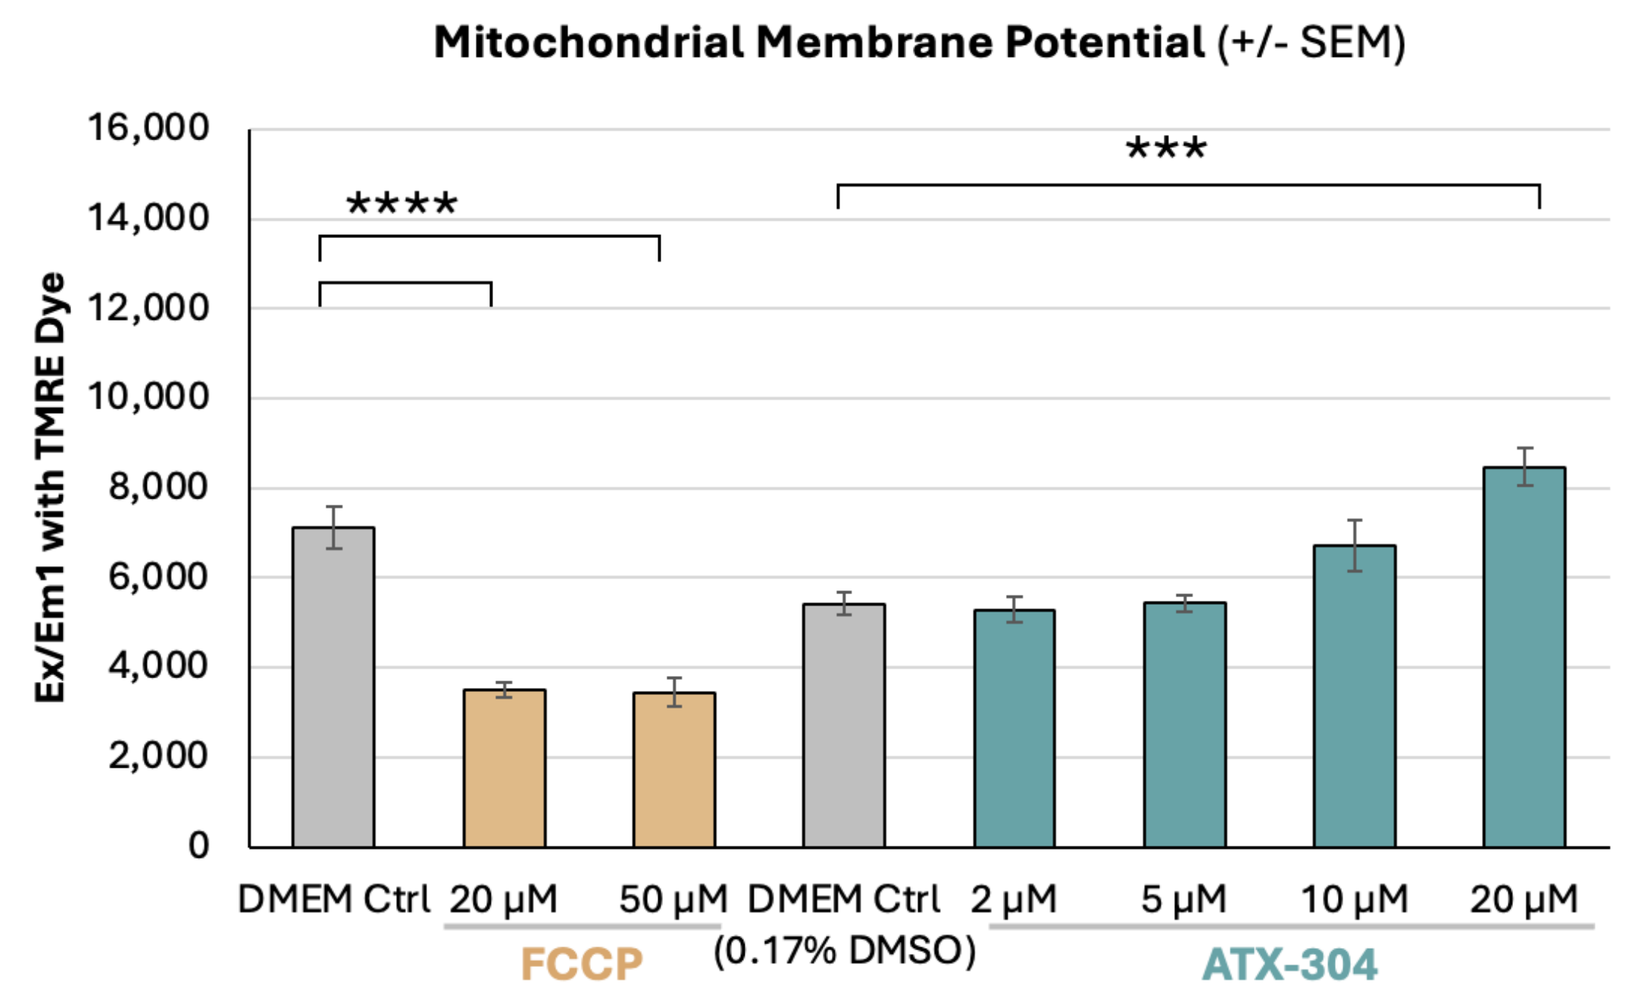
Mitochondrial Membrane Potential (+/- SEM)
Ex/Em1 with TMRE Dye

DMEM Ctrl
20 μM
50 μM
FCCP
DMEM Ctrl (0.17% DMSO)
2 μM
5 μM
10 μM
20 μM
ATX-304

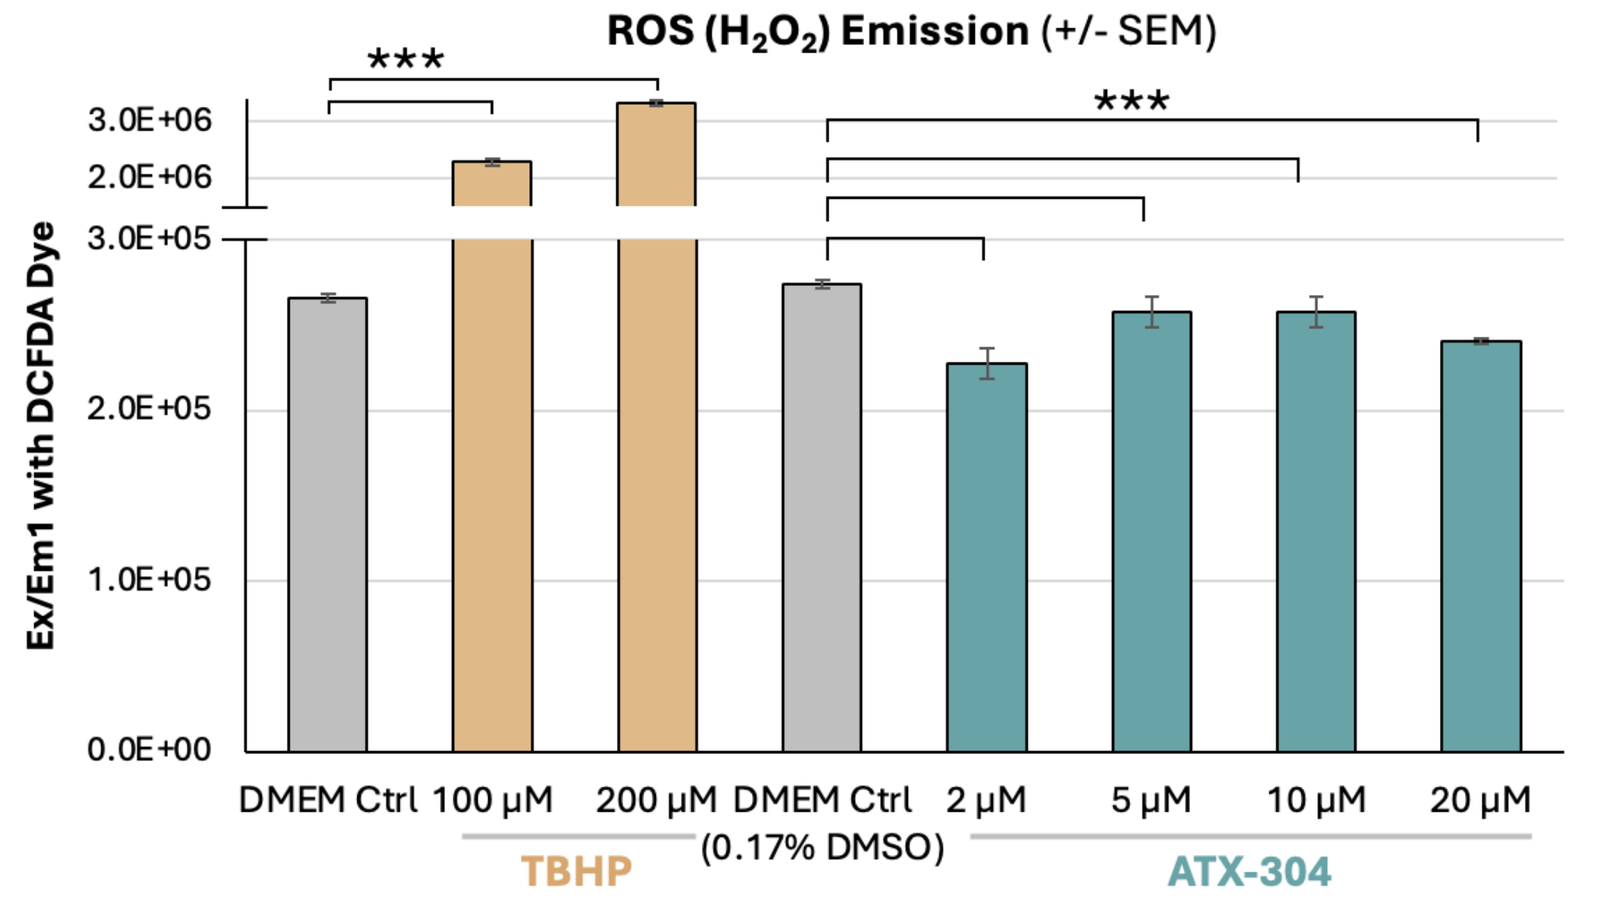
ROS (H2O2) Emission (+/- SEM)
Ex/Em1 with DCFDA Dye

DMEM Ctrl
100 μM
200 μM
TBHP
DMEM Ctrl (0.17% DMSO)
2 μM
5 μM
10 μM
20 μM
ATX-304

DMEM: Dulbecco's Modified Eagle Medium. ATX-304 maintained in 0.17% stock DMSO solution. Statistics: FCCP: vs DMEM only; ATX: vs DMEM Ctrl (+ 0.17% DMSO). One-way ANOVA with Dunnett's multiple comparison test. *** P-Value <0.01; ****P-Value<0.001. N=4 run in each experiment.

Proton leak decreases MMP unless substrate supply is increased commensurately

Key Takeaway:

- In cells with intact AMPK signaling, ATX-304 maintains MMP, indicative of increased substrate supply to the mitochondria
- ATX-304 reduces ROS production consistent with increased proton leak

ATX-304 in Intact Cells

Experimental Set-Up

- Test system assesses ATP levels in intact cells (primary human hepatocytes) for 48-hours of exposure
- ATP levels assayed via CellTiter-Glo® luciferin/luciferase assay; cell viability inferred from ATP levels
- Chlorpromazine positive control decreases ATP levels by inhibiting mitochondrial respiration

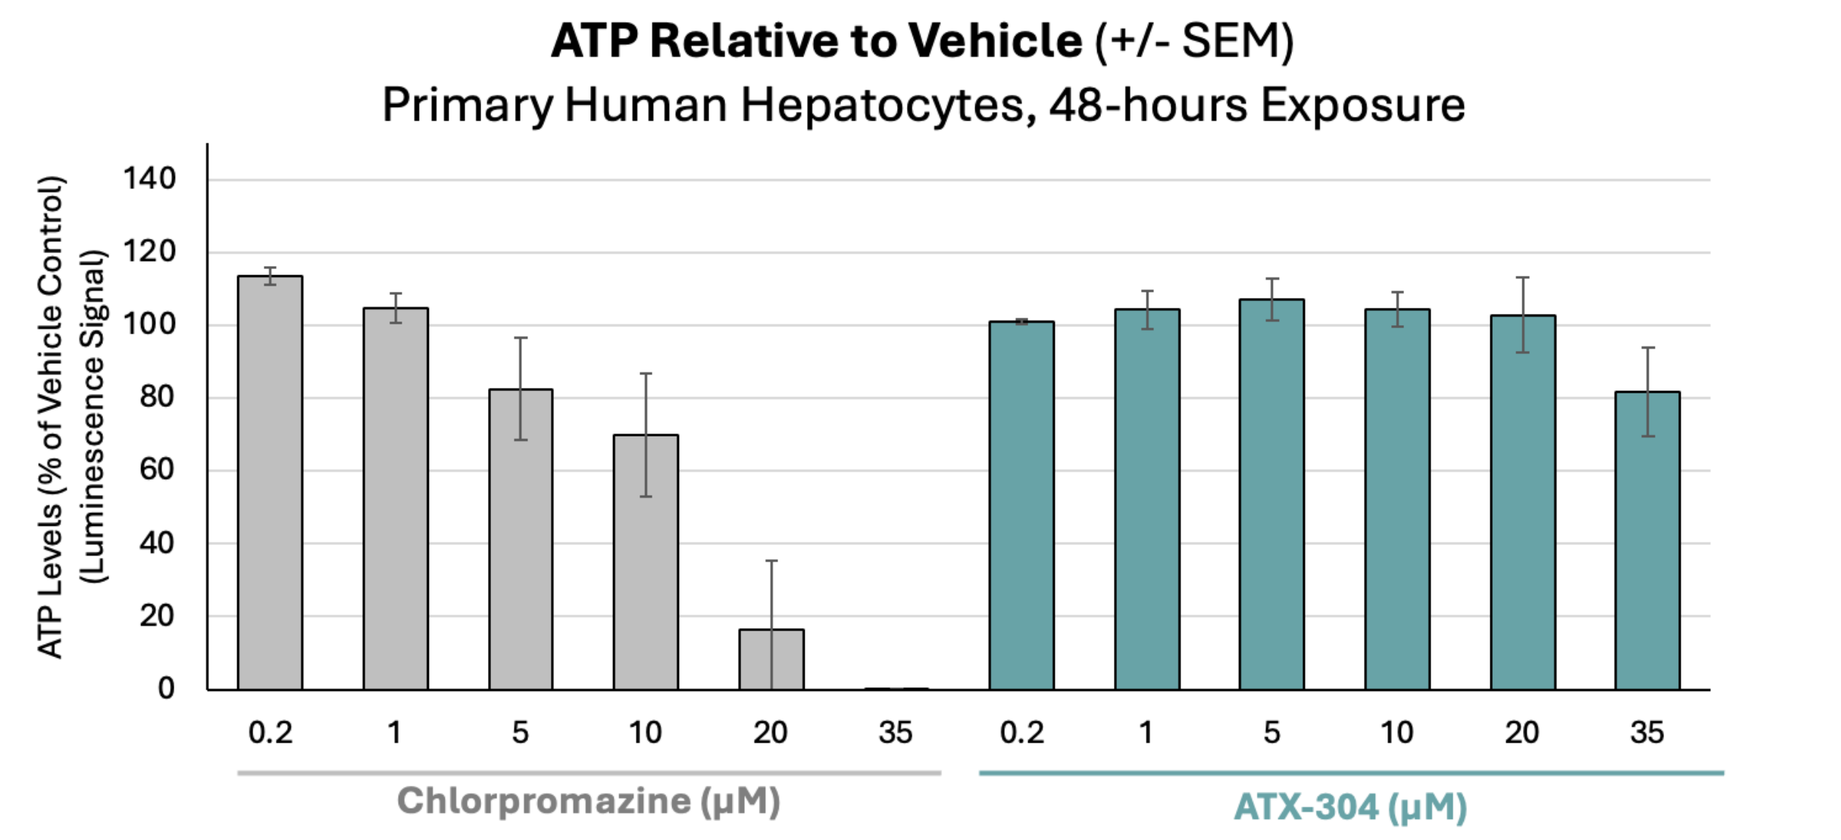
ATP Relative to Vehicle (+/- SEM)
Primary Human Hepatocytes, 48-hours Exposure
ATP Levels (% of Vehicle Control) (Luminescence Signal)
Chlorpromazine (μM)
ATX-304 (μM)

Inhibiting mitochondrial respiration dose-dependently reduces cellular ATPf solubility

Key Takeaway:

- In primary human hepatocytes, ATX-304 preserves ATP levels and cell viability up to the limit of solubility

AMPK Activation by ATX-304 in Intact Cells

Experimental Set-Up

- Test system assesses AMP-dependent AMPK activity in intact cells (HEK-293 expressing wild type (WT) AMPK or γ1 R299G (RG) mutant AMPK with reduced AMP sensitivity)
- AMPK activity assayed via synthetic peptide (SAMS) phosphorylation with isolated AMPK, or via increase in T172α phosphorylation via Western Blot
- 2-Deoxy-D-Glucose (2-DG; non-metabolizable glucose analogue) positive control for AMPK activation via ATP reduction and increase of AMP

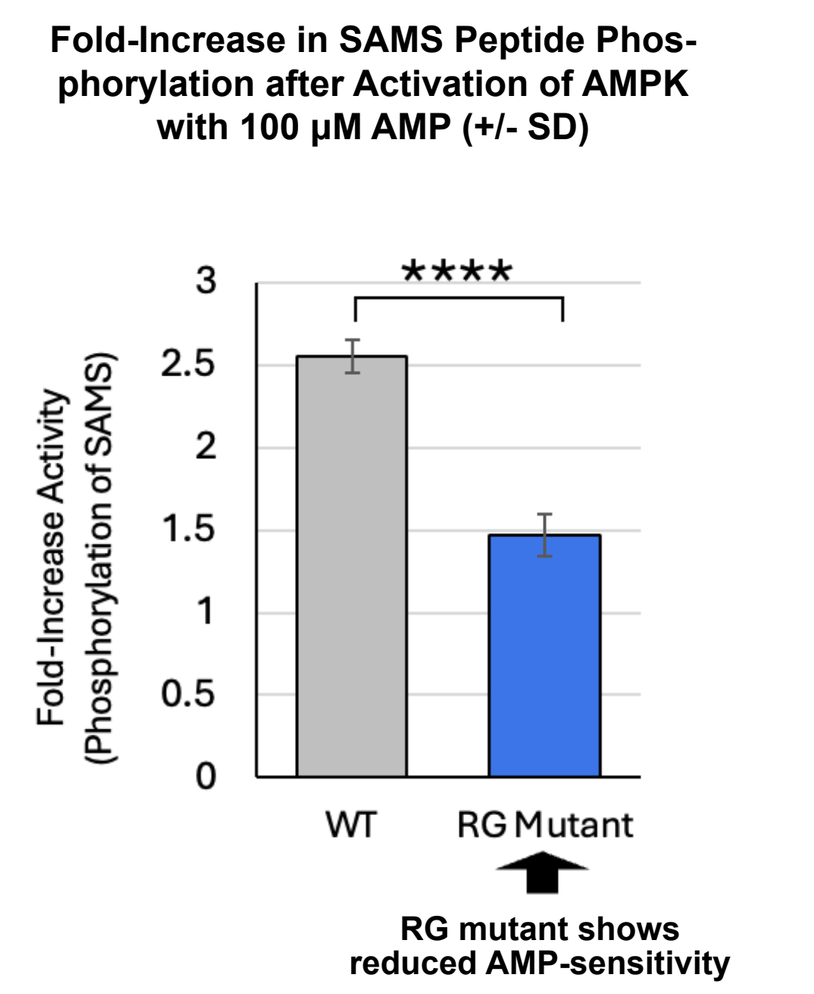
Fold-Increase in SAMS Peptide Phosphorylation after Activation of AMPK with 100 μM AMP (+/- SD)
Fold-Increase Activity (Phosphorylation of SAMS)

WT
RG Mutant
RG mutant shows reduced AMP-sensitivity

Data presented as mean fold change in SAMS peptide phosphorylation ± SD; Statistical analyses were performed by one way ANOVA with ordinary Dunnett's multiple comparison test vs WT; ****P<0.0001

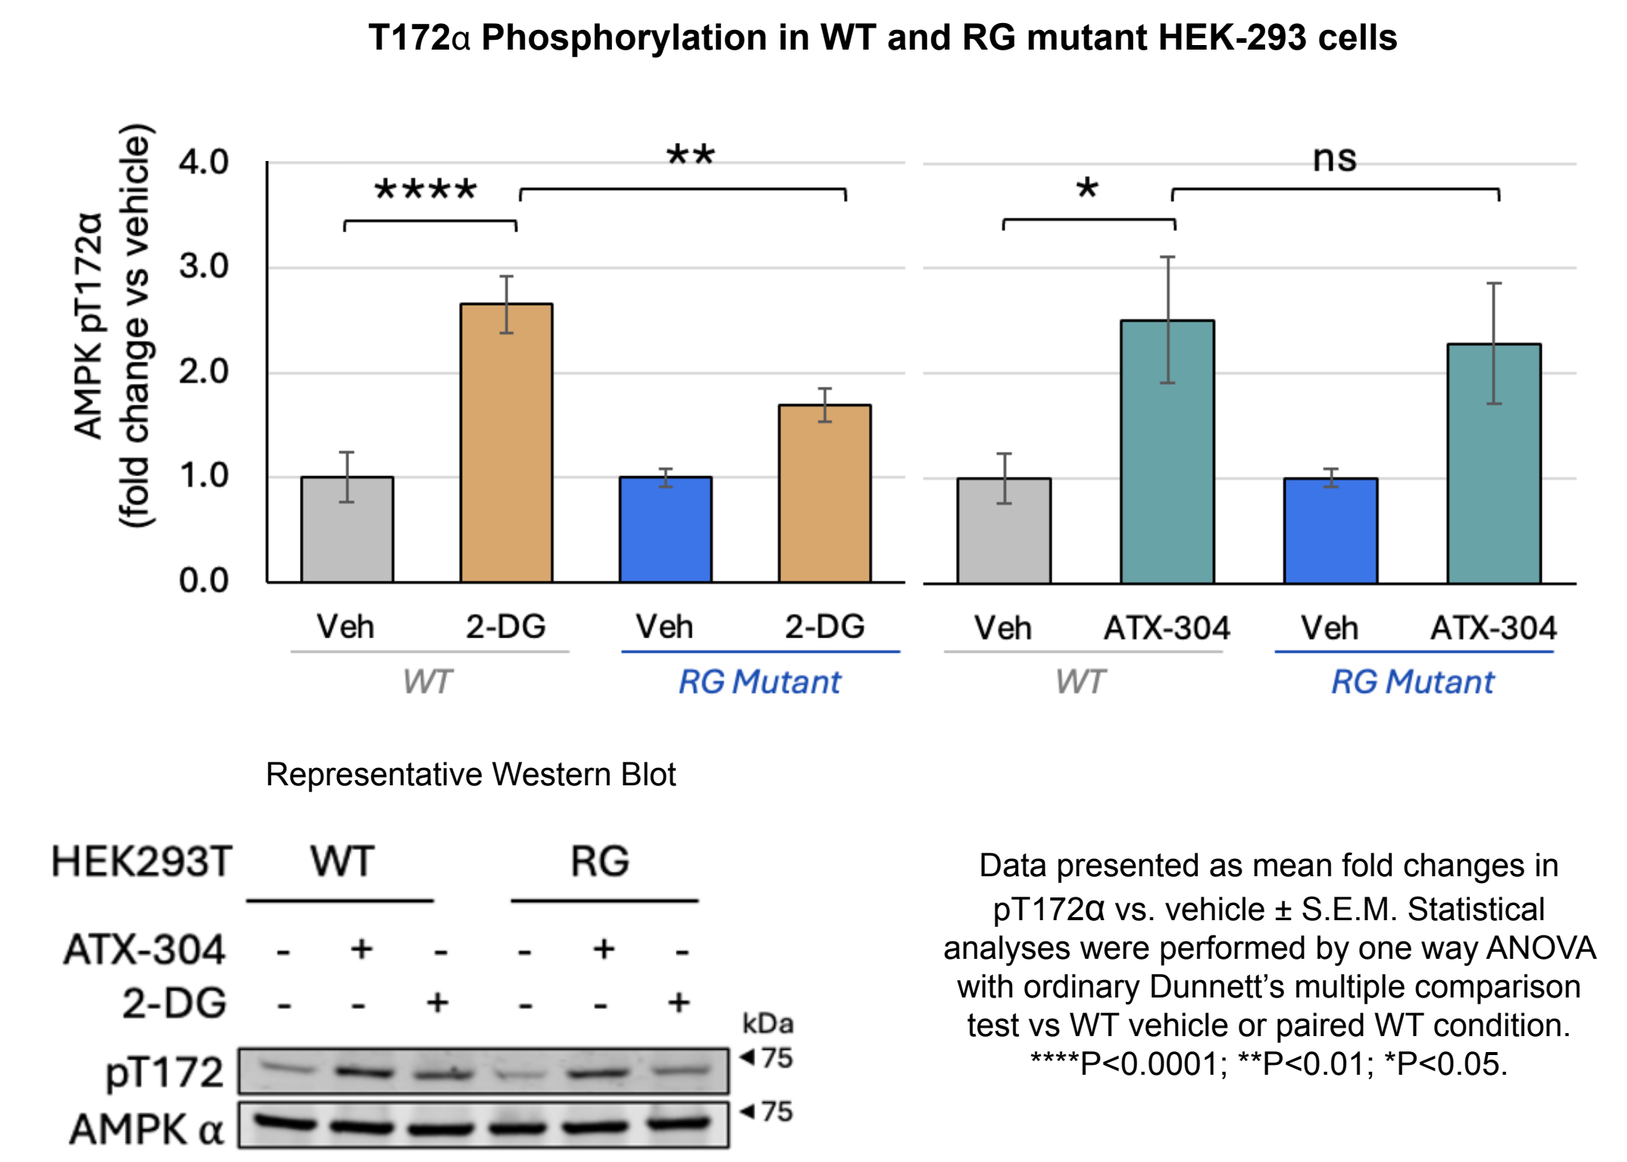
T172α Phosphorylation in WT and RG mutant HEK-293 cells
AMPK pT172α (fold change vs vehicle)

**
*
ns
Veh
2-DG
ATX-304
WT
RG Mutant
Representative Western Blot
HEK293T
WT
RG
ATX-304
2-DG
pT172
AMPK α
kDa
75

Data presented as mean fold changes in pT172α vs. vehicle ± S.E.M. Statistical analyses were performed by one way ANOVA with ordinary Dunnett's multiple comparison test vs WT vehicle or paired WT condition. ****P<0.0001; **P<0.01; *P<0.05.

Key Takeaway:

- Reduced activation of R299G mutant AMPK by AMP
- Reduced activation of R299G mutant AMPK with non-metabolizable glucose analogue, 2DG, consistent with AMPK activation dependent on reduced ATP and increased AMP levels
- Continued activation of R299G mutant AMPK by ATX-304, consistent with AMPK activation independent of the ATP / ADP / AMP binding site on the gamma subunit

CONCLUSIONS 4

- **ATX-304 increases uncoupled mitochondrial respiration by increasing proton leak**
 - In isolated mouse skeletal muscle mitochondria, ATX-304 increased O_2 consumption and decreased mitochondrial membrane potential, indicative of an increase in proton leak through the mitochondrial membrane
 - In mouse skeletal muscle fibers, ATX-304-driven decrease in ROS (indicative of increased proton leak) is reduced by CATR (indicative of ANT1 participation in proton leak)
- **In intact cells, ATX-304 activates AMPK, thereby increasing substrate provision and protecting against ATP reduction due to increased proton leak**
 - In intact mouse fibroblasts, ATX-304 did not decrease mitochondrial membrane potential, indicative of increased substrate supply to mitochondria
 - In intact primary human hepatocytes, ATX-304 did not reduce ATP levels compared to control
- **AMPK activation with ATX-304 is independent of the ATP / ADP / AMP binding site on the gamma subunit**
 - AMPK activation is reduced in cells expressing mutant AMPK with reduced AMP sensitivity
 - ATX-304 continued to activate AMPK in cells expressing mutated AMPK

Data suggest:

- **AMPK activation with ATX-304 is not secondary to a decrease in ATP levels**
- **ATX-304 increases mitochondrial substrate supply via AMPK activation and increases mitochondrial proton leak**

References

1. Jian C, et al. *Cell Metab.* 2020;31(5):892-908.e11. DOI: 10.1016/j.cmet.2020.04.011.
2. e.g. Sibille, B et al. *Biochem J.* 2001;355(1):231-235. DOI: 10.1042/bj3550231.

Disclosures

(1) Erik J Schneider, Ruth I Thieroff-Ekerdt, and James G Peyer are employees and shareholders of Cambrian Bio
(1) James G Peyer is the founder of Cambrian Bio
(2) James A Hall is an employee and shareholder of Betagenon AB

ATX-304 is an investigational drug agent that is not available for human use outside a clinical trial setting.

Get In Touch

Scan the QR code to visit the contact form on our website, or visit CambrianBio.Com

American Diabetes Association's 86th Scientific Sessions, June 5-8, 2026, New Orleans, LA