Analysis of tumor infiltrating lymphocytes and in vitro exhausted T cell models to identify unique Immuno-Oncology targets

James Ackroyd[1], Joanne Berry[1], Yaoyao Fu[2], Jason Allen[1], Martin Barnes[1], Alexander Patterson[1], Angelo Kaplan[2], Nickolas Attanasio[2], Rachel Dusek[2], Christian Rohlff[1], Haining Huang[2], Livija Deban[1], Robert Boyd[1]. [1]Oxford BioTherapeutics Ltd, Abingdon, United Kingdom; [2]Oxford BioTherapeutics Inc, San Jose, CA

AACR Abstract Number: 3715

OXFORD BioTherapeutics

Study rationale

T cells in tumors are exposed to persistent antigen stimulation and/or inflammatory signals. Such prolonged stimulation is often associated with deterioration of T cell effector functions resulting in a functional state termed 'exhaustion'. Identification of cell surface proteins involved in induction and maintenance of the exhausted state could inform novel immunomodulatory cancer therapies. We conducted in-depth expression profiling of membrane proteins from intact tumors (Colorectal, Pancreatic, Lung, Breast and Prostate cancer), different *in vitro* models of T cell exhaustion and primary tumor-derived lymphocytes (Colorectal, Breast and Lung cancer) using proteomics to identify novel Immuno-Oncology (IO) targets. Analysis of proteomic expression data revealed potential novel IO targets in primary tumor-derived lymphocytes (TILs) with significant cancer expression.

To rapidly validate IO targets emerging from the above approach we analyzed fresh tumors using an integrated evaluation methodology. Tumors were subject to multiple parallel analysis steps: (i) Unbiased proteomic analysis of the tumor and TILs; (ii) Flow cytometry analysis of TILs; (iii) Immuno-histochemical analysis of tumor sections; (iv) *in vitro* functional modulation of PBMCs; (v) *ex vivo* functional modulation of TILs and normal resident lymphocytes (NRLs). We show the data for one specific I-O target, OXBT185. Antibody modulation of this target results in potent T cell activation.

i. Exhausted T cell and TIL proteomics

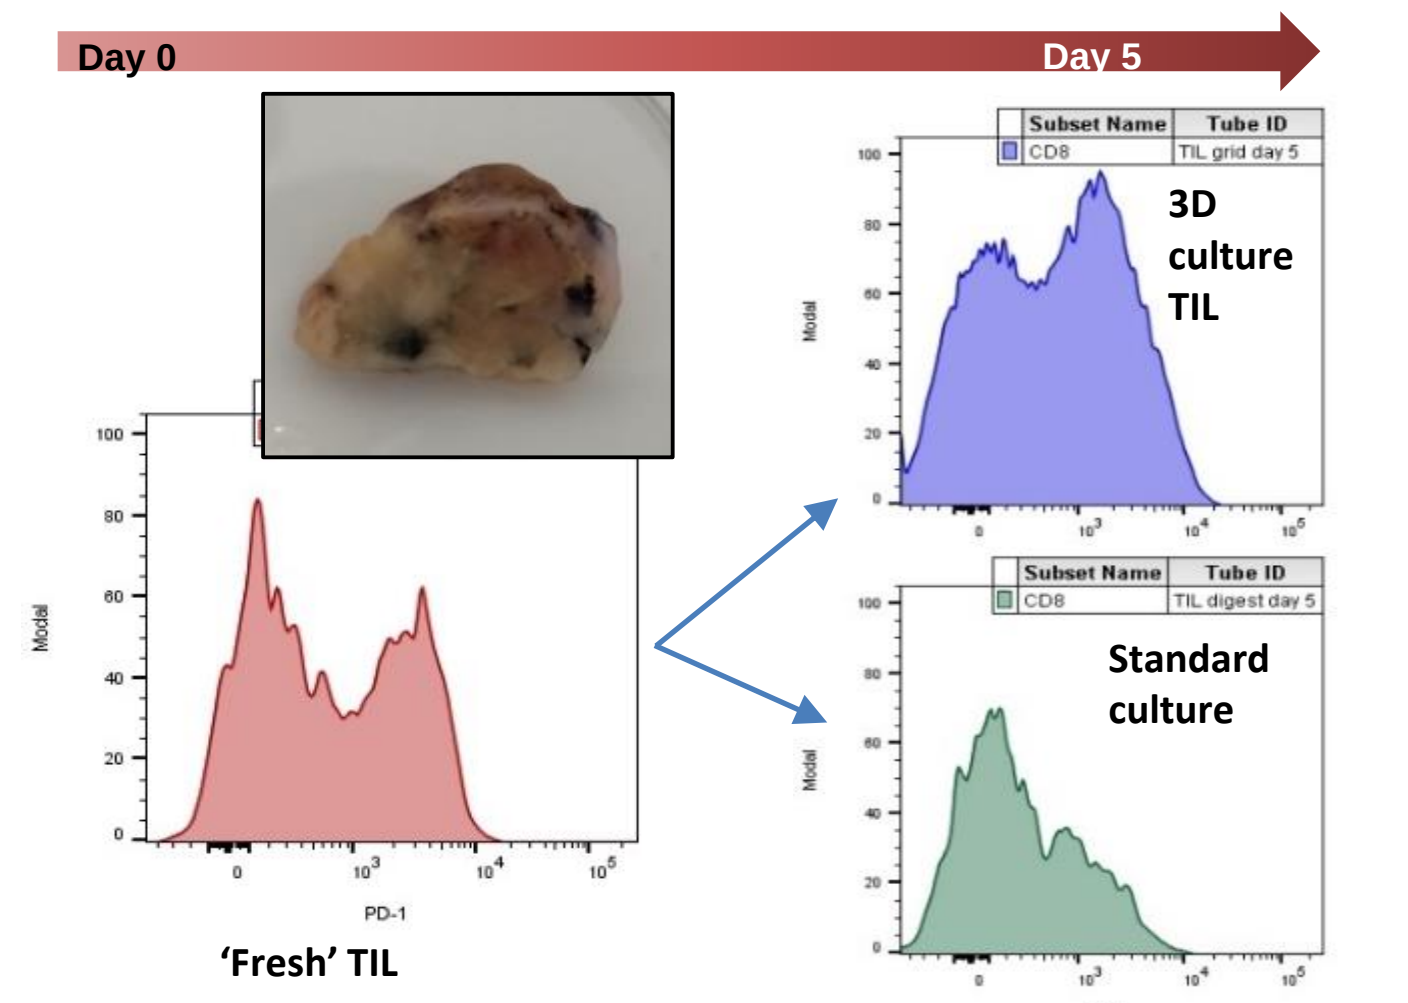
Day 0
Day 5
3D culture TIL
Standard culture
'Fresh' TIL
PD-1

Figure 1. FACS Analysis comparing PD-1 expression in fresh isolated TILs and 3D cultured TILs after 5 days.

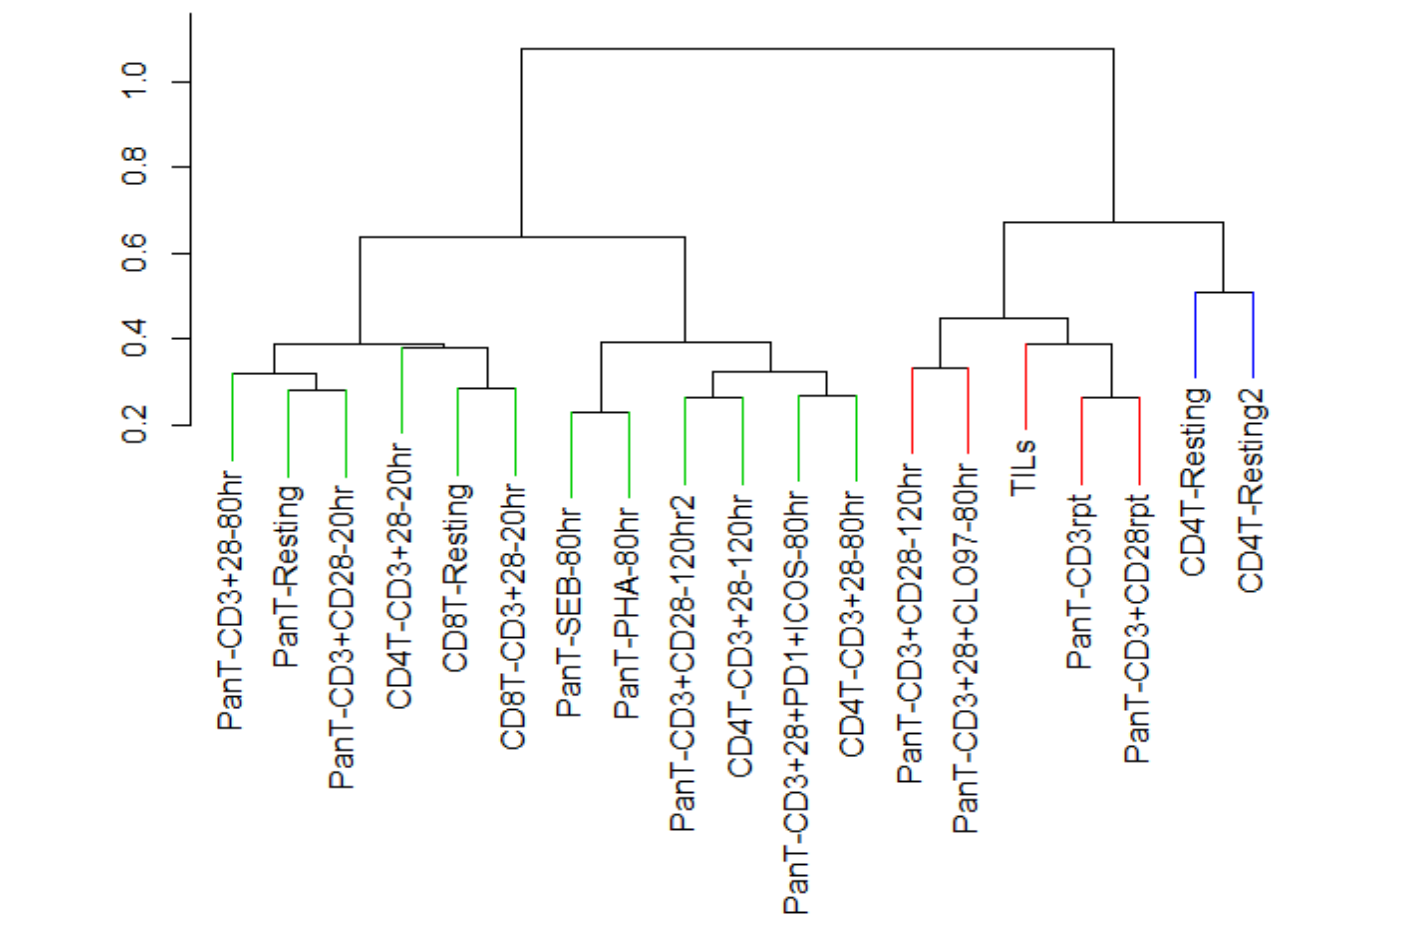
PanT-CD3+28-80hr
PanT-Resting
PanT-CD3+CD28-20hr
CD4T-CD3+28-20hr
CD8T-Resting
CD8T-CD3+28-20hr
PanT-SEB-80hr
PanT-PHA-80hr
PanT-CD3+CD28-120hr2
CD4T-CD3+28-120hr
PanT-CD3+28+PD1+ICOS-80hr
CD4T-CD3+28-80hr
PanT-CD3+CD28-120hr
PanT-CD3+28+CLO97-80hr
TILs
PanT-CD3rpt
PanT-CD3+CD28rpt
CD4T-Resting
CD4T-Resting2

Figure 2. Hierarchical clustering of TILs and exhausted T cell models. Generated using a binary distance matrix and Ward scoring methods available in R (Version 3.2.0). Exhausted T cell models exhibit close expression to TILs isolated from CRC.

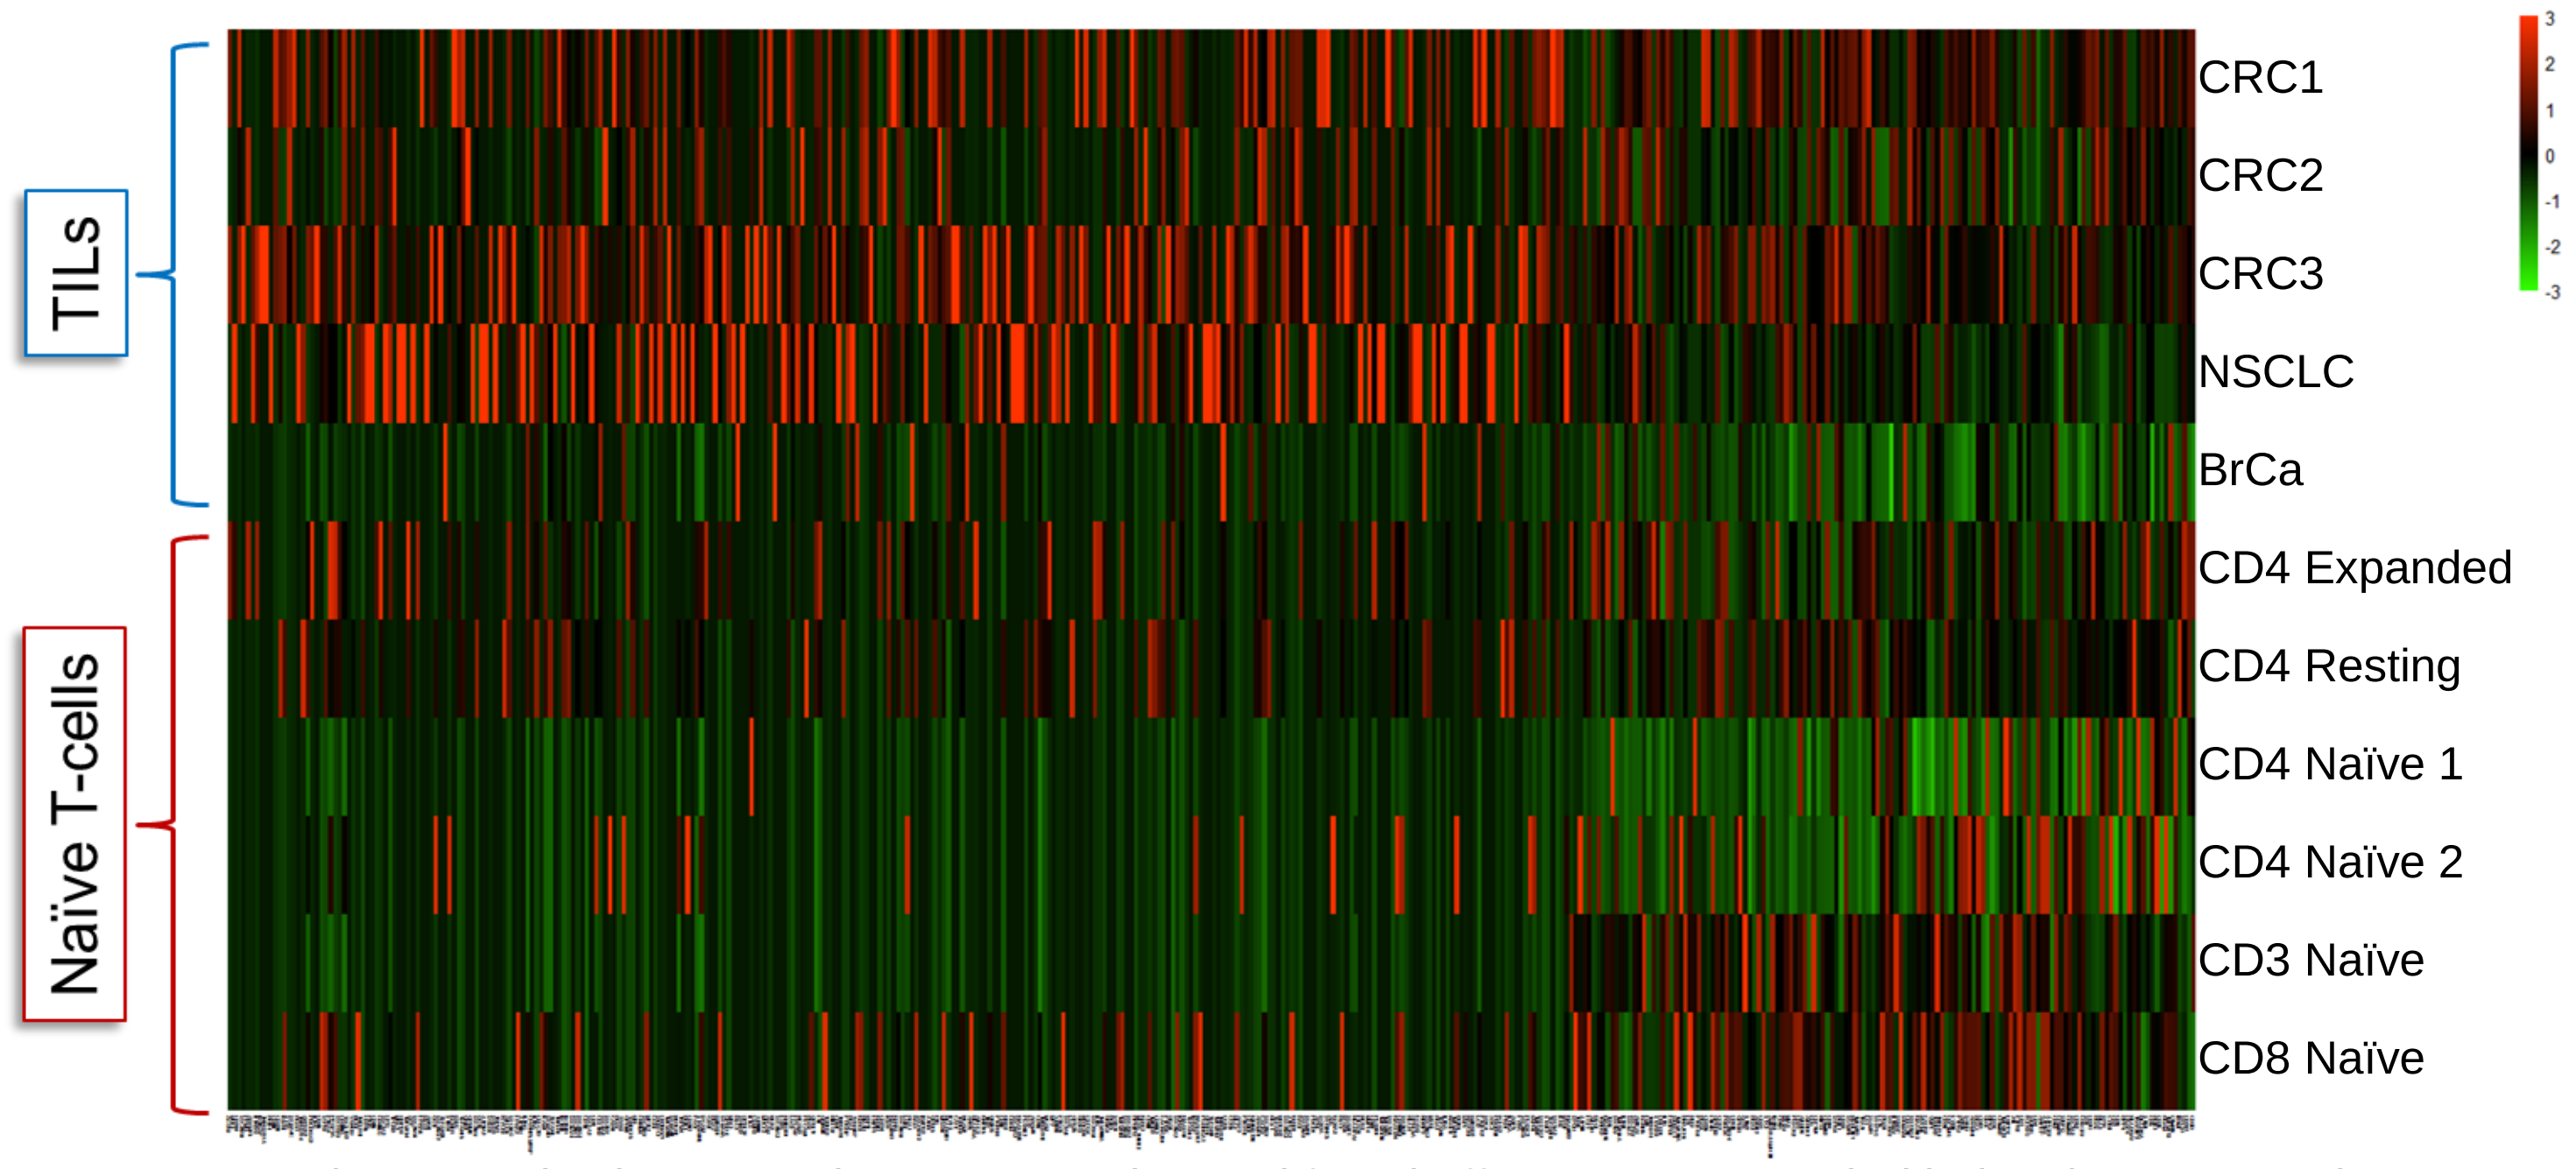
TILs
Naïve T-cells
CRC1
CRC2
CRC3
NSCLC
BrCa
CD4 Expanded
CD4 Resting
CD4 Naïve 1
CD4 Naïve 2
CD3 Naïve
CD8 Naïve

Figure 3. Proteomic expression in TILs and naïve T cells isolated from buffy coat. Generated with the pheatmap package available in R (Version 3.2.0) using a binary distance matrix and Ward scoring methods. TILs display unique proteomic signature as compared to naïve T cells allowing for discovery of unique immuno-oncology targets and biomarkers (Kast. J *et al.* (2013) ASMS 61st meeting Poster session MP25: 497).

ii. FACS Expression of OXBT185 in TILs

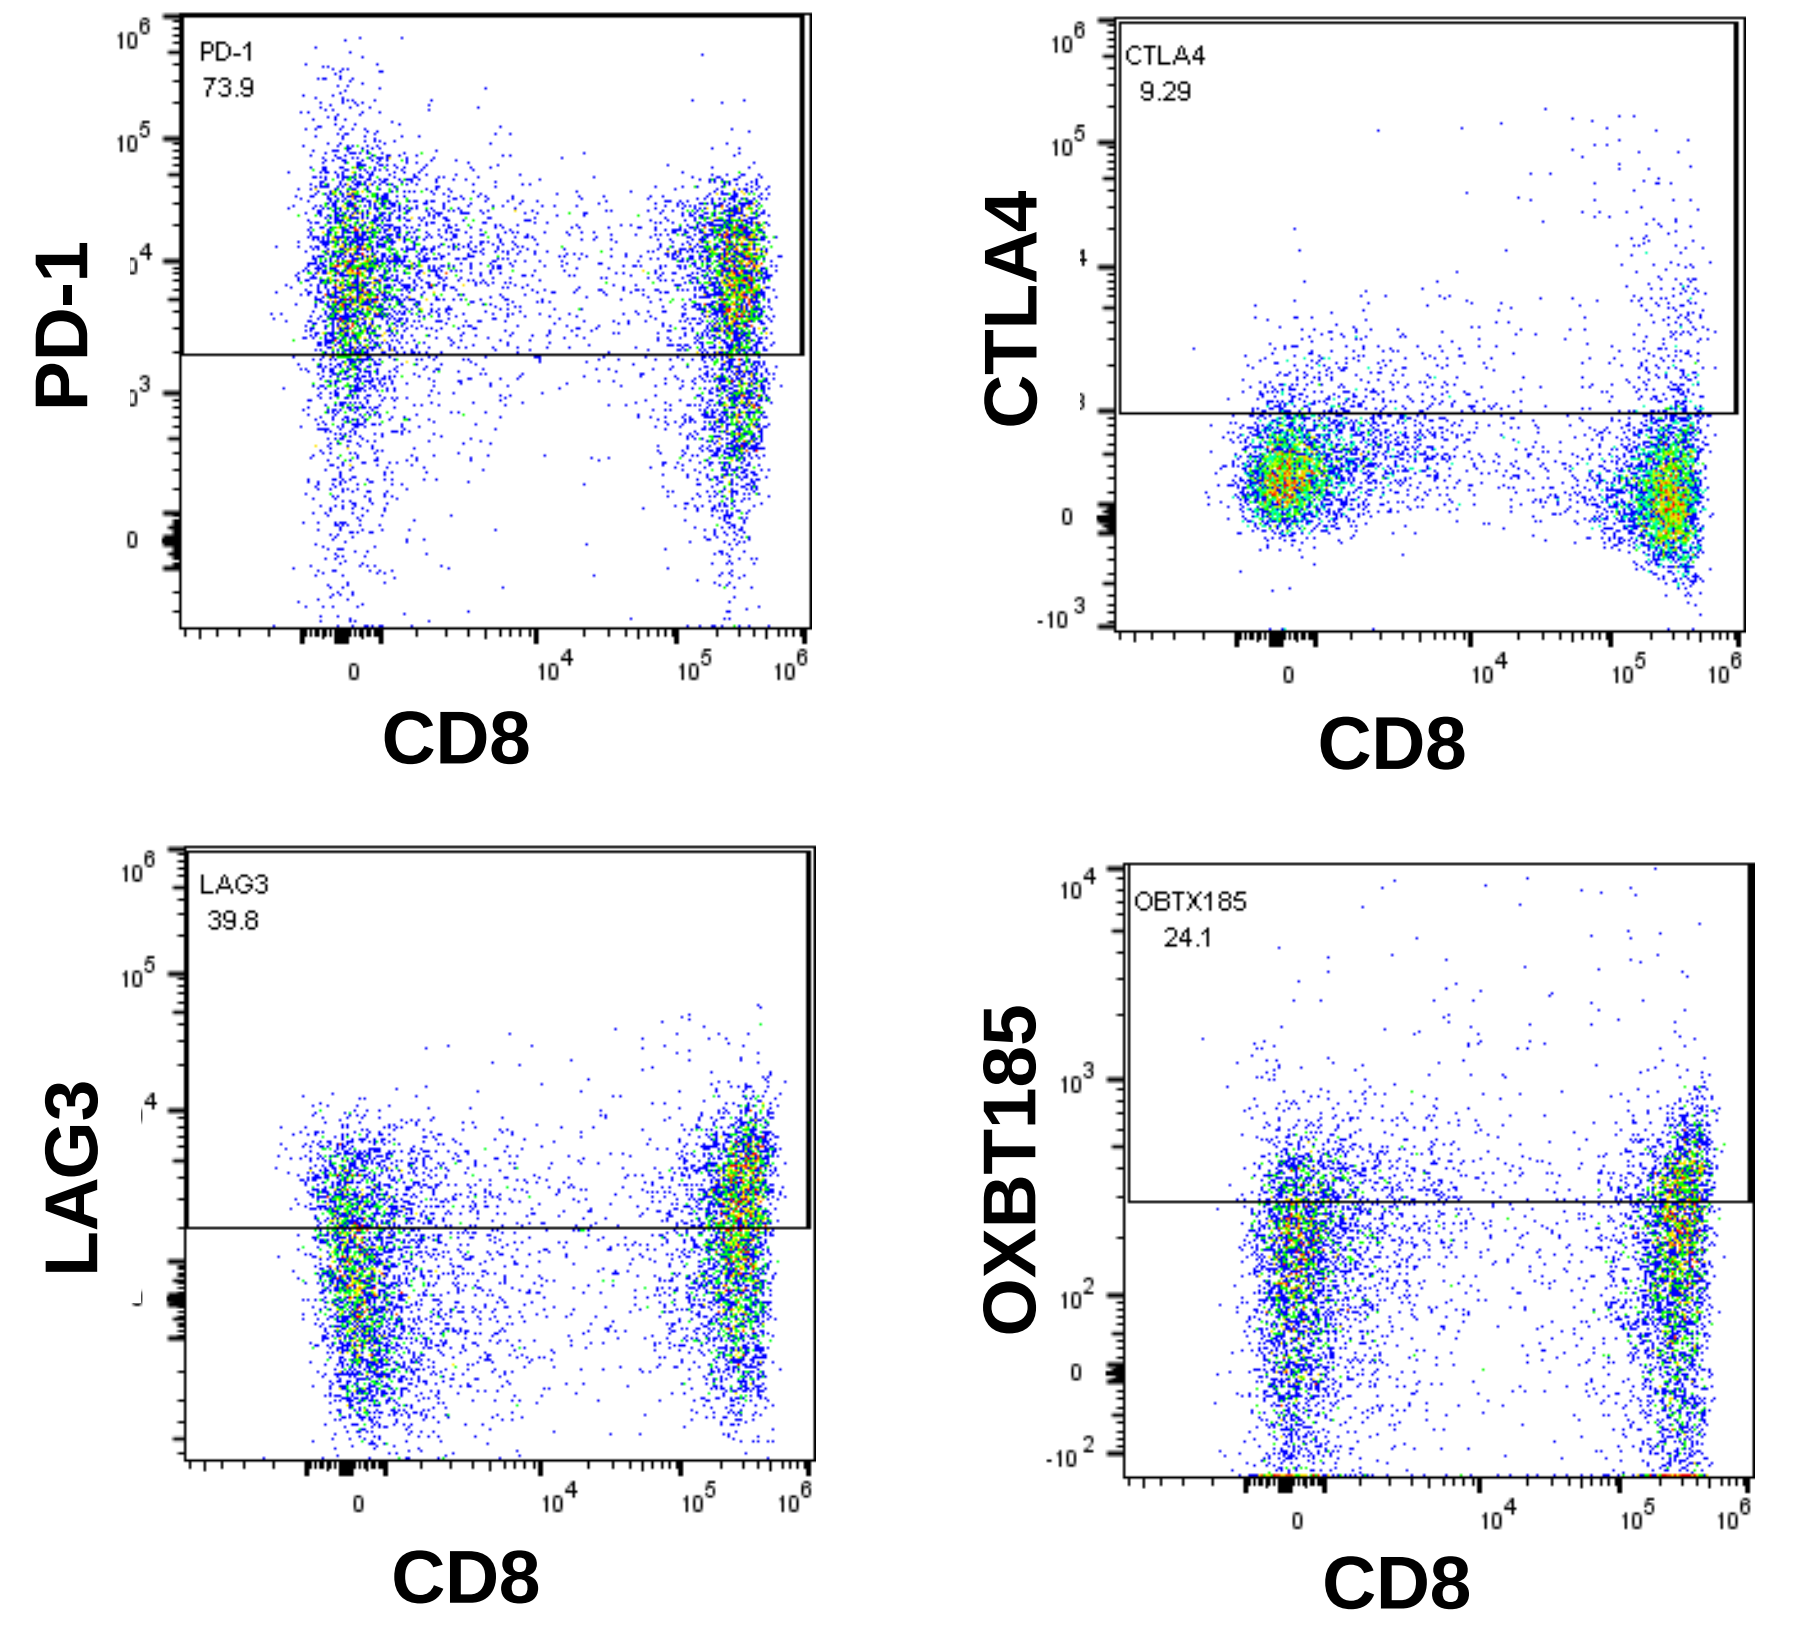
PD-1
73.9
CTLA4
9.29
LAG3
39.8
OBTX185
24.1
PD-1
CTLA4
LAG3
OXBT185
CD8

Figure 4. Protein expression of known IO targets and OXBT185 on TILs from NSCLC. Using a mouse anti-OXBT185 for FACS analysis, expression of OXBT185 was seen on TILs isolated from NSCLC.

iii. IHC Expression of OXBT185 in TILs

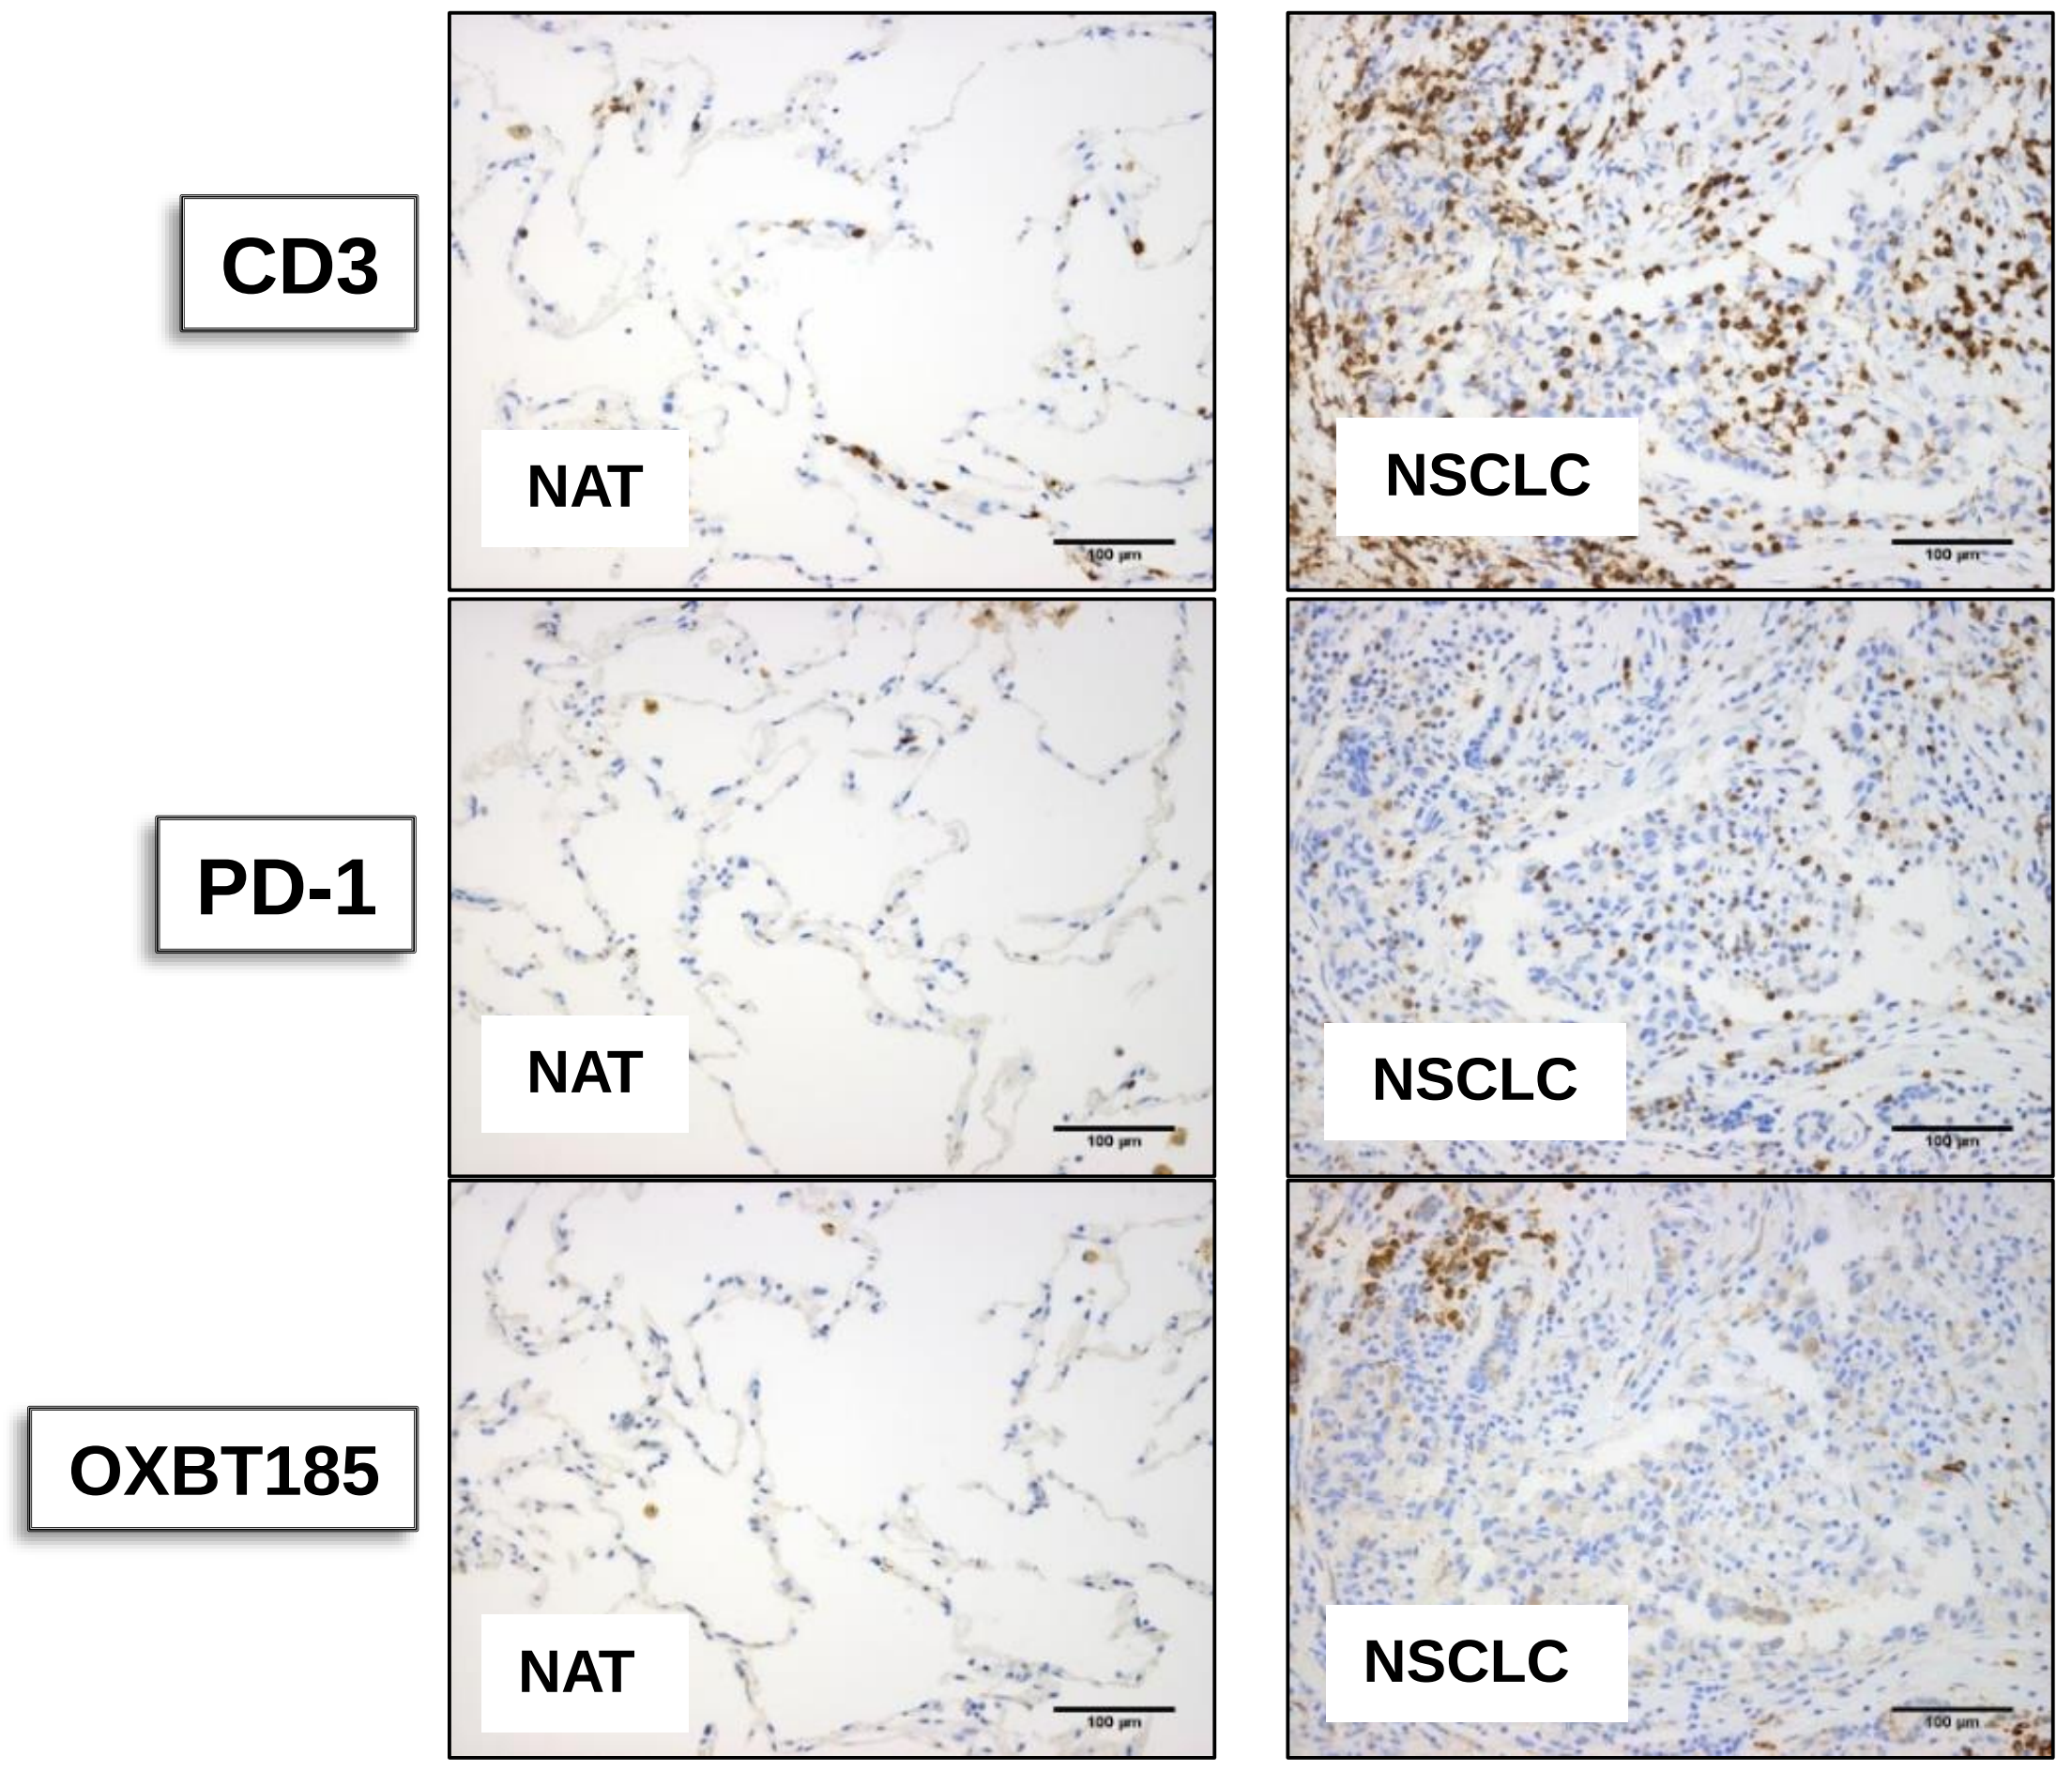
CD3
PD-1
OXBT185
NAT
NSCLC

Figure 5. IHC analysis of matched tumor and normal adjacent tissue. Expression of OXBT185 was confirmed by positive membrane staining in the immune infiltrate of NSCLC. While staining was observed in the NAT it was of weaker intensity.

iv. Functional anti-OXBT185 activates T cells

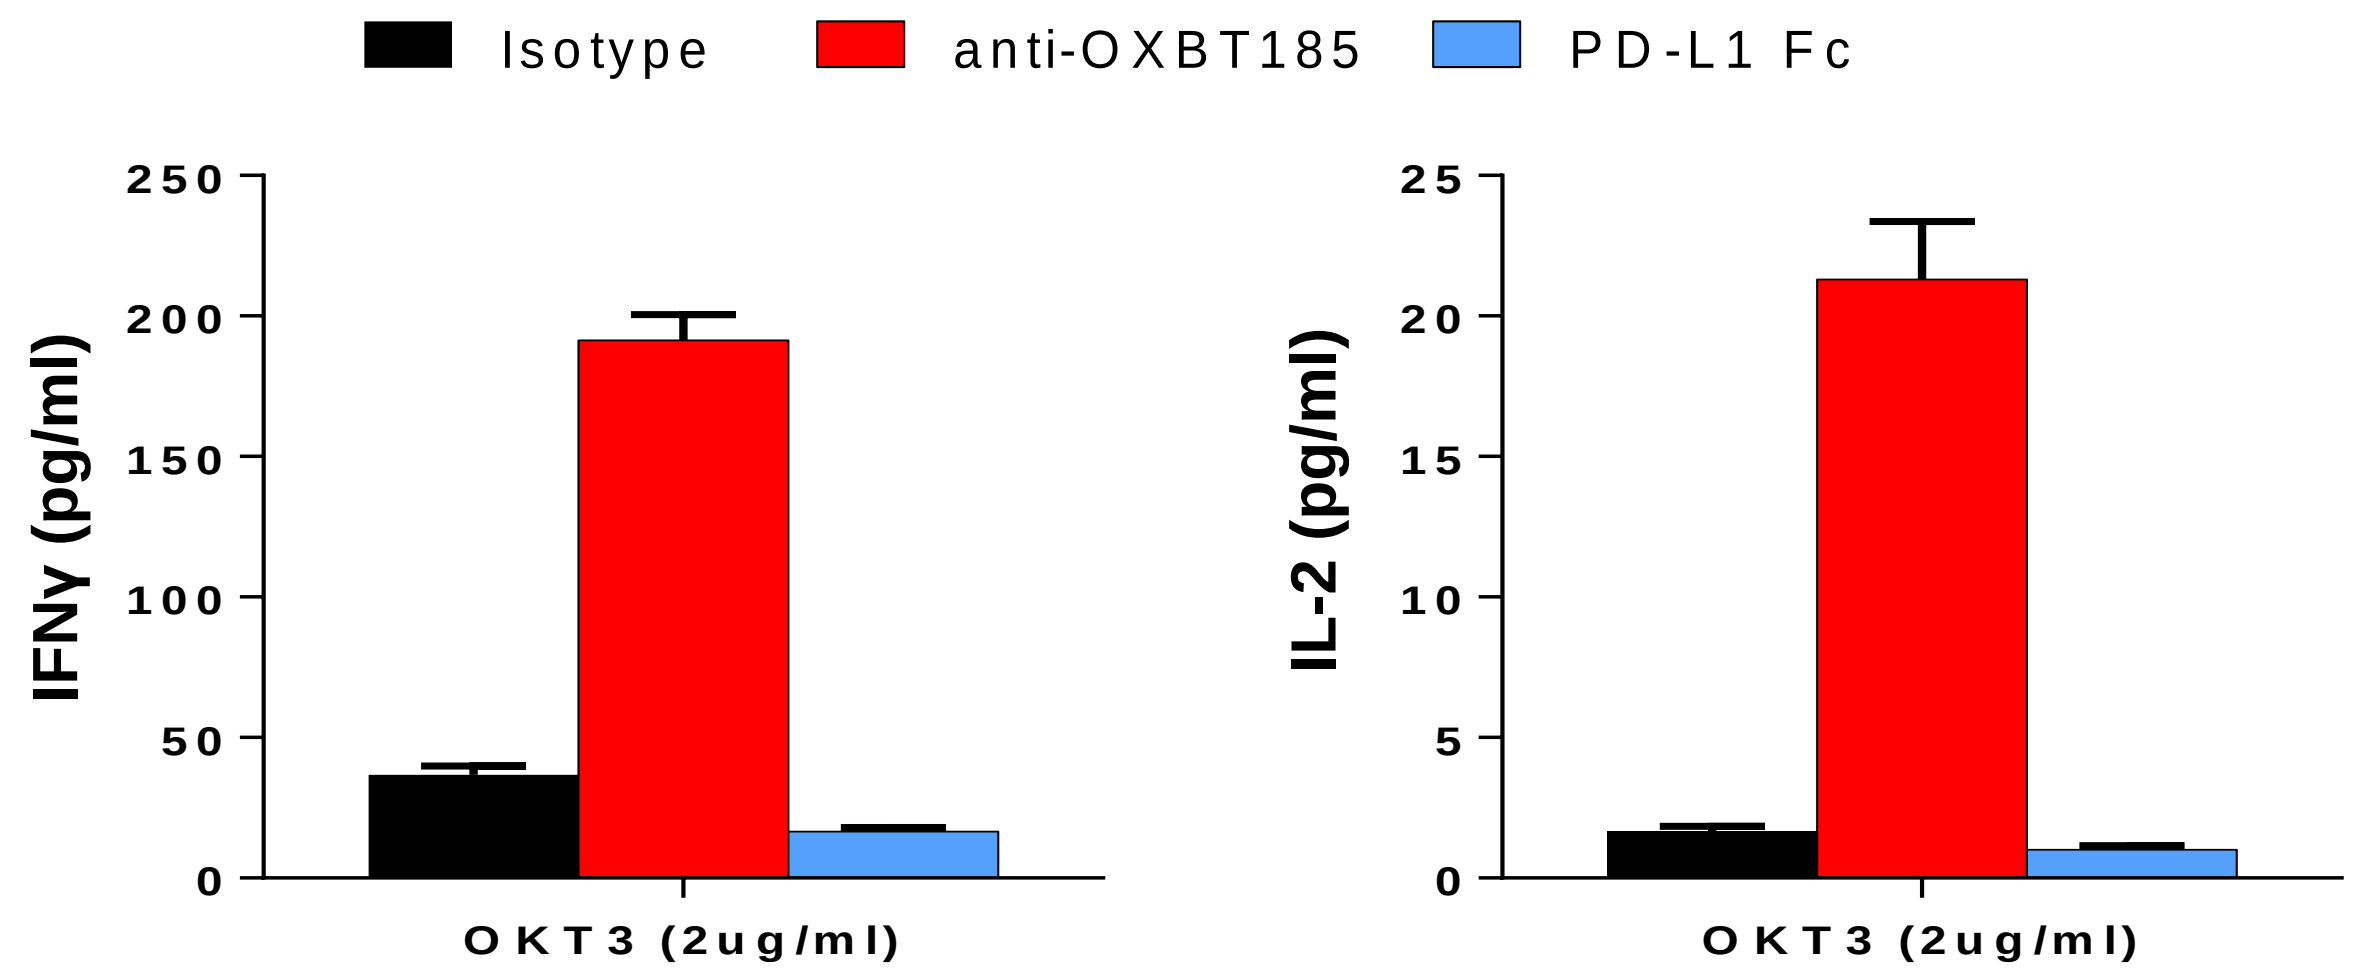
Isotype
anti-OXBT185
PD-L1 Fc
IFNγ (pg/ml)
IL-2 (pg/ml)
OKT3 (2ug/ml)

Figure 6. *In vitro* cytokine release using PBMCs primed with OKT3. IFNγ and IL-2 were measured using ELISA kits available from ebioscience. As expected PD-L1-Fc (R&D systems) inhibited cytokine release, while anti-OXBT185 stimulated production of IFNγ and IL-2.

v. Functional anti-OXBT185 activates TILs in tumor explant

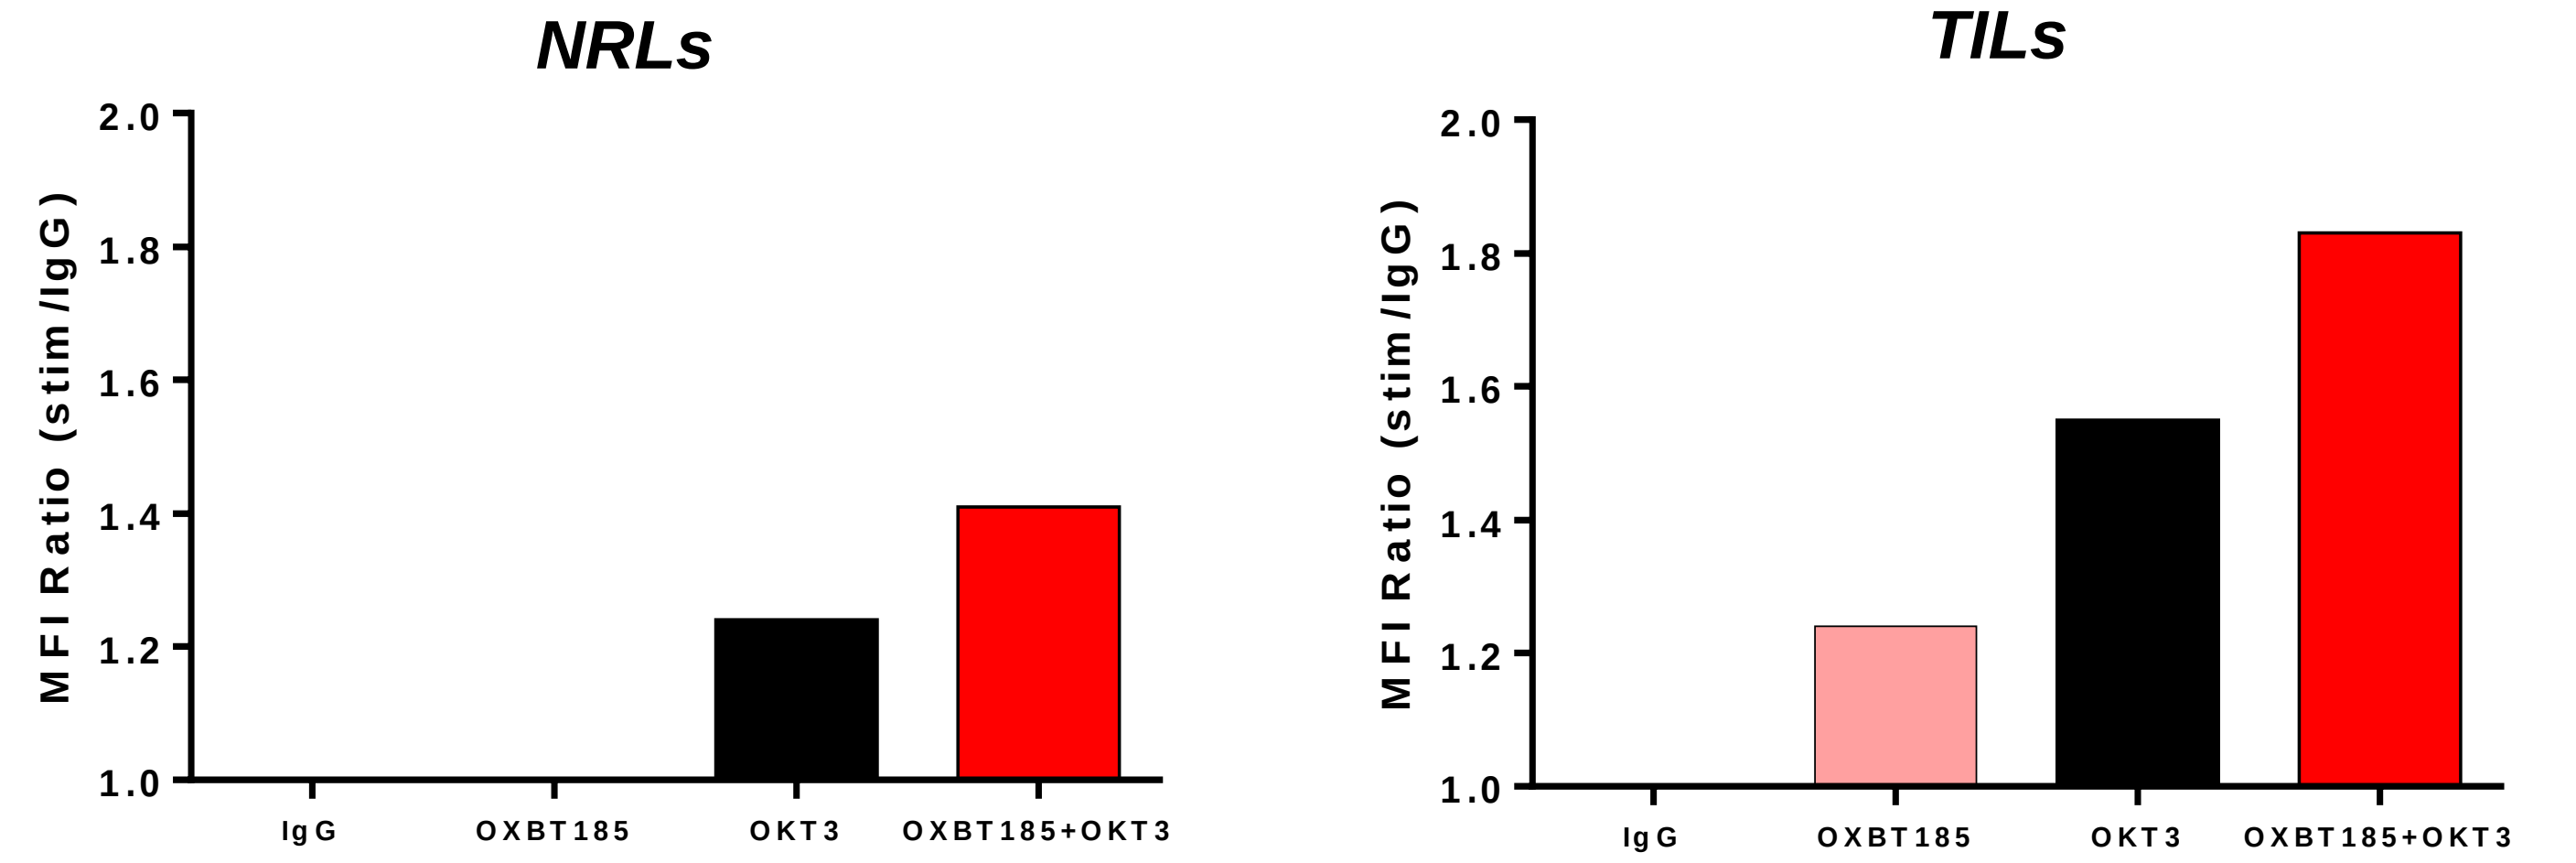
NRLs
TILs
MFI Ratio (stim/IgG)
IgG
OXBT185
OKT3
OXBT185+OKT3

Figure 7. *Ex vivo* cytokine release using NRLs (*A*) and TILS (*B*) primed with OKT3. Ratio against IgG isotype control indicated more IFNγ producing cells from TILs when stimulated with anti-OXBT185. Anti-OXBT185 stimulation induced IFNγ production in TILs but not in NRLs. Moreover, anti-OXBT185 synergised with low-level CD3 stimulation to boost IFNγ production.

Conclusions

- 3D culture of TILs maintains their tumor phenotype.
- Proteomic analysis of these TILS reveals an unique protein signature compared to naïve T cells allowing identification of IO targets and biomarkers.
- Agonist anti-OXBT185 antibody activates peripheral T cells and TILS from 3D tumor explant culture.

OBT's unique discovery path allows rapid identification and validation of IO targets.