Efficacy of zumilokibart (APG777) in patients with atopic dermatitis and evidence of Type 2 inflammatory comorbidities: results from the phase 2 APEX study

Poster L36

Linda Stein Gold[1], Jonathan I. Silverberg[2], Andrew Blauvelt[3], Melinda Gooderham[4], Kenji Kabashima[5], Catherine Maari[6], Susanna Wen[7], Olivia Choi[7], Christine Wang[7], Li Xie[7], Joshua Cirulli[7], Kristine Nograles[7], Amol Kamboj[7], Emma Guttman-Yassky[8]

[1]Henry Ford Health System, Detroit, MI, USA; [2]George Washington University School of Medicine and Health Sciences, Washington, DC, USA; [3]Blauvelt Consulting, LLC, Annapolis, MD, USA; [4]SKiN Centre for Dermatology, Peterborough, Ontario, Canada and Queen's University, Kingston, Ontario, Canada; [5]Kyoto University Graduate School of Medicine, Kyoto, Japan; [6]Innovaderm Research, Montréal, Québec, Canada; [7]Apogee Therapeutics, Inc., Waltham, MA, USA; [8]Icahn School of Medicine at Mount Sinai, New York, NY, USA

Scan to access digital poster

INTRODUCTION

- IL-13 is a pleiotropic cytokine with a dominant role in driving Type 2 inflammation, a hallmark of many Type 2 inflammatory conditions including atopic dermatitis (AD), asthma, and chronic rhinosinusitis.
 - These diseases are characterized by the presence of allergic and eosinophilic inflammation, where the Type 2 inflammatory response drives production of high-affinity IgE and recruitment, activation, and survival of eosinophils in tissues.[1,2]
- Zumilokibart (APG777) is a novel, half-life-extended anti-IL-13 monoclonal antibody designed to achieve early peak drug exposures that are sustained over an extended dosing interval.
- The ~77-day half life of zumilokibart is conferred through a YTE amino acid modification in the Fc region that increases FcRn-mediated antibody recycling.[3,4]
- The phase 2 APEX Part A study met the primary endpoint, showing significant improvement in AD lesions by Week 2 and a 71.0% decrease from baseline EASI at Week 16 in patients with moderate-to-severe AD.[5]

STUDY OBJECTIVE

This post hoc analysis evaluated the 16-week efficacy of zumilokibart in participants with AD with and without evidence of Type 2 inflammatory comorbidities in the phase 2 APEX Part A study, to support the rationale for further investigation of zumilokibart in AD and asthma.

STUDY DESIGN & METHODS

- APEX (NCT06395948) is a phase 2 study evaluating zumilokibart in adults (≥18 years) with moderate-to-severe AD (EASI ≥16, vIGA-AD score ≥3, BSA ≥10%; **Figure 1**).

Figure 1: Design of APEX Part A: proof of concept (N=123)

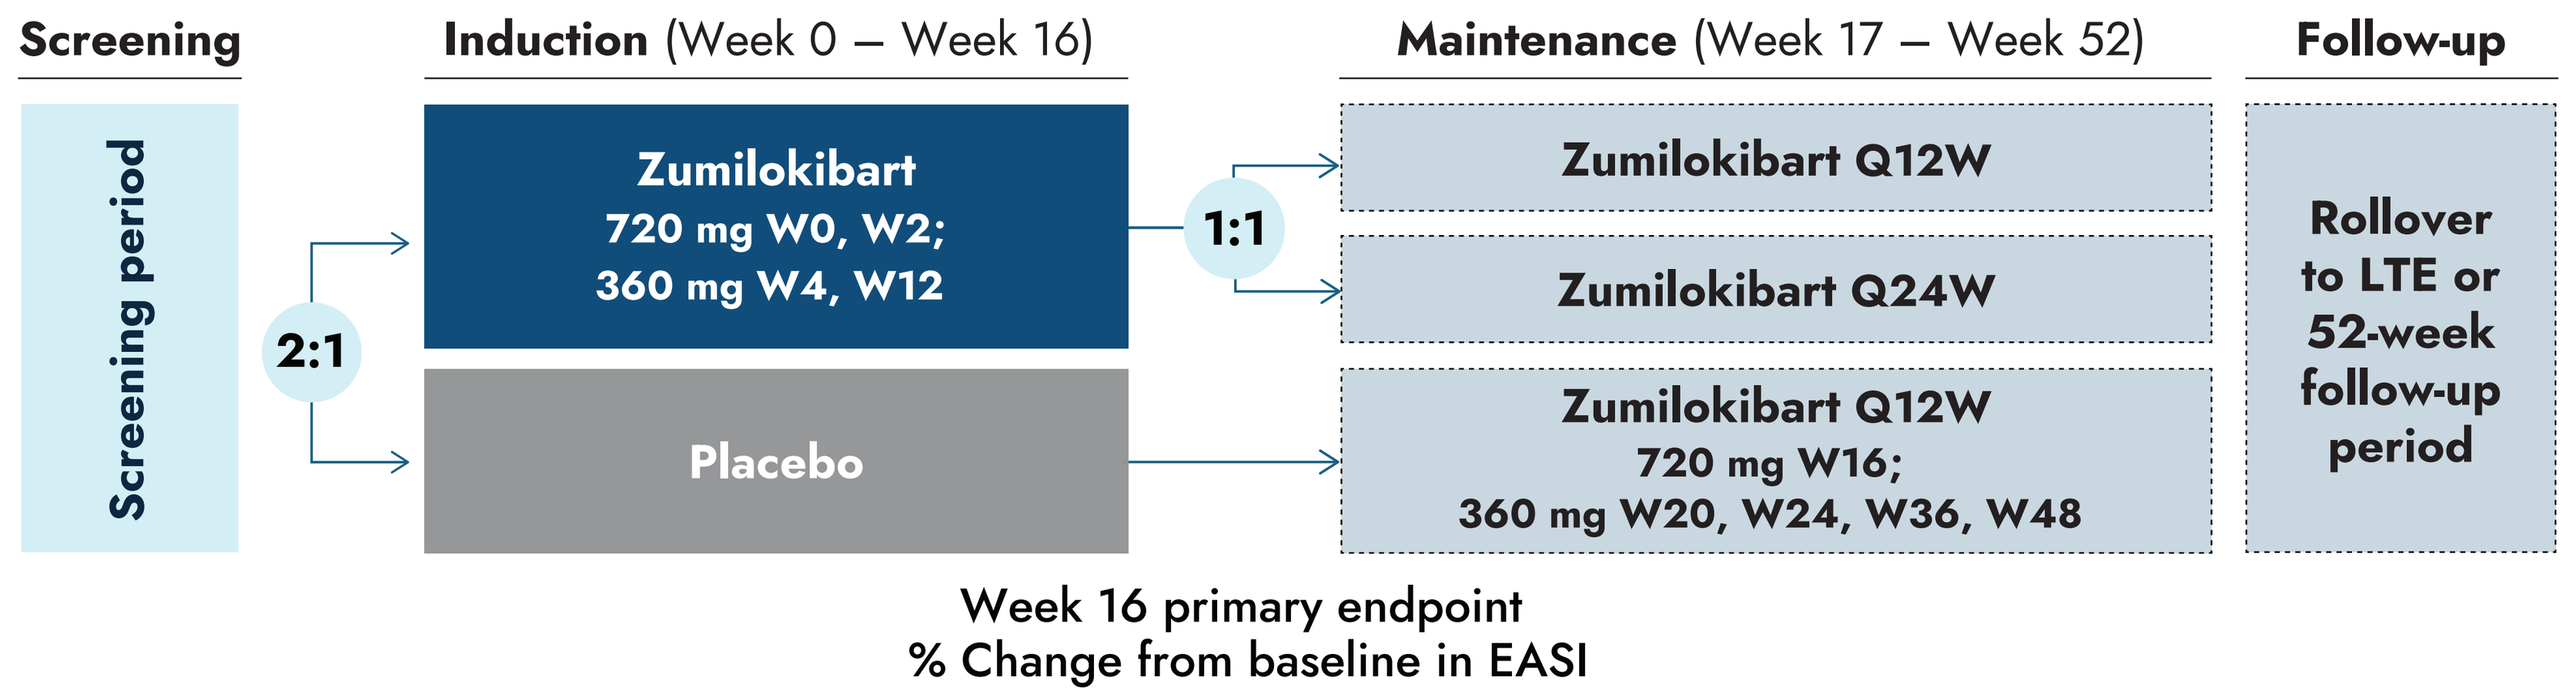

Screening
Induction (Week 0 – Week 16)
Maintenance (Week 17 – Week 52)
Follow-up
Screening period
2:1
Zumilokibart
720 mg W0, W2;
360 mg W4, W12
Placebo
1:1
Zumilokibart Q12W
Zumilokibart Q24W
Zumilokibart Q12W
720 mg W16;
360 mg W20, W24, W36, W48
Rollover to LTE or 52-week follow-up period
Week 16 primary endpoint
% Change from baseline in EASI

- For this post hoc, as-observed analysis, treatment comparisons in participants with and without evidence of Type 2 comorbidities at baseline were assessed by the percentage achieving 75% improvement in the Eczema Area and Severity Index (EASI-75) at Week 16.
- EASI-75 was evaluated in the overall study population and the following subgroups, defined by the presence or absence of Type 2 inflammation or comorbidities:
 - Self-reported history of asthma and/or sinonasal conditions (eg, allergic rhinitis or sinusitis, chronic rhinitis or sinusitis, nasal polyps, nasal turbinate hypertrophy) at baseline.
 - Baseline BEC above or below 300 cells/μL.
 - Baseline IgE level above or below 100 IU/mL.
- Descriptive statistics are provided; subgroup analyses were not powered for statistical comparisons between groups.

RESULTS

Baseline characteristics

- Baseline demographics and disease characteristics were generally similar between treatment groups.
 - Mean baseline EASI (± SD) was 25.3 (± 10.8) in the placebo group and 25.2 (± 10.8) in the zumilokibart group.

Overall safety & efficacy

- Treatment with zumilokibart led to a significantly greater proportion of participants achieving EASI-75 at Week 16 (74.7% vs 26.3% with placebo; p<0.001; **Figure 2**).
- Treatment with zumilokibart was well tolerated. AEs occurring in ≥5% of zumilokibart-treated participants included noninfective conjunctivitis and upper respiratory tract infection.[3]

Figure 2: Significant EASI-75 response at Week 16

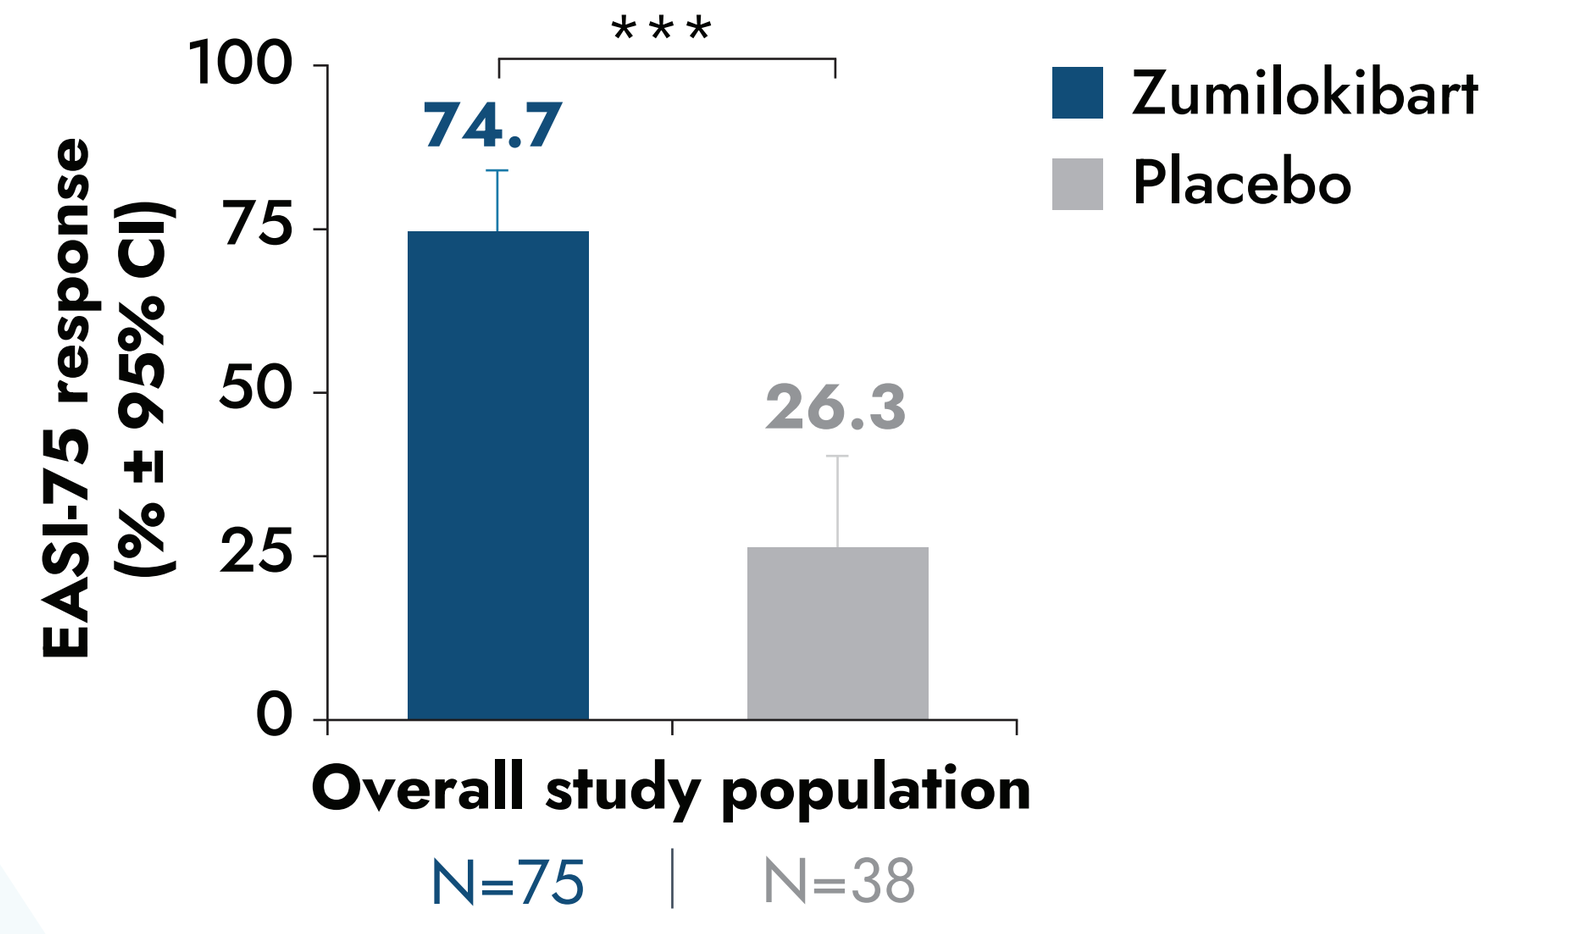

EASI-75 response (% ± 95% CI)
100
75
50
25
0

74.7
26.3
Zumilokibart
Placebo
Overall study population
N=75 | N=38

***p<0.001. N, number of participants with available data at Week 16; missing EASI response data are not imputed. All available data are used regardless of rescue medications or study drug discontinuation.

Type 2 comorbid subgroup analyses

- EASI-75 response rates were greater with zumilokibart than with placebo, regardless of clinical history of asthma and/or sinonasal conditions (**Figure 3A**), baseline IgE level (**Figure 3B**), or baseline blood eosinophil level (**Figure 3C**).
- Consistent results showing greater improvements in itch (as measured using I-NRS percentage change from baseline) were observed with zumilokibart versus placebo, regardless of how the Type 2 subgroup was defined (data not shown).
- No new safety signals were detected in patients with or without asthma and sinonasal conditions.

Figure 3: Higher EASI-75 response at Week 16 with zumilokibart regardless of coexisting Type 2 inflammatory conditions or biomarkers

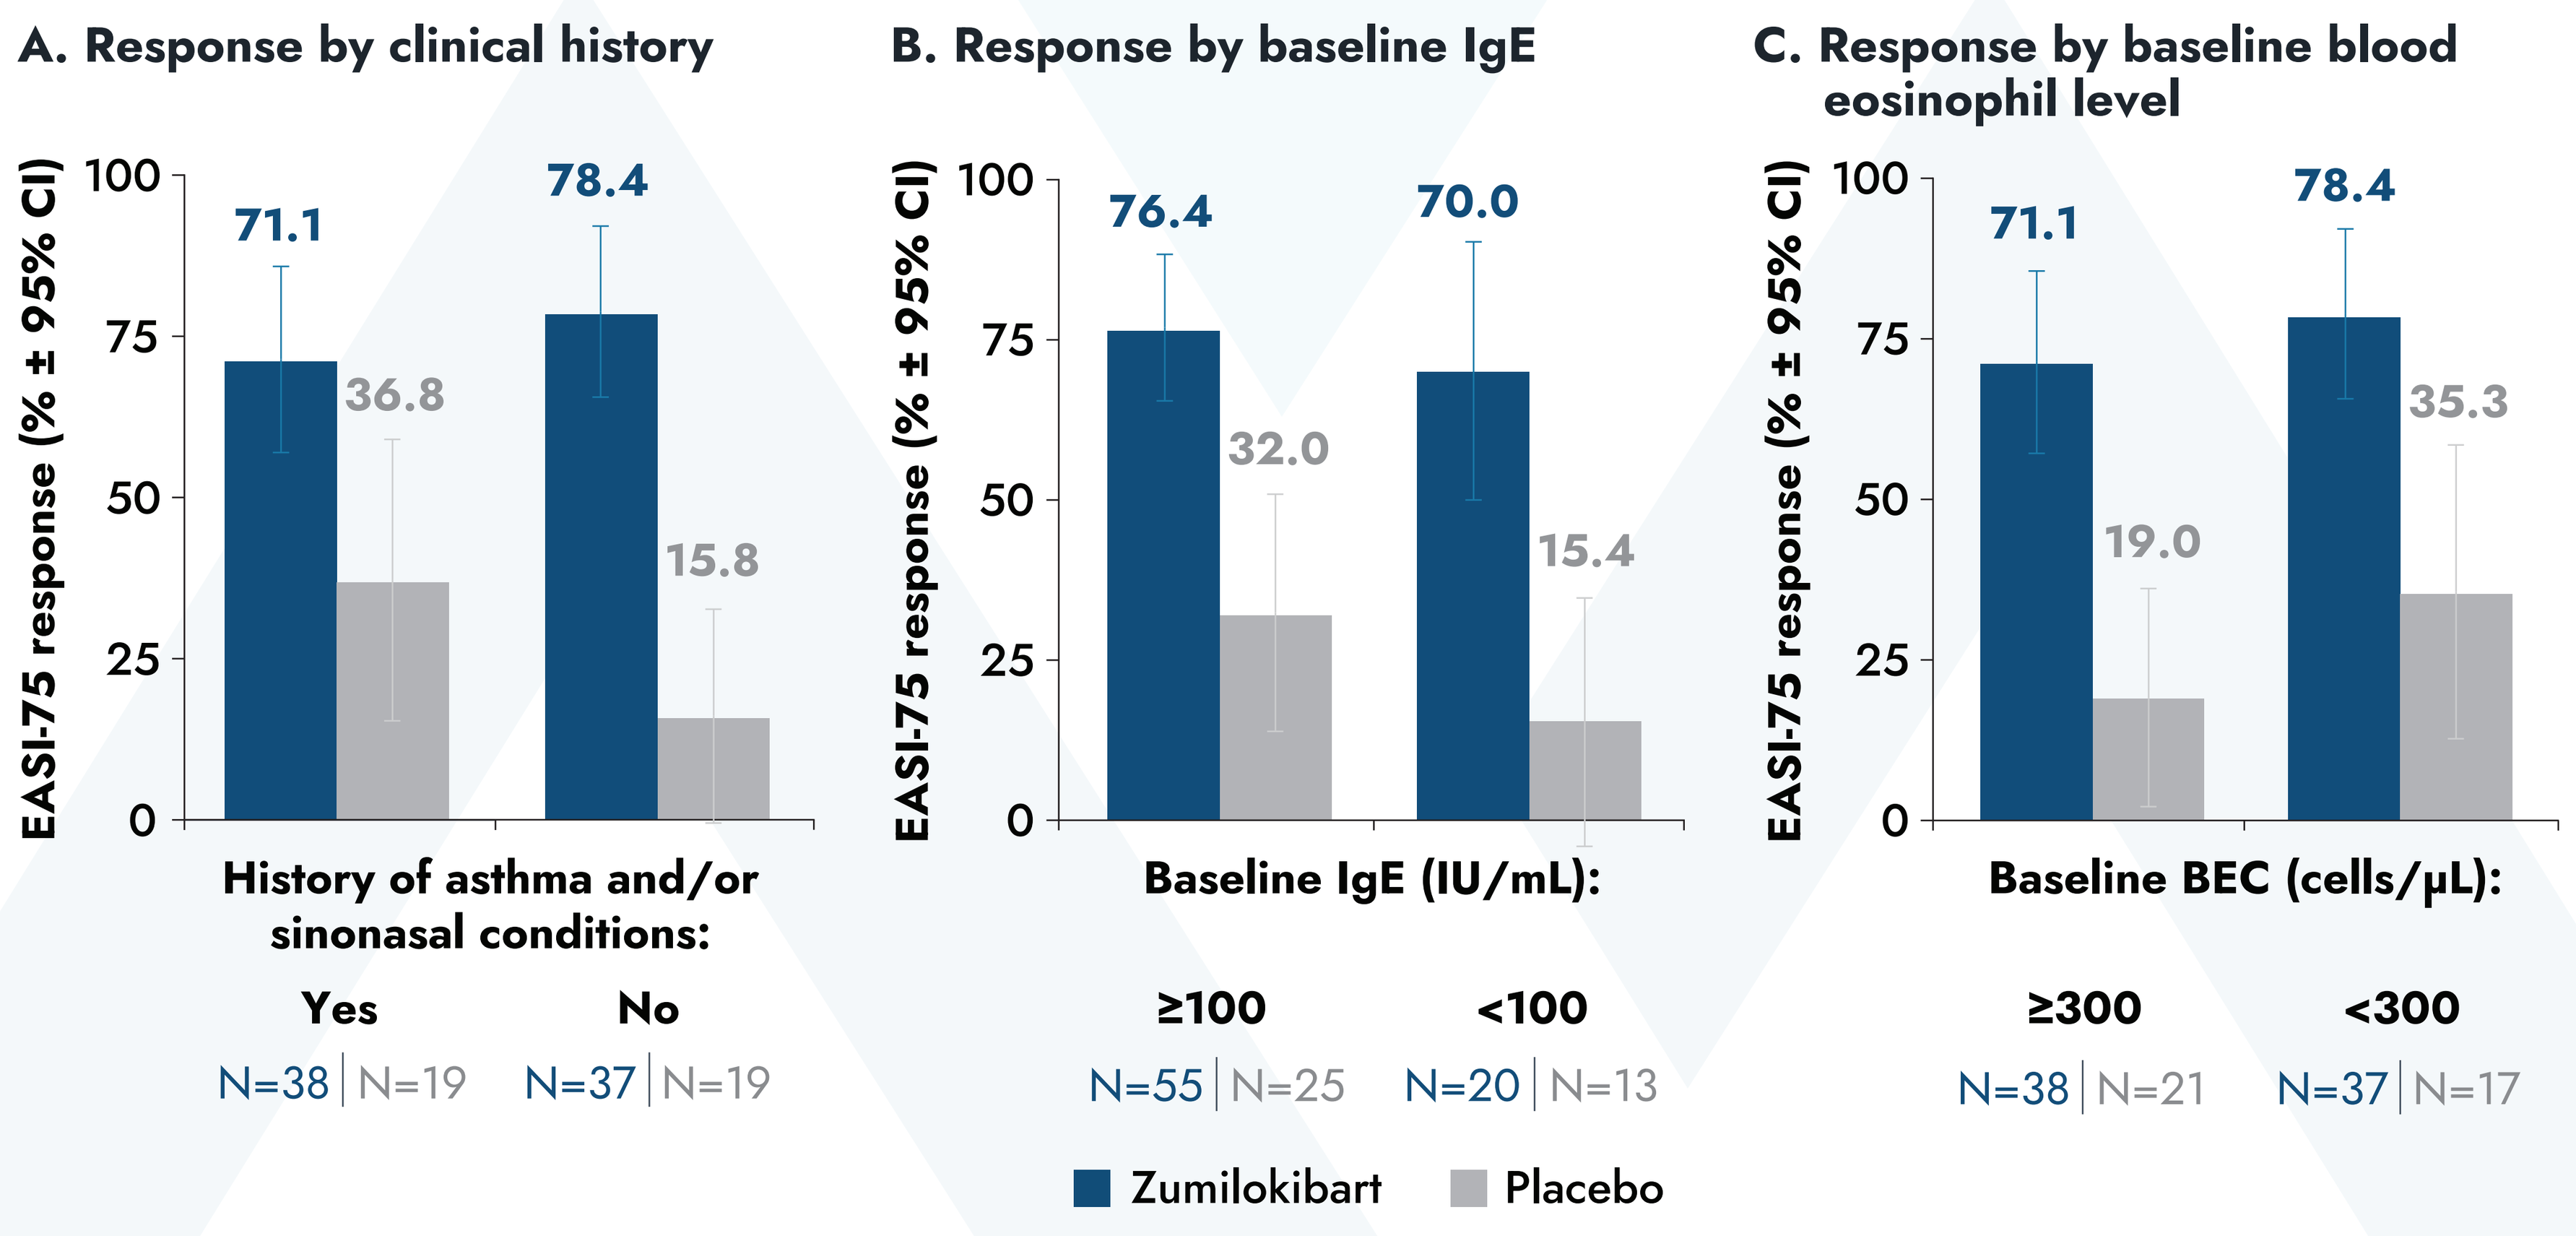

A. Response by clinical history
B. Response by baseline IgE
C. Response by baseline blood eosinophil level
EASI-75 response (% ± 95% CI)
100
75
50
25
0
71.1
36.8
78.4
15.8
History of asthma and/or sinonasal conditions:
Yes
No
N=38 | N=19
N=37 | N=19
76.4
32.0
70.0
15.4
Baseline IgE (IU/mL):
≥100
<100
N=55 | N=25
N=20 | N=13
71.1
19.0
78.4
35.3
Baseline BEC (cells/μL):
≥300
<300
N=38 | N=21
N=37 | N=17
Zumilokibart
Placebo

N, number of participants with available data at Week 16; missing EASI response data are not imputed. All available data are used regardless of rescue medications or study drug discontinuation.

CONCLUSIONS

- With only 4 dosing days during the 16-week induction period of the phase 2 APEX Part A study, treatment with zumilokibart led to clinically meaningful improvements in AD signs and symptoms at Week 16 regardless of coexisting Type 2 inflammatory conditions.
- Q12W and Q24W maintenance dosing is being evaluated in the ongoing APEX Part A study (readout expected in Q1 2026).
- Interim results of a phase 1b study in patients with mild-to-moderate asthma demonstrate early and sustained suppression of FeNO following a single dose of zumilokibart.
- These results, together with the improved PK of zumilokibart enabling less frequent dosing, support further clinical investigation with zumilokibart for the treatment of patients with diseases of Type 2 inflammation, including AD and asthma.

FUTURE DIRECTIONS

Zumilokibart in asthma

- Asthma is a common comorbidity among patients with AD, with 1 meta-analysis reporting that 28% of patients with moderate-to-severe AD had comorbid asthma.[6]
- IL-13 is a key driver of Type 2 airway inflammation and increased expression of FeNO, both of which are strongly associated with exacerbations and impaired lung function in patients with asthma.
- Safety, PK, and magnitude and durability of FeNO suppression following a single 720 mg dose of zumilokibart in adults with mild-to-moderate asthma and FeNO ≥25 ppb is being evaluated in an ongoing phase 1b study (NCT06920901).
 - Treatment with zumilokibart led to rapid suppression of FeNO, with maximal mean absolute reduction of 45 ppb (vs 11 ppb with placebo) and −60% change from baseline (vs −10% with placebo) at Week 2 after dosing (**Figure 4**).
 - Mean FeNO reduction >40 ppb from baseline was maintained at all timepoints from Week 2 through Week 16 (limit of available follow-up; **Figure 4**).

Figure 4: Early and sustained suppression of FeNO following a single dose of zumilokibart in patients with mild-to-moderate asthma

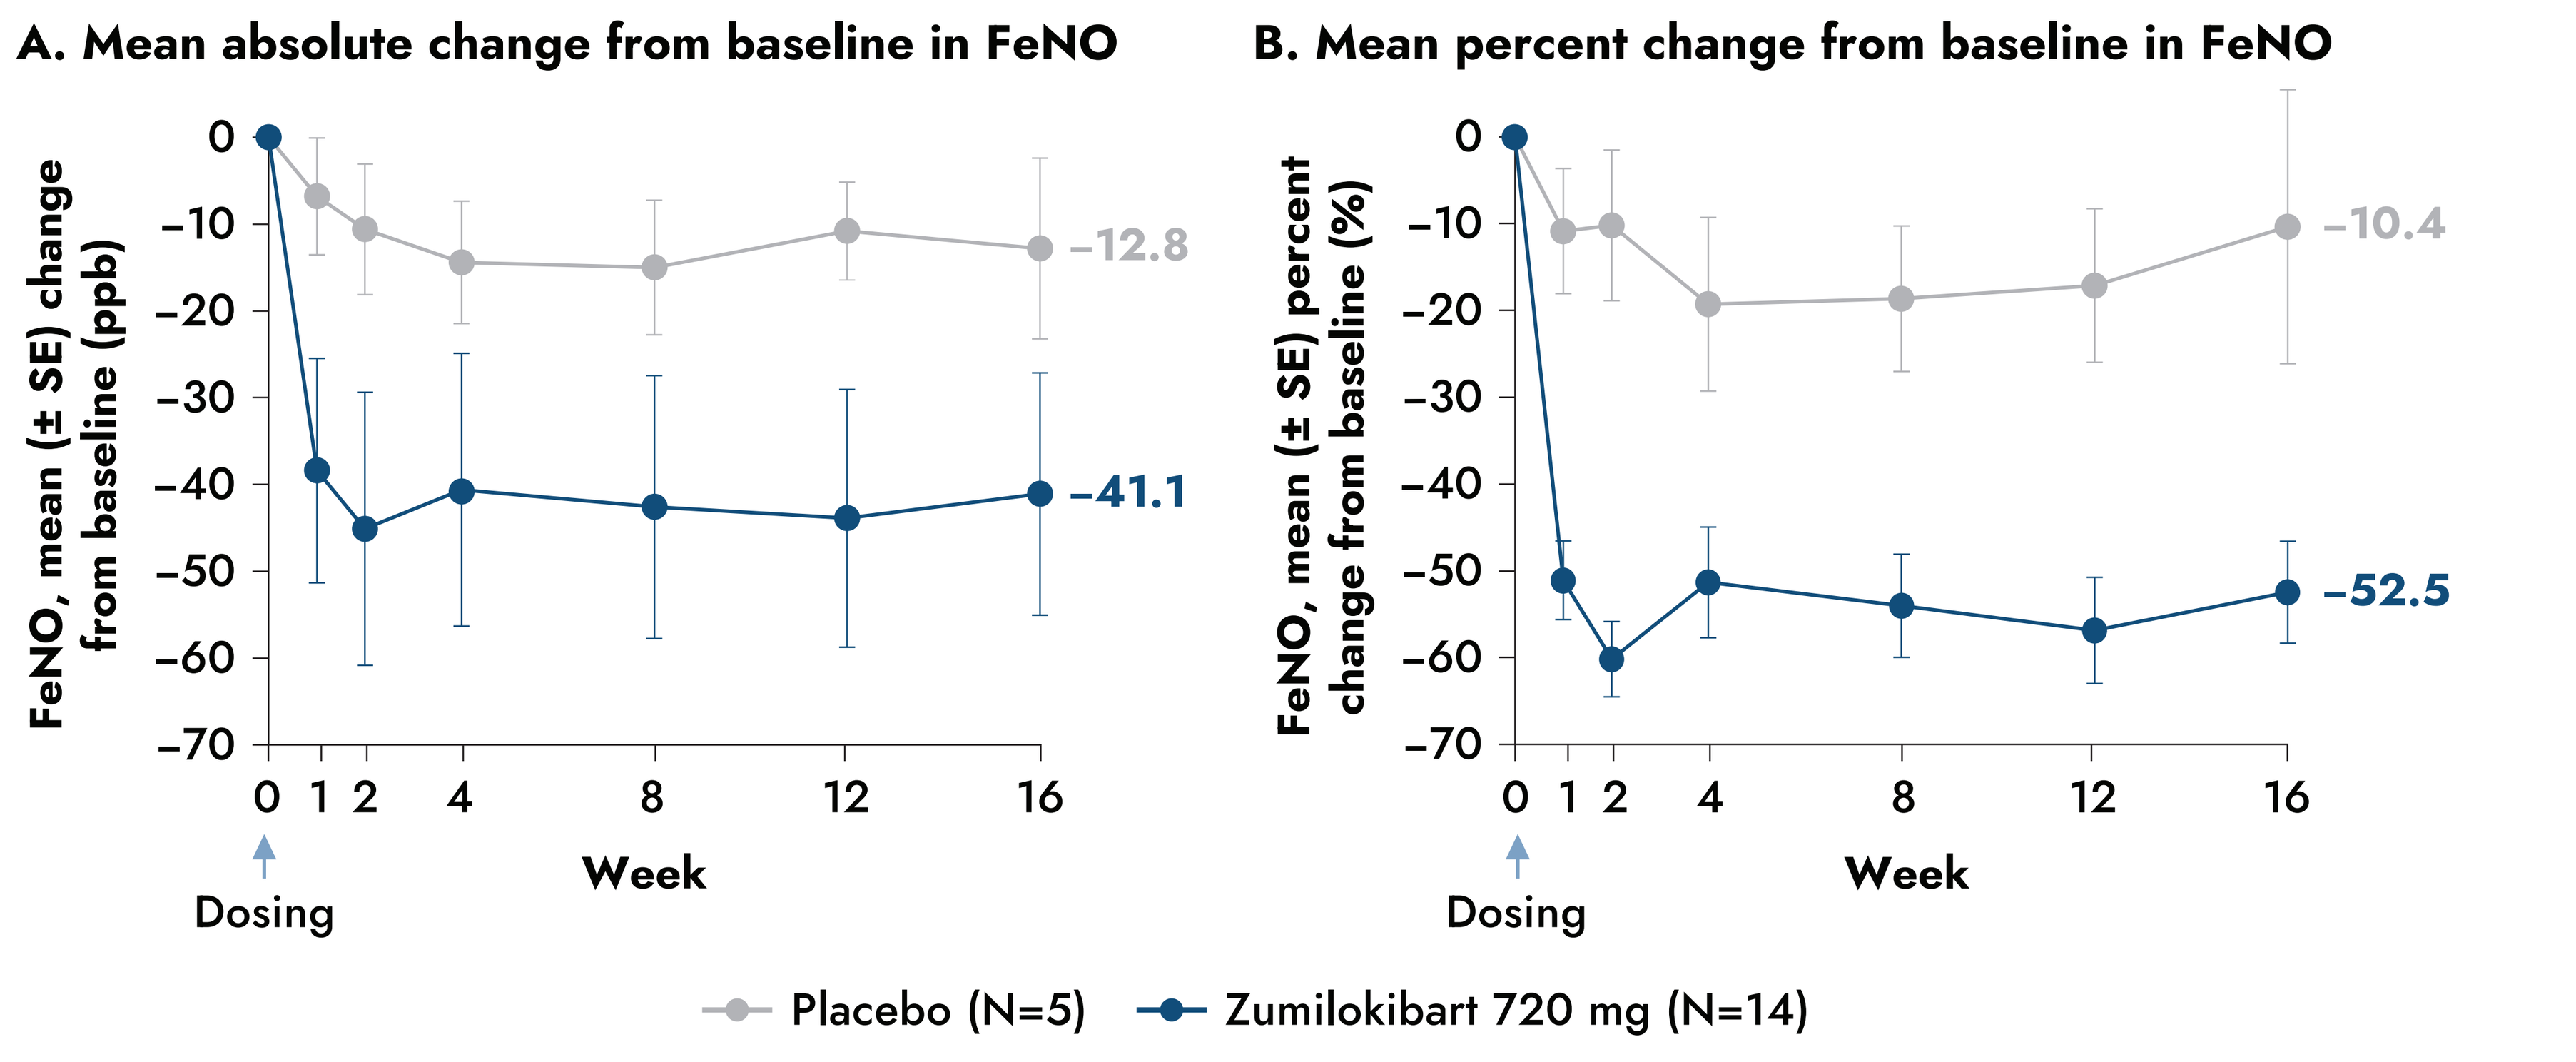

A. Mean absolute change from baseline in FeNO
FeNO, mean (± SE) change from baseline (ppb)
0
−10
−20
−30
−40
−50
−60
−70
0 1 2 4 8 12 16
Week
Dosing
−12.8
−41.1
B. Mean percent change from baseline in FeNO
FeNO, mean (± SE) percent change from baseline (%)
−10.4
−52.5
Placebo (N=5)
Zumilokibart 720 mg (N=14)

Mean ± SD FeNO levels at baseline were 57.2 ± 34.1 ppb in the placebo group and 66.9 ± 65.3 ppb in the zumilokibart group. One zumilokibart-treated participant discontinued following the Week 12 visit (lost to follow-up).

ABBREVIATIONS

AD, atopic dermatitis; AE, adverse event; BEC, blood eosinophil count; BSA, body surface area; CI, confidence interval; EASI, Eczema Area and Severity Index; EASI-75, achieving at least 75% reduction from baseline in EASI; Fc, fragment crystallizable; FcRn, neonatal Fc receptor; FeNO, fractional exhaled nitric oxide; IgE, immunoglobulin E; IL-13, interleukin-13; I-NRS, Itch-Numeric Rating Scale; IU, international units; LTE, long-term extension; Q12W, every 12 weeks; PK, pharmacokinetics; ppb, parts per billion; Q24W, every 24 weeks; SD, standard deviation; SE, standard error; vIGA-AD, Validated Investigator Global Assessment for AD; W, week; YTE, triple-amino acid modification in the Fc region that extends antibody half-life.

REFERENCES

1. Guttman-Yassky E, et al. *Lancet*. 2025;405:583-596.
2. Lindsley AW, et al. *J Asthma Allergy*. 2025;18:33-57.
3. Zhu E, et al. *J Allergy Clin Immunol Glob*. 2025;4:100545.
4. Lim XQ, et al. *Clin Transl Sci*. 2025;18:e70456.
5. Guttman-Yassky E, et al. Presented at the European Academy of Dermatology and Venerology (EADV) Congress 2025; Paris, France.
6. Ravnborg N, et al. *J Am Acad Dermatol*. 2021;84:471-478.

ACKNOWLEDGMENTS

Apogee Therapeutics, Inc. sponsored the studies reported in this poster. Medical writing and editorial support for this poster were provided by Kate Smigiel of Apogee Therapeutics, Inc., and Kevin Corcoran, PhD, and Heidi Reinholdt, MS, MA, both of Helios Global Group, on behalf of Apogee Therapeutics, Inc.

DISCLOSURES

LSG has served as an investigator, advisor, and/or speaker for AbbVie, Amgen, Apogee, Arcutis, Incyte, Kymera, Leo, Organon, Pfizer, Regeneron, and Sanofi. **JIS** has received honoraria as a consultant and/or advisory board member for AbbVie, Aldena, Amgen, AObiome, Apollo, Arcutis, Arena, Attovia, Boehringer-Ingelheim, Bristell-Meyers Squibb, Castle Biosciences, Connect Biopharma, Corevitas, Dermavant, Eli Lilly, FIDE, Formation Bio, Galderma, GlaxoSmithKline, Immunocore, Incyte, Inmagene, Invea, Leo Pharma, Merck, Nektar, Novartis, Pfizer, RAPT, Recludix, Regeneron, Sandoz, Sanofi-Genzyme, Shaperon, TARGET-RWE, Teva, Triveni, UCB, Union, UpToDate; speaker for AbbVie, Arcutis, Dermavant, Eli Lilly, Galderma, Leo Pharma, Pfizer, Regeneron, Sanofi-Genzyme; institution received grants from Galderma, Incyte, and Pfizer. **AB** has served as a speaker (received honoraria) for Almirall, Eli Lilly and Company, and UCB; has served as a scientific advisor (received honoraria) for AbbVie, Almirall, Alumis, Amgen, Anaptysbio, Apogee, Arcutis, Astria, Boehringer Ingelheim, Bristol Myers Squibb, Celltrion, Corvus, Dermavant, Eli Lilly and Company, Galderma, GlaxoSmithKline, Immunovant, Incyte, IQVIA, Janssen, Leo, Lipidio, Merck, Novartis, Oruka, Paragon, Pfizer, Rani Therapeutics, Regeneron, Sanofi, Spherix Global Insights, Sun Pharma, Syncona, Takeda, UCB, Union, and Zai Lab; has acted as a clinical study investigator (institution has received clinical study funds) for AbbVie, Acelyrin, Almirall, Alumis, Amgen, Arcutis, Boehringer Ingelheim, Bristol Myers Squibb, Dermavant, Eli Lilly and Company, Galderma, Incyte, Janssen, Leo, Merck, Novartis, Pfizer, Regeneron, Sanofi, Sun Pharma, Takeda, and UCB; and owns stock in Lipidio and Oruka. **MG** has served as an investigator, speaker and/or advisor for: AbbVie, Acelyrin, Alumis, Amgen, Akros, Arcutis, Aristea, AnaptysBio, Apogee, Bausch Health, BMS, Boehringer Ingelheim, Dermira, Dermavant, Eli Lilly, Galderma, GSK, Incyte, Inmagene, JAMP Pharma, Janssen, LEO Pharma, L'Oreal, MedImmune, Meiji, Moonlake, Nektar, Nimbus, Novartis, Organon, Oruka, Pfizer, Q32 Bio, Regeneron, Sanofi Genzyme, Sun Pharma, Tarsus, Takeda, UCB, Union, Ventyx, and Vyne. **KK** has received consulting fees, honoraria, grant support, and/or lecture fees from AbbVie, Amgen, Apogee Therapeutics, Inc., Eli Lilly, Kyowa Kirin, Japan Tobacco, LEO Pharma, Maruho, Mitsubishi Tanabe, Ono Pharmaceutical, Pfizer, Procter & Gamble, Sanofi, Taiho, and Torii Pharmaceutical. **CM** is a member of an advisory board or equivalent with AbbVie, Apogee Therapeutics, UCB, Boehringer Ingelheim, Sanofi, Eli Lilly, Galderma, Leo pharma, Bristol-Myers Squibb, Janssen, Bausch, and Pfizer; member of a speakers' bureau with AbbVie, Eli Lilly, Janssen, Bausch, and Pfizer; received a grant or honorarium from ICPDHM, McGill University; and participated in a clinical trial for AbbVie, Apogee Therapeutics, Arena, Concert, Bellus, Nimbus, Horizon, Incyte, Sanofi, Target Pharma, UCB, Boston, Cara, Boehringer Ingelheim, Eli Lilly, Dermavant, Galderma, Leo pharma, Bristol-Myers Squibb, Janssen, Bausch, and Pfizer. **EG-Y** is an employee of Mount Sinai and has received research grants (paid to the institution) from and/or is a consultant for: AbbVie, Aclaris Therapeutics, Almirall, Alumis, Amgen, AnaptysBio, Apogee Therapeutics, Apollo Therapeutics, Arcutis, Artax Biopharma, Astria, Boerhinger-Ingelhiem, Bristol Myers Squibb, Celldex, Centrexion Therapeutics, Connect Biopharm, Concerto Biosciences, Coty, DBV, Dualitas Therapeutics, Eli Lilly, Enveda Biosciences, Escient Pharmaceuticals, Galderma, Gate Bio, GSK, GSK Immunology, Incyte, Inmagene, Janssen Biotech, Jasper Therapeutics, Kymera Therapeutics, Kyowa Kirin, LEO Pharma, Matchpoint Therapeutics, Merck, Nektar Therapeutics, NUMAB Therapeutics, Obsidio, OTSUKA, Pfizer, Pharmaxis, Proteologix, Q32 Bio, RAPT, RayThera, Regeneron, Ribon Therapeutics, Sanofi, SATO, Schrödinger, Sitryx, Sun Pharma, Takeda, Teva, TRex Bio, UCB, VRG Therapeutics, and Xencor. **SW**, **OC**, **CW**, **LX**, **JC**, **KN**, and **AK** are employees of Apogee Therapeutics, Inc. and hold company stock/stock options.

APOGEE THERAPEUTICS®

Presented at the American Academy of Allergy, Asthma & Immunology (AAAAI) Annual Meeting, Philadelphia, PA, February 27 – March 2, 2026

For questions, please contact MedAffairs@apogeetherapeutics.com

©Apogee Therapeutics, Inc.