ART5803, A First-in-Class Precision Medicine to Treat Anti-NMDA Receptor Encephalitis: First-in-Human Phase 1 Clinical Safety and Pharmacokinetic Profile

Mickey Matsumoto[1], Mari Maurer[1], April Purcell[1], Amanda Singh[2], Sanela Bilic[2], Martin Jefson[1], Peter Flynn[1]

[1]Arialys Therapeutics, Inc. [2]Vanadro, LLC

BACKGROUND

- Anti-NMDA receptor encephalitis (ANRE) is a rare, potentially lethal, poorly managed, and often misdiagnosed neurological disease caused by pathogenic autoantibodies that bind to and crosslink NMDARs in the brain leading to receptor internalization and synaptic dysfunction.
- ANRE results in a range of debilitating neuropsychiatric symptoms including psychiatric and behavioral alterations, cognitive decline, seizures, coma, and diminished autonomic function. A significant proportion of patients require admittance to intensive care units followed by longer-term hospitalization.
- There are no approved therapies for this disease. Current Standard of Care (SoC) is the off-label use of anti-inflammatory and immunosuppressive therapies. First-line therapy for ANRE typically includes corticosteroids, IVIG, and potentially PLEX. Non-responders/inadequate responders are then treated with rituximab, cyclophosphamide, and other therapies.
- Historical disease assessments demonstrate that the use of anti-inflammatory and immunosuppressive therapies results in a lowering of autoantibody titer and gradual improvement in symptoms in the majority of patients. However, recovery is often protracted with persistent cognitive, behavioral, and neurological symptoms.
- There is a significant and growing body of evidence to suggest that the speed at which a patient responds to therapy and is relieved of the autoantibody pathogenic activity, correlates with their longer-term positive outcomes.
- Arialys has developed ART5803 as a therapeutic with the potential to rapidly treat the symptoms of ANRE and other anti-NMDAR autoantibody–associated psychiatric diseases.
- ART5803 is a novel humanized monovalent (one-armed) IgG1 (silent Fc) antibody that binds to the amino (N)-terminal extracellular domain of the NMDAR subunit 1 (NR1) blocking the ability of the pathogenic anti-NMDAR autoantibodies to bind to and crosslink the receptors.
- Nonclinical studies have demonstrated that ART5803 can penetrate the blood-brain barrier, bind to NMDARs, and exert a central nervous system (CNS) effect. In a primate model system, abnormal behavioral, motor, and cognitive symptoms were induced via the direct and constant administration of high-dose human pathogenic autoantibodies. ART5803 bolus systemic administration rapidly resolved these symptoms, with efficacy seen within days.
- ART5803 may offer a novel treatment for patients with ANRE, resulting in a swifter, and more comprehensive recovery than with SoC immunosuppressive therapy alone.

METHODS

Study 101:

- A Phase 1, single-center, randomized, double-blind, placebo-controlled, two-part, SAD and MAD, study conducted to assess the safety, tolerability, PK, and immunogenicity of ART5803 in adult HVs.
- ART5803 was administered IV as a single dose across dose levels ranging from 3 to 100 mg/kg and as multiple doses at 30 mg/kg, 60 mg/kg and 100 mg/kg Q1W for 4 weeks. 6 HVs received ART5803 and 2 received placebo per dose level in both SAD and MAD phases of the study.
- The PK profile of ART5803 in serum and CSF was measured with the goal of assessing blood-brain penetrance.
- Immunogenicity was evaluated for persistence of ADA and their potential impact on the PK profile.

Study 102:

- A Phase 1, single-center, open-label study conducted to assess the safety, tolerability, and PK of ART5803 following prior IVIG administration in adult HVs.
- Participants received two consecutive daily doses of IVIG (total dose of 1 g/kg) followed by a single IV dose of ART5803 (60 mg/kg) administered 6 days after the final IVIG dose.
- The endpoints and assessments were the same as for Study 101.
- Post study comparison of the PK profile from the Study 101 60 mg/kg SAD dose level and the Study 102 60mg/kg dose was made for both serum and CSF.

RESULTS

Safety:

- ART5803 was generally well tolerated at doses up to 60 mg/kg. In the MAD cohorts, the incidence of TEAEs generally increased with ascending dose through Cohort C (100 mg/kg), without evidence of a dose-limiting safety signal.
- Four SAEs occurred in Study 101, including one death. All SAEs were assessed by both the study Primary Investigator (PI) and the Safety Review Committee (SRC) as unrelated to ART5803.
- Adverse events of special interest (AESIs) for the studies were either CNS-related or Immune-related AEs. Five CNS-related AESIs (across three participants) were reported in the 100 mg/kg dose level of the MAD phase of the study and within the ART5803-treated participants. No AESIs were observed in the 30 mg/kg or 60 mg/kg SAD or MAD dose levels nor in the SAD 100 mg/kg dose level.

Pharmacology:

- Both serum and CSF concentrations of ART5803 increased in a dose-proportional manner.
- $T_{1/2}$ was estimated to be up to 350 hours (after the last dose at 100 mg/kg, MAD).
- Significant accumulation (>2-fold) was observed in the CSF following repeat doses.
- Potentially efficacious levels of ART5803 seen in CSF at 30 mg/kg and above.
- No significant difference was seen in the PK profile of ART5803 in both serum and CSF on comparison of the Study 101 60 mg/kg SAD dose level and the Study 102 60mg/kg dose, suggesting that ART5803 can be administered post IVIG without the requirement for dose adjustment.
- Immunogenicity: 7 participants (14.6%) in study 101 were confirmed ADA positive, but this was not associated with clinically meaningful changes in the PK profile of ART5803. None of the Study 102 participants were ADA positive.

CONCLUSIONS

- These Phase 1 data support the continued development of ART5803 for the treatment of ANRE and other anti-NMDAR autoantibody associated disorders.
- Assessment of ART5803 therapeutic efficacy is planned and currently comprises two Phase 2 studies: an open label signal seeking study (Korea) and a randomized, placebo-controlled study (USA, international)

ANRE and ART5803

ANRE Disease Progression

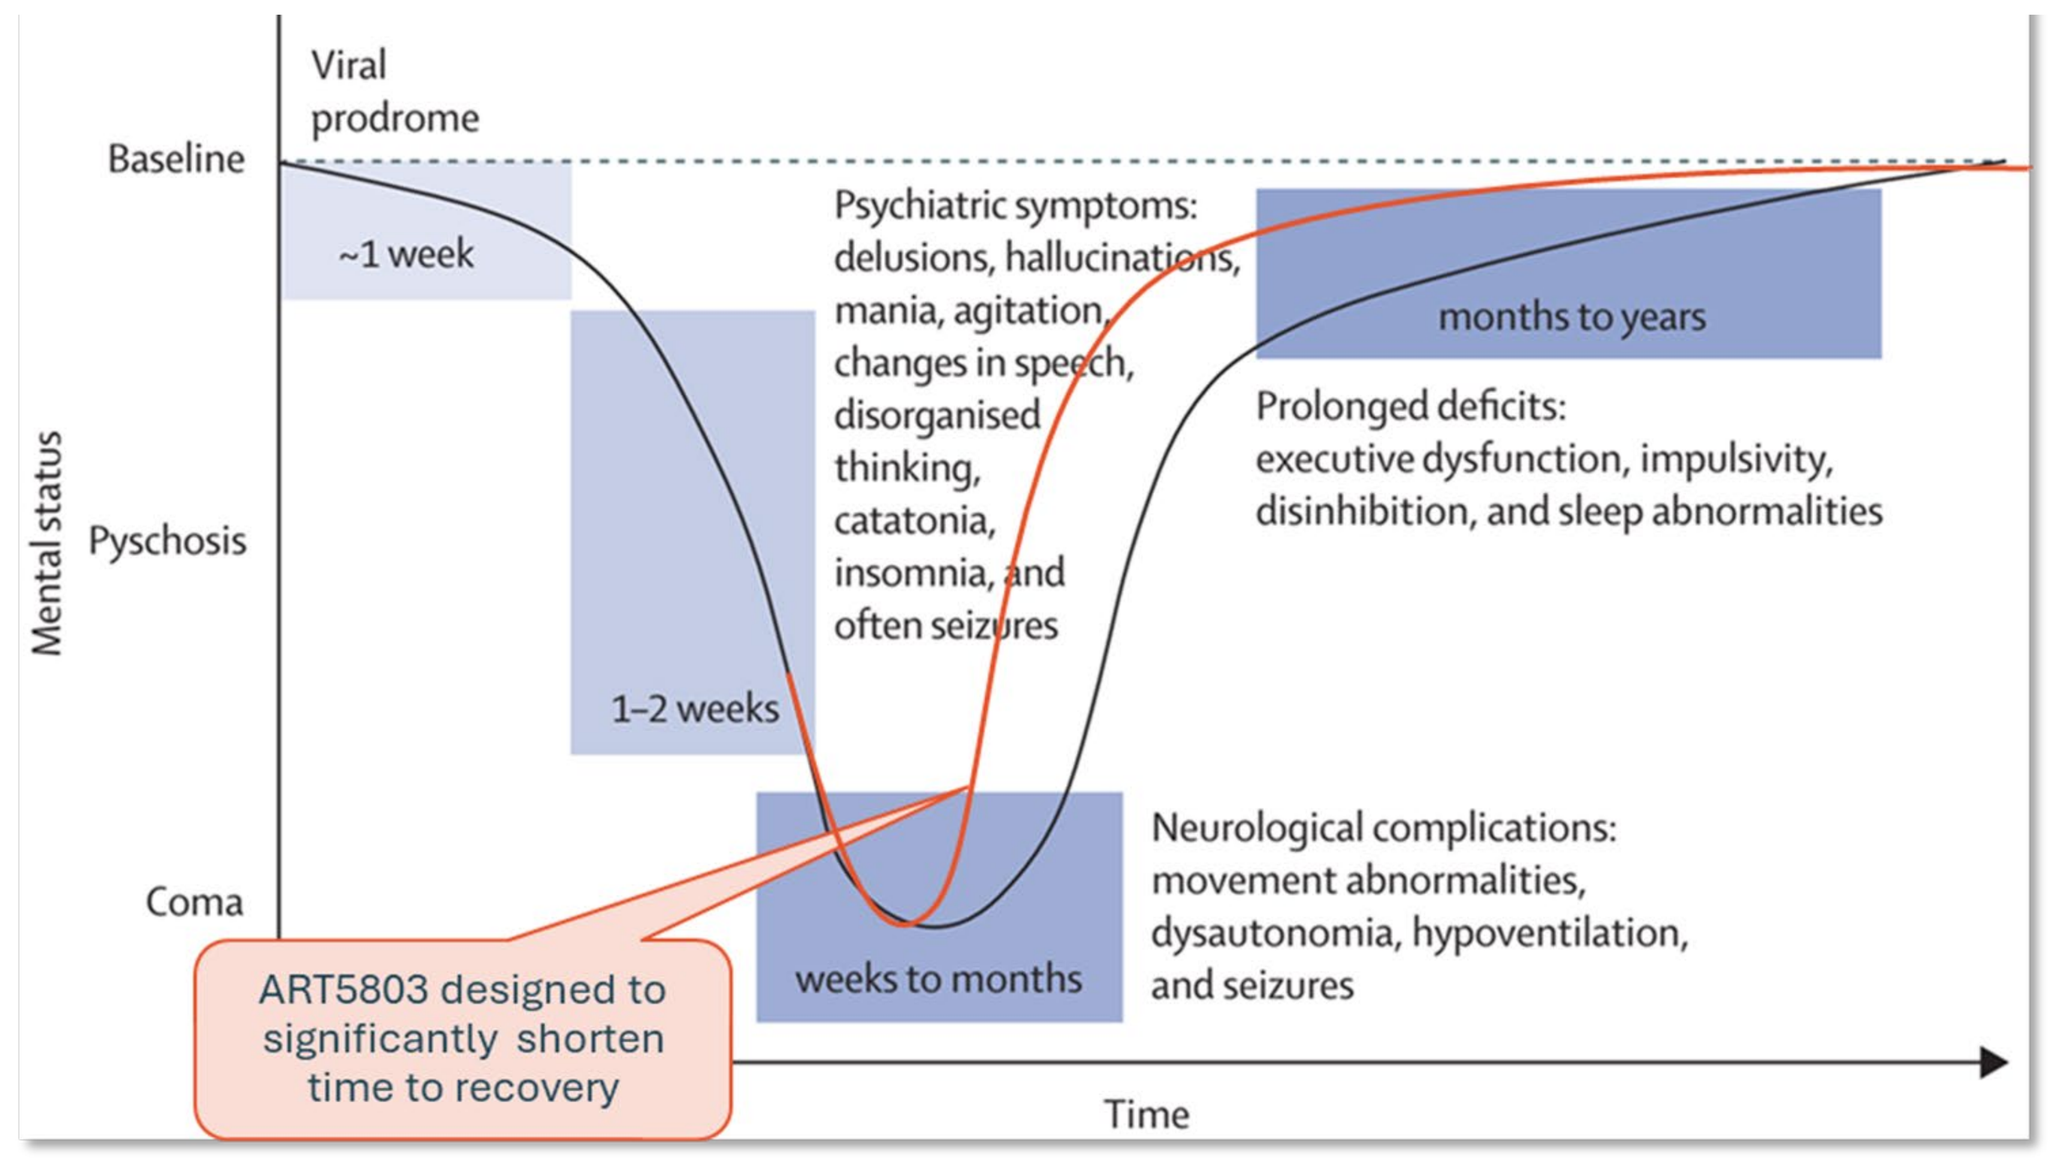
Viral prodrome
Baseline
Psychosis
Coma
Mental status
~1 week
1–2 weeks
weeks to months
months to years
Psychiatric symptoms: delusions, hallucinations, mania, agitation, changes in speech, disorganised thinking, catatonia, insomnia, and often seizures
Prolonged deficits: executive dysfunction, impulsivity, disinhibition, and sleep abnormalities
Neurological complications: movement abnormalities, dysautonomia, hypoventilation, and seizures
ART5803 designed to significantly shorten time to recovery
Time

Adapted from Graus & Dalmau (2019)

ART5803 Mode of Action

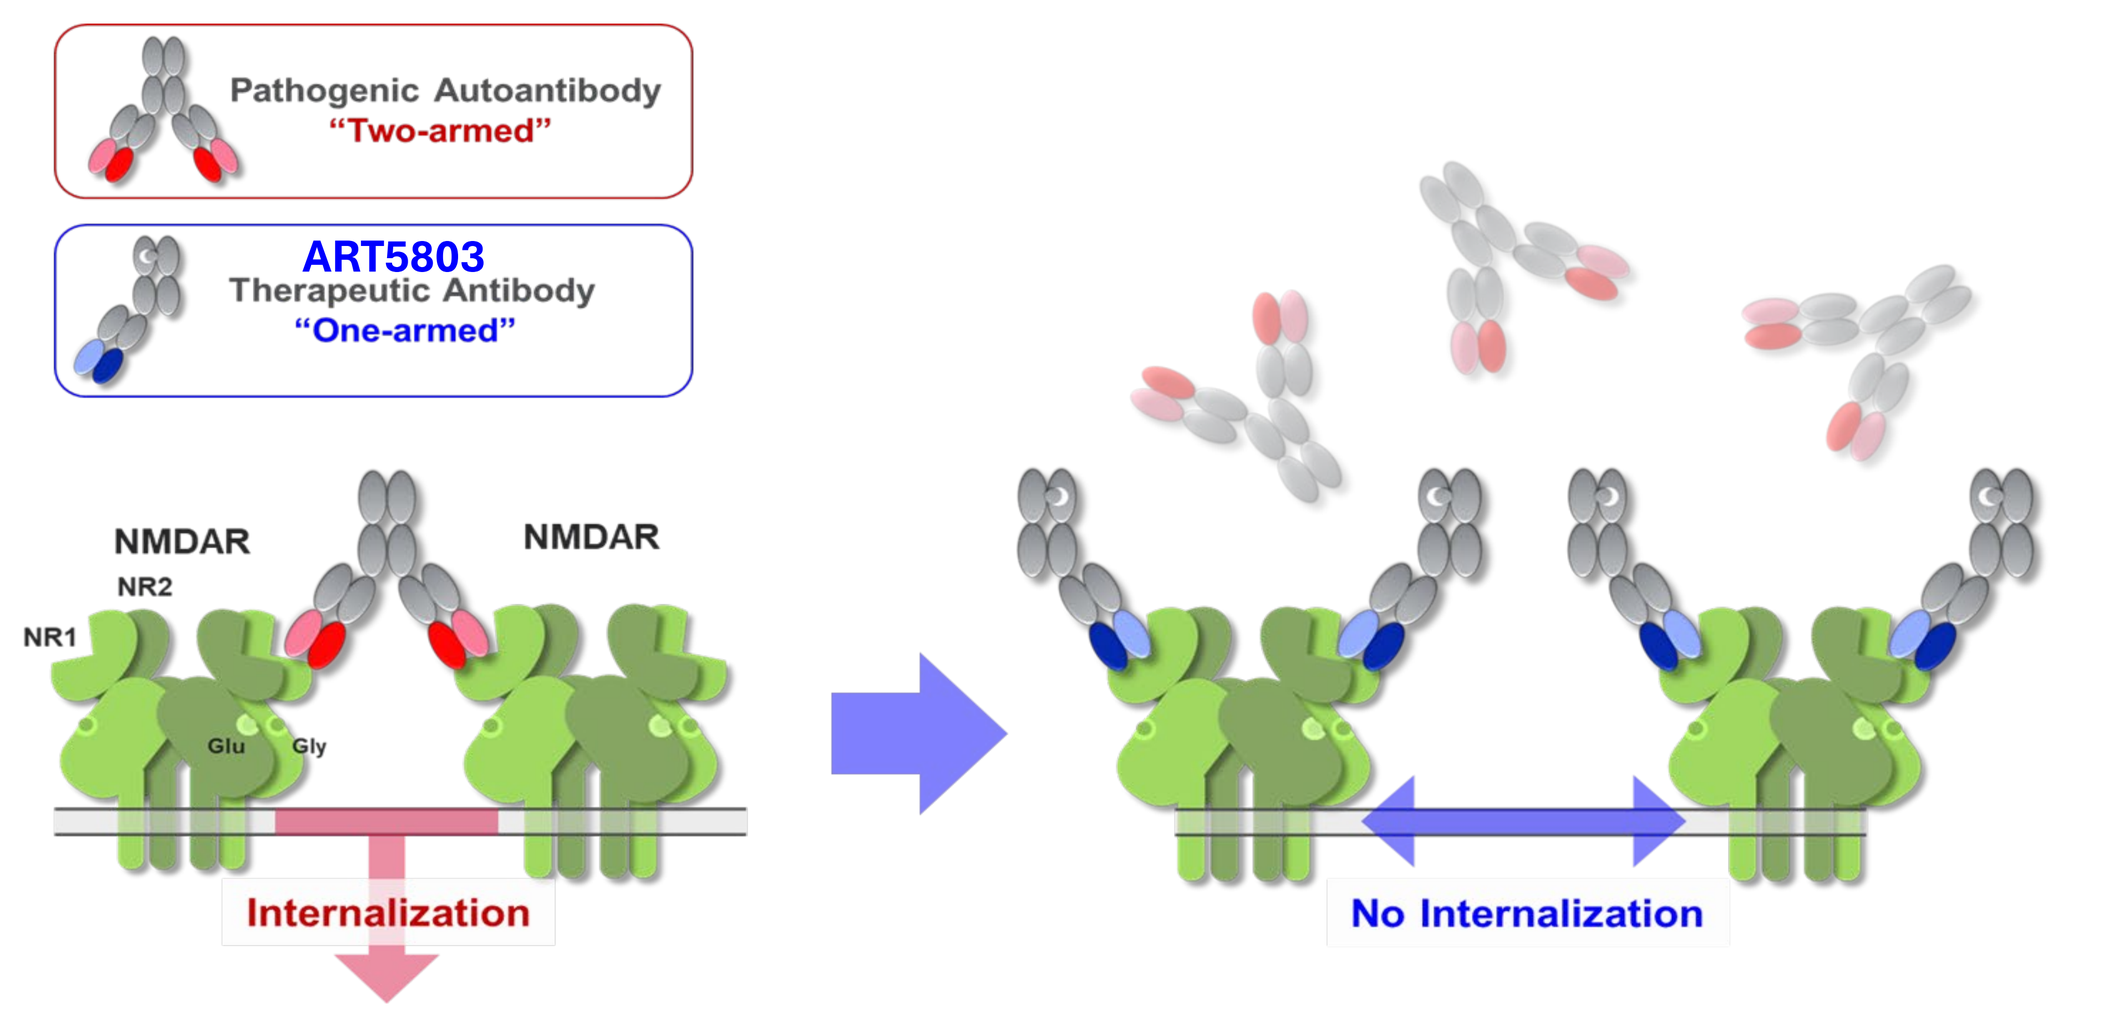
Pathogenic Autoantibody
"Two-armed"
ART5803
Therapeutic Antibody
"One-armed"
NMDAR
NR2
NR1
Glu
Gly
NMDAR
Internalization
No Internalization

ART5803 Preclinical Data

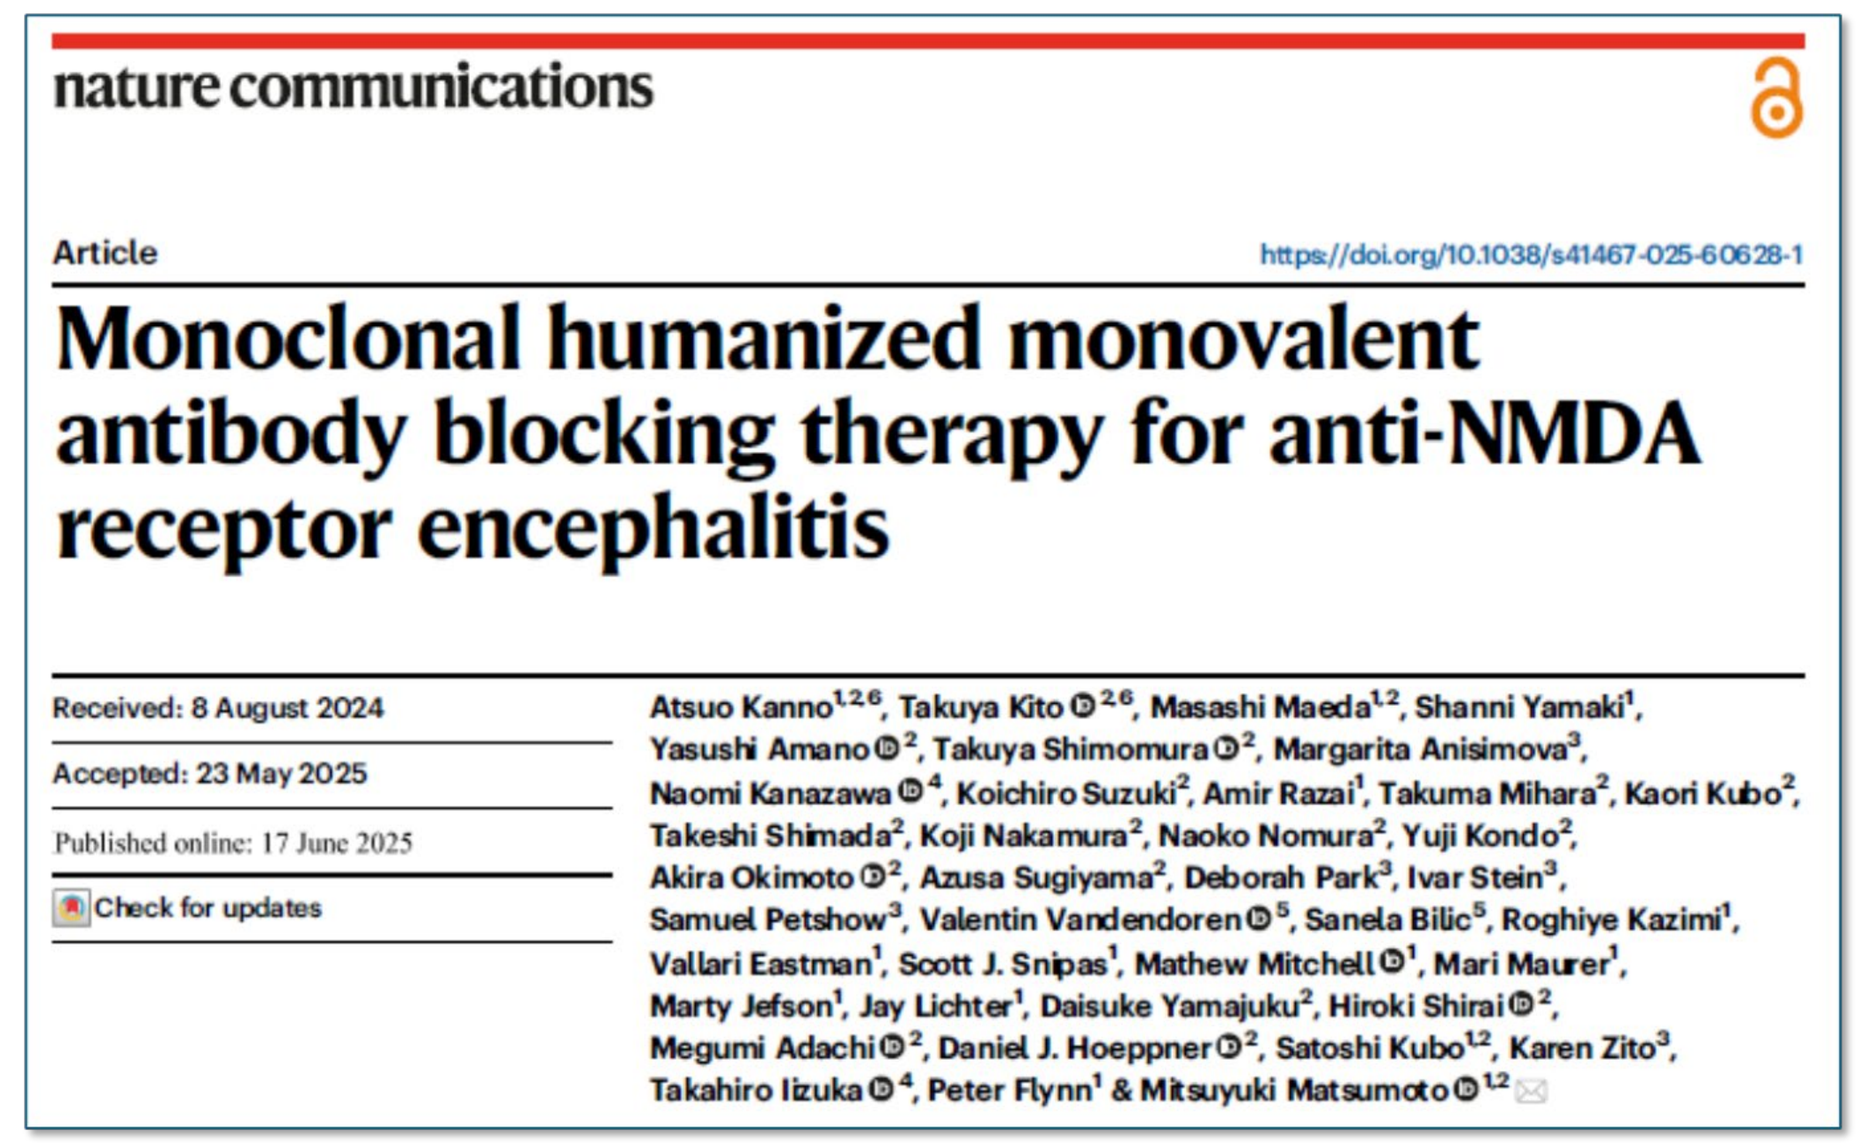
nature communications

Article

https://doi.org/10.1038/s41467-025-60628-1

Monoclonal humanized monovalent antibody blocking therapy for anti-NMDA receptor encephalitis

Received: 8 August 2024

Accepted: 23 May 2025

Published online: 17 June 2025

Check for updates

Atsuo Kanno[1,2,6], Takuya Kito[2,6], Masashi Maeda[1,2], Shanni Yamaki[1], Yasushi Amano[2], Takuya Shimomura[2], Margarita Anisimova[3], Naomi Kanazawa[4], Koichiro Suzuki[2], Amir Razai[1], Takuma Mihara[2], Kaori Kubo[2], Takeshi Shimada[2], Koji Nakamura[2], Naoko Nomura[2], Yuji Kondo[2], Akira Okimoto[2], Azusa Sugiyama[2], Deborah Park[3], Ivar Stein[3], Samuel Petshow[3], Valentin Vandendoren[5], Sanela Bilic[5], Roghiye Kazimi[1], Vallari Eastman[1], Scott J. Snipas[1], Mathew Mitchell[1], Mari Maurer[1], Marty Jefson[1], Jay Lichter[1], Daisuke Yamajuku[2], Hiroki Shirai[2], Megumi Adachi[2], Daniel J. Hoeppner[2], Satoshi Kubo[1,2], Karen Zito[3], Takahiro Iizuka[4], Peter Flynn[1] & Mitsuyuki Matsumoto[1,2]

Study ART5803-101 Schematic

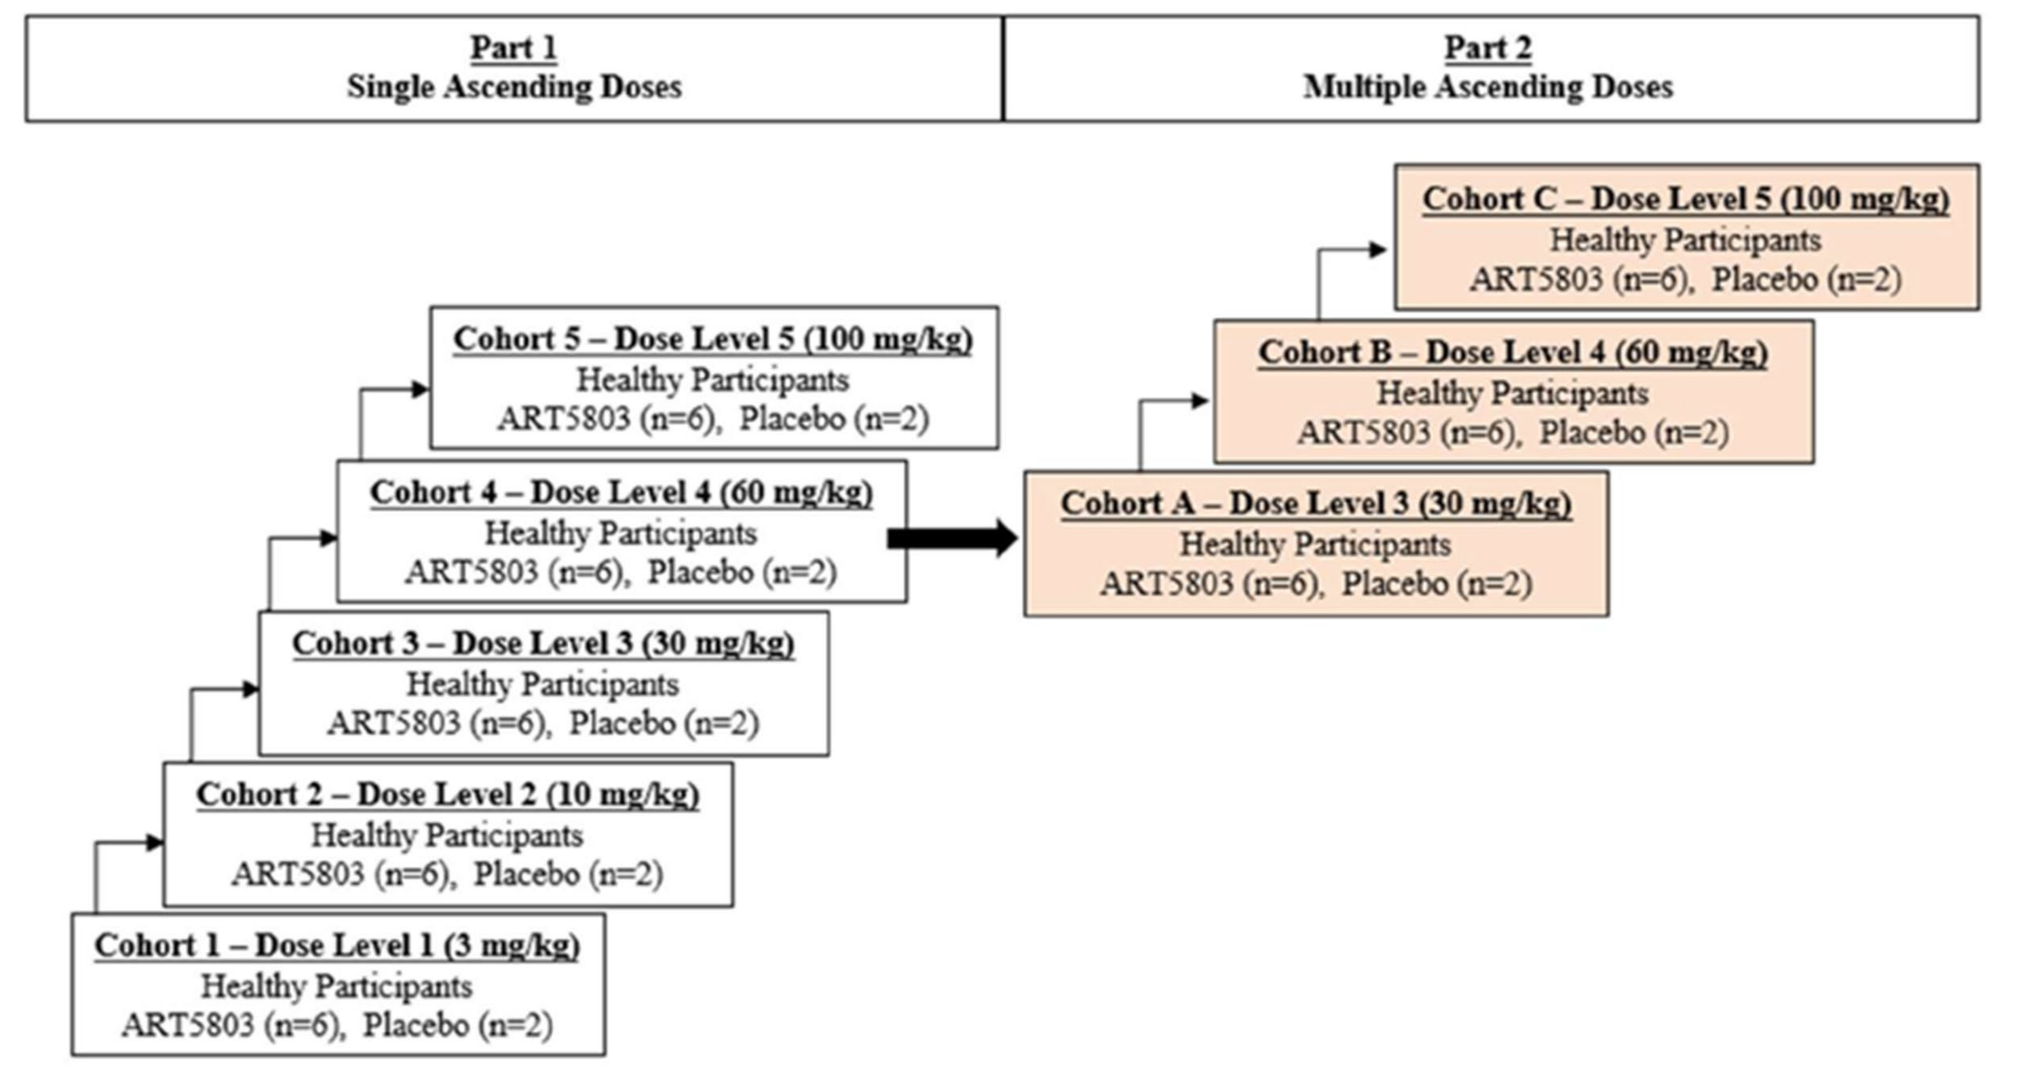
Part 1
Single Ascending Doses
Part 2
Multiple Ascending Doses
Cohort 1 – Dose Level 1 (3 mg/kg)
Healthy Participants
ART5803 (n=6), Placebo (n=2)
Cohort 2 – Dose Level 2 (10 mg/kg)
Healthy Participants
ART5803 (n=6), Placebo (n=2)
Cohort 3 – Dose Level 3 (30 mg/kg)
Healthy Participants
ART5803 (n=6), Placebo (n=2)
Cohort 4 – Dose Level 4 (60 mg/kg)
Healthy Participants
ART5803 (n=6), Placebo (n=2)
Cohort 5 – Dose Level 5 (100 mg/kg)
Healthy Participants
ART5803 (n=6), Placebo (n=2)
Cohort A – Dose Level 3 (30 mg/kg)
Healthy Participants
ART5803 (n=6), Placebo (n=2)
Cohort B – Dose Level 4 (60 mg/kg)
Healthy Participants
ART5803 (n=6), Placebo (n=2)
Cohort C – Dose Level 5 (100 mg/kg)
Healthy Participants
ART5803 (n=6), Placebo (n=2)

Phase 1 Safety Data

Study ART5803-101 SAD TEAEs

Event	Cohort 1 3 mg/kg (N=6)	Cohort 2 10 mg/kg (N=6)	Cohort 3 30 mg/kg (N=6)	Cohort 4 60 mg/kg (N=6)	Cohort 5 100 mg/kg (N=6)	Placebo (N=10)	Overall (N=40)
	Number (%) of Participants Reporting, Number of Events						
Any AE	4 (66.7), 7	5 (83.3), 13	5 (83.3), 14	4 (66.7), 8	4 (66.7), 9	8 (80.0), 21	30 (75.0), 72
Any TEAE	4 (66.7), 7	5 (83.3), 13	5 (83.3), 12	4 (66.7), 8	4 (66.7), 8	8 (80.0), 21	30 (75.0), 69
Any Treatment-Related TEAE	0	0	2 (33.3), 2	0	1 (16.7), 1	2 (20.0), 2	5 (12.5), 5
Any Serious TEAE	0	0	0	0	0	1 (10.0), 1	1 (2.5), 1
Any Serious Treatment-Related TEAE	0	0	0	0	0	0	0
Any TEAE Leading to Early Withdrawal	0	0	0	0	0	0	0
Any TEAE Leading to Death	0	0	0	0	0	0	0
Dose Limiting Toxicities	0	0	0	0	0	0	0
Adverse Events of Special Interest	0	0	0	0	0	0	0
Central Nervous System - related AEs	0	0	0	0	0	0	0
Immune-mediated AEs	0	0	0	0	0	0	0
Infusion-related AEs	0	0	0	0	0	0	0

Study ART5803-101 MAD TEAEs

Event	Cohort A 30 mg/kg (N=6)	Cohort B 60 mg/kg (N=6)	Cohort C 100 mg/kg (N=6)	Placebo (N=6)	Overall (N=24)
	Number (%) of Participants Reporting, Number of Events				
Any AE	6 (100.0), 13	6 (100.0), 18	6 (100.0), 29	4 (66.7), 15	22 (91.7), 75
Any TEAE	6 (100.0), 13	6 (100.0), 18	6 (100.0), 29	4 (66.7), 14	22 (91.7), 74
Any Treatment-Related TEAE	4 (66.7), 5	5 (83.3), 11	3 (50.0), 10	2 (33.3), 4	14 (58.3), 30
Any Serious TEAE	1 (16.7), 1	0	2 (33.3), 2	0	3 (12.5), 3
Any Serious Treatment-Related TEAE	0	0	0	0	0
Any TEAE Leading to Early Withdrawal	0	2 (33.3), 2	2 (33.3), 3	0	4 (16.7), 5
Any TEAE Leading to Death	1 (16.7), 1	0	0	0	1 (4.2), 1
Dose Limiting Toxicities	0	0	0	0	0
Adverse Events of Special Interest	0	0	3 (50.0), 5	0	3 (12.5), 5
Central Nervous System - related AEs	0	0	3 (50.0), 5	0	3 (12.5), 5
Immune-mediated AEs	0	0	0	0	0
Infusion-related AEs	1 (16.7), 1	1 (16.7), 1	0	2 (33.3), 2	4 (16.7), 4

Study ART5803-101 SAEs

Study Part	Participant Number	Dose (mg/kg)	SOC / PT / VT	Event Description	Relationship to ART5803	Outcome
SAD	201-002	Placebo	Musculoskeletal and connective tissue disorders / Rhabdomyolysis / Rhabdomyolysis	Elevation of creatine kinase / Rhabdomyolysis	Not related	Recovered /Resolved
MAD	201-172	30 mg/kg	Respiratory, thoracic and mediastinal disorders / Asphyxia / Asphyxiation	Asphyxiation	Not related	Fatal
MAD	201-227	100 mg/kg	Psychiatric disorders / Bipolar disorder / Relapsed bipolar affective disorder with psychotic features	Relapsed bipolar affective disorder with psychotic features	Not related	Recovered /Resolved
MAD	201-209	100 mg/kg	Psychiatric disorders / Acute psychosis / Acute psychosis	Acute psychosis	Not related	Recovered /Resolved

Study ART5803-101 AESIs

Study Part	Participant Number	Dose (mg/kg)	SOC / PT / VT	Relationship to ART5803	Severity	Resolution
MAD	201-213	100	Nervous system disorders / Visual perseveration / Palinopsia	Probably related	Moderate / Grade 2	Recovered/Resolved
MAD	201-213	100	Psychiatric disorders / Mental fatigue / Mental fatigue	Possibly related	Mild / Grade 1	Recovered/Resolved
MAD	201-213	100	Nervous system disorders / Ataxia / Mild ataxia	Possibly related	Mild / Grade 1	Recovered/Resolved
MAD	201-237	100	Psychiatric disorders / Mood altered / Mood fluctuation	Possibly related	Mild / Grade 1	Recovered/Resolved
MAD	201-209	100	Psychiatric disorders / Acute psychosis / Acute psychosis	Not related	Severe / Grade 3	Recovered/Resolved

Phase 1 Pharmacology

Study ART5803-101 Pharmacokinetics

Mean (±SD) Concentrations of ART5803 in Serum and CSF Following MAD of 30, 60, and 100 mg/kg (Study 101)

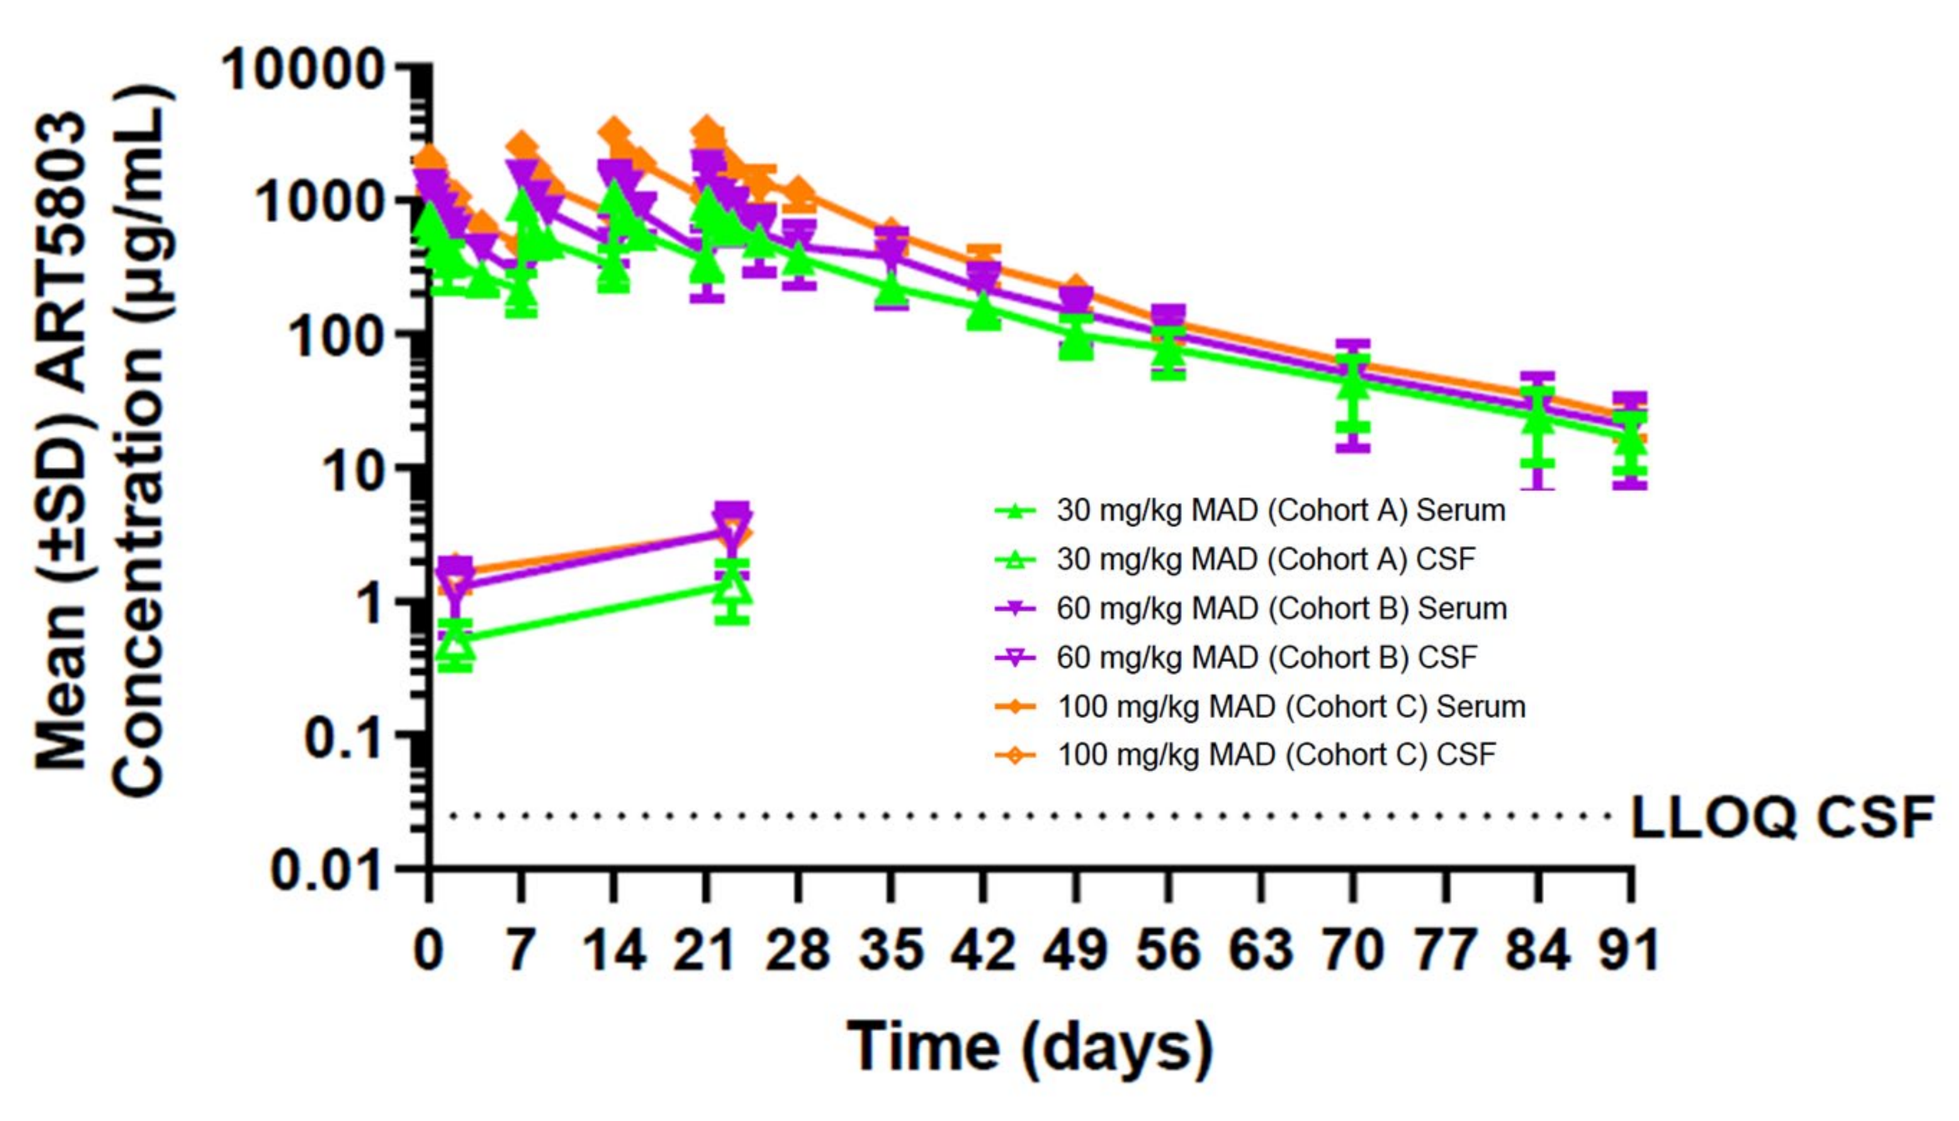
Mean (±SD) ART5803 Concentration (µg/mL)
10000
1000
100
10
1
0.1
0.01
0 7 14 21 28 35 42 49 56 63 70 77 84 91
Time (days)
LLOQ CSF
30 mg/kg MAD (Cohort A) Serum
30 mg/kg MAD (Cohort A) CSF
60 mg/kg MAD (Cohort B) Serum
60 mg/kg MAD (Cohort B) CSF
100 mg/kg MAD (Cohort C) Serum
100 mg/kg MAD (Cohort C) CSF

ART5803 PK in Presence of IVIG

Mean (±SD) Concentrations of ART5803 in Serum and CSF Following a Single Dose of ART5803 60 mg/kg (Studies 101 and 102)

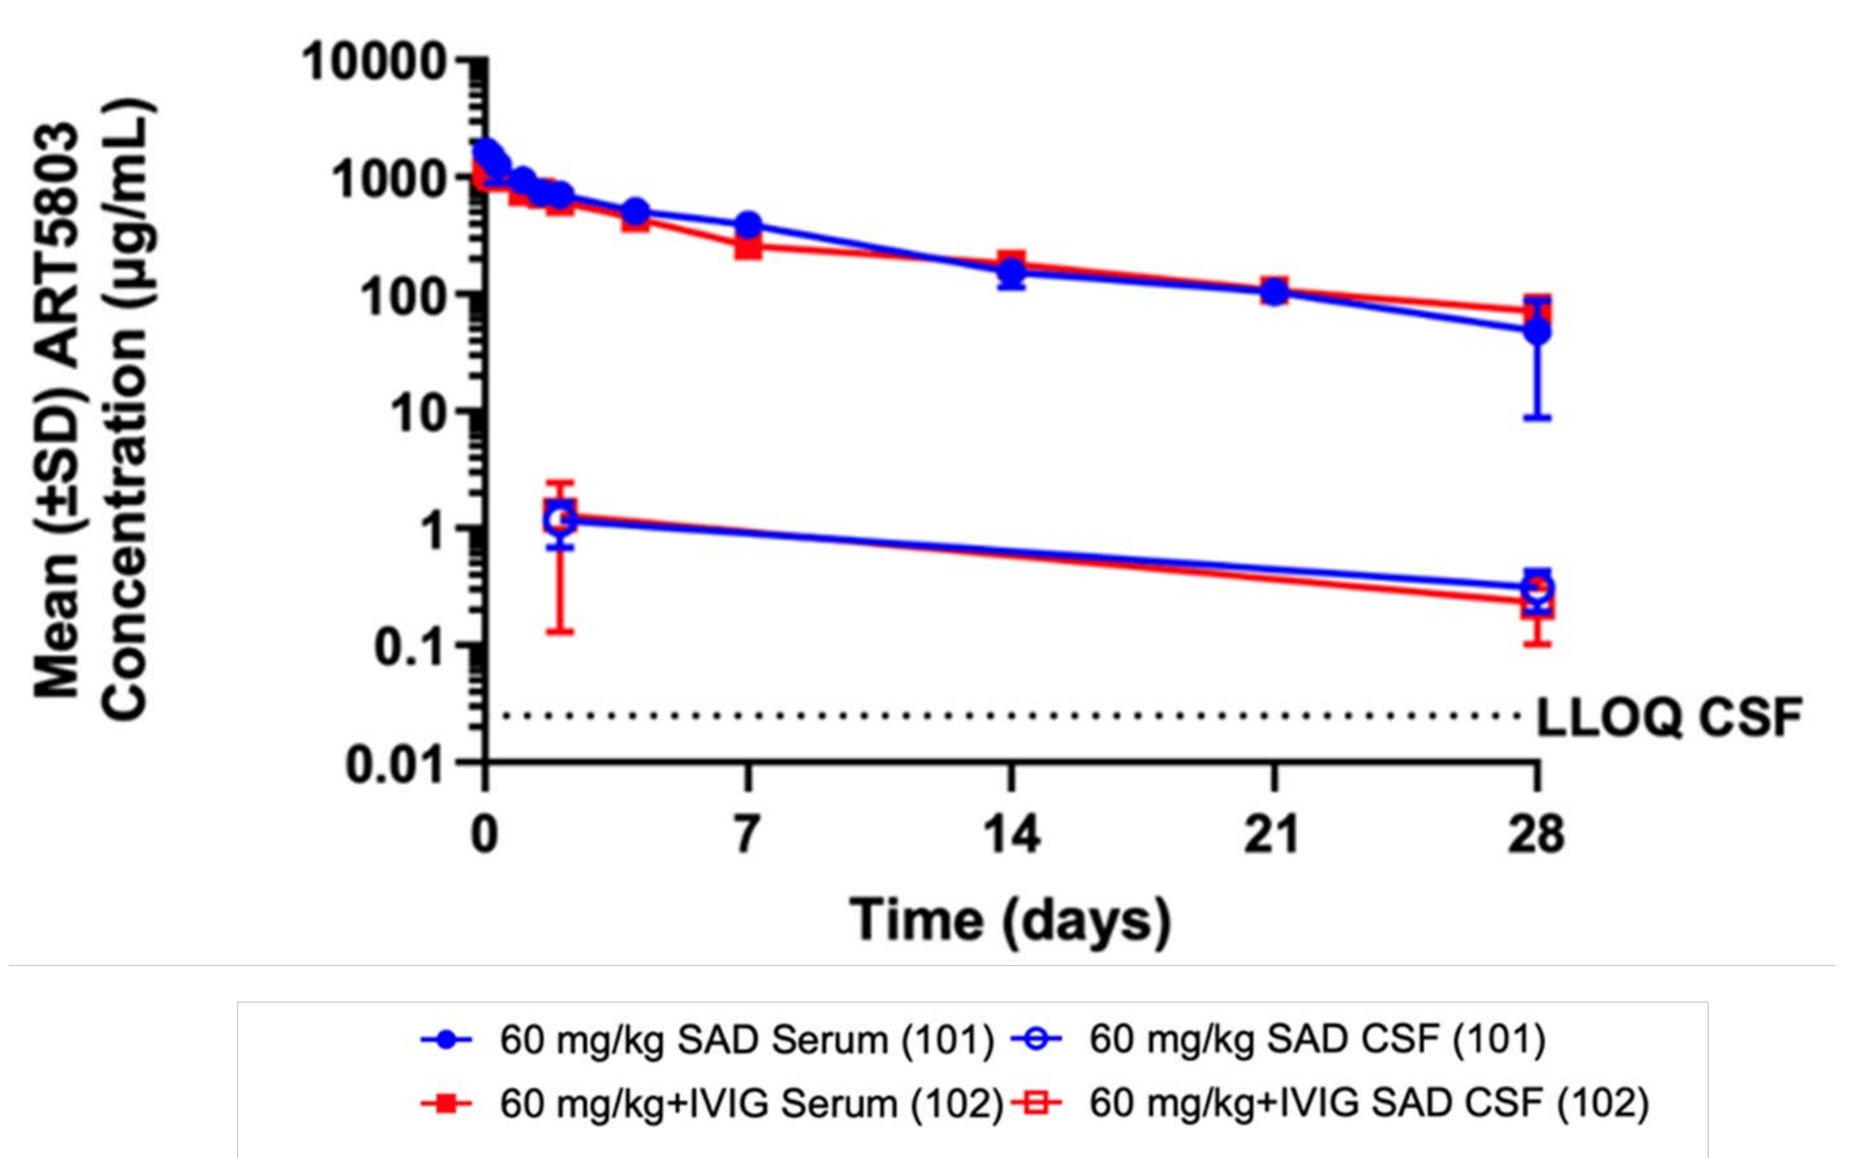
Mean (±SD) ART5803 Concentration (µg/mL)
10000
1000
100
10
1
0.1
0.01
0 7 14 21 28
Time (days)
LLOQ CSF
60 mg/kg SAD Serum (101)
60 mg/kg SAD CSF (101)
60 mg/kg+IVIG Serum (102)
60 mg/kg+IVIG SAD CSF (102)

Planned Phase 2 Studies

Phase 2a Signal Seeking: Korea

A Phase 2a, Open-label Study to Evaluate the Safety, Tolerability, and Preliminary Efficacy of ART5803 in Participants with Anti-NMDAR Encephalitis and Anti-NMDAR Autoantibody-Associated Psychiatric Disease

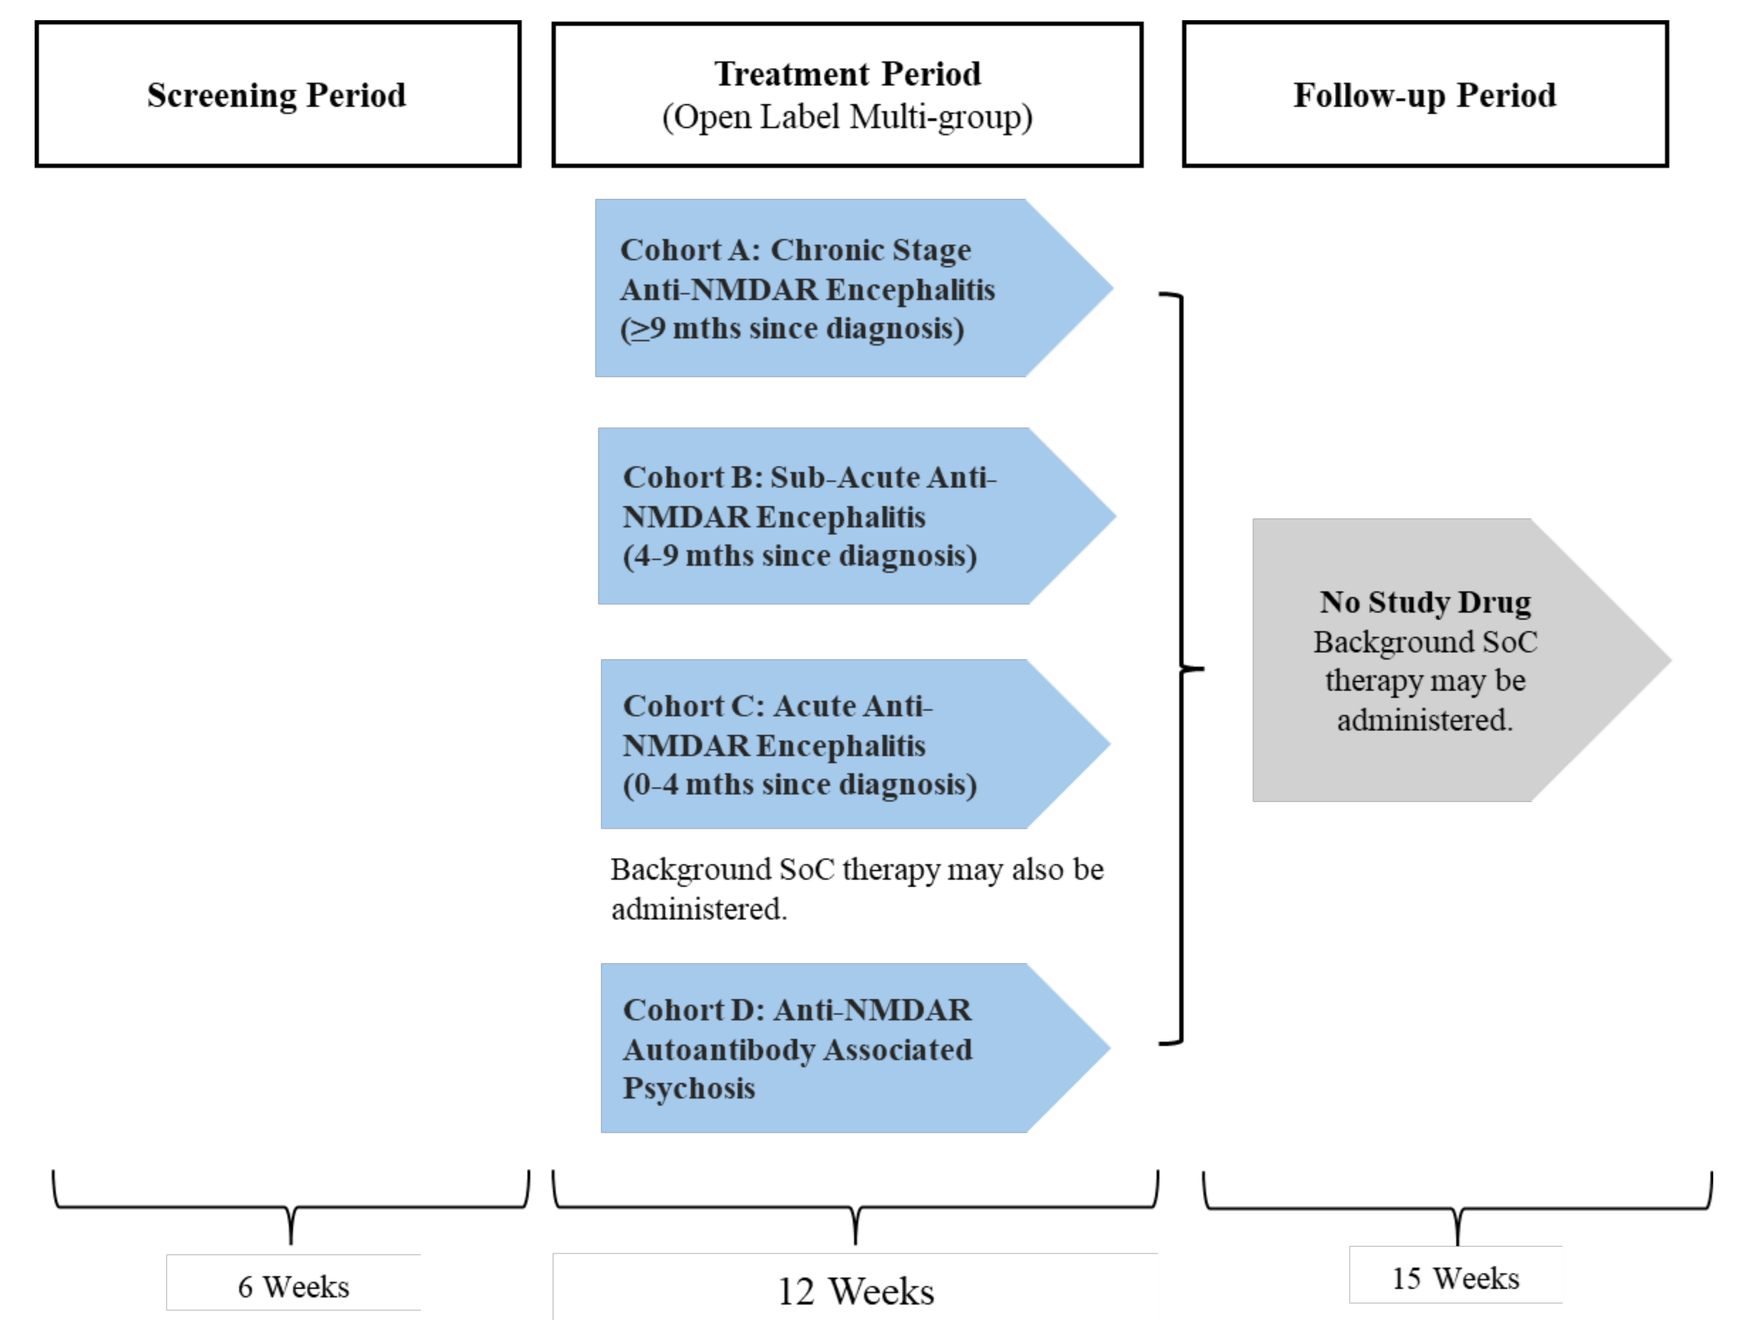
Screening Period
Treatment Period
(Open Label Multi-group)
Follow-up Period
Cohort A: Chronic Stage Anti-NMDAR Encephalitis (≥9 mths since diagnosis)
Cohort B: Sub-Acute Anti-NMDAR Encephalitis (4-9 mths since diagnosis)
Cohort C: Acute Anti-NMDAR Encephalitis (0-4 mths since diagnosis)
Background SoC therapy may also be administered.
Cohort D: Anti-NMDAR Autoantibody Associated Psychosis
No Study Drug
Background SoC therapy may be administered.
6 Weeks
12 Weeks
15 Weeks

Phase 2: USA/International

A Phase 2, Randomized, Double-blind, Placebo-Controlled, Parallel Group Study to Evaluate the Safety, Tolerability, Dose, and Efficacy of ART5803 in Participants with Anti-NMDAR Encephalitis

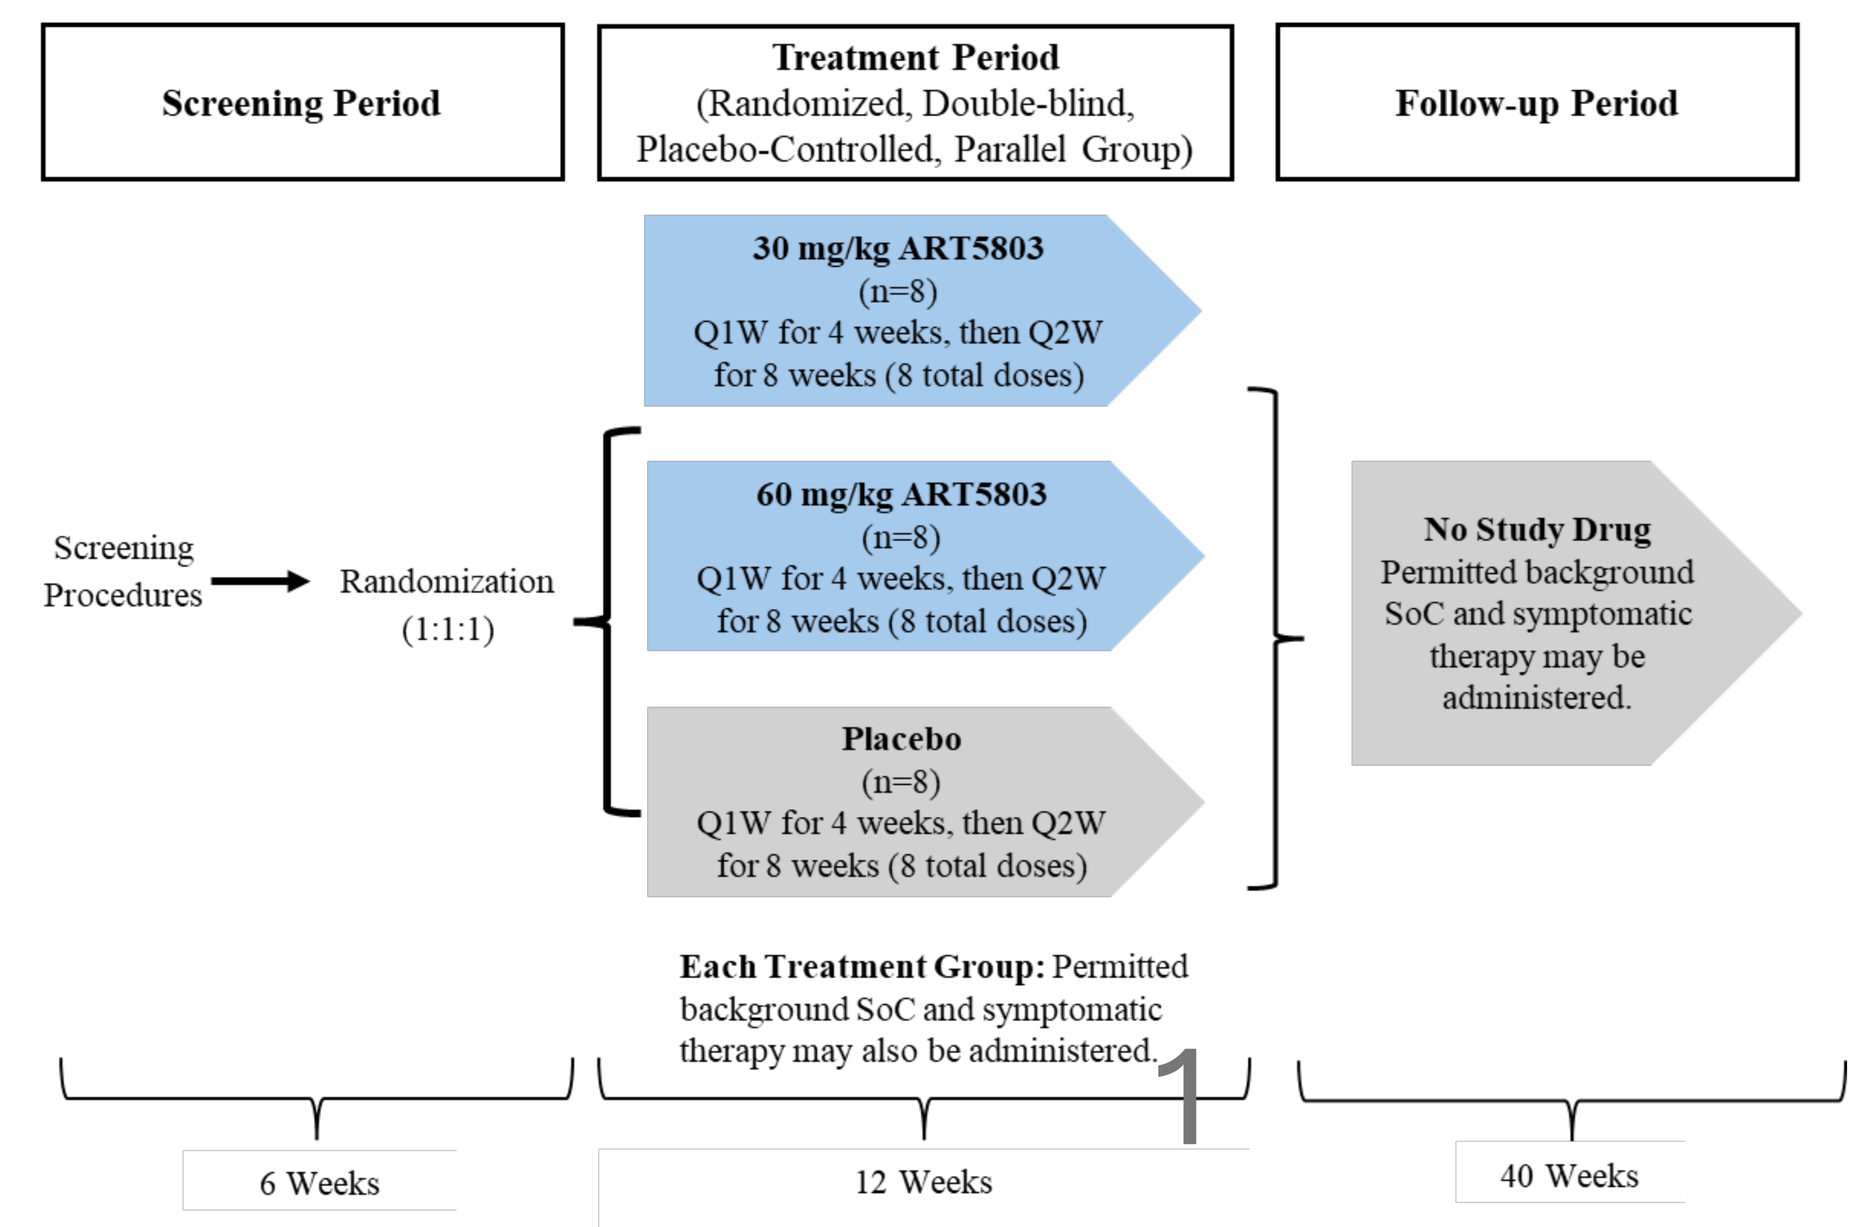
Screening Period
Treatment Period
(Randomized, Double-blind, Placebo-Controlled, Parallel Group)
Follow-up Period
Screening Procedures
Randomization
(1:1:1)
30 mg/kg ART5803
(n=8)
Q1W for 4 weeks, then Q2W for 8 weeks (8 total doses)
60 mg/kg ART5803
(n=8)
Q1W for 4 weeks, then Q2W for 8 weeks (8 total doses)
Placebo
(n=8)
Q1W for 4 weeks, then Q2W for 8 weeks (8 total doses)
Each Treatment Group: Permitted background SoC and symptomatic therapy may also be administered.
No Study Drug
Permitted background SoC and symptomatic therapy may be administered.
6 Weeks
12 Weeks
40 Weeks

1