NeurIPS 2025 | December 3
San Diego

NeuralPLexer3: Accurate Biomolecular Complex Structure Prediction with Flow Models

Iambic

Zhuoran Qiao, Feizhi Ding, Thomas Dresselhaus, Mia A. Rosenfeld, Xiaotian Han, Owen Lewis Howell, Aniketh Iyengar, Stephen Opalenski, Anders S. Christensen, Sai Krishna Sirumalla, Fred Manby, Thomas F. Miller III, Matthew Welborn

Iambic Therapeutics, San Diego, CA

Paper NPBench

NeuralPLexer3 is a physics-inspired flow model that predicts full biomolecular complex structures—proteins, nucleic acids, ligands, ions and PTMs—with higher accuracy and better physical realism than prior methods, while running in seconds.

Motivation

Experimental structures are slow/expensive, but most drug discovery programs still depend on them.

AlphaFold2/3 revolutionized folding, but:

- can hallucinate unphysical ligands / geometries,
- are expensive to run at scale,
- and are not systematically evaluated on drug-discovery-relevant tasks.

Goal: accurate, physically valid, *fast* prediction of protein–ligand and other biomolecular complexes for real drug-discovery workflows.

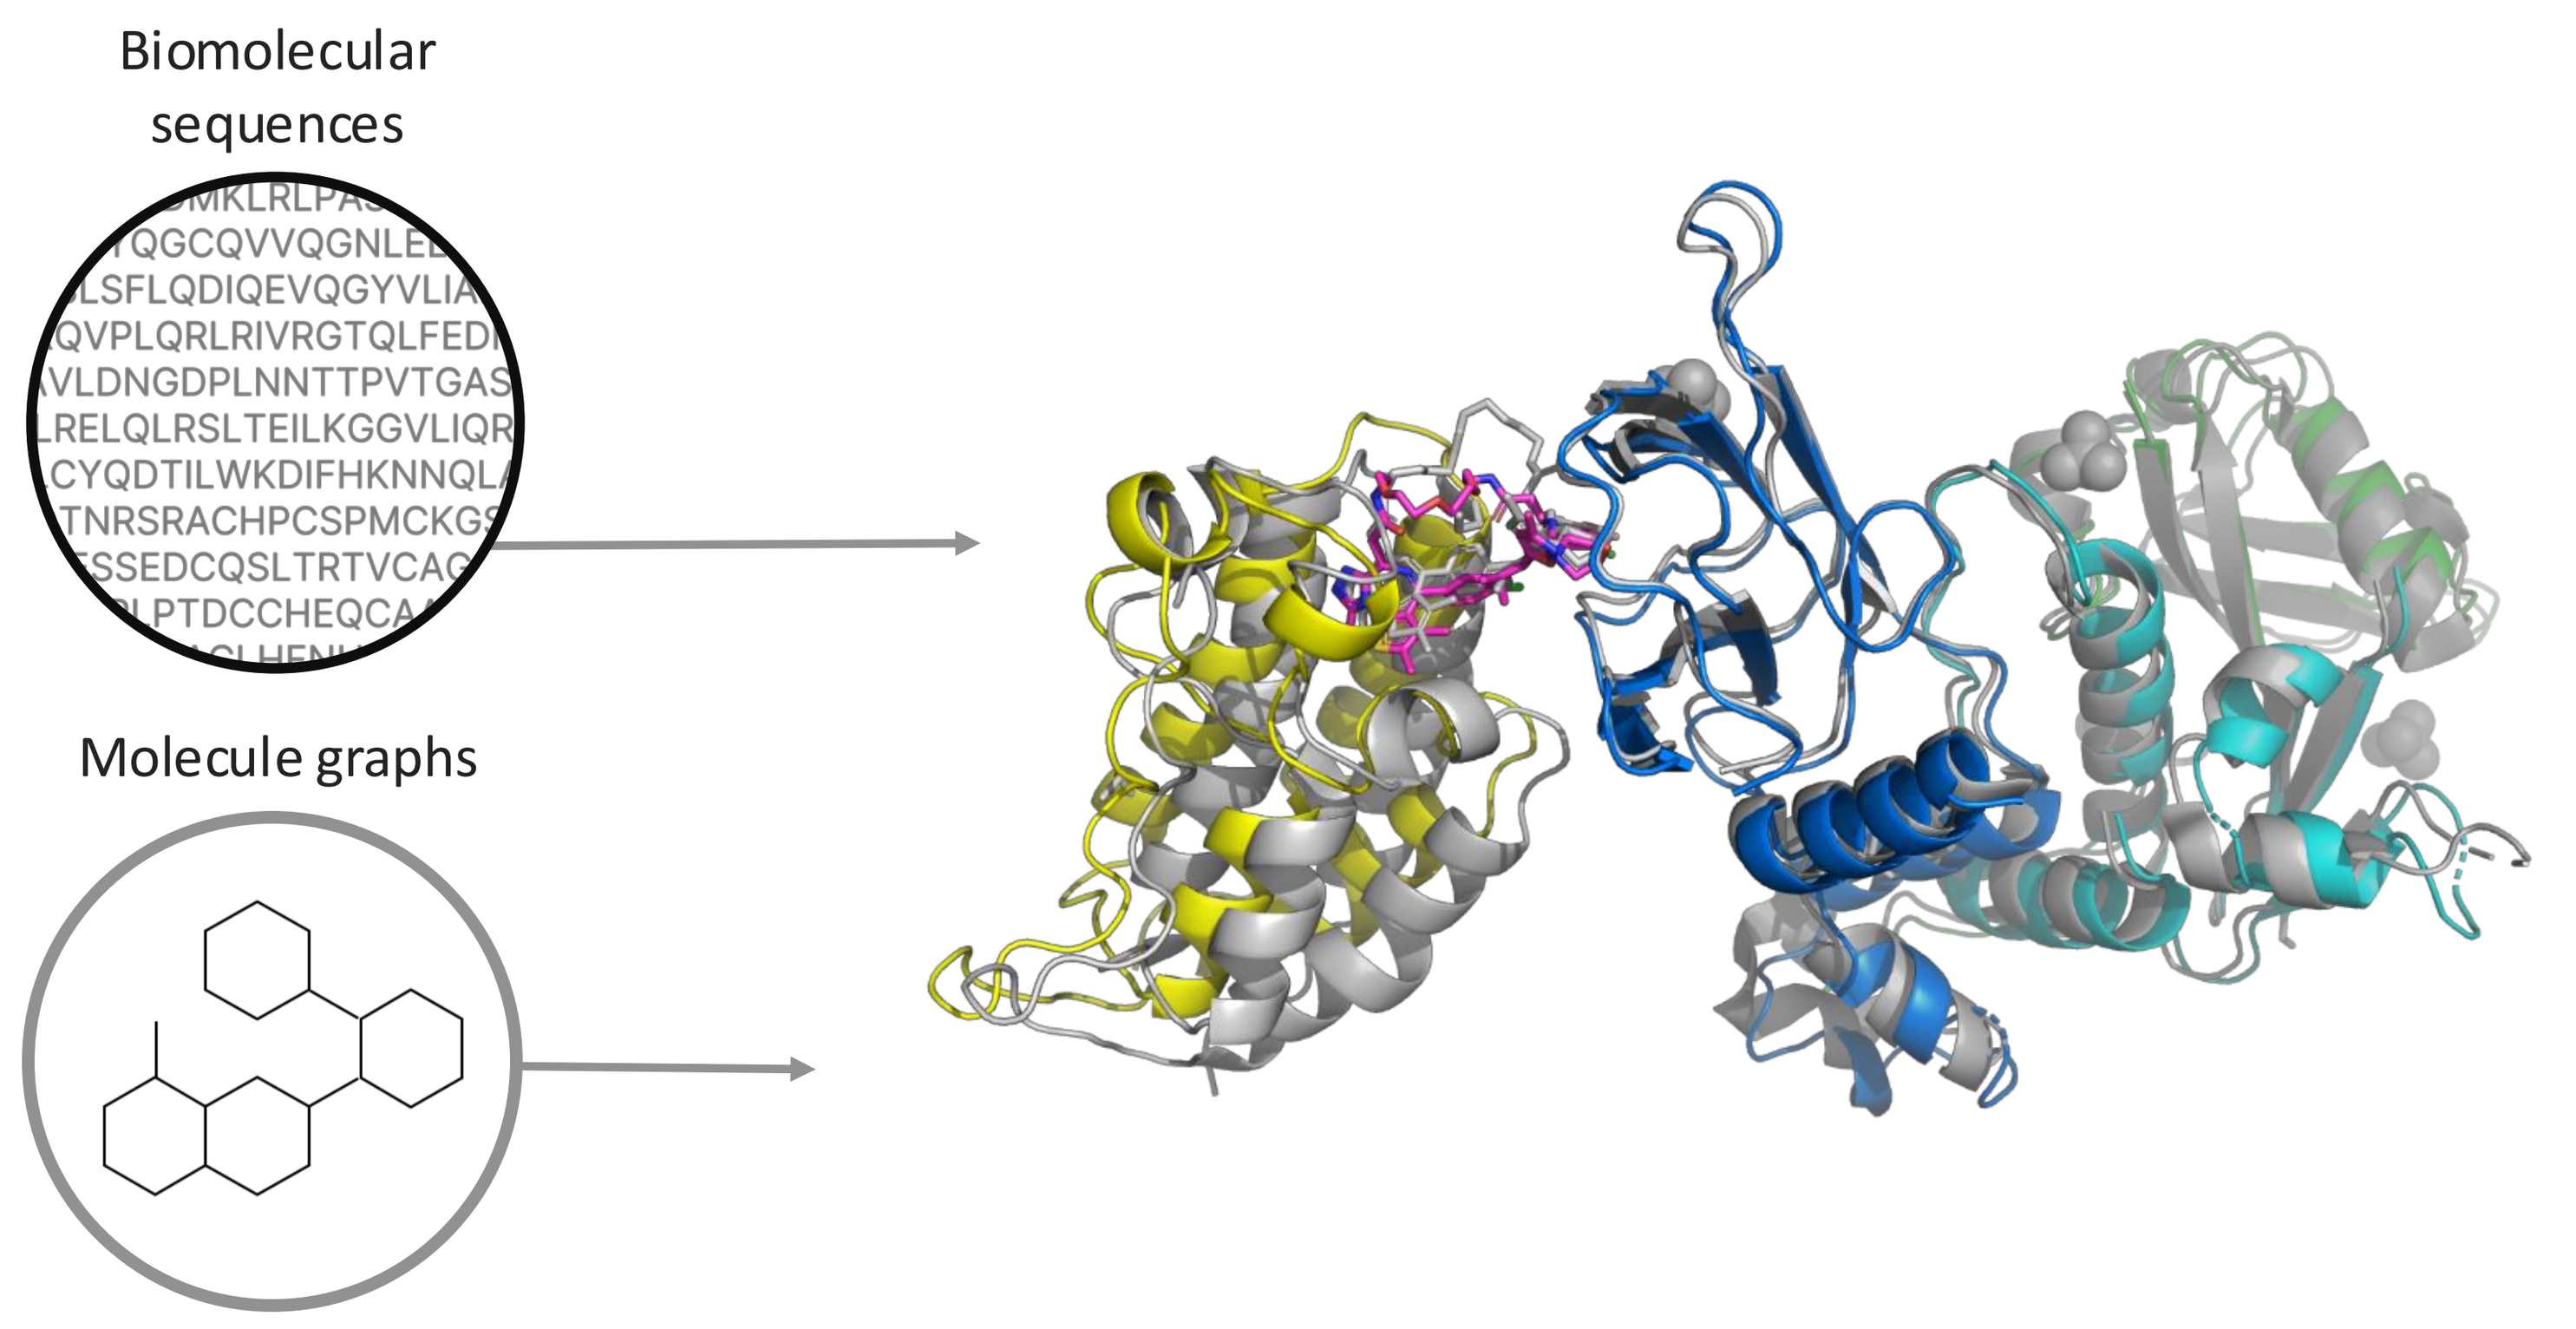
Biomolecular sequences
Molecule graphs

Model

NeuralPLexer3 is a conditional flow-matching model with a physics-informed polymer prior.

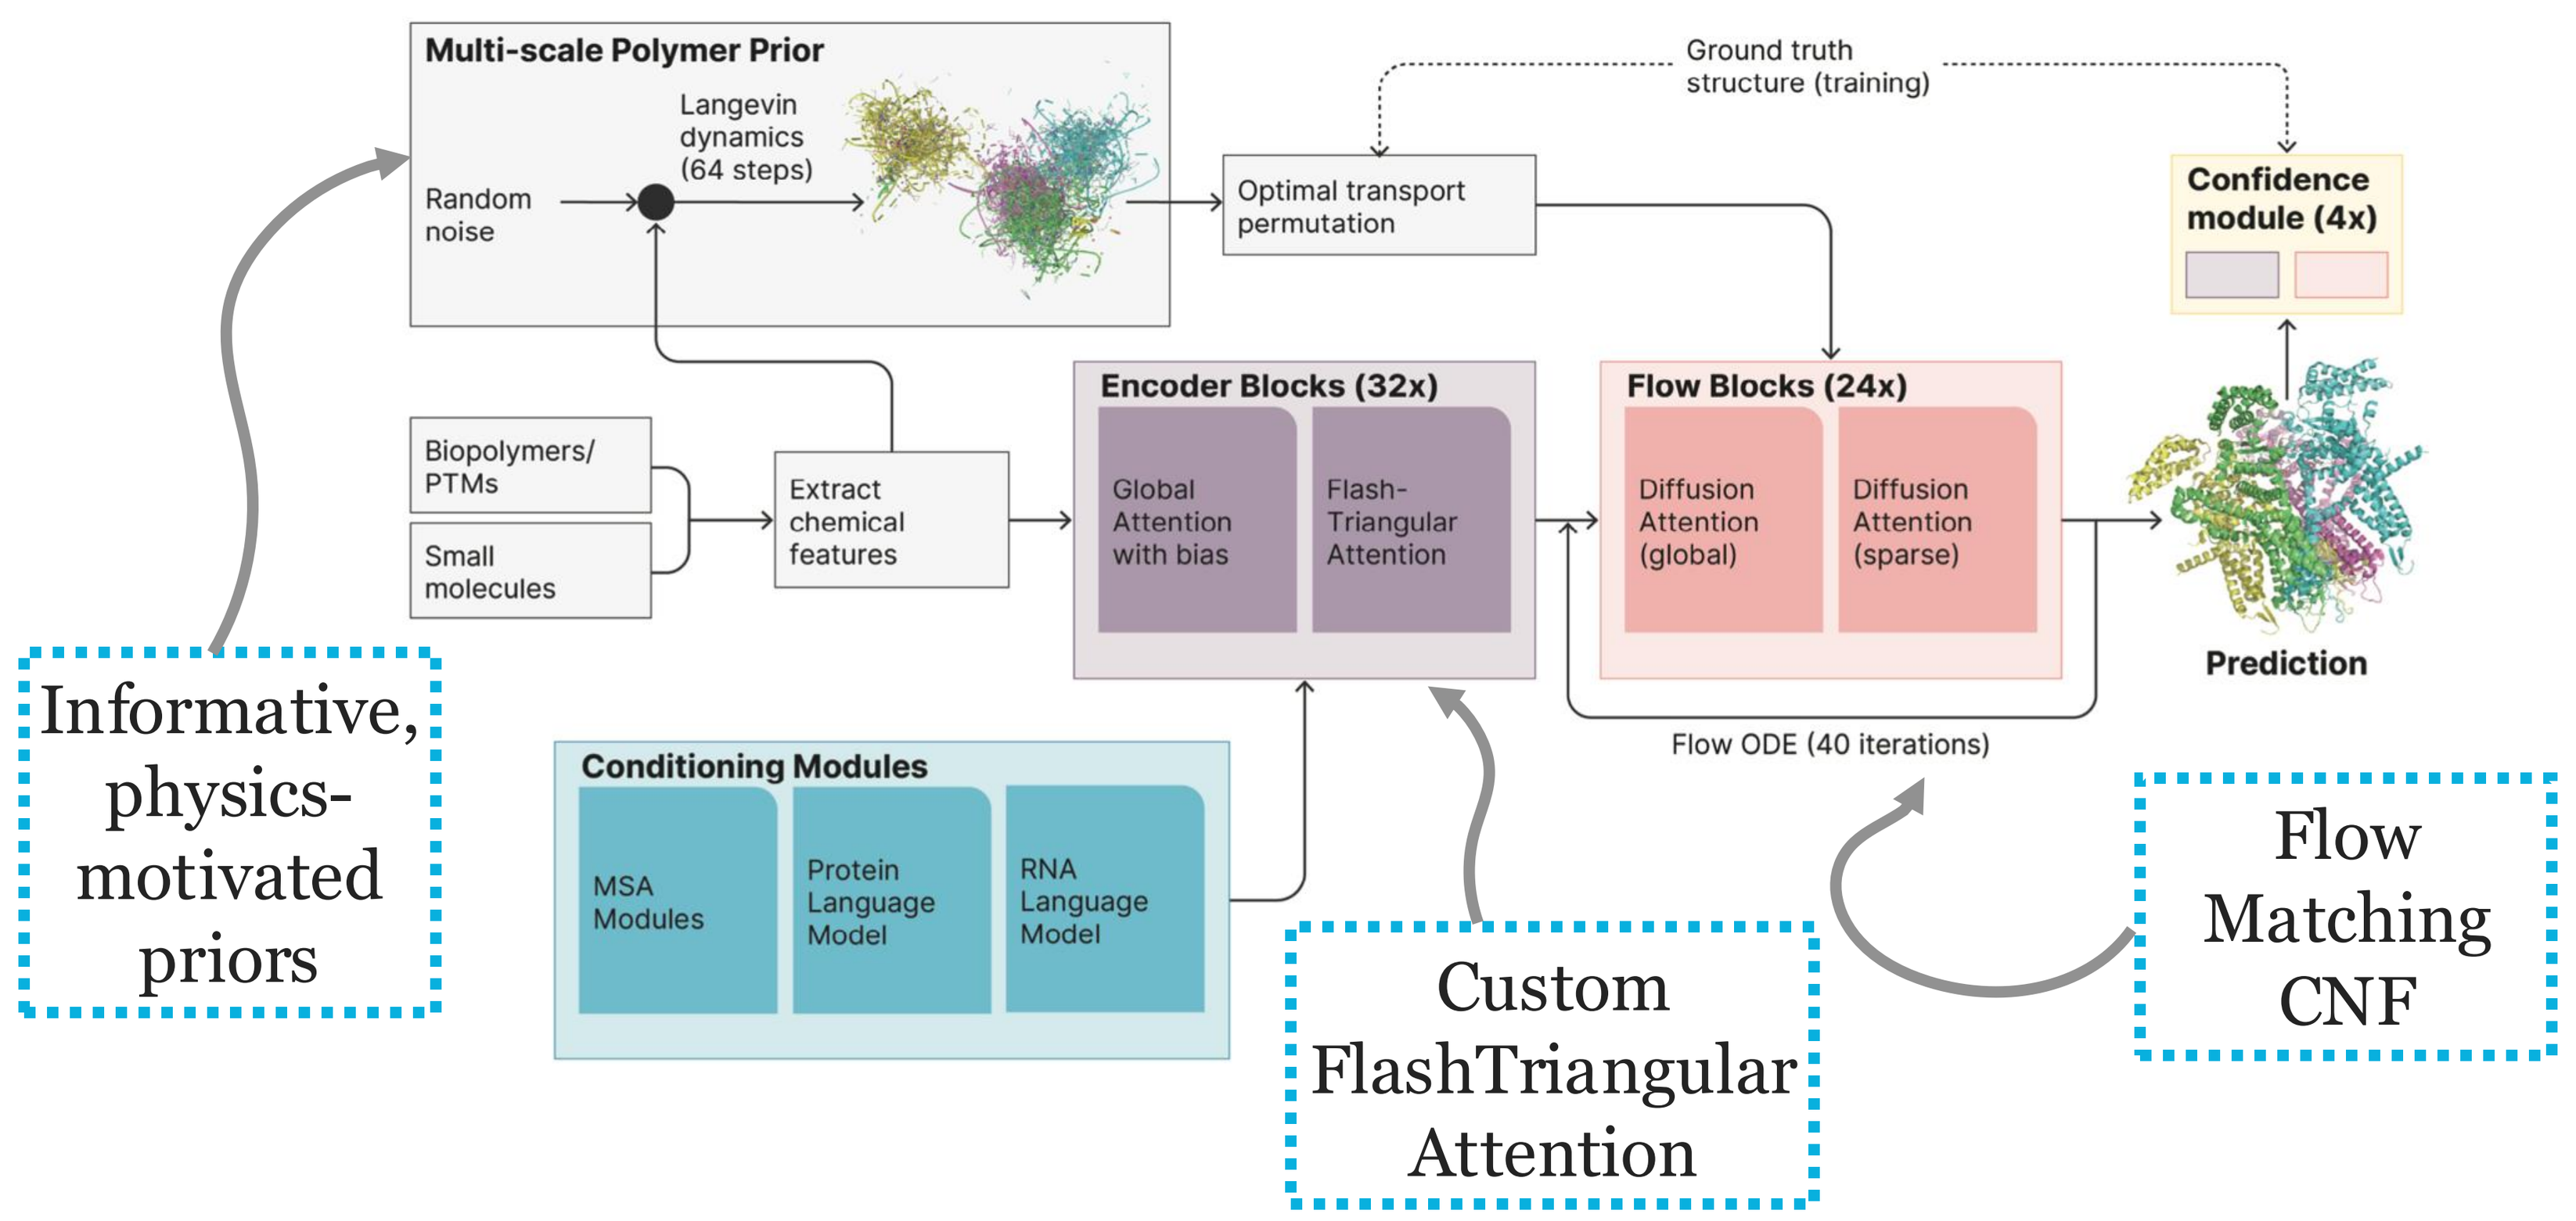
Multi-scale Polymer Prior
Random noise
Langevin dynamics (64 steps)
Ground truth structure (training)
Optimal transport permutation
Confidence module (4x)
Biopolymers/ PTMs
Small molecules
Extract chemical features
Encoder Blocks (32x)
Global Attention with bias
Flash-Triangular Attention
Flow Blocks (24x)
Diffusion Attention (global)
Diffusion Attention (sparse)
Prediction
Flow ODE (40 iterations)
Conditioning Modules
MSA Modules
Protein Language Model
RNA Language Model
Informative, physics-motivated priors
Custom FlashTriangular Attention
Flow Matching CNF

Training NeuralPLexer3

Compute optimal encoder-decoder scaling, and training and inference improvements led to NP3's high accuracy.

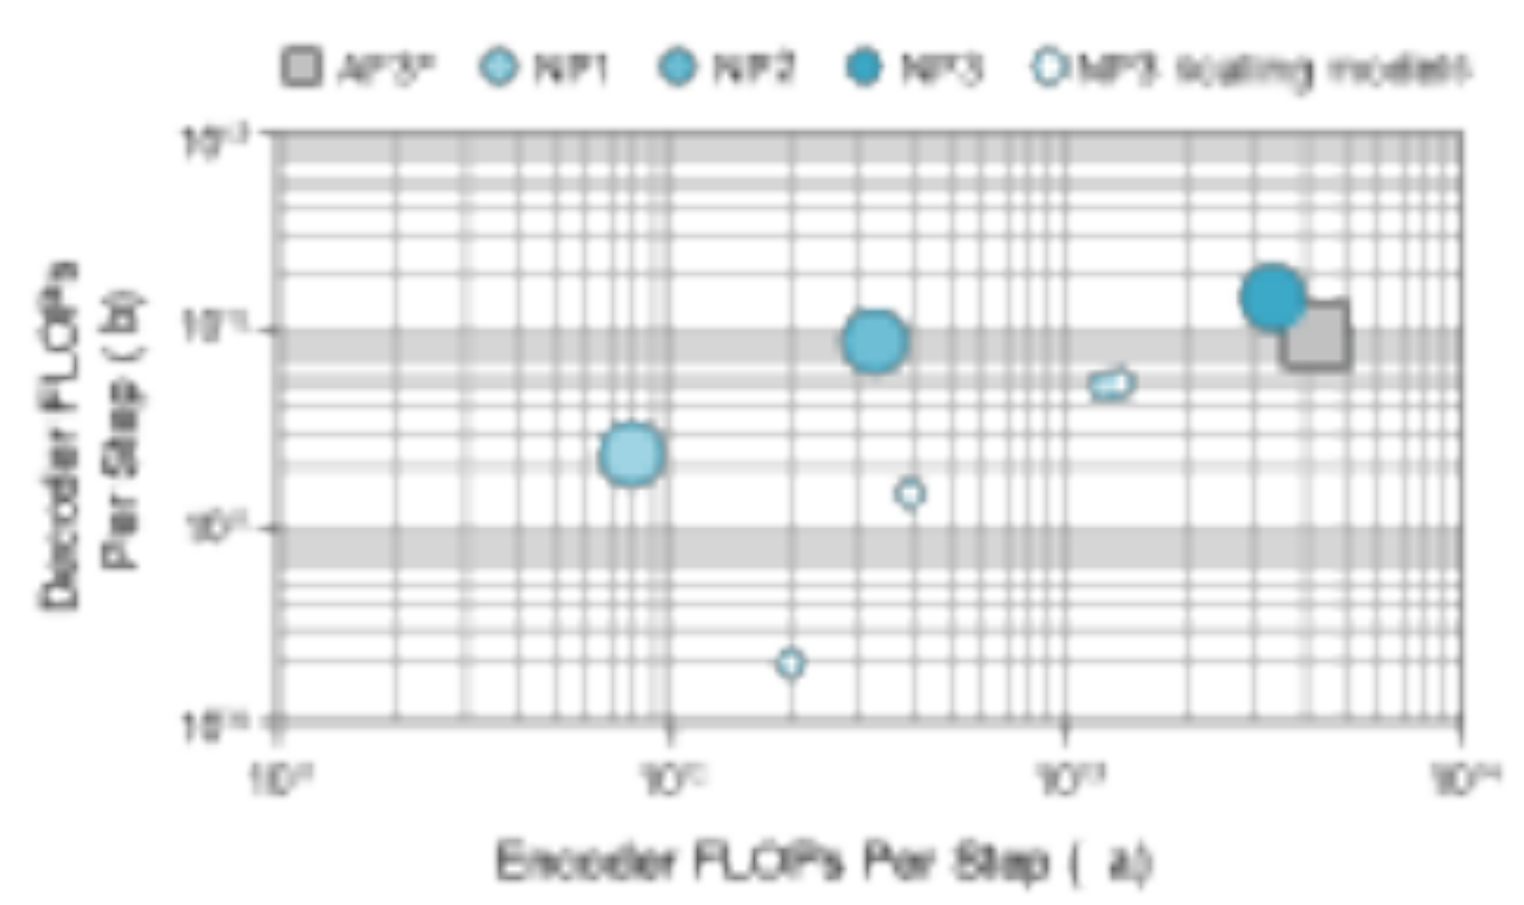
AF3* NP1 NP2 NP3 NP3 scaling models
Decoder FLOPs Per Step
Encoder FLOPs Per Step

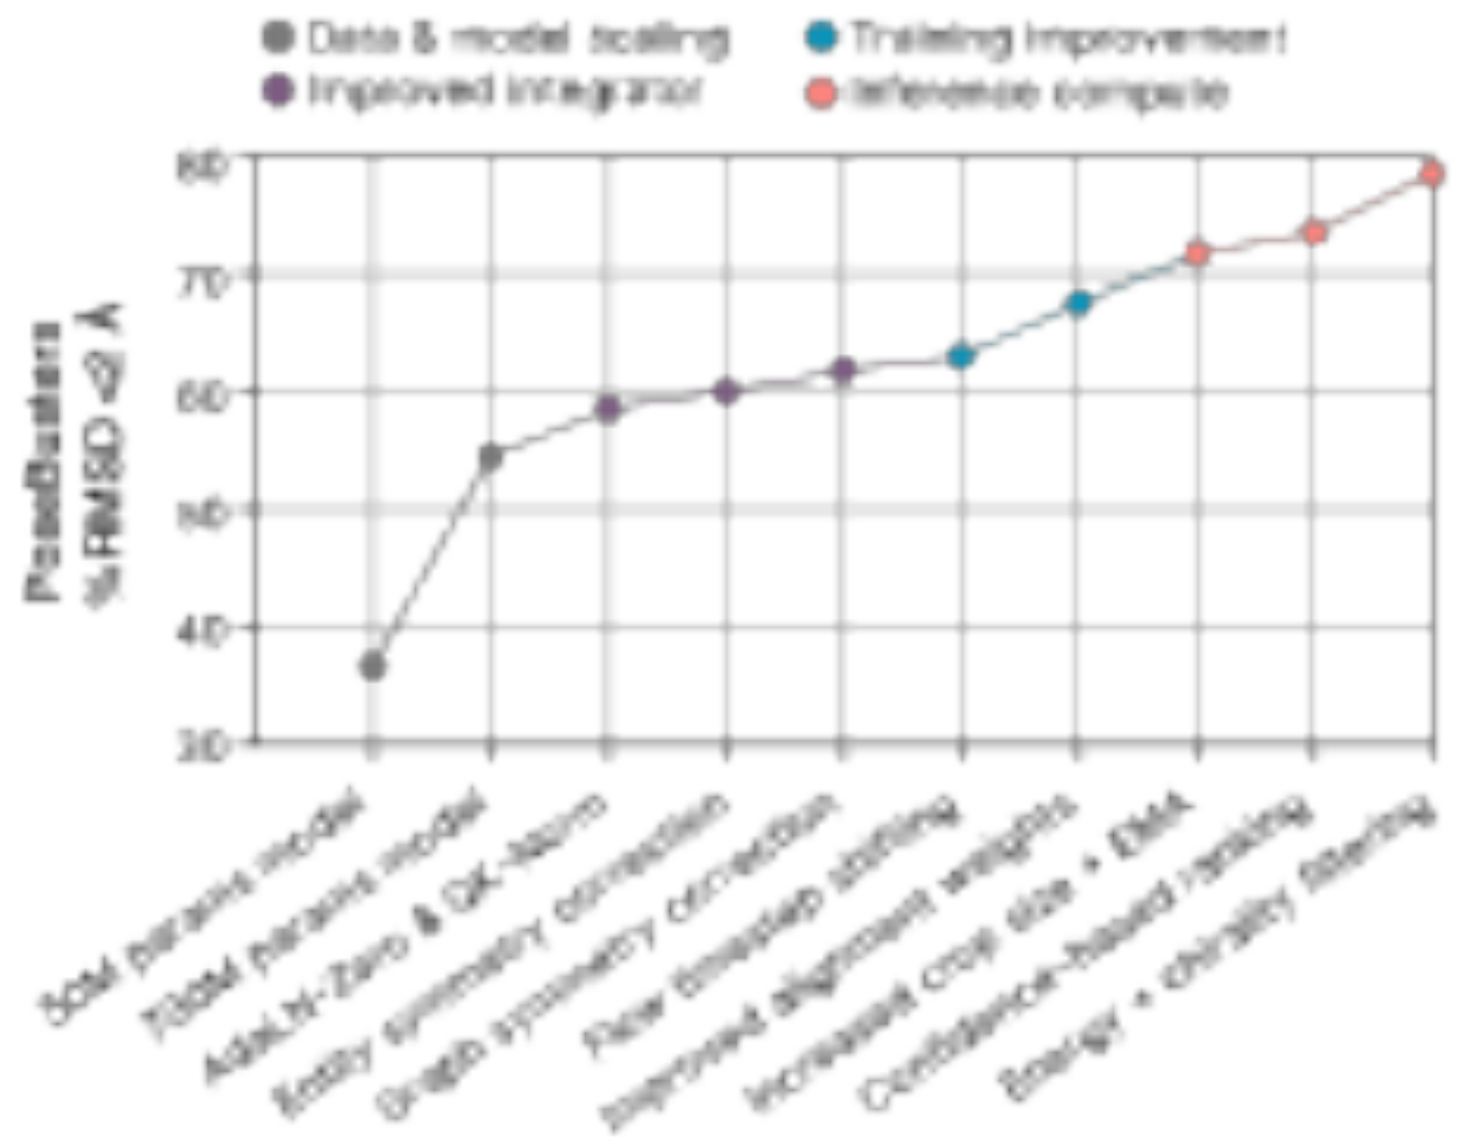
Data & model scaling
Training improvement
Improved integrator
Inference compute
PoseBusters %RMSD <2 Å

Our custom triangular attention kernel cuts peak memory by ~5× and gives ~40% speedup at 1k residues.

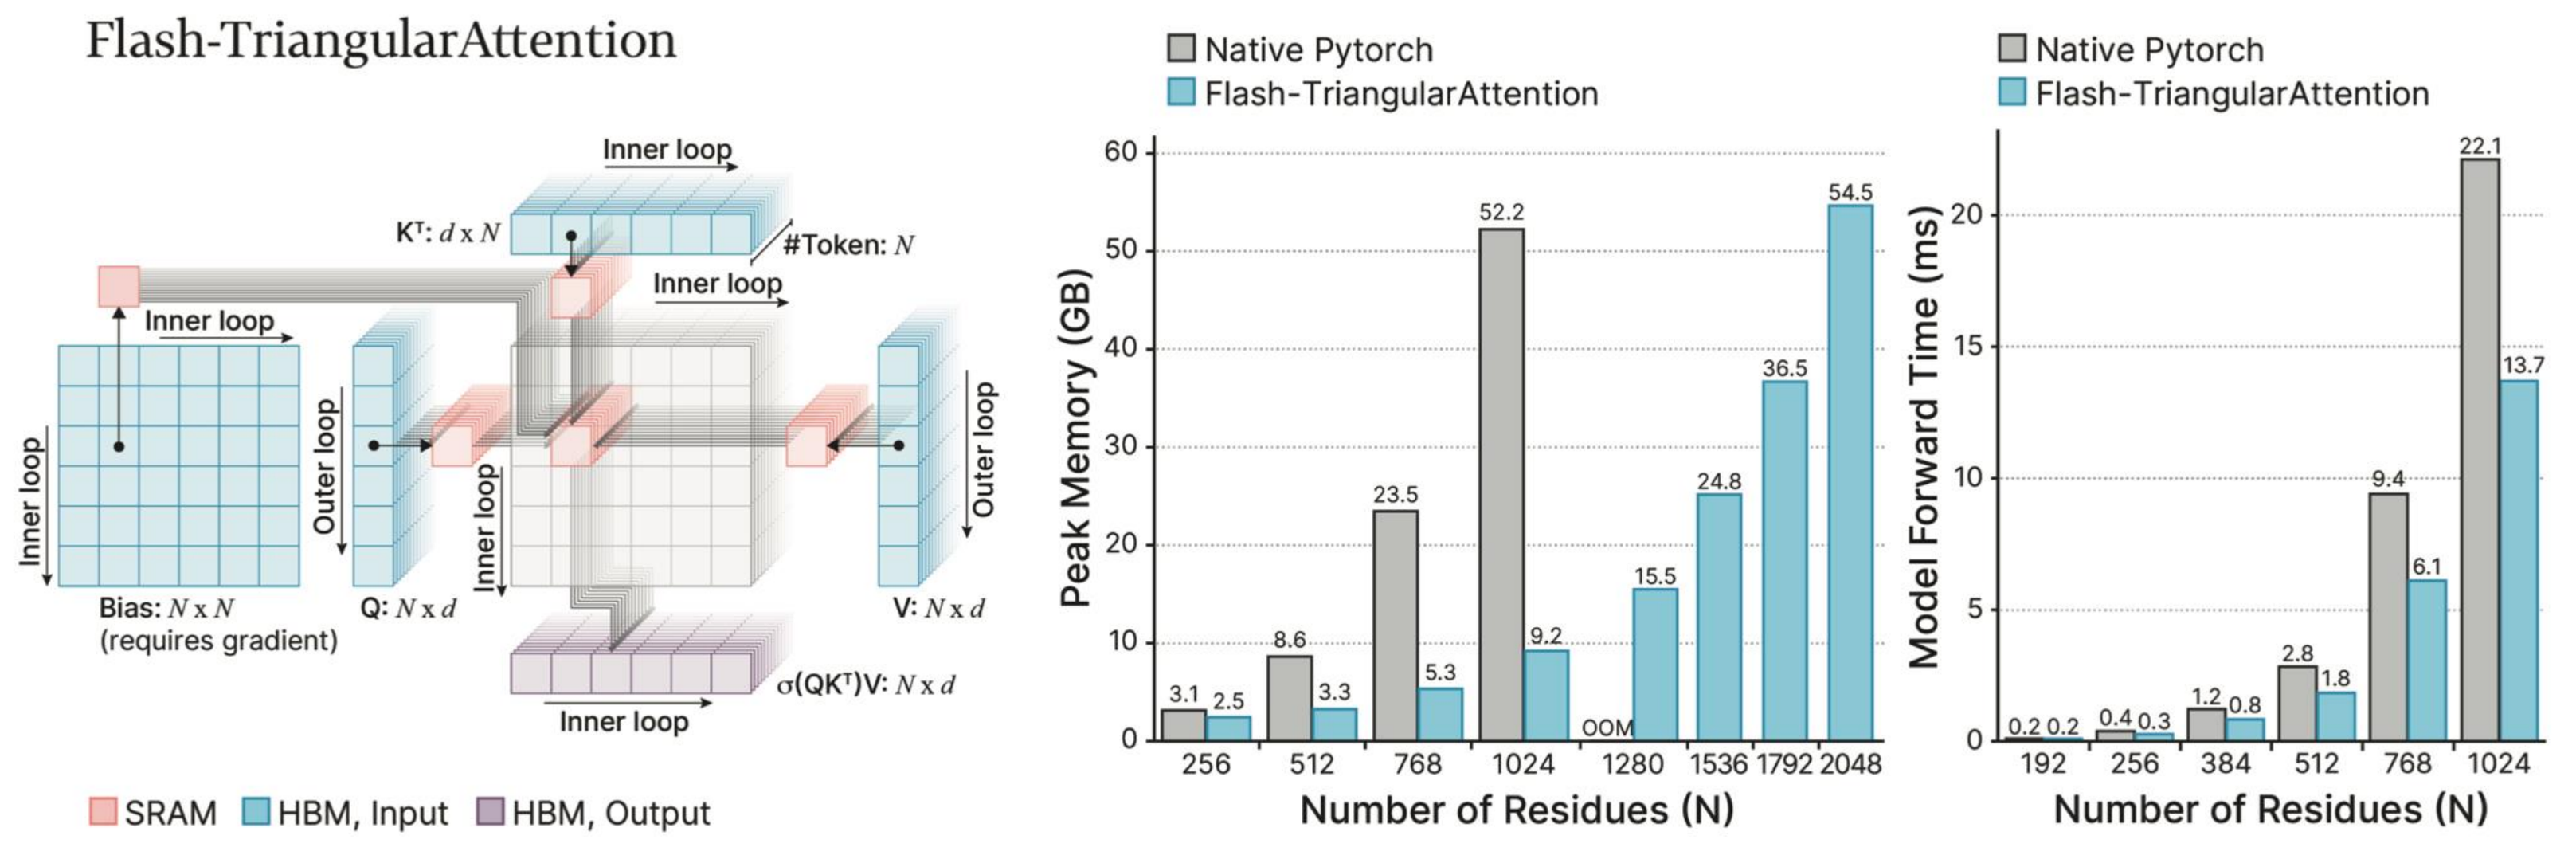
Flash-TriangularAttention
Inner loop
Outer loop
Kᵀ: d x N
#Token: N
Bias: N x N (requires gradient)
Q: N x d
V: N x d
σ(QKᵀ)V: N x d
SRAM HBM, Input HBM, Output
Native Pytorch
Flash-TriangularAttention
Peak Memory (GB)
Model Forward Time (ms)
Number of Residues (N)
3.1 2.5 8.6 3.3 23.5 5.3 52.2 9.2 OOM 15.5 24.8 36.5 54.5
256 512 768 1024 1280 1536 1792 2048
0.2 0.2 0.4 0.3 1.2 0.8 2.8 1.8 9.4 6.1 22.1 13.7
192 256 384 512 768 1024

Results

ConfBench is a novel benchmark for ligand-induced conformational changes in structure prediction models. NeuralPLexer3 shows good ability to predict holo-like structures upon ligand binding.

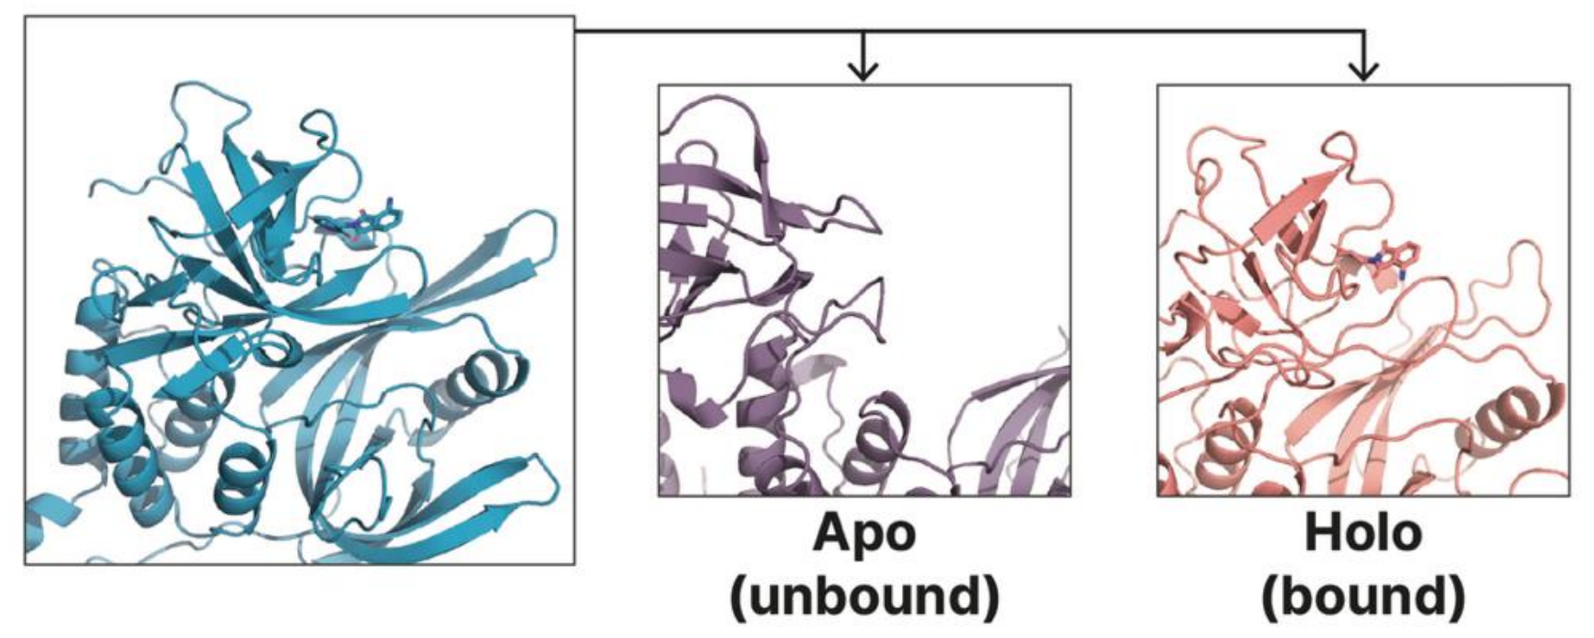
Apo (unbound)
Holo (bound)

$$\mathrm{Score}_{\mathrm{ConfBench}} = \frac{\mathrm{RMSD}_{\mathrm{query_alt}} - \mathrm{RMSD}_{\mathrm{query_ref}}}{\sqrt{\left(\mathrm{RMSD}^2_{\mathrm{query_alt}} + \mathrm{RMSD}^2_{\mathrm{query_ref}} + \mathrm{RMSD}^2_{\mathrm{alt_ref}}\right) \times \frac{1}{2}}}$$

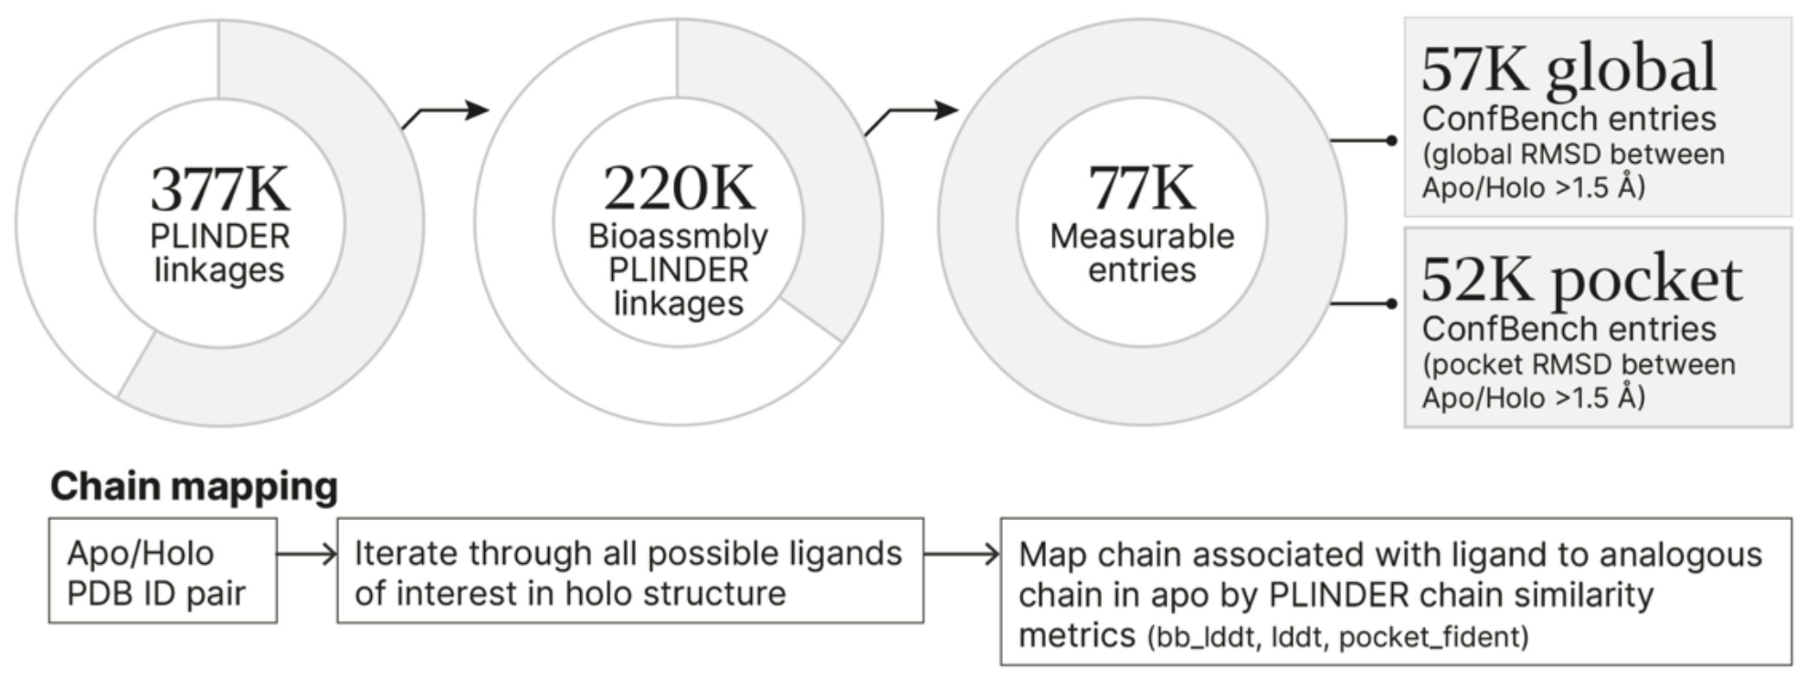
377K PLINDER linkages
220K Bioassmbly PLINDER linkages
77K Measurable entries
57K global ConfBench entries (global RMSD between Apo/Holo >1.5 Å)
52K pocket ConfBench entries (pocket RMSD between Apo/Holo >1.5 Å)
Chain mapping
Apo/Holo PDB ID pair
Iterate through all possible ligands of interest in holo structure
Map chain associated with ligand to analogous chain in apo by PLINDER chain similarity metrics (bb_lddt, lddt, pocket_fident)

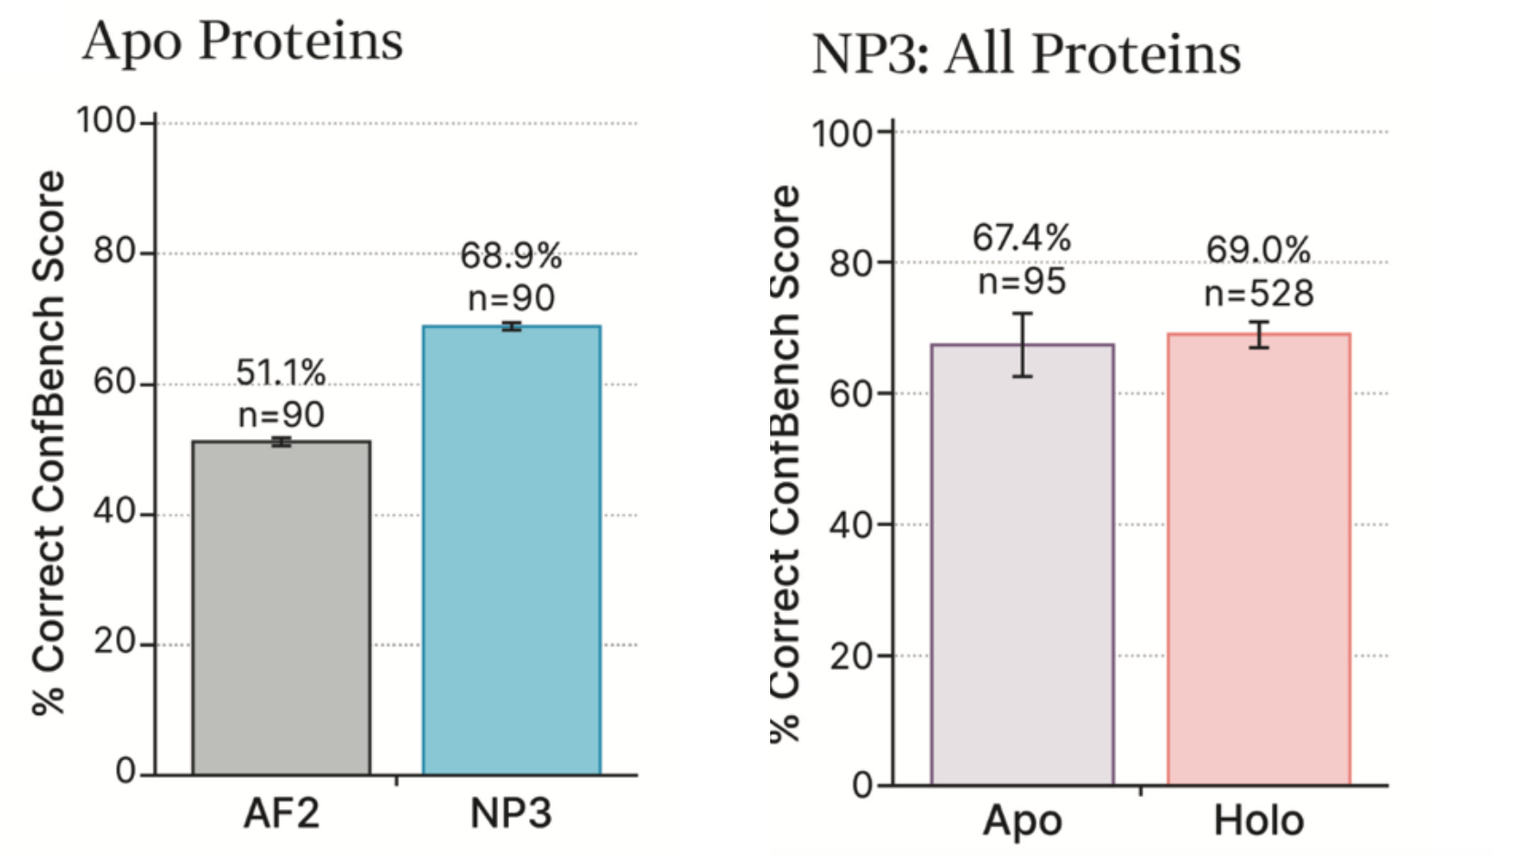
Apo Proteins
NP3: All Proteins
% Correct ConfBench Score
51.1% n=90
68.9% n=90
AF2 NP3
67.4% n=95
69.0% n=528
Apo Holo

Results

PoseBusters is a benchmark for small-molecule structure-based drug design. NeuralPLexer3 gives more accurate structures, gives more rigorous stereochemistry, and is much faster than AlphaFold 3.

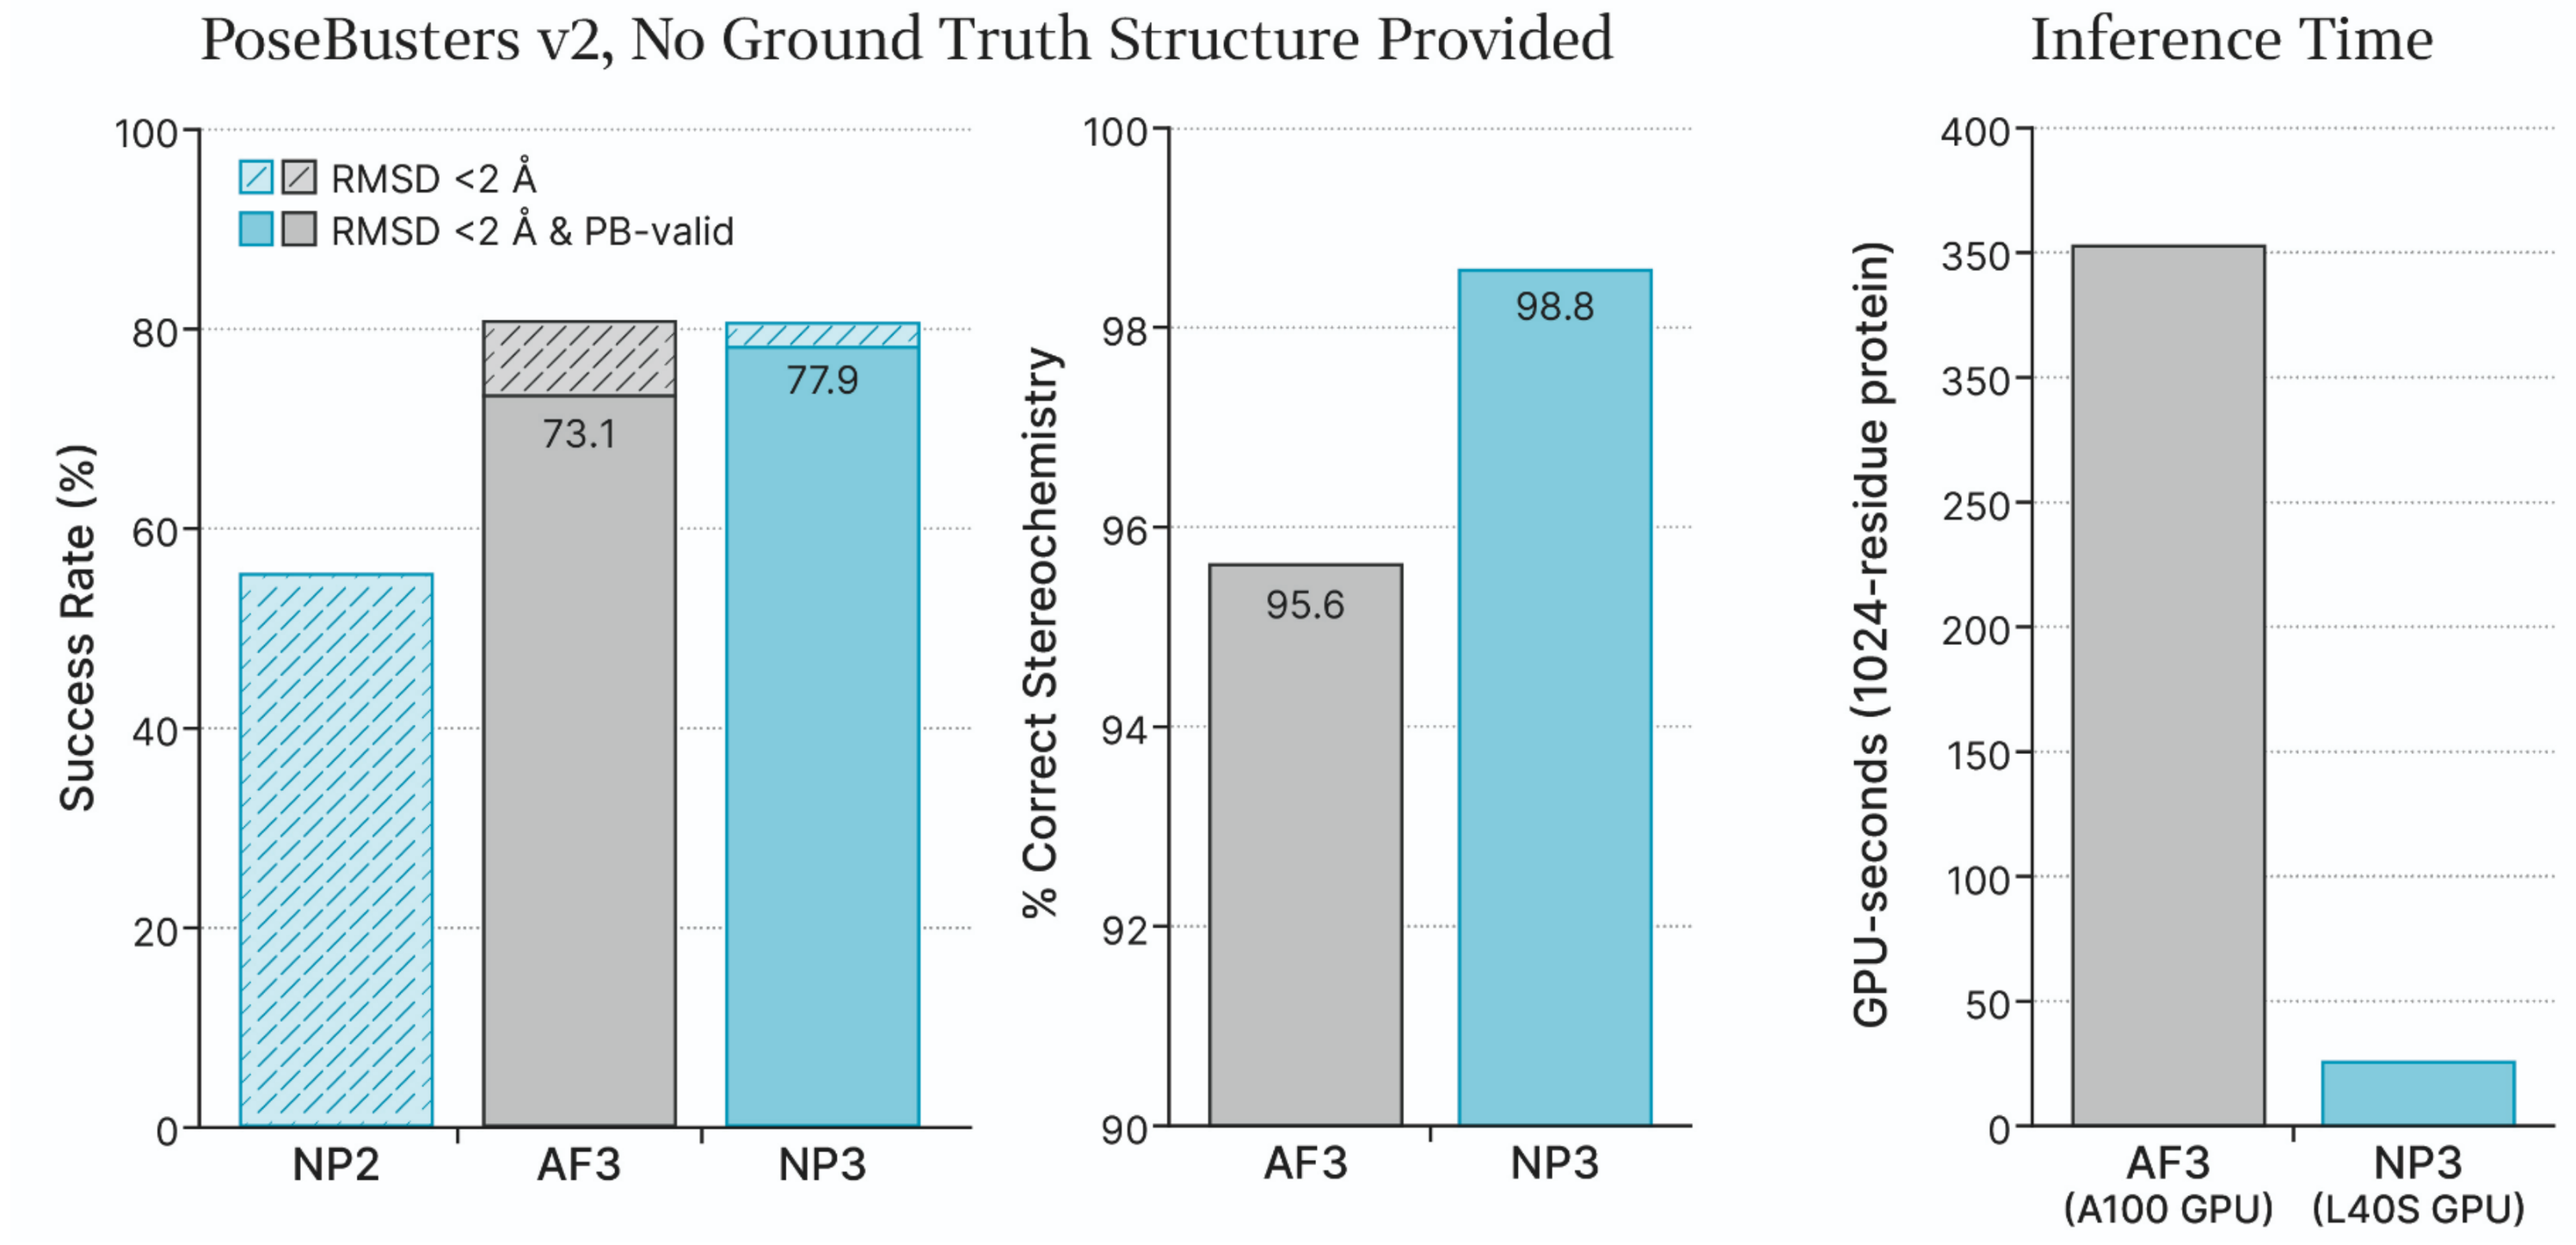
PoseBusters v2, No Ground Truth Structure Provided
Inference Time
RMSD <2 Å
RMSD <2 Å & PB-valid
Success Rate (%)
73.1
77.9
NP2 AF3 NP3
% Correct Stereochemistry
95.6
98.8
AF3 NP3
GPU-seconds (1024-residue protein)
AF3 (A100 GPU)
NP3 (L40S GPU)

NeuralPLexer3 is accurate across diverse categories of biomolecular interactions. Test structures are low-homology, low-Tanimoto-similarity, recent PDB entries.

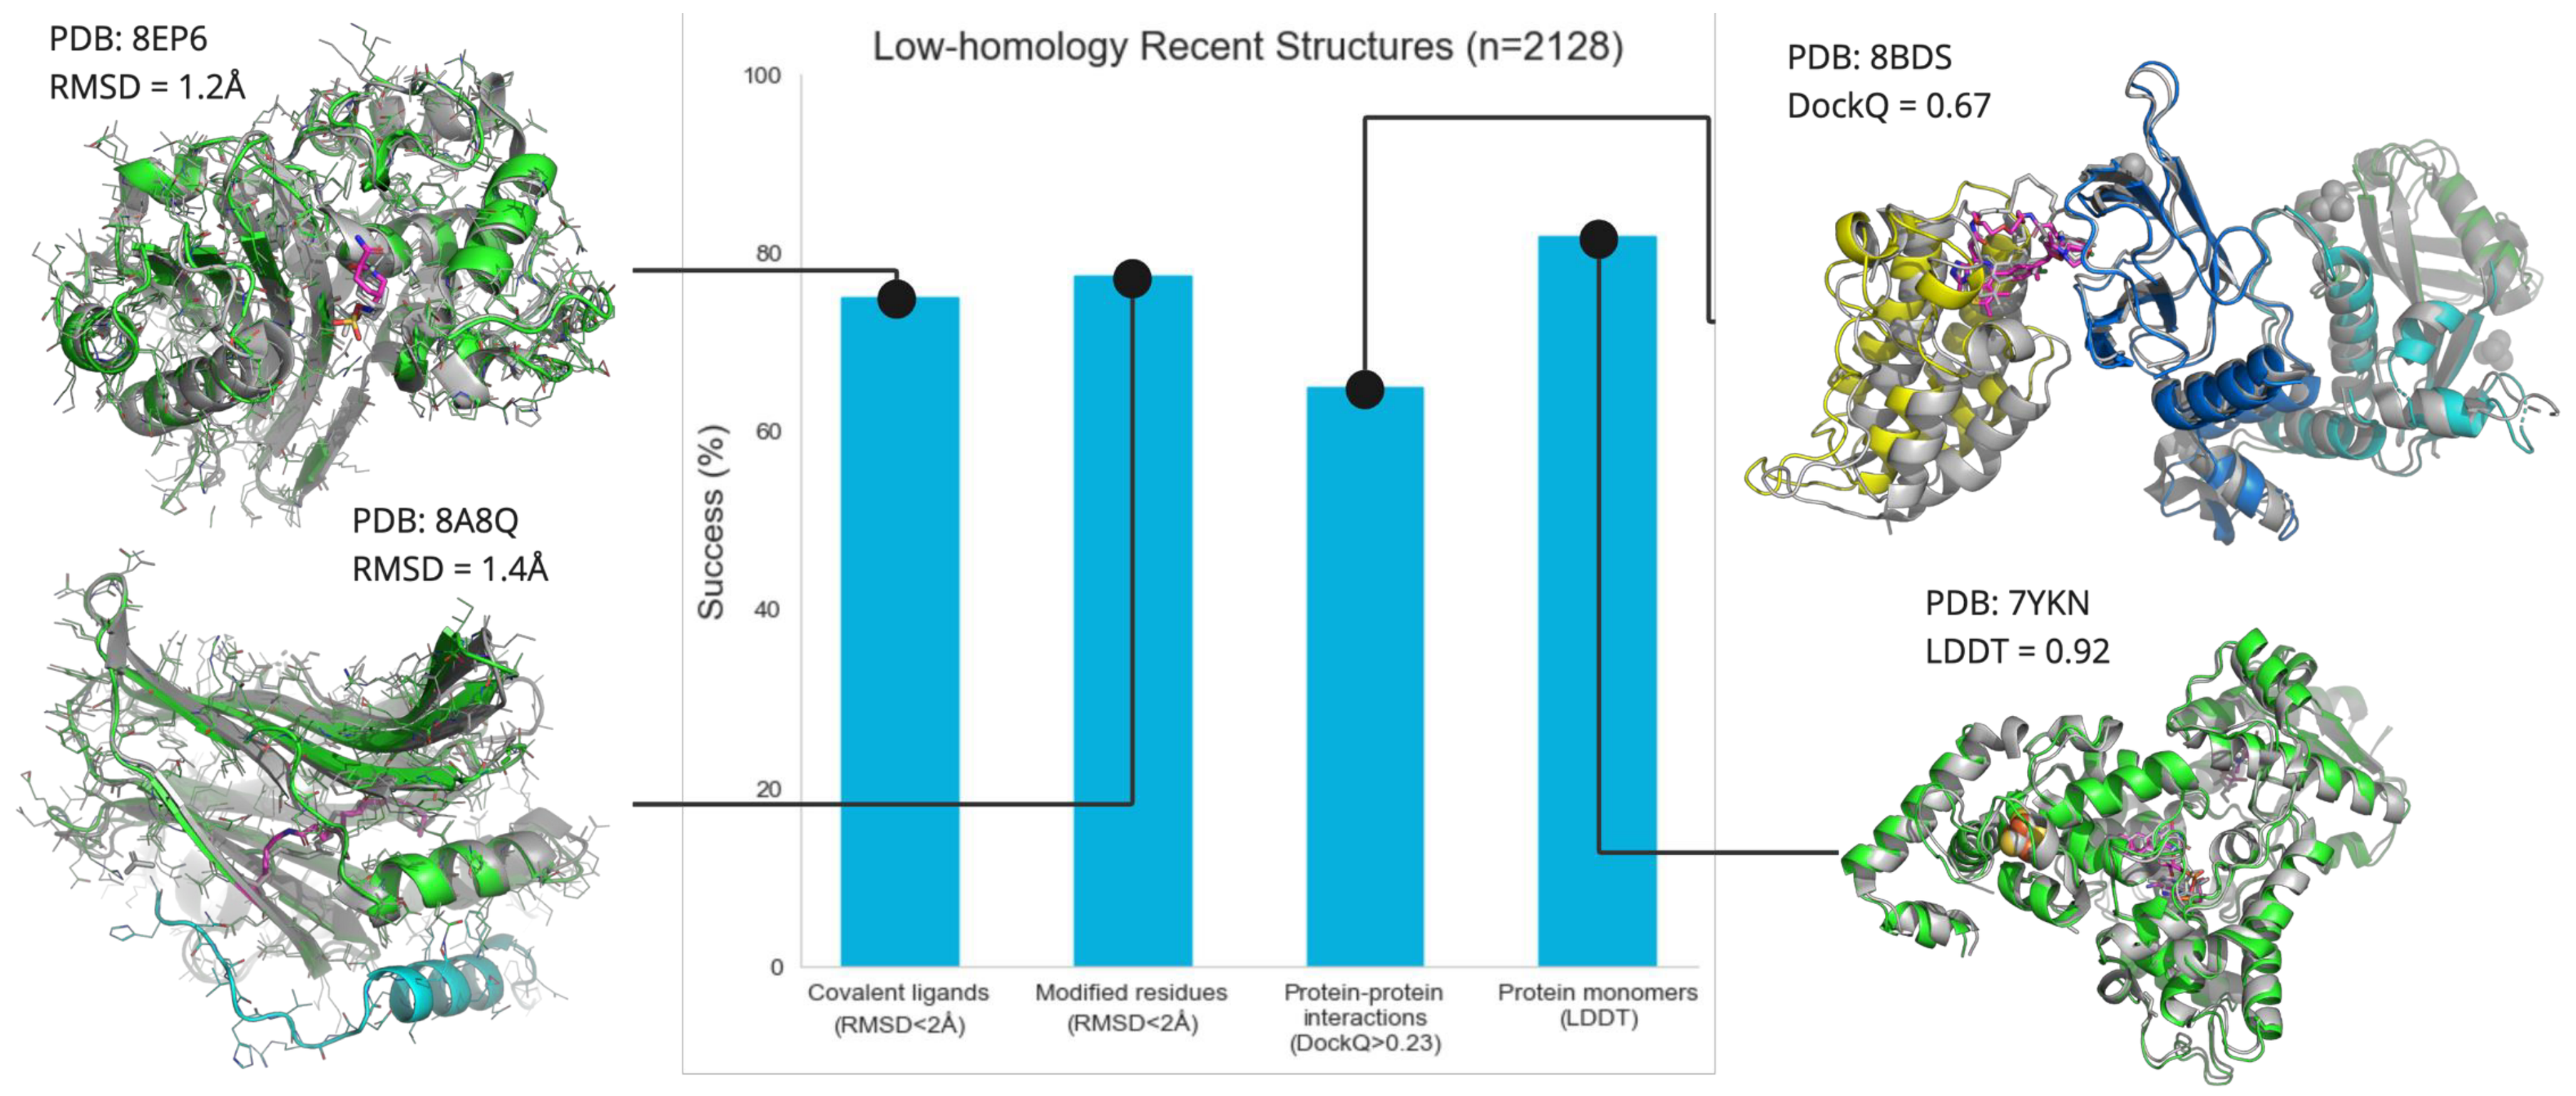
PDB: 8EP6
RMSD = 1.2Å
PDB: 8A8Q
RMSD = 1.4Å
Low-homology Recent Structures (n=2128)
Success (%)
Covalent ligands (RMSD<2Å)
Modified residues (RMSD<2Å)
Protein-protein interactions (DockQ>0.23)
Protein monomers (LDDT)
PDB: 8BDS
DockQ = 0.67
PDB: 7YKN
LDDT = 0.92

NeuralPLexer3 provides well callibraed confidence estimates for protein-ligand binding (ligand pLDDT) and protein-protein interfaces (pDockQ).

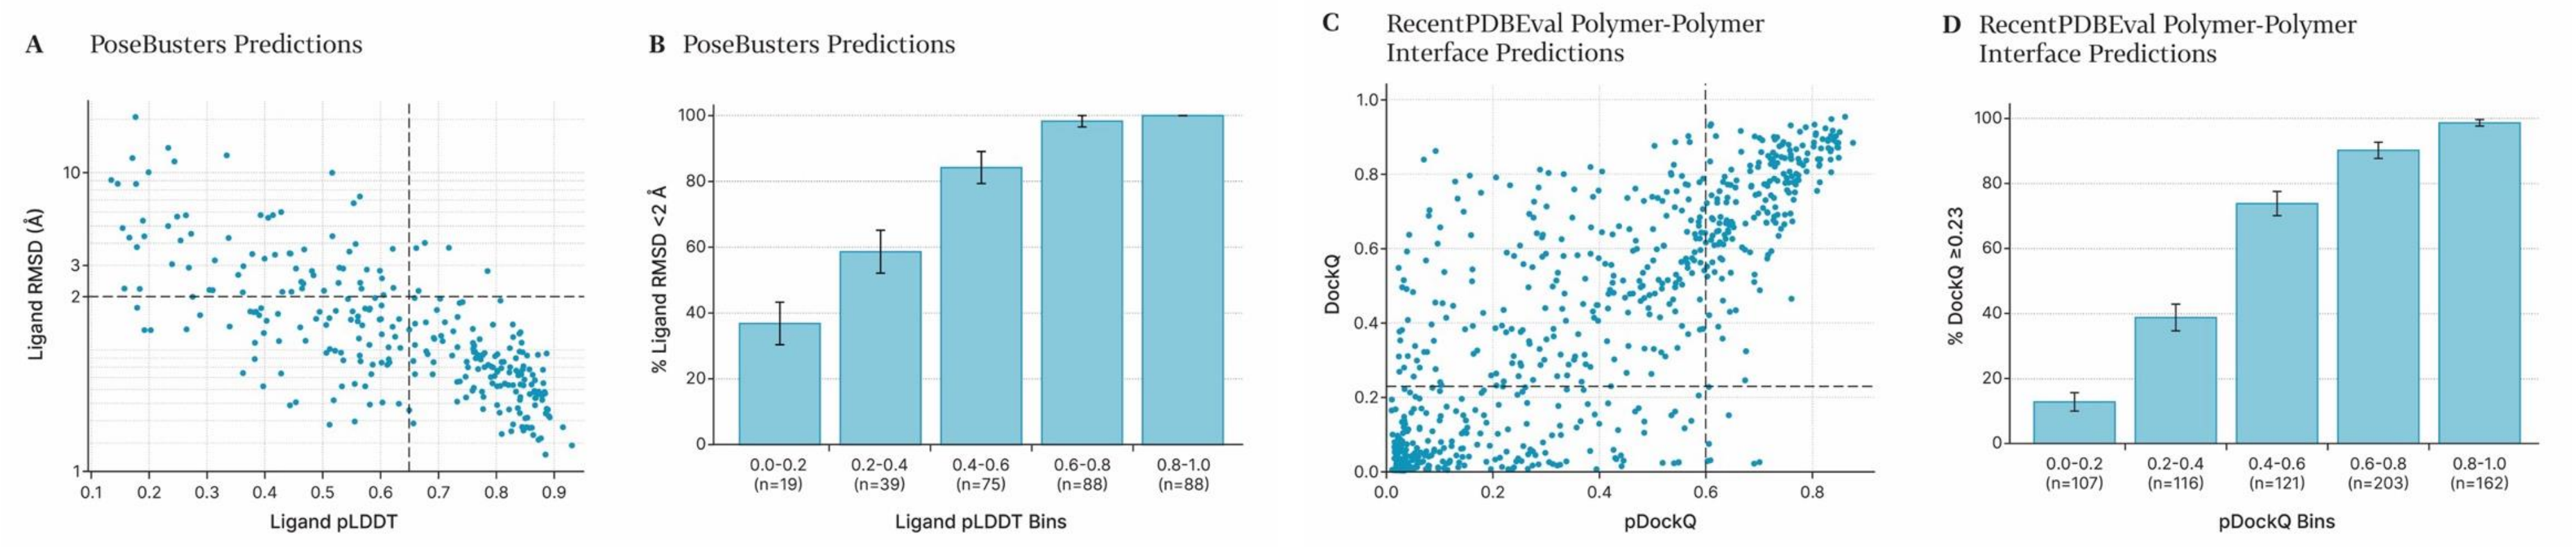
A PoseBusters Predictions
Ligand RMSD (Å)
Ligand pLDDT
B PoseBusters Predictions
% Ligand RMSD <2 Å
Ligand pLDDT Bins
C RecentPDBEval Polymer-Polymer Interface Predictions
DockQ
pDockQ
D RecentPDBEval Polymer-Polymer Interface Predictions
% DockQ ≥0.23
pDockQ Bins

Iambic | iambic.ai @iambic_ai | NON-CONFIDENTIAL