Early and sustained suppression of FeNO with a single dose of zumilokibart (APG777), a novel half-life-extended anti-IL-13 antibody, in patients with mild-to-moderate asthma

Dave Singh[1,2], Jeremy Cole[3], James DeAngelo[4], Erica Winter[5], Abby Chen[5], Glenn Whelan[5], Myreen Tomas[5], Claudia Halaby[5], Carl L. Dambkowski[5], Amol Kamboj[5], Mario Castro[6]

[1]Medicines Evaluation Unit, an IQVIA business, Manchester, United Kingdom; [2]University of Manchester, Manchester University NHS Foundation Trust, Manchester, United Kingdom; [3]OK Clinical Research, LLC, Edmond, OK, USA; [4]Allergy and Clinical Immunology Associates, Pittsburgh, PA, USA; [5]Apogee Therapeutics, Inc., Waltham, MA, USA; [6]University of Kansas School of Medicine, Kansas City, KS, USA

INTRODUCTION

- IL-13 is a key driver of Type 2 airway inflammation and increased expression of fractional exhaled nitric oxide (FeNO), both of which are strongly associated with exacerbations and impaired lung function in patients with asthma.[1,2]
- Zumilokibart (APG777) is a novel, half-life-extended anti-IL-13 monoclonal antibody that achieves early peak therapeutic drug levels that are sustained over an extended dosing interval.
- The phase 2 APEX Part A study (NCT06395948) in moderate-to-severe atopic dermatitis (AD) met the primary endpoint, with zumilokibart producing a 71.0% decrease from baseline in EASI at Week 16 (vs 33.8% placebo; p<0.001).[3]
 - APEX Part A results through Week 52 demonstrated sustained AD disease control with every 12- and 24-week dosing, supporting evaluation of extended dosing intervals in other chronic inflammatory diseases including asthma.[4]
- Asthma is a common comorbidity among patients with AD, with 1 meta-analysis reporting that 28% of patients with moderate-to-severe AD have comorbid asthma.[5]

STUDY OBJECTIVES & DESIGN

- **This phase 1b study (NCT06920901) is evaluating the safety, PK, and magnitude and durability of FeNO suppression following a single dose of zumilokibart in adults with asthma.**
- **Participants were required to have FeNO ≥25 ppb to enrich for asthma with Type 2 inflammation.**

Figure 1: Design of the single-dose zumilokibart phase 1b study in mild-to-moderate asthma

N=19[a]

Key inclusion criteria
- Adults aged 18 to 65 years
- Mild-to-moderate asthma (GINA 2023 criteria)
- FeNO ≥25 ppb

Objectives
- Safety of zumilokibart in patients with asthma
- Change in FeNO as a surrogate marker of treatment response in Type 2 asthma

Duration: ~52 weeks

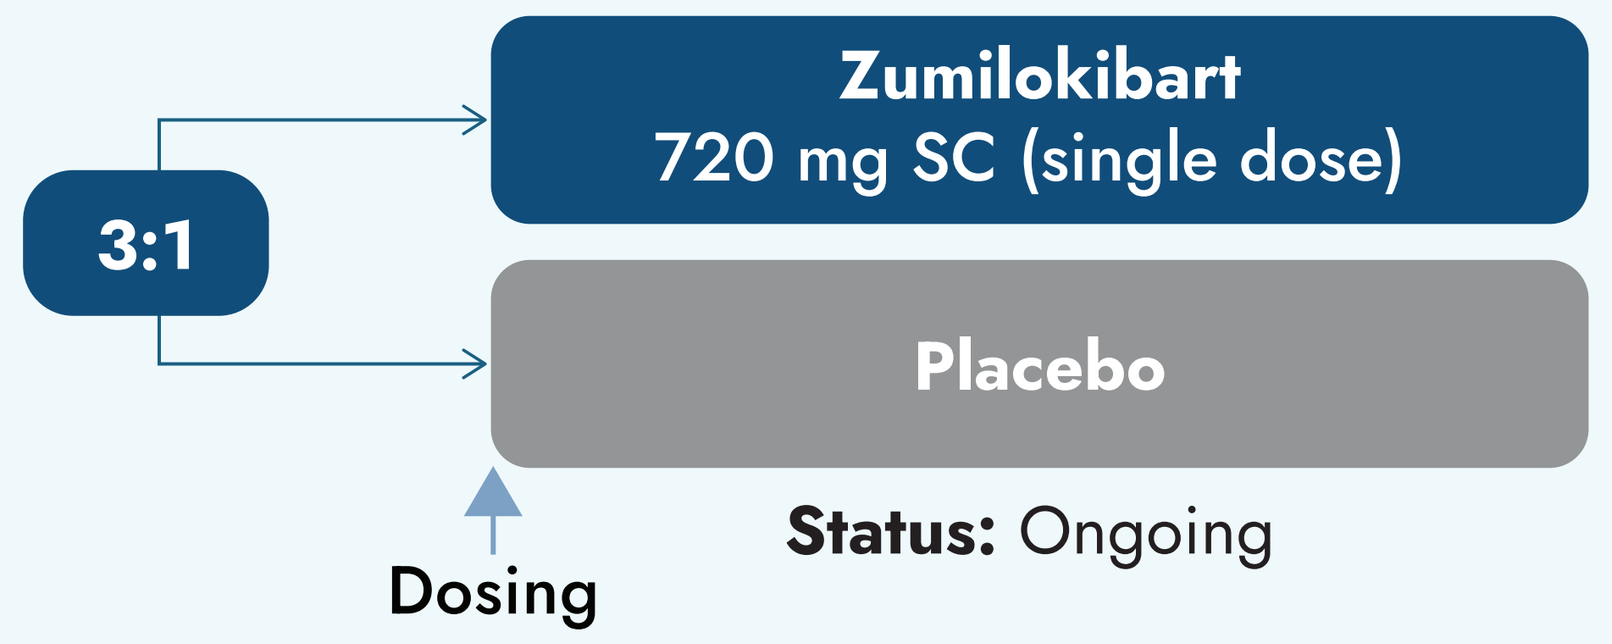
3:1
Zumilokibart
720 mg SC (single dose)
Placebo
Dosing

Status: Ongoing

[a]Study enrolled a total of 31 participants; 19 met the prespecified criteria for the subgroup with FeNO ≥25 ppb, representative of asthma with Type 2 inflammation. This study reports an interim analysis with a data cutoff date of December 23, 2025.

CONCLUSIONS

- Zumilokibart was well tolerated, with safety results in line with expectations for therapies targeting IL-13 in asthma.
- The early peak serum concentration of zumilokibart and its extended half-life enabled rapid suppression of FeNO that was maintained through 16 weeks in all patients, and through 32 weeks for patients with available follow-up, after a single dose.
 - This profile supports evaluation of every 12- and 24-week (and potentially less frequent) dosing in patients with moderate-to-severe asthma.
- These results reinforce the therapeutic potential of IL-13 inhibition in asthma and support further clinical investigation with zumilokibart, as monotherapy or in combination with other half-life-extended monoclonal antibodies, for the treatment of obstructive airway diseases.

Poster number: P1431

Scan to access Digital Poster

For questions, please contact MedAffairs@apogeetherapeutics.com

Presented at the American Thoracic Society (ATS) Annual Scientific Meeting, Orlando, FL, USA, May 15 - 20, 2026

RESULTS

Baseline characteristics

- Interim results were available for all patients through Week 16; this analysis included 19 adults (mean age 44.5 years; 42.1% female) with mild-to-moderate asthma and FeNO ≥25 ppb.
- Baseline demographics and disease characteristics were generally similar between treatment groups (**Table 1**).

Table 1: Demographics and baseline characteristics

	Placebo (N=5)	Zumilokibart (N=14)
Age (years), mean (SD)	51.2 (8.2)	42.1 (13.3)
Female, %	40.0%	42.9%
Caucasian, %	80.0%	92.9%
Weight (kg), mean (SD)	92.5 (16.2)	81.0 (12.8)
Patients on daily ICS ± LABA, %	100.0%	78.6%
Blood eosinophil count, mean (SD)	176 (108)	304 (205)
FeNO (ppb), mean (SD)	57.2 (34.1)	66.9 (65.2)

Pharmacokinetics

- In a first-in-human phase 1 study, the half-life of zumilokibart ranged from 75.3 to 77.5 days across the doses tested.[6]
 - The extended half-life of zumilokibart is conferred by a YTE amino acid modification in the Fc region enhancing FcRn-mediated antibody recycling (**Figure 2**).[7]
- In participants with mild-to-moderate asthma, zumilokibart demonstrated a PK profile consistent with previous studies.
 - The maximum serum concentration (C_{max}) of zumilokibart was achieved 15 days after administration of a single dose.

Figure 2: YTE modification extends the half-life of zumilokibart

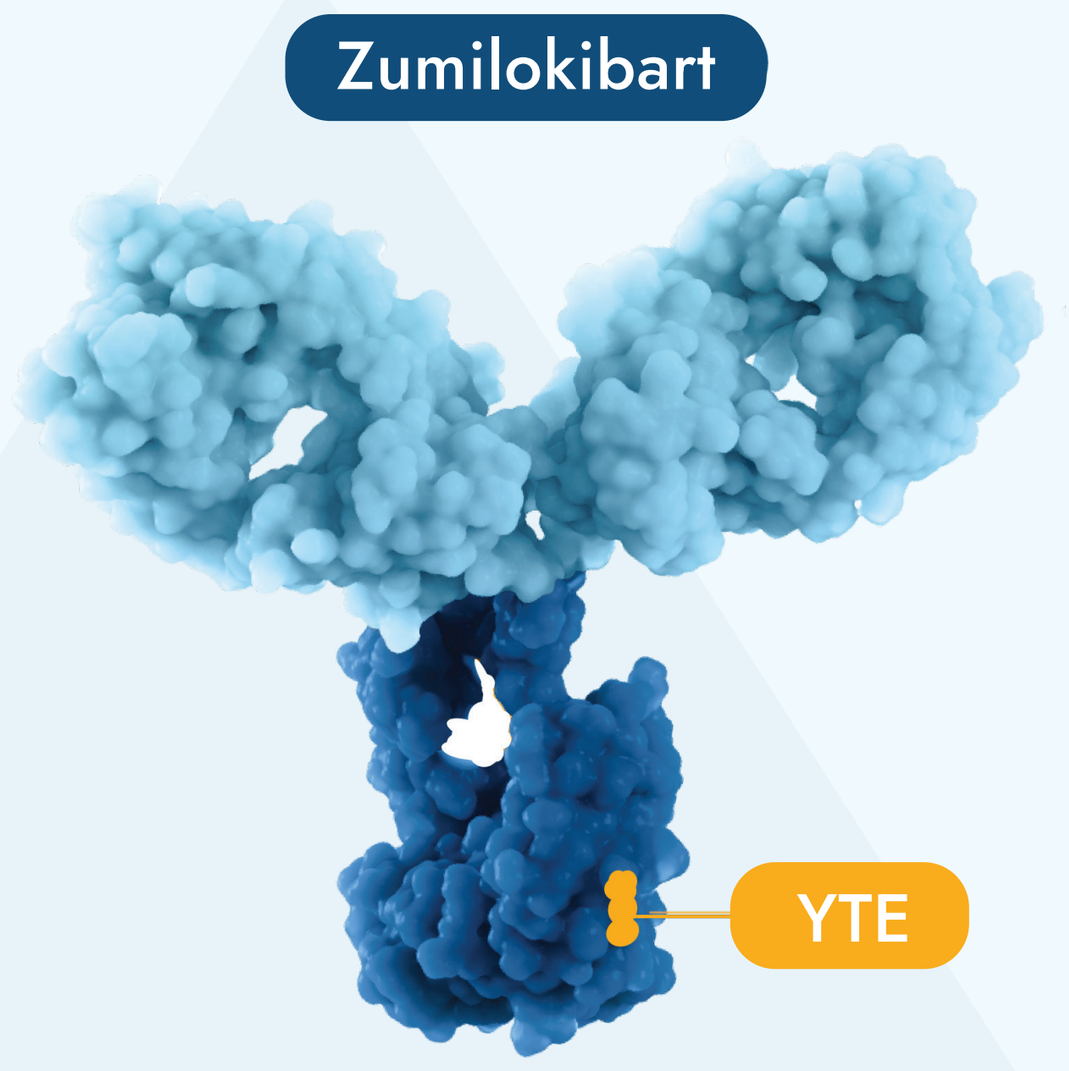
Zumilokibart
YTE

Safety

- Zumilokibart was well tolerated, with no serious or grade 3/4 TEAEs (**Table 2**), and a safety profile consistent with expectations for an IL-13-targeted therapy in asthma.

Table 2: Treatment-emergent adverse events

N (%)	Placebo (N=5)	Zumilokibart (N=14)
≥1 TEAE	4 (80.0%)	9 (64.3%)
≥1 serious TEAE	0	0
≥1 Grade 3 or 4 TEAE	0	0
Discontinued study due to TEAE	0	0

Data cutoff date: December 23, 2025.

FeNO reduction

- Treatment with zumilokibart led to rapid suppression of FeNO, with maximal mean ± SE absolute reduction of 45.1 ± 15.7 ppb (vs 10.6 ± 7.5 ppb with placebo) and −60.2 ± 4.3% change from baseline (vs −10.2 ± 8.7% with placebo) at Week 2 after dosing (**Figure 3**).
- Mean FeNO reduction >40 ppb from baseline was maintained through Week 16 (limit of planned interim follow-up) following a single dose of zumilokibart.
- In participants with extended follow-up available (Weeks 20-32; N=2-11), FeNO suppression remained evident through Week 32.

Figure 3: Single dose of zumilokibart led to durable FeNO suppression through Week 16

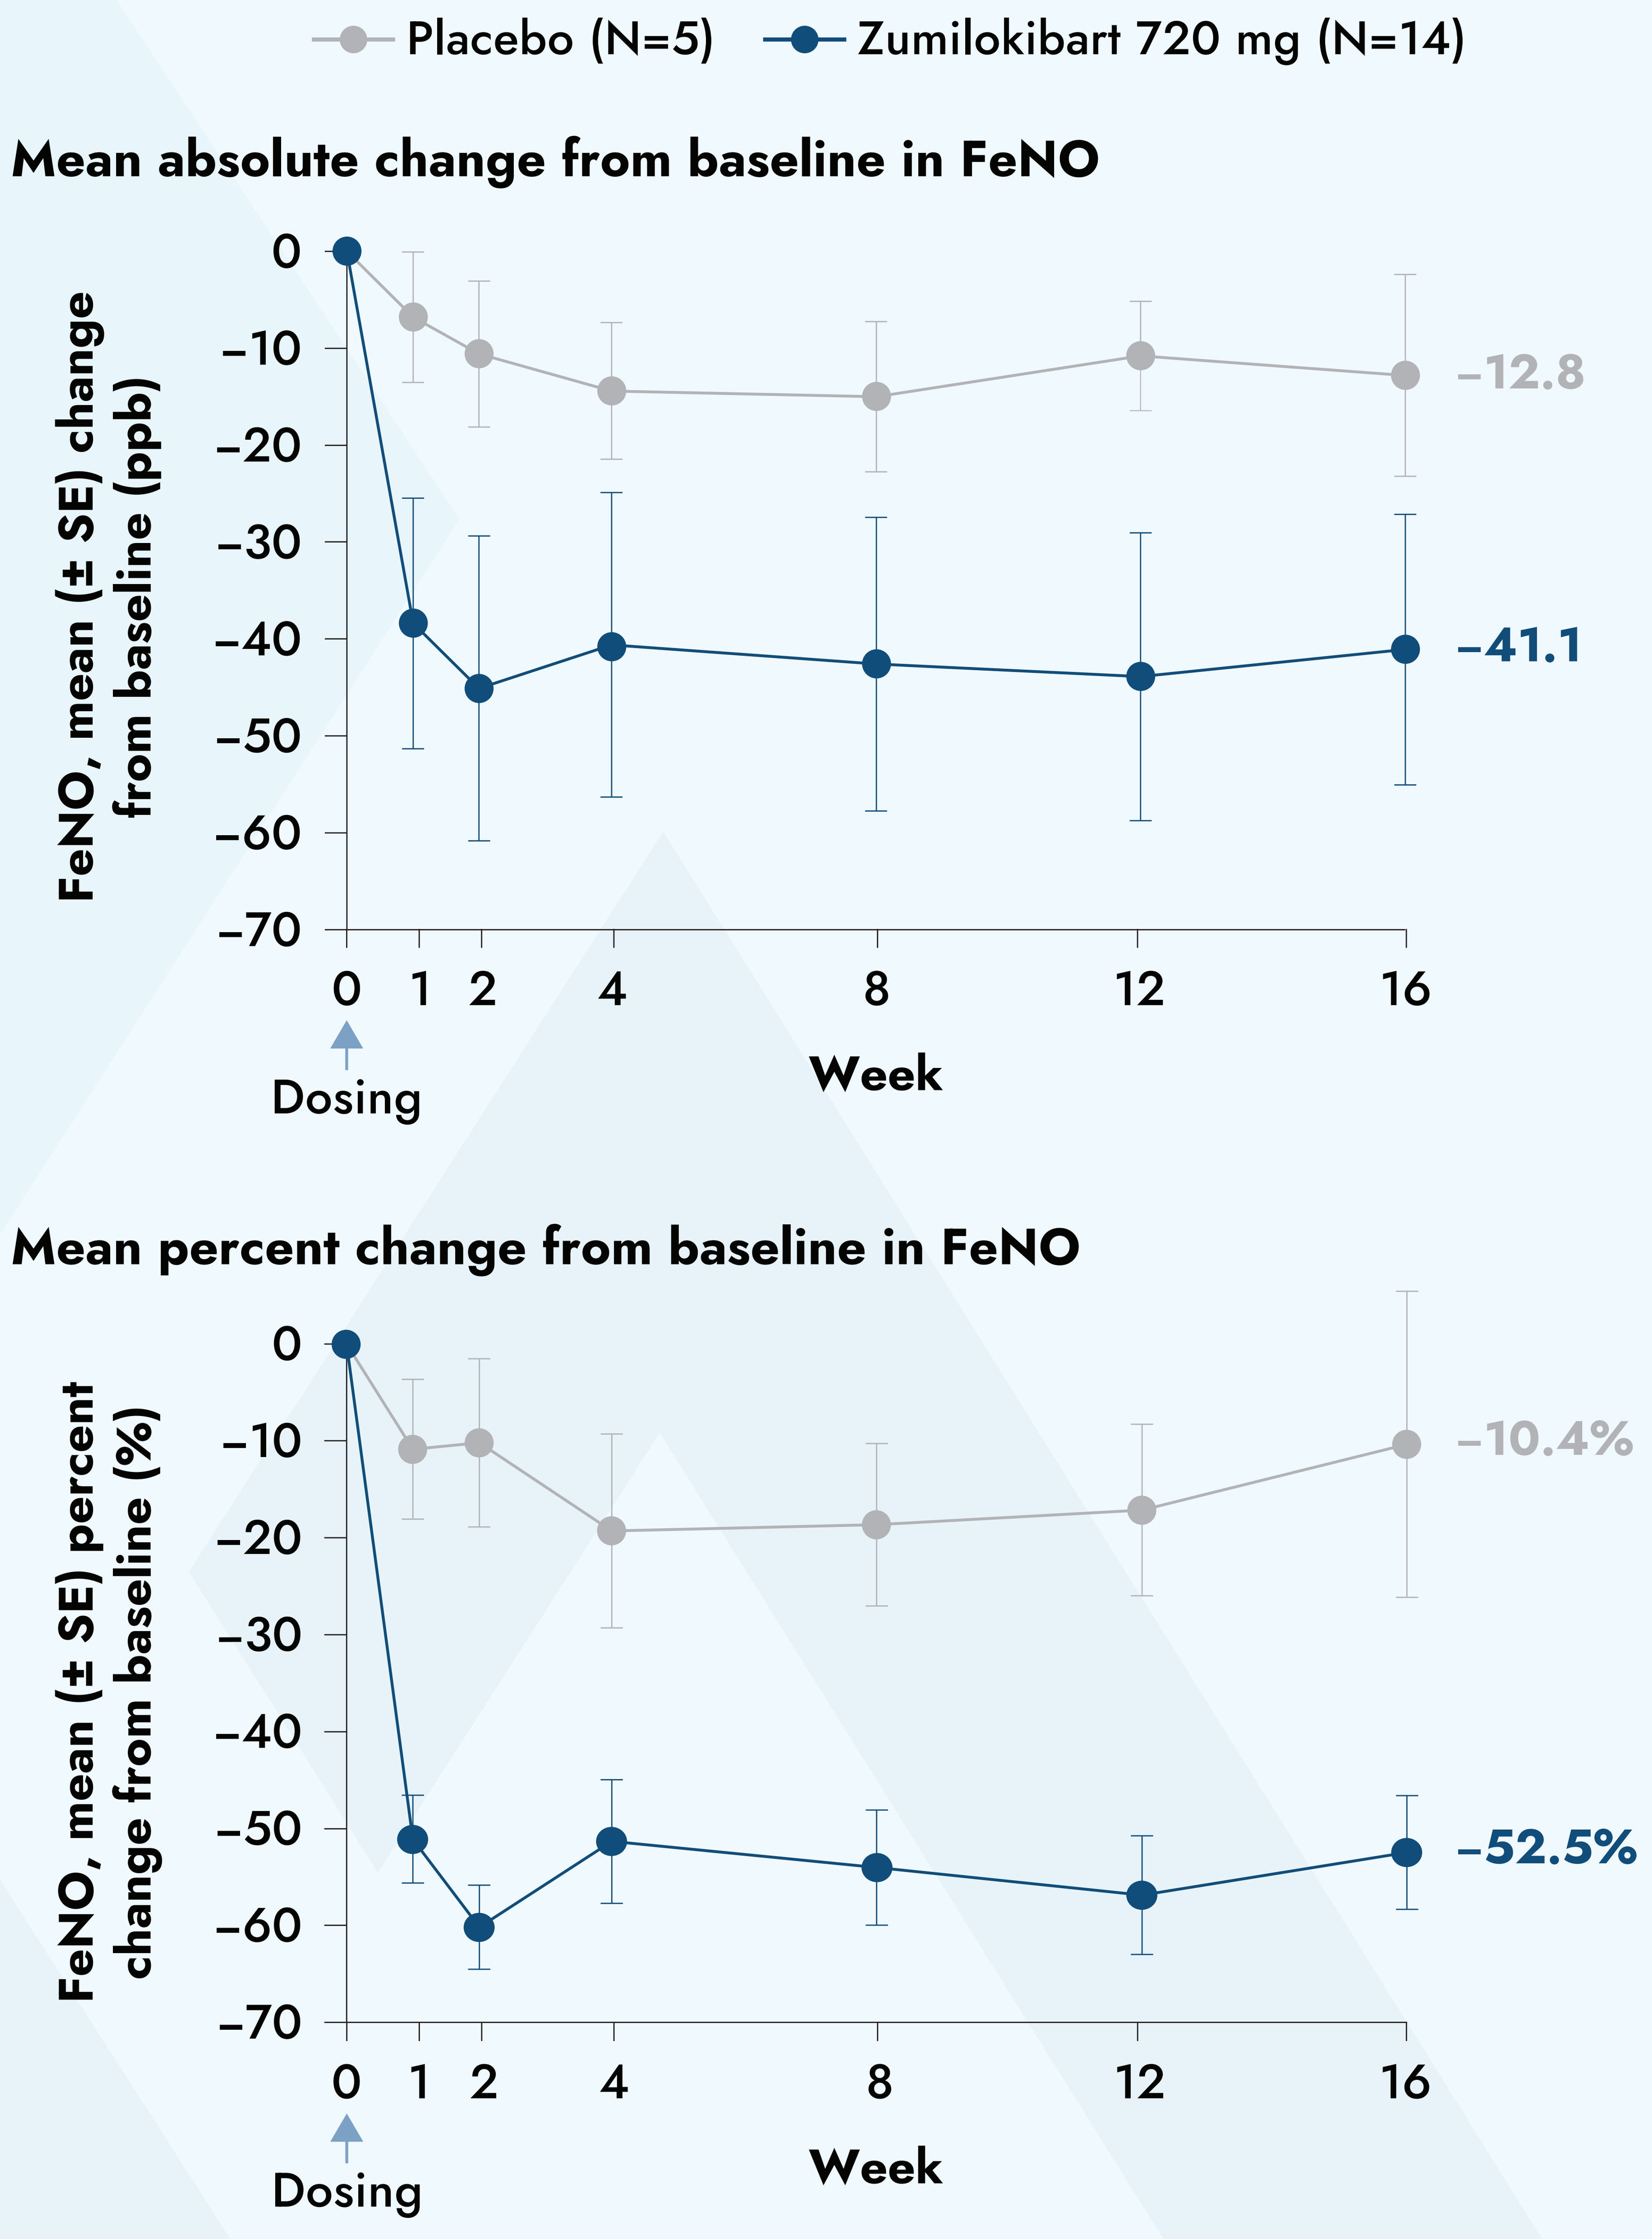
Placebo (N=5)
Zumilokibart 720 mg (N=14)
Mean absolute change from baseline in FeNO
FeNO, mean (± SE) change from baseline (ppb)
0
−10
−20
−30
−40
−50
−60
−70
0 1 2 4 8 12 16
Week
Dosing
−12.8
−41.1
Mean percent change from baseline in FeNO
FeNO, mean (± SE) percent change from baseline (%)
0
−10
−20
−30
−40
−50
−60
−70
0 1 2 4 8 12 16
Week
Dosing
−10.4%
−52.5%

One zumilokibart-treated participant discontinued following the Week 12 visit (lost to follow-up).

ABBREVIATIONS

AD, atopic dermatitis; C_{max}, maximum serum concentration; EASI, Eczema Area Severity Index; Fc, fragment crystallizable; FcRn, neonatal Fc receptor; FeNO, fractional exhaled nitric oxide; GINA, Global Initiative for Asthma; ICS, inhaled corticosteroids; IL, interleukin; LABA, long-acting beta-agonist; ppb, parts per billion; PK, pharmacokinetics; SC, subcutaneous; SD, standard deviation; SE, standard error; TEAE, treatment-emergent adverse event; YTE, triple-amino acid modification in the Fc region that extends antibody half-life.

REFERENCES

1. Mansur AH, et al. *Respir Med.* 2018;143:31-38.
2. Kraft M, et al. *Eur Respir J.* 2021;58(6):2100413.
3. Guttman-Yassky E, et al. Presented at the European Academy of Dermatology and Venerology (EADV) Congress, September 17-20, 2025; Paris, France.
4. Guttman-Yassky E, et al. Presented at the American Academy of Dermatology (AAD) Congress, March 27-31, 2026; Denver, CO, USA.
5. Ravnborg N, et al. *J Am Acad Dermatol.* 2021;84:471-478.
6. Lim XQ, et al. *Clin Transl Sci.* 2025;18:e70456.
7. Zhu E, et al. *J Allergy Clin Immunol Glob.* 2025;4:100545.

ACKNOWLEDGMENTS AND DISCLOSURES

- Apogee Therapeutics, Inc. sponsored the studies reported in this poster. Medical writing and editorial support for this poster were provided by Kate Smigiel of Apogee Therapeutics, Inc., and Kevin Corcoran and Heidi Reinholdt, both of Helios Global Group, on behalf of Apogee Therapeutics, Inc.
- **DS** has received sponsorship to attend and speak at international meetings, honoraria for lecturing or attending advisory boards from the following companies: Adovate, Almirall, Anaveon, Apogee, Arcutis Biotherapeutics, Arrowhead, AstraZeneca, Belenos Biosciences, Bial, Celldex, Chiesi, Cipla, CONNECT Biopharm, Covis, DevPro Biopharma LCC, Elpen, Empirico, EpiEndo, Generate Biomedicines, GlaxoSmithKline, Glenmark, Jasper, Kinaset Therapeutics, KOLON, Kymera, Lupin, Melodia, Menarini, MicroA, OM Pharma, OrientEuroPharma, Recipharm, Revolo, RIGImmune Inc, Roche, Roivant Sciences, Sanofi, Sitryx, Synairgen, Tetherex, UCB, Upstream, Verona Pharma, Winward, Zura Bio, and Zymeworks. **JC** and **JDA** have no relevant disclosures. **EW**, **AC**, **GW**, **MT**, **CH**, and **AK** are employees of Apogee Therapeutics, Inc. and hold company stock/stock options. **CLD** is an employee of Apogee Therapeutics, Inc. and holds company stock/stock options; serves on the Board of Directors of Oruka Therapeutics, Inc.; and holds stock/stock options in Oruka, Spyre, and Junevity. **MC** reports grants/research support from American Lung Association, AstraZeneca, Gala Therapeutics, Genentech, GSK, NIH, Nocion, Novartis, PCORI, Pfizer, Pulmatrix, Regeneron, Sanofi-Aventis; receives consulting fees from Allakos, Amgen, Apogee, Apreo Health, Arrowhead Pharmaceuticals, AstraZeneca, Blueprint Medicines, Connect BioPharma, Eli Lilly, Enveda, Evommune, Generate Biomedicines, GSK, Jasper Therapeutics, Kinaset, Kymera, Novartis, Om Pharma, Pfizer, Pioneering Medicines, Polarean, Regeneron, Sanofi-Aventis, Syneos, Teva, Third Rock Ventures, Uniquity Bio, Upstream Bio, and Verona Pharmaceuticals; receives payment for speaker's bureau activities from Amgen, AstraZeneca, MedLearningGroup, Regeneron, and Sanofi-Aventis; and receives stock options from Aer Therapeutics, less than $25 USD value.

©Apogee Therapeutics, Inc.