Early and sustained effect of zumilokibart (APG777) in atopic dermatitis and asthma: 16-week results from the phase 2 APEX study in moderate-to-severe AD and a phase 1b study in mild-to-moderate asthma

Emma Guttman-Yassky[1], Jonathan I. Silverberg[2], Melinda Gooderham[3], Andrew Blauvelt[4], Kenji Kabashima[5], Erica Winter[6], Li Xie[6], Abby Chen[6], Xiaoye Ma[6], Susanna Wen[6], Olivia Choi[6], Glenn Whelan[6], Myreen Tomas[6], Claudia Halaby[6], Amol Kamboj[6], Kristine Nograles[6], Carl L. Dambkowski[6], Jeremy Cole[7], James DeAngelo[8], Mario Castro[9], Dave Singh[10,11]

[1]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [2]George Washington University School of Medicine and Health Sciences, Washington, DC, USA; [3]SKiN Centre for Dermatology, Peterborough, Ontario, Canada and Queen's University, Kingston, Ontario, Canada; [4]University of Maryland School of Medicine, Baltimore, MD, USA; [5]Kyoto University Graduate School of Medicine, Kyoto, Japan; [6]Apogee Therapeutics, Inc., Waltham, MA, USA; [7]OK Clinical Research, LLC, Edmond, OK, USA; [8]Allergy and Clinical Immunology Associates, Pittsburgh, PA, USA; [9]University of Kansas School of Medicine, Kansas City, KS, USA; [10]Medicines Evaluation Unit, an IQVIA business, Manchester, United Kingdom; [11]University of Manchester, Manchester University NHS Foundation Trust, Manchester, United Kingdom

INTRODUCTION

- IL-13 is a key mediator of Type 2 inflammation in both atopic dermatitis (AD) and asthma.
 - In AD, IL-13 drives skin barrier dysfunction and the persistence of itch associated with the disease.[1]
 - In asthma, IL-13 promotes increased expression of fractional exhaled nitric oxide (FeNO), which is strongly linked to exacerbations and impaired lung function.[2,3]
- Asthma is a common comorbidity among patients with AD, with 1 meta-analysis reporting that 28% of patients with moderate-to-severe AD have comorbid asthma.[4]
- Zumilokibart (APG777) is a novel, half-life-extended anti-IL-13 monoclonal antibody that achieves early peak therapeutic drug levels that are sustained over an extended dosing interval.
- Zumilokibart is currently being investigated in:
 - The 2-part phase 2 APEX study (NCT06395948) in participants with moderate-to-severe AD.
 - A phase 1b study (NCT06920901) in participants with mild-to-moderate asthma.

OBJECTIVE

To assess the speed and durability of effect with zumilokibart from separate clinical trials in participants with moderate-to-severe AD (APEX Part A) and mild-to-moderate asthma (phase 1b)

- In a first-in-human phase 1 study, the half-life of zumilokibart ranged from 75.3 to 77.5 days across the doses tested.[5]
- The extended half-life of zumilokibart is conferred by a YTE amino acid modification in the Fc region, enhancing FcRn-mediated antibody recycling (**Figure 1**).[6]
- The pharmacokinetics of zumilokibart support evaluation of every 12-week and 24-week dosing.

Figure 1: YTE modification extends the half-life of zumilokibart

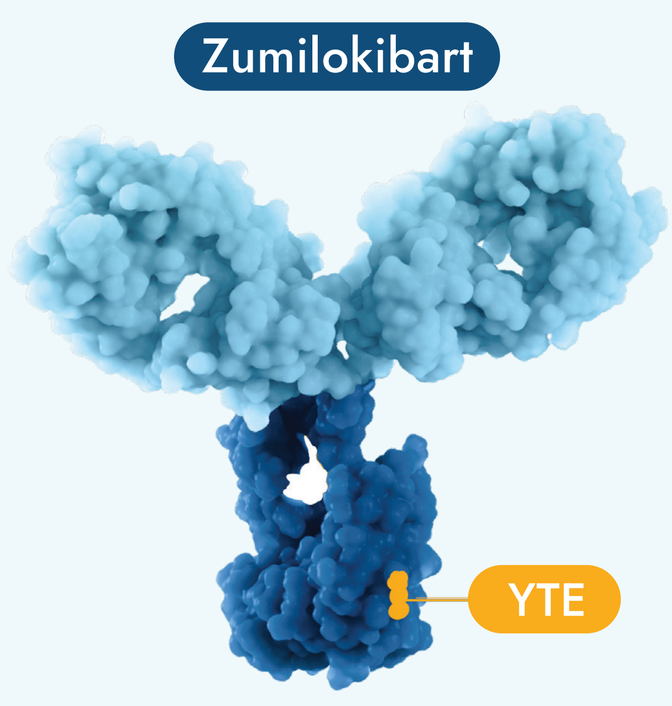
Zumilokibart
YTE

CONCLUSIONS

- Treatment with zumilokibart led to early and sustained improvements in markers of disease activity in both AD and asthma:
 - In AD, treatment with zumilokibart resulted in significant improvements in AD lesions by Week 2 and itch by 48 hours after the first dose. Both every 12- and 24-week maintenance dosing with zumilokibart led to sustained responses through Week 52.
 - In asthma, a single dose of zumilokibart led to rapid suppression of FeNO that was maintained through 16 weeks in all participants and through 32 weeks for participants with available follow-up.
- Zumilokibart was well tolerated in both studies, with safety results in line with expectations for therapies targeting IL-13.
- These results support continued investigation of zumilokibart as a novel therapy for AD and asthma with the potential to reduce injection burden for patients.

Poster number: D3.164

Scan to access Digital Poster

For questions, please contact MedAffairs@apogeetherapeutics.com

Presented at the European Academy of Allergy and Clinical Immunology (EAACI) Annual Scientific Meeting, Istanbul, Turkey, 12–15 June, 2026

RESULTS

Phase 2 APEX Part A in participants with moderate-to-severe AD

Study design & methods

- APEX Part A evaluated zumilokibart in adults (≥18 years) with moderate-to-severe AD (EASI ≥16, vIGA-AD score ≥3, BSA ≥10%; **Figure 2**).
- At baseline, participants (N=123) were randomized 2:1 to an induction regimen of zumilokibart or placebo. Upon completing Week 16, participants initially randomized to zumilokibart (N=72) were rerandomized 1:1 to receive zumilokibart Q12W or Q24W during maintenance.
- The primary endpoint of percent change from baseline in EASI was evaluated at Week 16. Efficacy and safety were evaluated through Week 52.

Figure 2: Design of the phase 2 APEX Part A study in moderate-to-severe AD

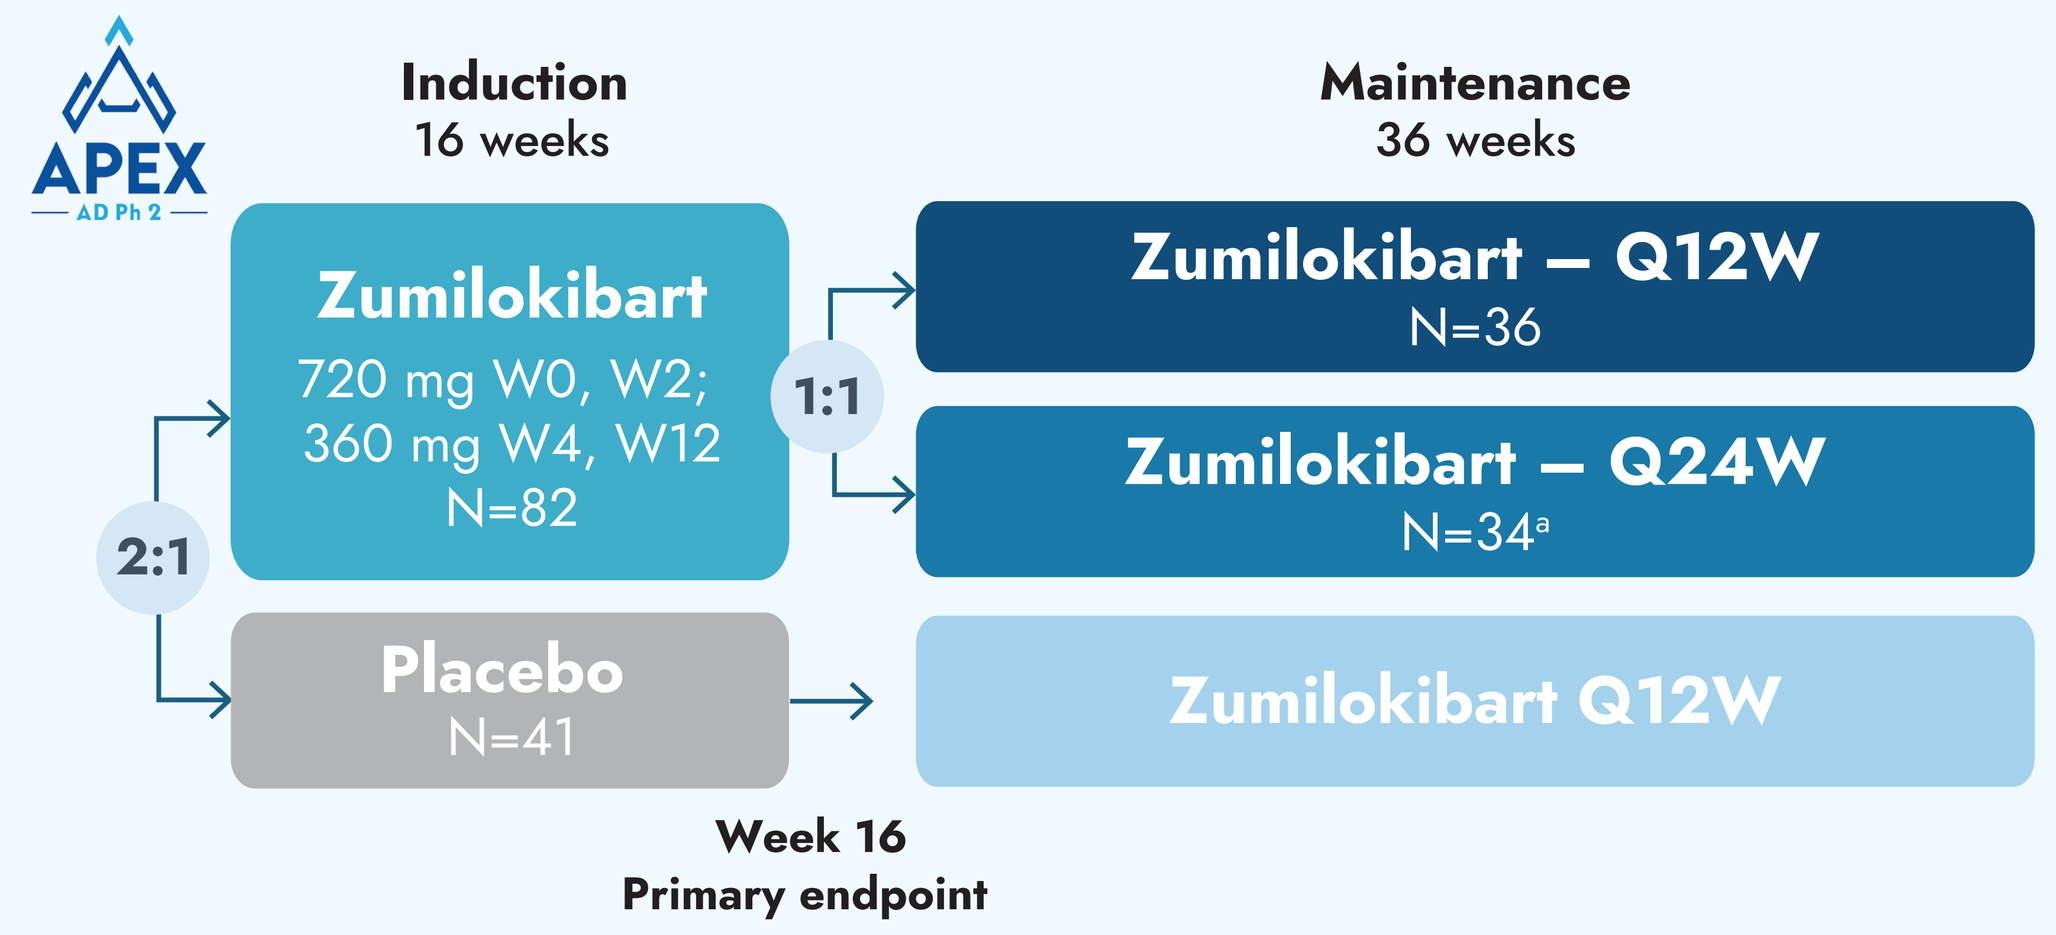
APEX
AD Ph 2
Induction
16 weeks
Maintenance
36 weeks
2:1
Zumilokibart
720 mg W0, W2;
360 mg W4, W12
N=82
1:1
Zumilokibart – Q12W
N=36
Zumilokibart – Q24W
N=34a
Placebo
N=41
Zumilokibart Q12W
Week 16
Primary endpoint

Q12W: 360 mg zumilokibart at Weeks 24, 36, and 48; Q24W: 360 mg at Weeks 24 and 48.
[a]Excludes 2 participants who discontinued the maintenance period before receiving zumilokibart.
For induction, N = number of participants randomized and received ≥1 dose of study drug during induction. For maintenance, N = number randomized to zumilokibart during induction and received ≥1 dose of study drug during maintenance.

Baseline characteristics

- Baseline demographics and disease characteristics were generally comparable between groups and in line with expectations for a population with moderate-to-severe AD.

Primary and key secondary endpoints at Week 16

- APEX Part A met the primary endpoint at Week 16. The reduction from baseline in EASI was significantly greater with zumilokibart than placebo at Week 2 (−35.4% vs −20.8%, respectively; p=0.01) and all subsequent timepoints through Week 16 (**Table 1**).
- A significantly greater percentage of participants achieved ≥75% improvement in EASI (EASI-75) with zumilokibart than placebo as early as Week 4, and approximately two-thirds of participants treated with zumilokibart achieved EASI-75 at Week 16 (**Table 1**).
- Treatment with zumilokibart led to significant improvements in secondary endpoints, including percentages of participants achieving EASI-90 and vIGA-AD 0/1 responses at Week 16 (**Table 1**).

Table 1: Primary and secondary endpoints were significant at Week 16

Efficacy endpoints at Week 16	Placebo (N=41)	Zumilokibart (N=82)	P value
Primary outcome, LS mean % change from baseline in EASI	−33.8	−71.0	<0.001
Secondary outcomes			
EASI-75, %	24.6	66.9	<0.001
EASI-90, %	14.7	33.9	<0.05
vIGA-AD 0/1, %	17.3	34.9	<0.05

P values vs placebo were determined using ANCOVA (for continuous variables) and CMH (for categorical variables). vIGA-AD 0/1 response was defined as achieving a vIGA-AD score of 0 or 1 with a ≥2-point reduction from baseline. For EASI % change from baseline, missing data were imputed with Markov Chain Monte Carlo Multiple Imputation (MCMC-MI), including the data set to missing after rescue medication use or study treatment discontinuation. For EASI-75, EASI-90, and vIGA-AD, missing data were imputed with MCMC-MI, and rescue medication use or treatment discontinuation due to lack of efficacy was imputed as nonresponder for all subsequent timepoints.

Efficacy through Week 52

- Among participants rerandomized to continue zumilokibart during maintenance, 87.5% and 80.6% of those in the Q12W and Q24W arms, respectively, achieved EASI-75 at Week 52 (**Figure 3**).

Participant-reported itch

- Significant improvement in itch, as measured by the LS mean % change from baseline in I-NRS,[a] was observed 48 hours after the first dose of zumilokibart (corresponding to study Day 3).
 - Progressive improvement in itch continued through Week 16 (−50.7% zumilokibart vs −23.2% placebo; p=0.001). At Week 48, participants in the Q12W and Q24W arms reached −72.5% and −64.1% change from baseline in I-NRS.

[a]Daily I-NRS results in Week 1 and I-NRS results at Week 48 were evaluated as-observed without imputation for missing data or rescue medication use. For I-NRS results at Week 16, missing data were imputed with MCMC-MI.

Figure 3: Progressive improvement in EASI-75 response over 52 weeks

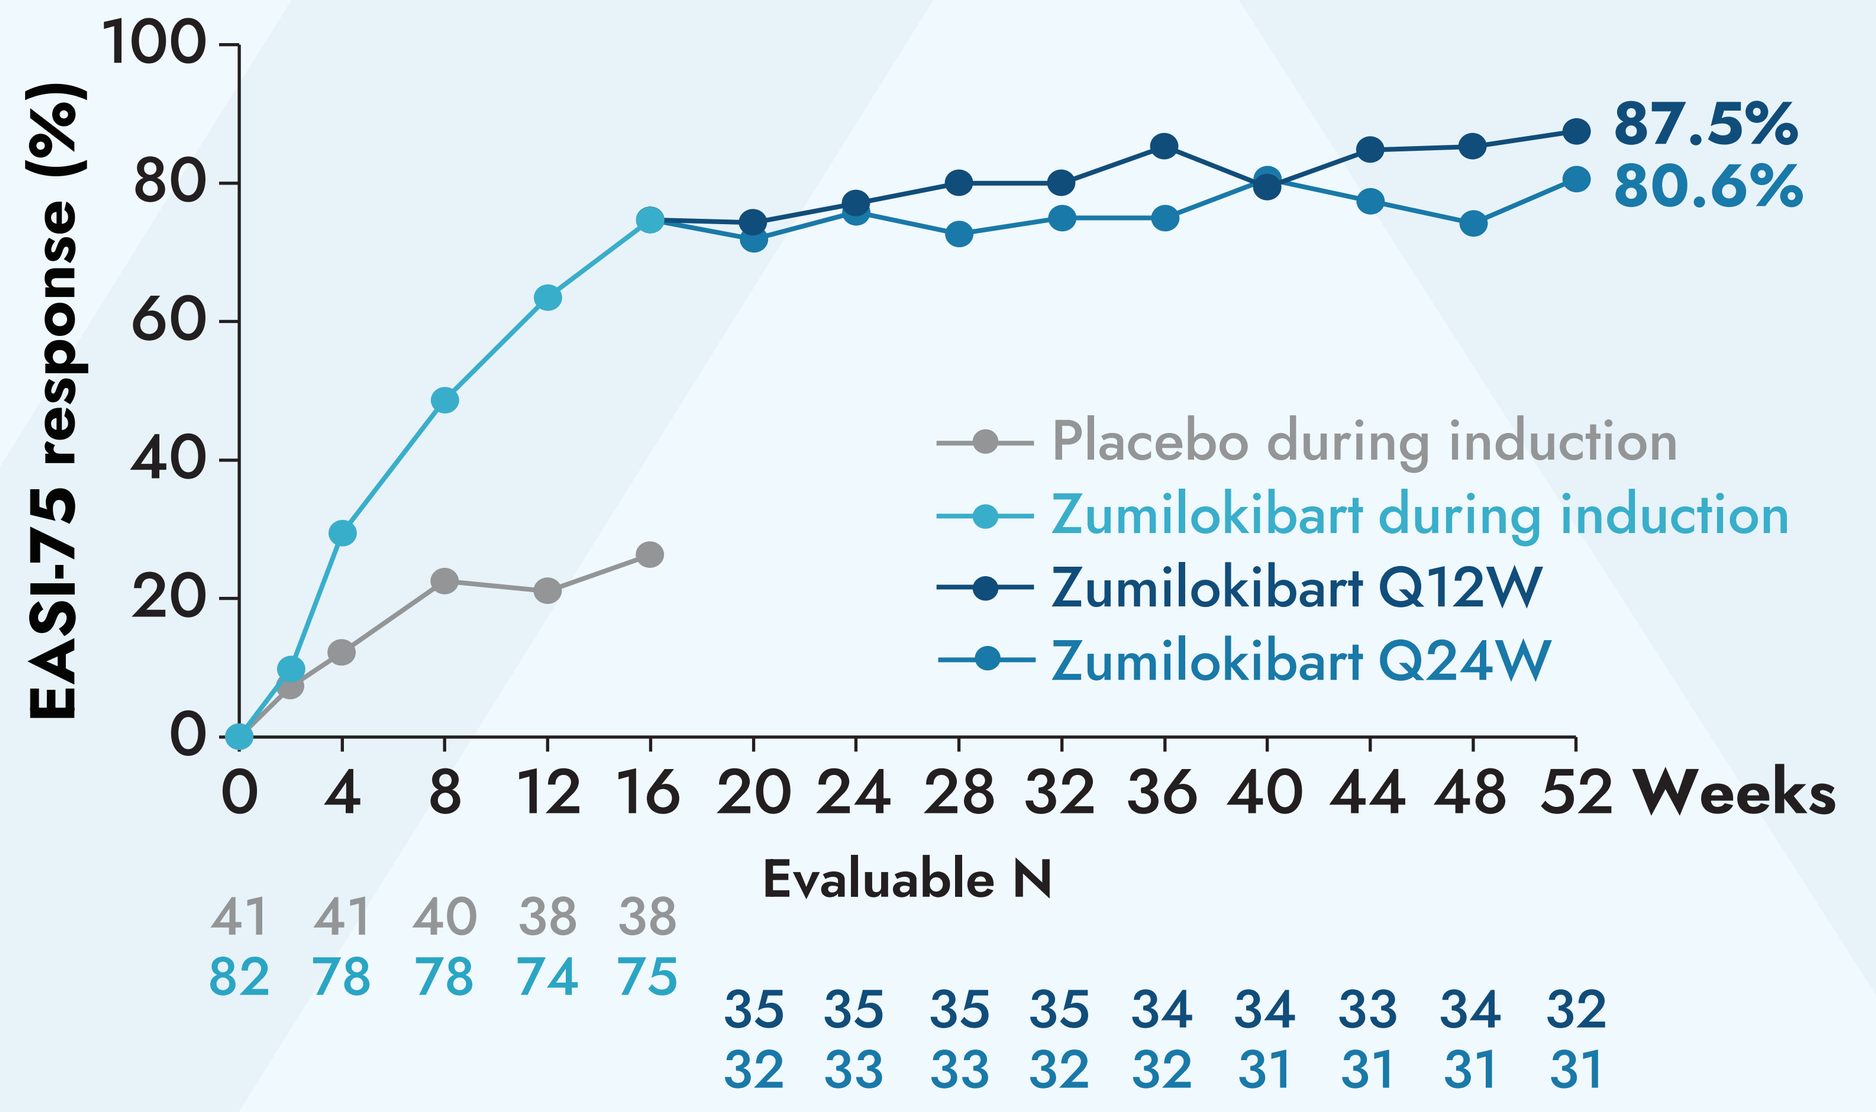
EASI-75 response (%)
100
80
60
40
20
0
0 4 8 12 16 20 24 28 32 36 40 44 48 52 Weeks
87.5%
80.6%
Placebo during induction
Zumilokibart during induction
Zumilokibart Q12W
Zumilokibart Q24W
Evaluable N
41 41 40 38 38
82 78 78 74 75
35 35 35 35 34 34 33 34 32
32 33 33 32 32 31 31 31 31

For induction, N = number of participants randomized and received ≥1 dose of study drug in induction. For maintenance, N = number randomized to zumilokibart in induction and received ≥1 dose of study drug in maintenance. Efficacy data were evaluated as-observed without imputation for missing data or rescue medication use.

Safety

- Treatment with zumilokibart was well tolerated during the 52-week treatment period (**Table 2**).

Table 2: Safety summary through Week 52

Event, n (%)	Zumilokibart mITT (N=119) (All participants receiving ≥1 dose of zumilokibart)[a]
≥1 adverse event	85 (71.4)
≥1 serious adverse event	1 (0.8)[b]
Adverse events leading to discontinuation through Week 52	4 (3.4)
Deaths	0
Most common adverse events through Week 52 (≥5%)	
Noninfective conjunctivitis[c]	16 (13.4)
Upper respiratory tract infection	15 (12.6)
Nasopharyngitis	11 (9.2)
Dermatitis atopic	6 (5.0)

[a]Safety data are summarized for all randomized participants who received ≥1 dose of zumilokibart (N=82 randomized to zumilokibart and N=37 who crossed over from placebo to zumilokibart at Week 16). [b]Case of Sézary syndrome; not considered related to study treatment. [c]The total conjunctivitis rate was 20.2%, including the following preferred terms: noninfective conjunctivitis, conjunctivitis allergic, conjunctivitis bacterial and conjunctivitis viral, with <1% discontinuations over the 52-week treatment period. Most cases were mild to moderate, and the median duration of conjunctivitis was 27.5 days. No relationship between zumilokibart exposure and conjunctivitis was observed.

Phase 1b study in participants with mild-to-moderate asthma

Study design & methods

- This phase 1b study evaluated zumilokibart in adults (18-65 years) with mild-to-moderate asthma (GINA 2023 criteria) and elevated FeNO (≥25 ppb; **Figure 4**).
- Participants (N=19) were randomized 3:1 to receive a single dose of zumilokibart 720 mg or placebo.
- The primary objectives were to evaluate safety and the change in FeNO as a surrogate marker of treatment response in Type 2 asthma.

Figure 4: Design of the single-dose zumilokibart phase 1b study in mild-to-moderate asthma

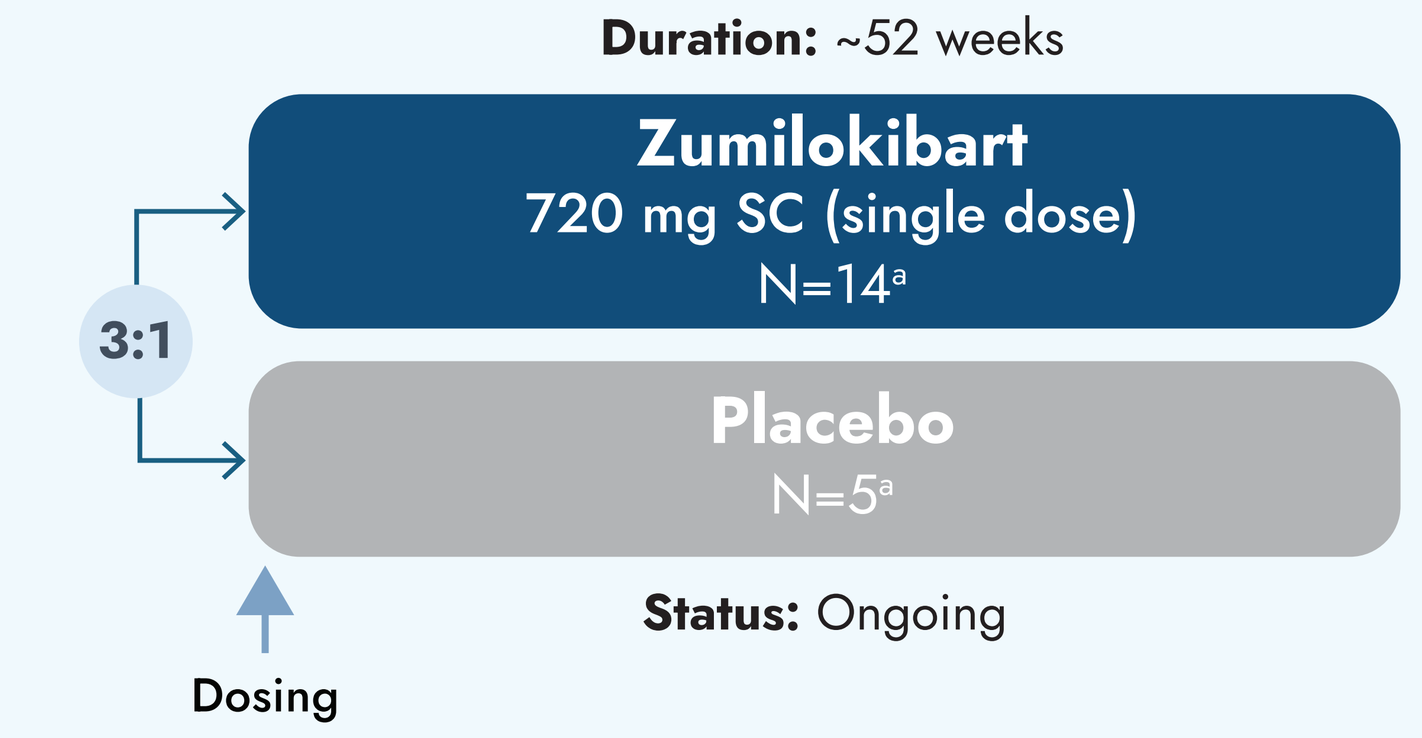
Duration: ~52 weeks
3:1
Zumilokibart
720 mg SC (single dose)
N=14a
Placebo
N=5a
Status: Ongoing
Dosing

[a]Study enrolled a total of 31 participants; 19 met the prespecified criteria for the subgroup with FeNO ≥25 ppb, representative of asthma with Type 2 inflammation. This study reports an interim analysis with a data cutoff date of December 23, 2025.

Baseline characteristics

- Interim results were available through Week 16 for all participants; this analysis included 19 adults (mean age 44.5 years; 42.1% female) with mild-to-moderate asthma and FeNO ≥25 ppb.
- Baseline demographics and FeNO levels were generally similar between treatment groups.

FeNO reduction

- Treatment with zumilokibart led to rapid suppression of FeNO, with maximal mean ± SE absolute reduction from baseline of 45.1 ± 15.7 ppb (vs 10.6 ± 7.5 ppb with placebo) and −60.2 ± 4.3% change from baseline to Week 2 after dosing (vs −10.2 ± 8.7% with placebo; **Figure 5**).
- Mean FeNO reduction of >40 ppb from baseline was maintained through Week 16 (limit of planned interim follow-up) after a single dose of zumilokibart.
- In participants with extended follow-up available (Weeks 20-32; N=2-11), FeNO suppression remained evident through Week 32.

Figure 5: Single dose of zumilokibart led to durable FeNO suppression through Week 16

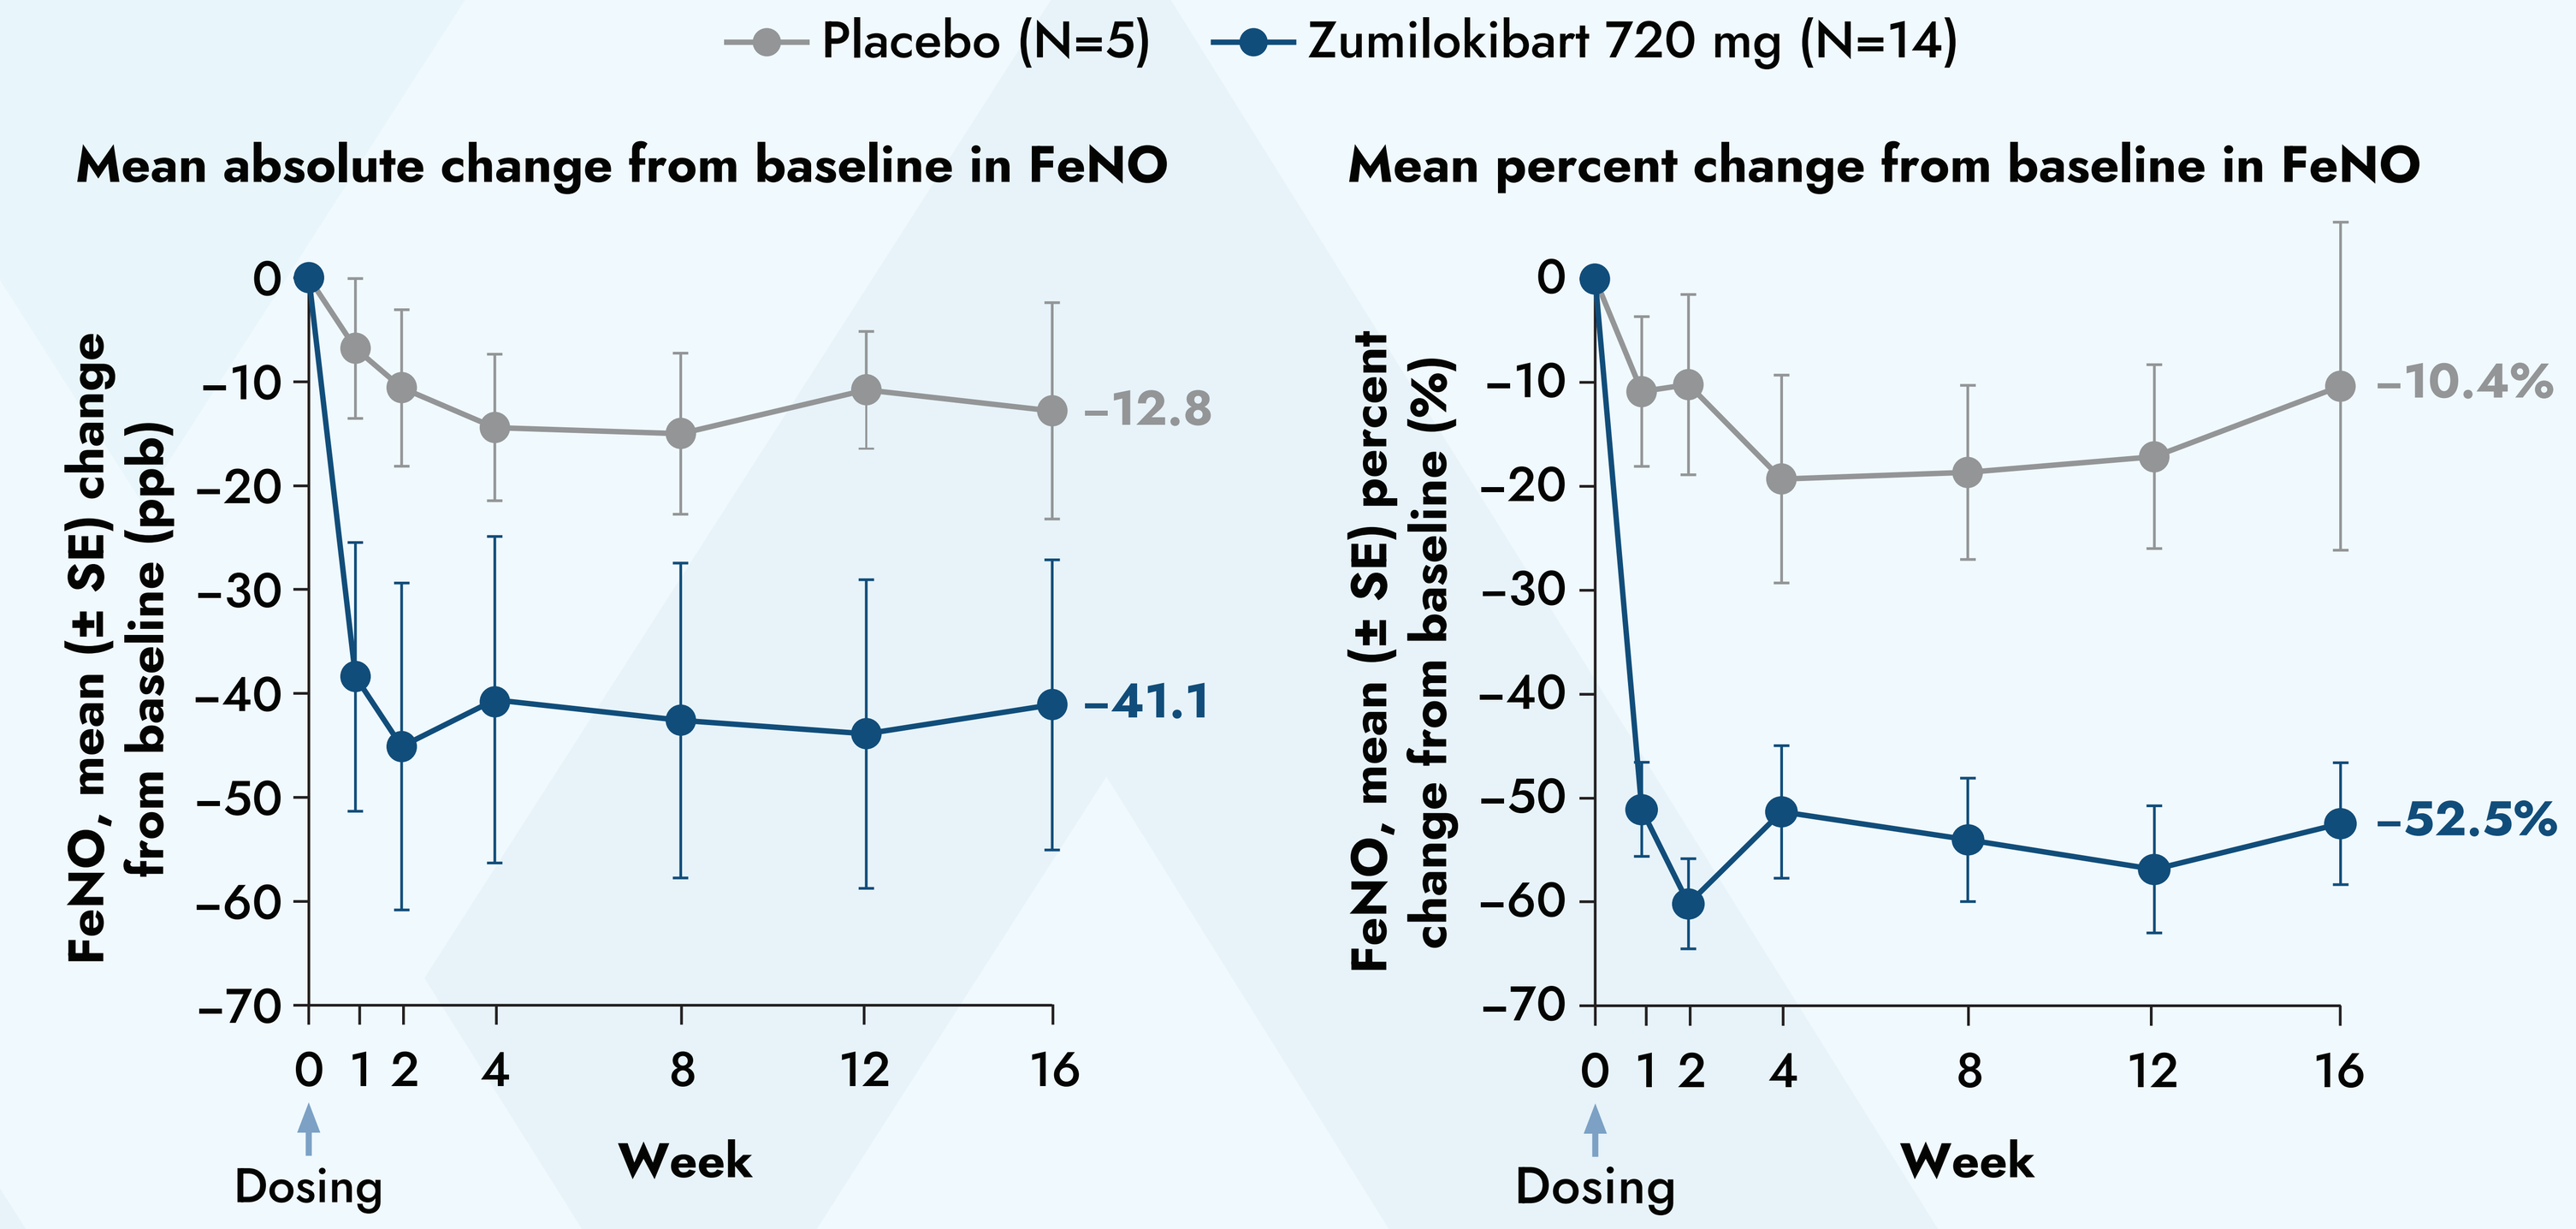
Placebo (N=5)
Zumilokibart 720 mg (N=14)
Mean absolute change from baseline in FeNO
FeNO, mean (± SE) change from baseline (ppb)
0
−10
−20
−30
−40
−50
−60
−70
−12.8
−41.1
0 1 2 4 8 12 16
Dosing
Week
Mean percent change from baseline in FeNO
FeNO, mean (± SE) percent change from baseline (%)
−10.4%
−52.5%

Mean ± SD FeNO levels at baseline were 57.2 ± 34.1 ppb in the placebo group and 66.9 ± 65.3 ppb in the zumilokibart group. One zumilokibart-treated participant discontinued following the Week 12 visit (lost to follow-up).

Safety

- Zumilokibart was well tolerated, with no serious grade 3 or 4 TEAEs (**Table 3**), and a safety profile consistent with expectations for an IL-13-targeted therapy in asthma.

Table 3: Safety summary from the phase 1b study in asthma

Event, n (%)	Placebo (N=5)	Zumilokibart (N=14)
≥1 adverse event	4 (80.0)	9 (64.3)
≥1 serious adverse event	0	0
≥1 Grade 3 or 4 adverse event	0	0
Adverse events leading to discontinuation	0	0

Data cutoff date: December 23, 2025.

REFERENCES

1. Guttman-Yassky E, et al. *Lancet.* 2025;405:583-596.
2. Mansur AH, et al. *Respir Med.* 2018;143:31-38.
3. Kraft M, et al. *Eur Respir J.* 2021;58(6):2100413.
4. Ravnborg N, et al. *J Am Acad Dermatol.* 2021;84:471-478.
5. Lim XQ, et al. *Clin Transl Sci.* 2025;18:e70456.
6. Zhu E, et al. *J Allergy Clin Immunol Glob.* 2025;4:100545.

ABBREVIATIONS

AD, atopic dermatitis; ANCOVA, analysis of covariance; BSA, body surface area; CMH, Cochran-Mantel-Haenszel test; EASI, Eczema Area Severity Index; EASI-75, achieving at least 75% reduction from baseline in EASI; EASI-90, achieving at least 90% reduction from baseline in EASI; Fc, fragment crystallizable; FcRn, neonatal Fc receptor; FeNO, fractional exhaled nitric oxide; GINA, Global Initiative for Asthma; IL, interleukin; I-NRS, Itch Numeric Rating Scale; LS, least squares; MCMC-MI, Markov Chain Monte Carlo Multiple Imputation; mITT, modified intent-to-treat; ppb, parts per billion; Q12W, every 12 weeks; Q24W, every 24 weeks; SC, subcutaneous; SD, standard deviation; SE, standard error; TEAE, treatment-emergent adverse event; vIGA-AD, Validated Investigator Global Assessment for AD; YTE, triple-amino acid modification in the Fc region that extends antibody half-life.

ACKNOWLEDGMENTS

Apogee Therapeutics, Inc. sponsored the study reported in this poster. Medical writing and editorial support were provided by Kate Smigiel of Apogee Therapeutics, Inc., and Kevin Corcoran and Heidi Reinholdt, both of Helios Global Group, on behalf of Apogee Therapeutics, Inc.

DISCLOSURES

EG-Y is an employee of Mount Sinai and has received research grants (paid to the institution) from and/or is a consultant for: AbbVie, Aclaris Therapeutics, Almirall, Alumis, Amgen, AnaptysBio, Apogee Therapeutics, Apollo Therapeutics, Arcutis, Artax Biopharma, Astria, Boehringer Ingelhiem, BMS, Celldex, Centrexion Therapeutics, Connect Biopharm, Concerto Biosciences, Coty, DBV, Dualitas Therapeutics, Eli Lilly, Enveda Biosciences, Escient Pharmaceuticals, Galderma, Gate Bio, GSK, GSK Immunology, Incyte, Inmagene, Janssen Biotech, Jasper Therapeutics, Kymera Therapeutics, Kyowa Kirin, LEO Pharma, Matchpoint Therapeutics, Merck, Nektar Therapeutics, NUMAB Therapeutics, Obsidio, Otsuka, Pfizer, Pharmaxis, Proteologix, Q32 Bio, RAPT, RayThera, Regeneron, Ribon Therapeutics, Sanofi, SATO, Schrödinger, Sitryx, Sun Pharma, Takeda, Teva, TRex Bio, UCB, VRG Therapeutics, and Xencor. **JIS** has received honoraria as a consultant and/or advisory board member for AbbVie, Aldena, Amgen, AObiome, Apollo, Arcutis, Arena, Attovia, Boehringer Ingelheim, BMS, Castle Biosciences, Connect Biopharma, Corevitas, Dermavant, Eli Lilly, FIDE, Formation Bio, Galderma, GSK, Immunocore, Incyte, Inmagene, Invea, Leo Pharma, Merck, Nektar, Novartis, Pfizer, RAPT, Recludix, Regeneron, Sandoz, Sanofi Genzyme, Shaperon, TARGET-RWE, Teva, Triveni, UCB, Union, and UpToDate; speaker for AbbVie, Arcutis, Dermavant, Eli Lilly, Galderma, LEO Pharma, Pfizer, Regeneron, and Sanofi Genzyme; institution received grants from Galderma, Incyte, and Pfizer. **MG** has served as an investigator, speaker and/or advisor for AbbVie, Acelyrin, Alumis, Amgen, Akros, Arcutis, Aristea, AnaptysBio, Apogee Therapeutics, Bausch Health, BMS, Boehringer Ingelheim, Dermira, Dermavant, Eli Lilly, Galderma, GSK, Incyte, Inmagene, JAMP Pharma, Janssen, LEO Pharma, L'Oreal, MedImmune, Meiji, Moonlake, Nektar, Nimbus, Novartis, Organon, Oruka, Pfizer, Q32 Bio, Regeneron, Sanofi Genzyme, Sun Pharma, Tarsus, Takeda, UCB, Union, Ventyx, and Vyne. **AB** has served as a speaker (received honoraria) for Almirall, Eli Lilly, Leo, Sanofi, and UCB; has served as a scientific adviser (received honoraria) for AbbVie, Almirall, Alumis, Amgen, Anaptysbio, Apogee Therapeutics, Arcutis, Eli Lilly, Incyte, Janssen, Khanda Therapeutics, LEO Pharma, Novartis, Oruka, Pfizer, Regeneron, Sanofi, Sun Pharma, Takeda, and UCB; and owns stock in Lipidio and Oruka. **KK** has received consulting fees, honoraria, grant support, and/or lecture fees from AbbVie, Amgen, Apogee Therapeutics, Inc., Eli Lilly, Kyowa Kirin, Japan Tobacco, LEO Pharma, Maruho, Mitsubishi Tanabe, Ono Pharmaceutical, Pfizer, Procter & Gamble, Sanofi, Taiho, and Torii Pharmaceutical. **EW**, **LX**, **AC**, **XM**, **SW**, **OC**, **GW**, **MT**, **CH**, **AK**, and **KN** are employees of Apogee Therapeutics, Inc. and hold company stock/stock options. CLD is an employee of Apogee Therapeutics, Inc. and holds company stock/stock options; serves on the Board of Directors of Oruka Therapeutics, Inc.; and holds stock/stock options in Oruka, Spyre, and Junevity. **JC** and **JD** have no relevant disclosures. **MC** has received grants/research support from American Lung Association, AstraZeneca, Gala Therapeutics, Genentech, GSK, NIH, Nocion, Novartis, PCORI, Pfizer, Pulmatrix, Regeneron, Sanofi-Aventis; consulting fees from Allakos, Amgen, Apogee Therapeutics, Apreo Health, Arrowhead Pharmaceuticals, AstraZeneca, Blueprint Medicines, Connect BioPharma, Eli Lilly, Enveda, Evommune, Generate Biomedicines, GSK, Jasper Therapeutics, Kinaset, Kymera, Novartis, Om Pharma, Pfizer, Pioneering Medicines, Polarean, Regeneron, Sanofi-Aventis, Syneos, Teva, Third Rock Ventures, Uniquity Bio, Upstream Bio, and Verona Pharmaceuticals; payment for speaker's bureau activities from Amgen, AstraZeneca, MedLearningGroup, Regeneron, and Sanofi-Aventis; and stock options from Aer Therapeutics, less than $25 USD value. **DS** has received sponsorship to attend and speak at international meetings, honoraria for lecturing or attending advisory boards from the following companies: Adovate, Almirall, Anaveon, Apogee Therapeutics, Arcutis, Arrowhead, AstraZeneca, Belenos Biosciences, Bial, Celldex, Chiesi, Cipla, Connect Biopharm, Covis, DevPro Biopharma LCC, Elpen, Empirico, EpiEndo, Generate Biomedicines, GSK, Glenmark, Jasper, Kinaset, KOLON, Kymera, Lupin, Melodia, Menarini, MicroA, OM Pharma, OrientEuroPharma, Recipharm, Revolo, RIGImmune Inc, Roche, Roivant Sciences, Sanofi, Sitryx, Synairgen, Tetherex, UCB, Upstream, Verona Pharma, Winward, Zura Bio, and Zymeworks.

©Apogee Therapeutics, Inc.