Zumilokibart (APG777) provides robust, progressive improvement in the signs and symptoms of atopic dermatitis: 52-week results from the phase 2 APEX Part A study

Emma Guttman-Yassky[1], Andrew Blauvelt[2], Melinda Gooderham[3], Kenji Kabashima[4], Susanna Wen[5], Olivia Choi[5], Jiahong Xu[5], Cindy Wu[5], Amol Kamboj[5], Carl L. Dambkowski[5], Kristine Nograles[5], Jonathan I. Silverberg[6]

[1]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [2]University of Maryland School of Medicine, Baltimore, MD, USA; [3]SKiN Centre for Dermatology, Peterborough, Ontario, Canada and Queen's University, Kingston, Ontario, Canada; [4]Kyoto University Graduate School of Medicine, Kyoto, Japan; [5]Apogee Therapeutics, Inc., Waltham, MA, USA; [6]George Washington University School of Medicine and Health Sciences, Washington, DC, USA

Scan to access digital poster and supplemental material

INTRODUCTION

- Optimal management of atopic dermatitis (AD) aims to achieve treatment targets in both clinician-reported outcomes, such as skin clearance, and patient-reported outcomes, such as itch.[1]
- Increased expression of IL-13 drives both skin barrier dysfunction and persistent itch in patients with AD.[2,3]
- Zumilokibart (APG777) is a novel, half-life-extended anti-IL-13 monoclonal antibody that achieves early peak and sustained drug exposures, which allows for only 4 dosing days during induction and 2 or 4 dosing days per year in maintenance.[4,5]
- The ~77-day half life of zumilokibart is conferred through a YTE amino acid modification in the Fc region that increases FcRn-mediated antibody recycling.[4,5]
- The phase 2 APEX Part A study (NCT06395948) met the primary endpoint at Week 16, with significant improvements in AD lesions by Week 2 and itch by 48 hours after the first dose of zumilokibart, in patients with moderate-to-severe AD.[6]
 - Both every 12- and 24-week maintenance dosing with zumilokibart led to durable maintenance and deepening of response across all lesional endpoints through Week 52.[7]

STUDY OBJECTIVE

To evaluate achievement of clinician- and patient-reported treatment thresholds with zumilokibart administered every 12 or 24 weeks during maintenance over 52 weeks in the APEX Part A study.

STUDY DESIGN & METHODS

- The 2-part phase 2 APEX study is evaluating zumilokibart in adults (≥18 years) with moderate-to-severe AD (EASI ≥16, vIGA-AD score ≥3, BSA ≥10%).
- APEX Part A included a total of 123 biologic-naïve participants (**Figure 1**).
- This analysis reports prespecified clinician-reported and patient-reported outcomes through Week 52 of APEX Part A (**Figure 2**).
- All analyses were conducted as-observed. Missing data were not imputed; all available data were used regardless of rescue medications or study drug discontinuation.

Figure 1: Design of the phase 2 APEX Part A study in moderate-to-severe AD

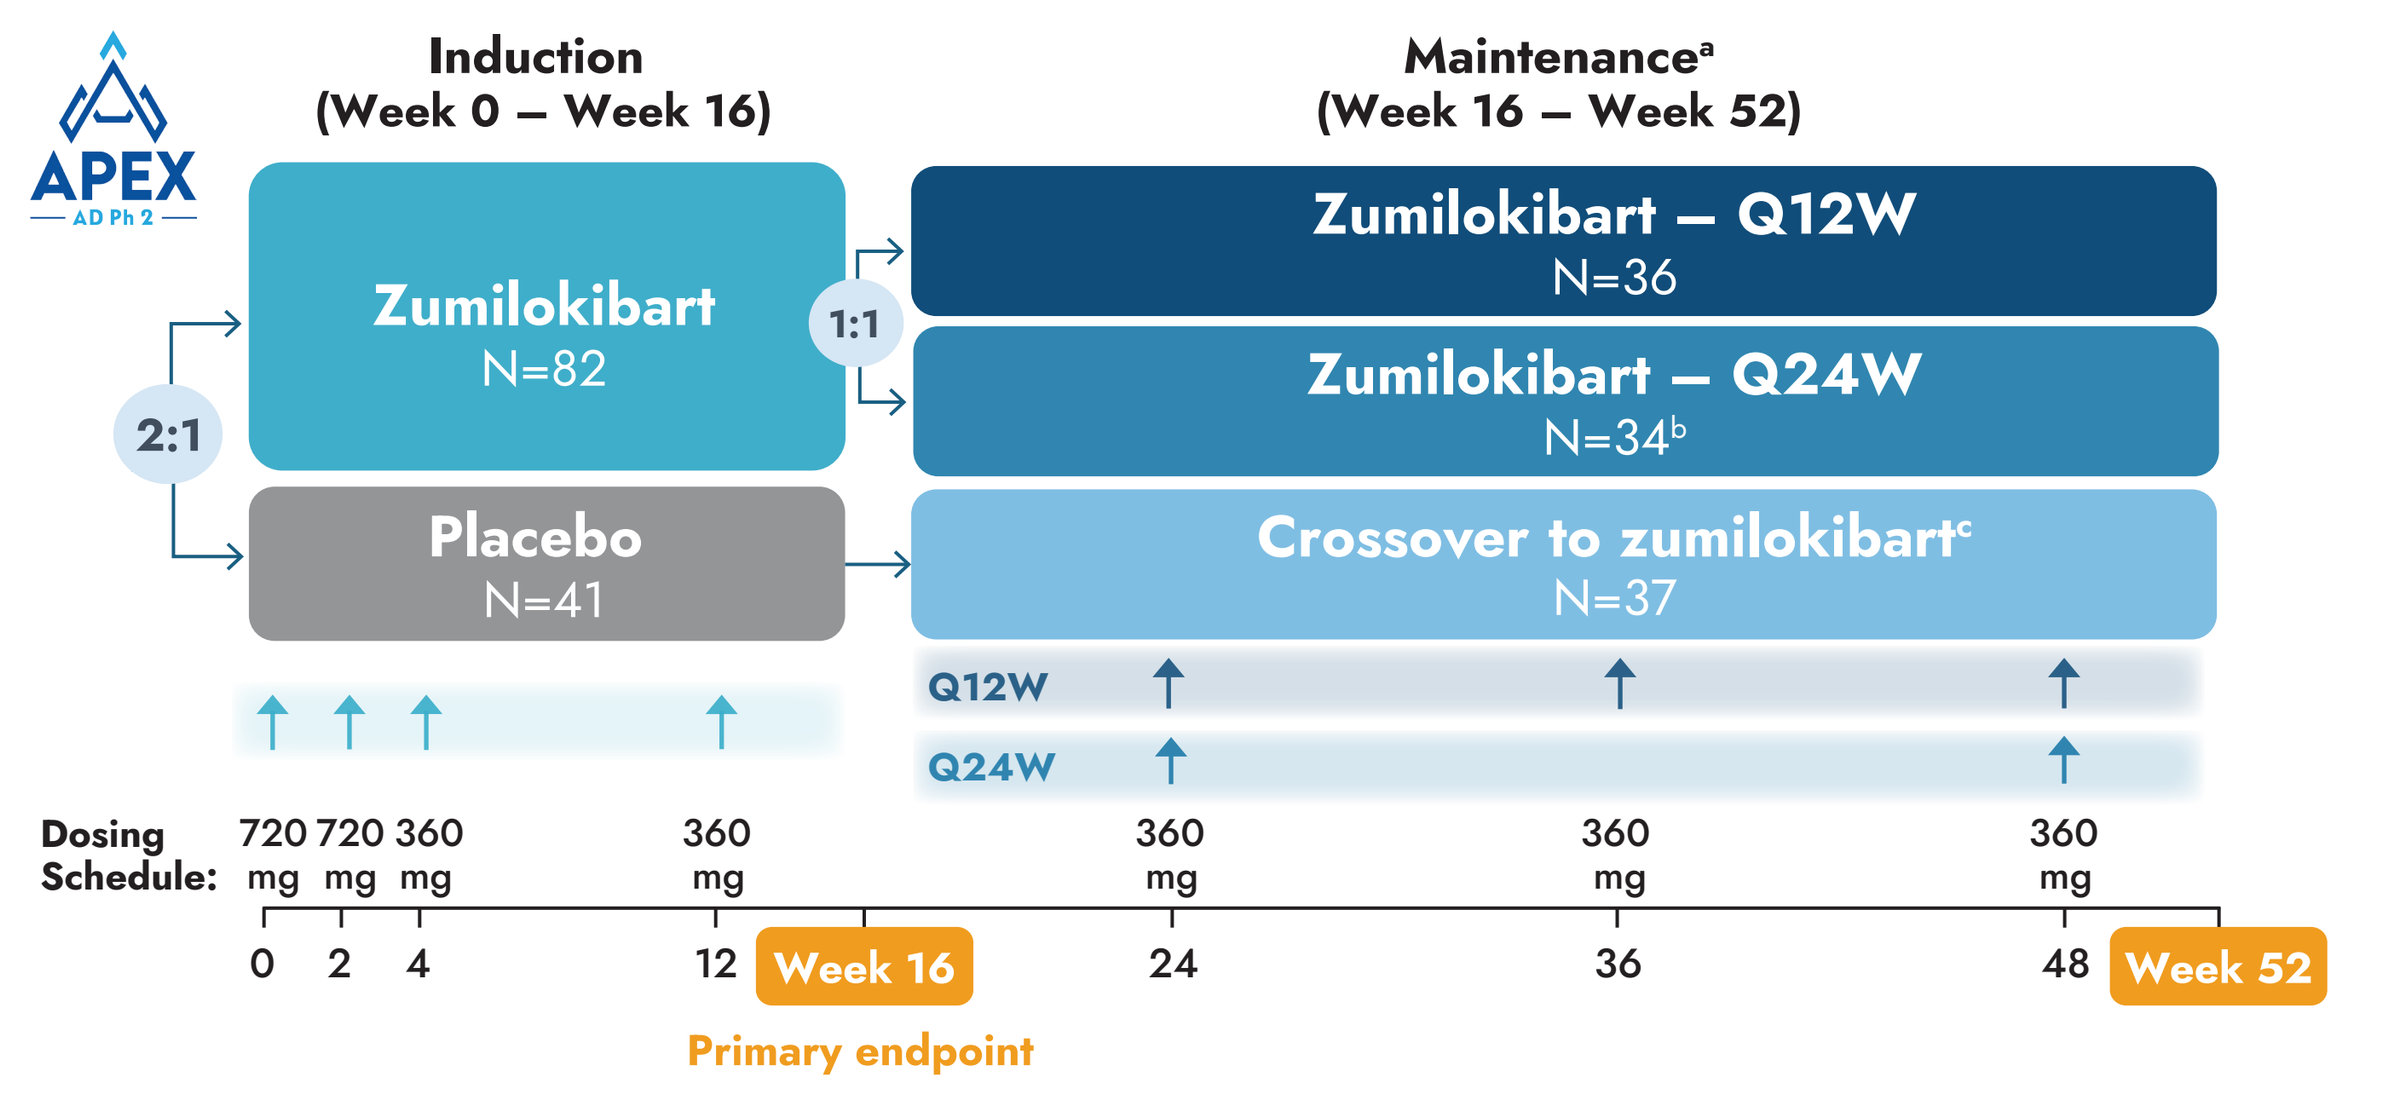
APEX
AD Ph 2
Induction
(Week 0 – Week 16)
Maintenance[a]
(Week 16 – Week 52)
2:1
Zumilokibart
N=82
Placebo
N=41
1:1
Zumilokibart – Q12W
N=36
Zumilokibart – Q24W
N=34[b]
Crossover to zumilokibart[c]
N=37
Q12W
Q24W
Dosing Schedule:
720 mg 720 mg 360 mg 360 mg 360 mg 360 mg 360 mg
0 2 4 12 Week 16 24 36 48 Week 52
Primary endpoint

[a]Double-blind with placebo-matched injections for dose masking. [b]Excluded 2 participants who discontinued the maintenance period before receiving zumilokibart. [c]Modified induction dosing at crossover: 720 mg W16; 360 mg W20, W24, W36, W48.
For induction, N = number of participants randomized and received ≥1 dose of study drug in induction. For maintenance, N = number randomized in induction and received ≥1 dose of study drug in maintenance.

Figure 2: Clinician- and patient-reported outcomes evaluated through Week 52

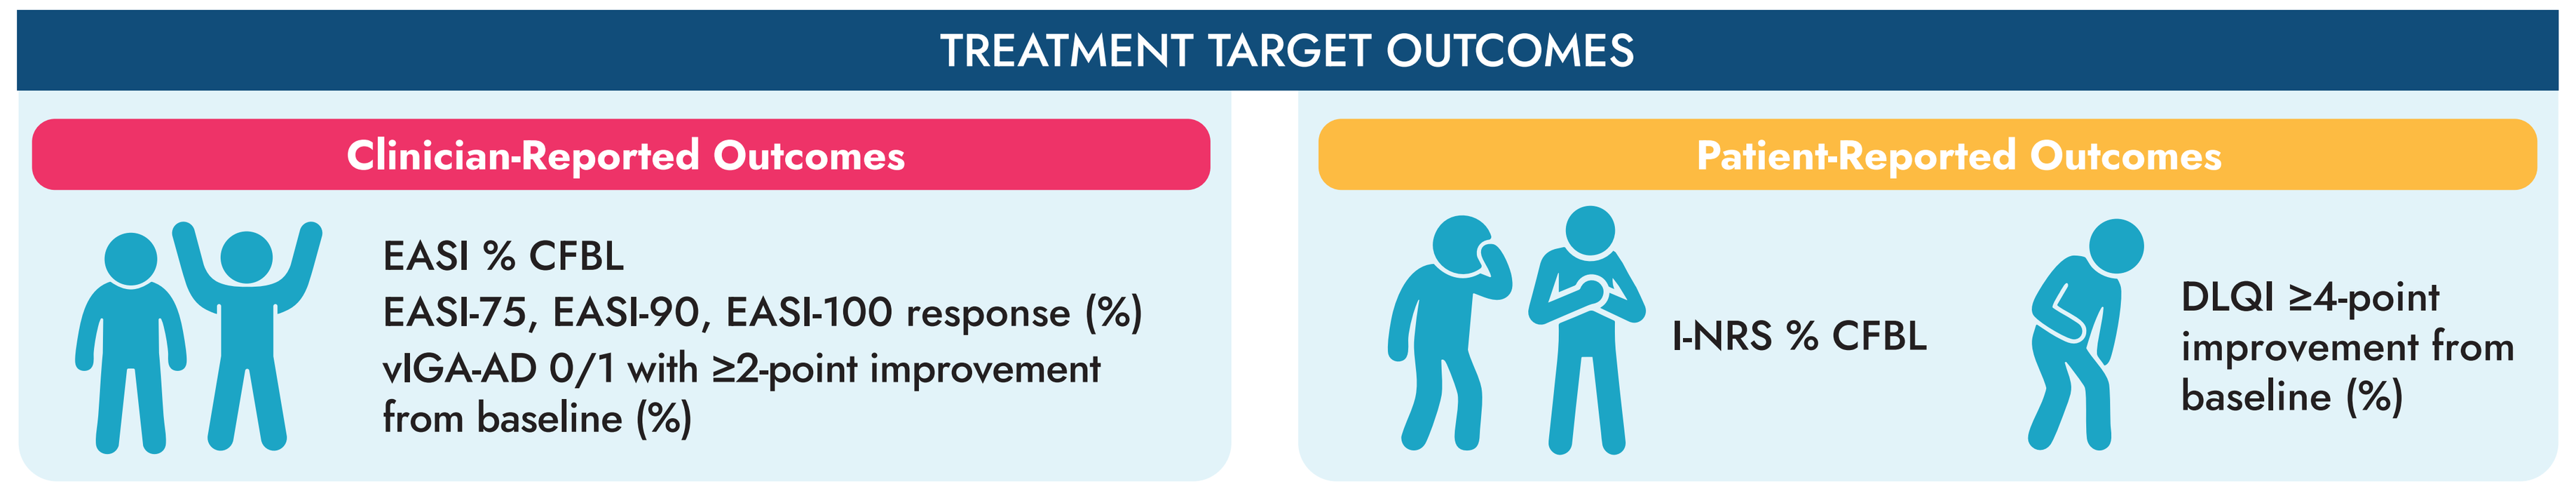
TREATMENT TARGET OUTCOMES
Clinician-Reported Outcomes
EASI % CFBL
EASI-75, EASI-90, EASI-100 response (%)
vIGA-AD 0/1 with ≥2-point improvement from baseline (%)
Patient-Reported Outcomes
I-NRS % CFBL
DLQI ≥4-point improvement from baseline (%)

All measures evaluated through Week 52, with the exception of I-NRS, for which the final data collection was performed at Week 48.
Figure adapted from Williams HC, et al. *J Allergy Clin Immunol.* 2022;149(6):1899-1911 under Creative Commons CC-BY.

RESULTS

Participant disposition

- In total, 109 (88.6%) participants completed the induction period and rolled over into maintenance (N=72 zumilokibart induction arm, N=37 placebo induction arm).
- Among all patients who entered the maintenance period and received ≥1 dose of zumilokibart (N=107), 88.8% completed the maintenance period (**Supplemental Figure 1**).

Baseline characteristics

- Baseline demographics and disease characteristics were generally comparable between induction arms (**Table 1**).
- The maintenance arms were also well balanced following rerandomization at Week 16 (**Supplemental Table 1**).

Clinician-reported lesional endpoints

- Treatment with zumilokibart led to continued reduction in EASI over time, with participants in the Q12W and Q24W arms achieving a −90.8% and −86.0% LS mean change from baseline in EASI, respectively, at Week 52 (**Figure 3**).
 - Participants crossing over from placebo to zumilokibart achieved a similar −85.2% LS mean change from baseline in EASI at Week 52 (**Figure 3**).
- Among participants rerandomized to continue zumilokibart in maintenance, 87.5% and 80.6% of those in the Q12W and Q24W arms, respectively, achieved EASI-75 at Week 52 (**Figure 4A**).
- The higher threshold of EASI-90 was met by 75.0% and 48.4% of participants in the Q12W and Q24W zumilokibart arms, respectively, at Week 52, showing further improvement of response during the maintenance period (**Figure 4B**).
 - Similarly, 71.9% and 51.6% of participants in the Q12W and Q24W arms, respectively, achieved clear/almost clear skin with a vIGA-AD 0/1 response at Week 52 (**Supplemental Figure 2**).
- Complete skin clearance (EASI-100) at Week 52 was achieved in 40.6% and 19.4% of participants in the Q12W and Q24W zumilokibart arms, respectively (**Figure 4C**).

Table 1: Select baseline characteristics

	Placebo (N=41)	Zumilokibart (N=82)
Age, mean ± SD, years	36.0 ± 13.7	38.7 ± 15.6
Female, n (%)	19 (46.3)	41 (50.0)
Previous use of systemic treatment, n (%)	11 (26.8)	35 (42.7)
EASI, mean ± SD	25.3 ± 10.8	25.2 ± 10.8
BSA affected, mean ± SD, %	33.2 ± 22.6	37.2 ± 22.3
vIGA-AD score=4, n (%)	14 (34.1)	27 (32.9)
I-NRS score, mean ± SD	6.7 ± 1.9	6.4 ± 2.1
DLQI, mean ± SD	13.3 ± 7.1	14.3 ± 7.0

Figure 3: Rapid and sustained EASI reduction through Week 52

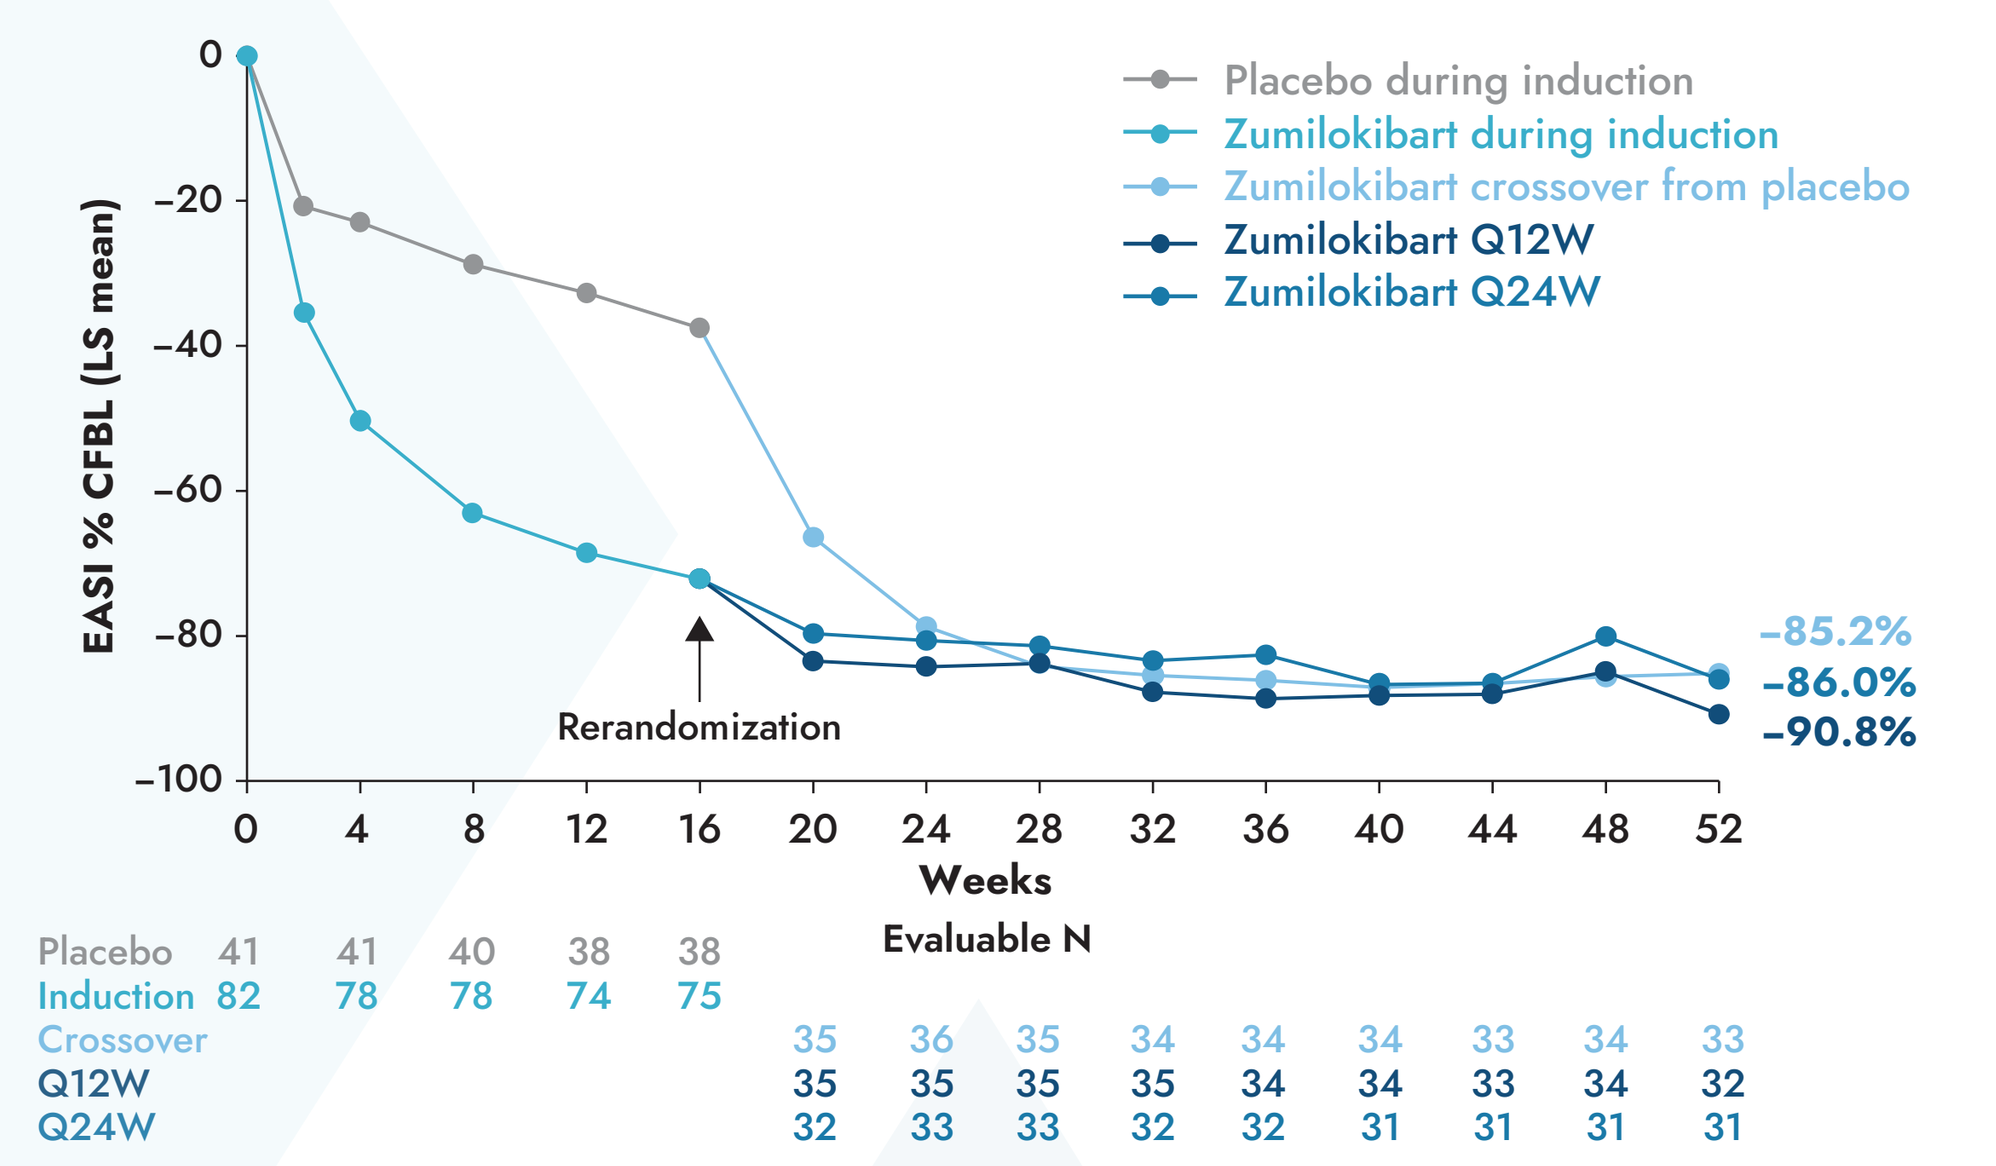
EASI % CFBL (LS mean)
Placebo during induction
Zumilokibart during induction
Zumilokibart crossover from placebo
Zumilokibart Q12W
Zumilokibart Q24W
Rerandomization
−85.2%
−86.0%
−90.8%
Weeks
Evaluable N
Placebo 41 41 40 38 38
Induction 82 78 78 74 75
Crossover 35 36 35 34 34 34 33 34 33
Q12W 35 35 35 35 34 34 33 34 32
Q24W 32 33 33 32 32 31 31 31 31

Efficacy data were evaluated as-observed without imputation for missing data or rescue medication use. For induction, N = number of participants randomized and received ≥1 dose of study drug in induction. For maintenance, N = number randomized in induction and received ≥1 dose of study drug in maintenance.

Figure 4: Robust improvement in EASI response rates with zumilokibart over 52 weeks of treatment

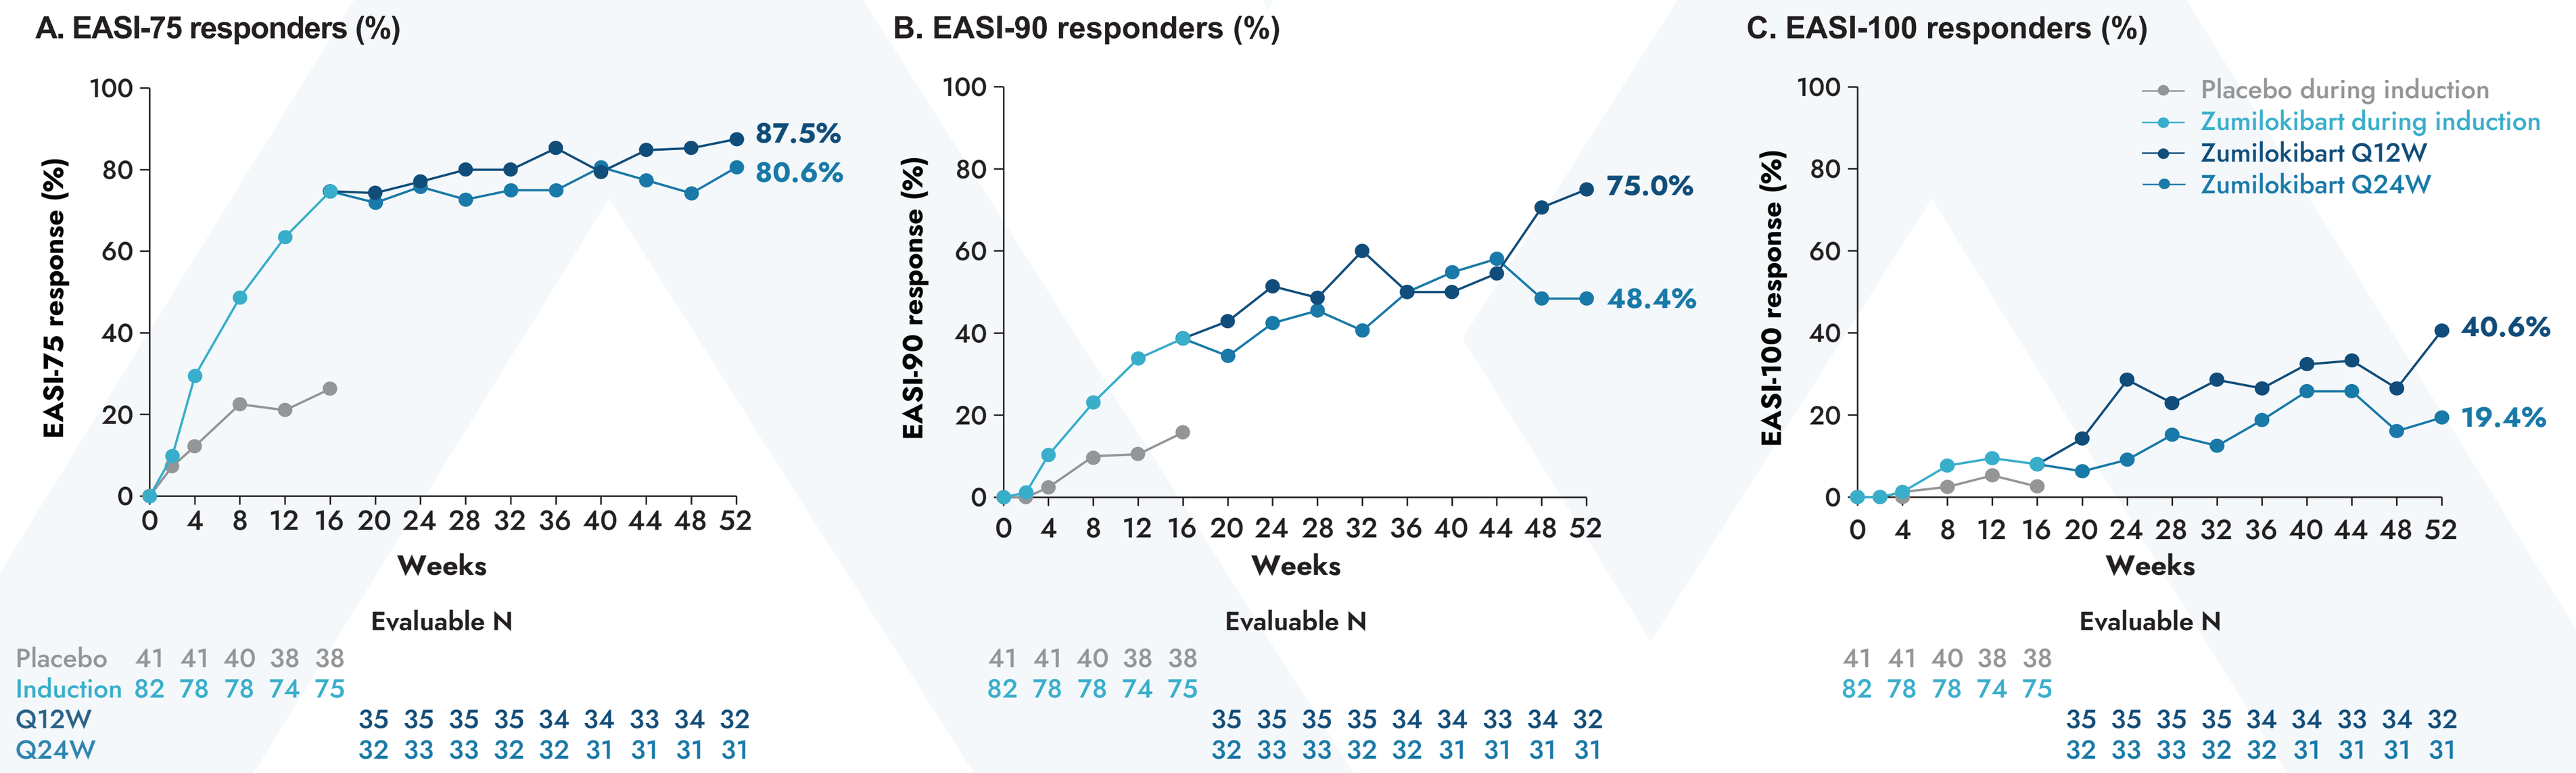
A. EASI-75 responders (%)
EASI-75 response (%)
87.5%
80.6%
B. EASI-90 responders (%)
EASI-90 response (%)
75.0%
48.4%
C. EASI-100 responders (%)
EASI-100 response (%)
40.6%
19.4%
Placebo during induction
Zumilokibart during induction
Zumilokibart Q12W
Zumilokibart Q24W
Weeks
Evaluable N
Placebo 41 41 40 38 38
Induction 82 78 78 74 75
Q12W 35 35 35 35 34 34 33 34 32
Q24W 32 33 33 32 32 31 31 31 31

Efficacy data were evaluated as-observed without imputation for missing data or rescue medication use.
For induction, N = number of participants randomized and received ≥1 dose of study drug in induction. For maintenance, N = number randomized in induction and received ≥1 dose of study drug in maintenance.

Patient-reported itch and quality of life

- Progressive improvement in itch continued throughout the maintenance period, with participants in the Q12W and Q24W arms reaching a −72.5% and −64.1% LS mean change from baseline in I-NRS at Week 48 (**Figure 5A**).
- By the end of the 52-week treatment period, ≥4-point improvement in DLQI was achieved by 92.9% and 88.5% of participants in the Q12W and Q24W arms, respectively (**Figure 5B**).

Safety

- Treatment with zumilokibart was well tolerated during the 52-week treatment period of APEX Part A (**Table 2**).

Figure 5: Improvements in itch and quality of life with zumilokibart over 52 weeks of treatment

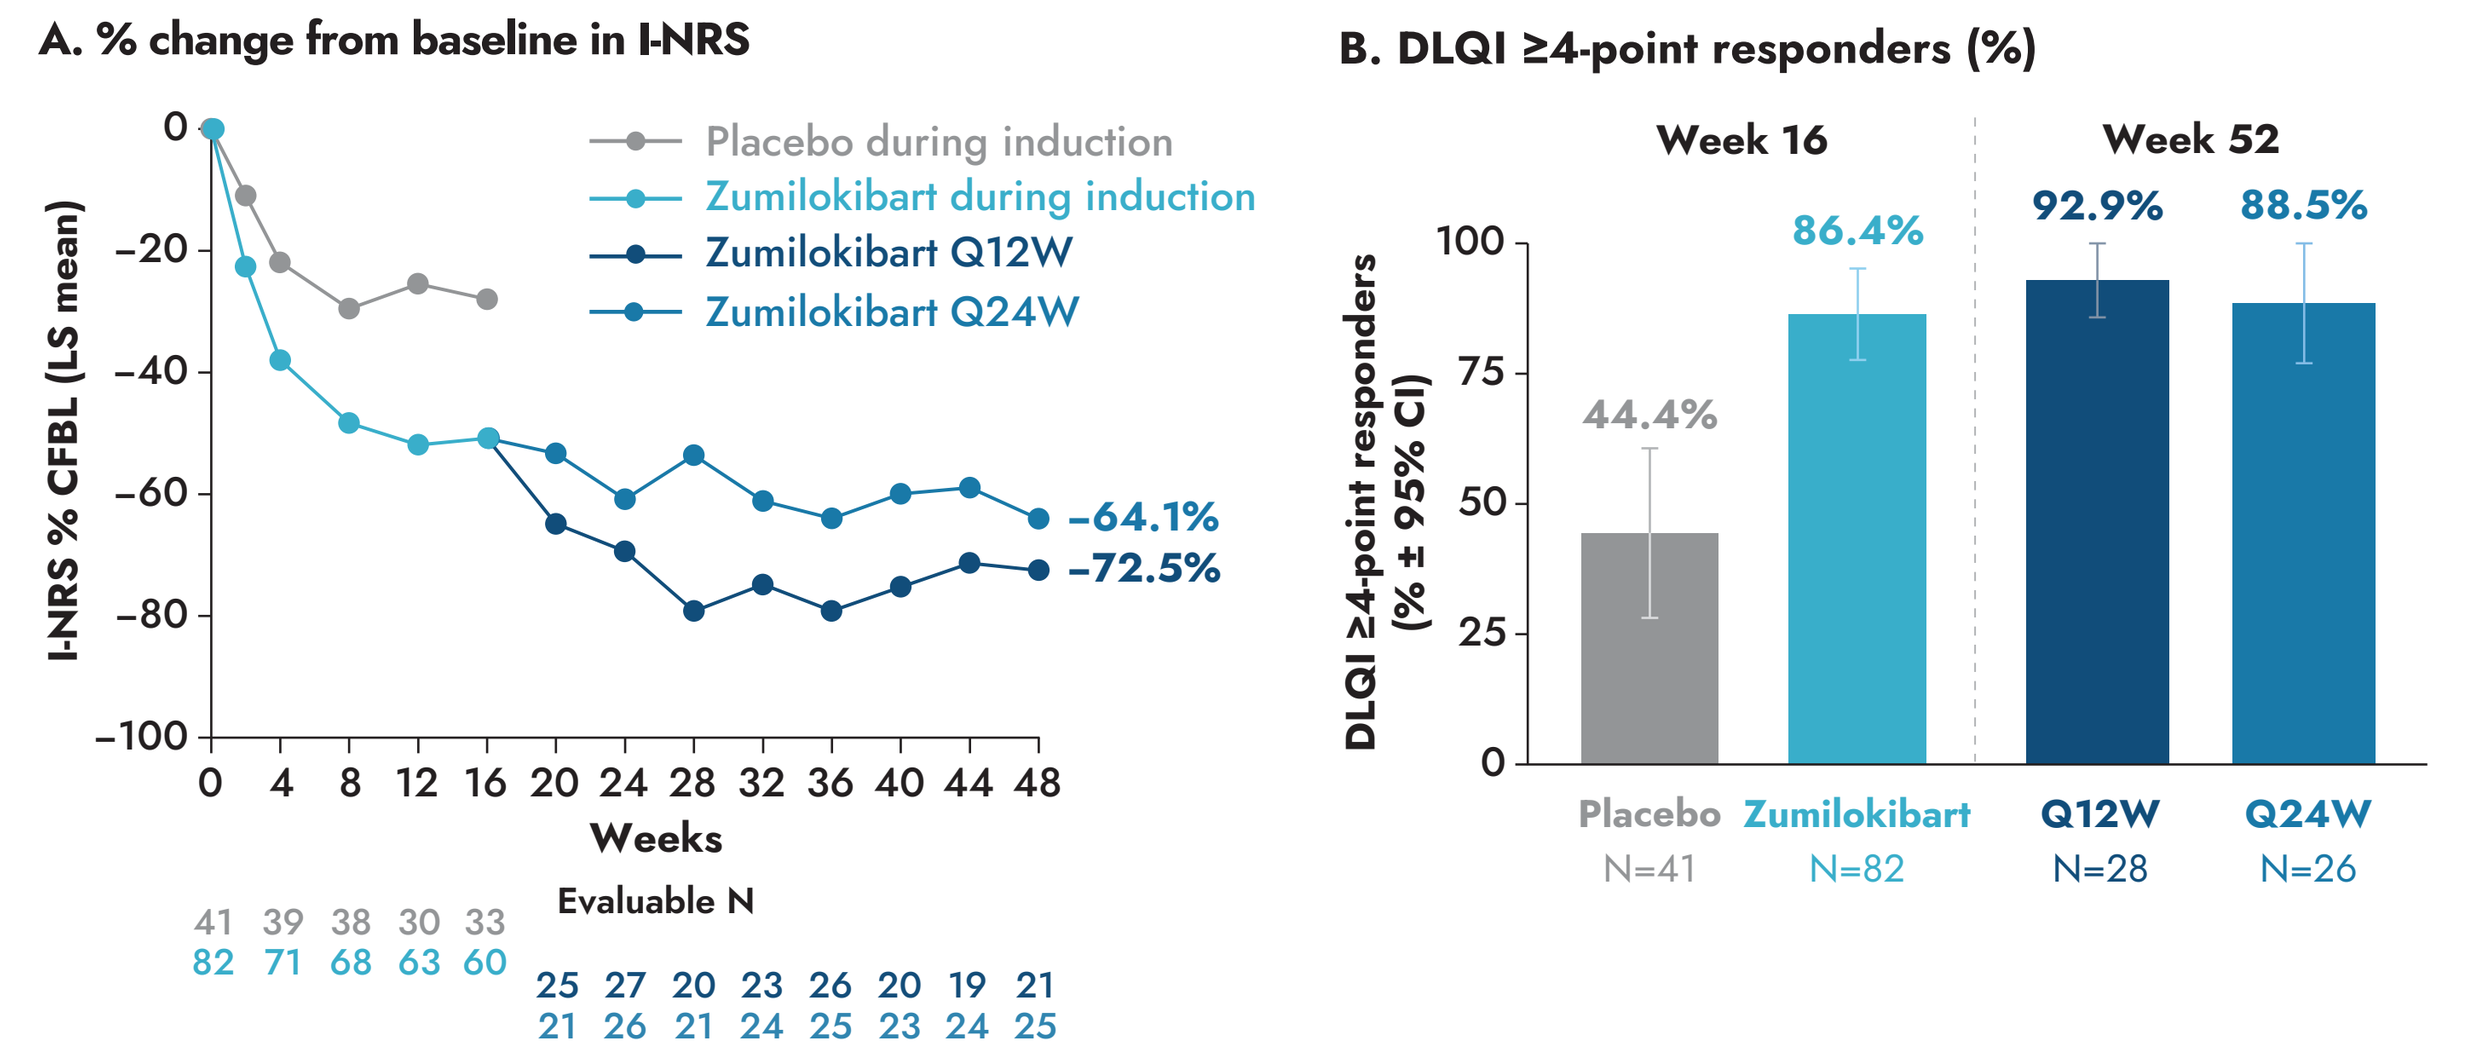
A. % change from baseline in I-NRS
I-NRS % CFBL (LS mean)
Placebo during induction
Zumilokibart during induction
Zumilokibart Q12W
Zumilokibart Q24W
−64.1%
−72.5%
Weeks
Evaluable N
41 39 38 30 33
82 71 68 63 60
25 27 20 23 26 20 19 21
21 26 21 24 25 23 24 25
B. DLQI ≥4-point responders (%)
Week 16
Week 52
DLQI ≥4-point responders (% ± 95% CI)
44.4%
86.4%
92.9%
88.5%
Placebo N=41
Zumilokibart N=82
Q12W N=28
Q24W N=26

A. Analysis was conducted as-observed without imputation for missing data or rescue medication use. For induction, N = number of participants randomized and received ≥1 dose of study drug in induction. For maintenance, N = number randomized to induction and received ≥1 dose of study drug in maintenance. **B.** N = number of participants with available data at Week 16 (left) and Week 52 (right).

Table 2: Safety summary through Week 52

Event, n (%)	Zumilokibart mITT (N=119) (All participants receiving ≥1 dose of zumilokibart)
≥1 adverse event	85 (71.4)
≥1 serious adverse event	1 (0.8)[a]
Adverse events leading to discontinuation through Week 52	4 (3.4)
Deaths	0
Most common adverse events through Week 52 (≥5%)	
Noninfective conjunctivitis[b]	16 (13.4)
Upper respiratory tract infection	15 (12.6)
Nasopharyngitis	11 (9.2)
Dermatitis atopic	6 (5.0)

Safety data are summarized for all randomized participants who received ≥1 dose of zumilokibart (N=82 randomized to zumilokibart and N=37 who crossed over from placebo to zumilokibart at Week 16).
[a]Case of Sézary syndrome; not considered related to study treatment.
[b]The total conjunctivitis rate was 20.2%, including the following preferred terms: noninfective conjunctivitis, conjunctivitis allergic, conjunctivitis bacterial and conjunctivitis viral, with <1% discontinuations over the 52-week treatment period. Most cases were mild to moderate. The median duration of conjunctivitis was 27.5 days. No relationship between exposure and conjunctivitis was observed.

CONCLUSIONS

- The pharmacokinetics of zumilokibart allow for less frequent dosing, with only 4 dosing days during induction and 2 or 4 dosing days per year in maintenance.
- Treatment with zumilokibart was associated with progressive improvements in AD signs, symptoms, and quality of life over 52 weeks.
- Robust clinical responses were observed through Week 52 with both every 12-week and every 24-week maintenance dosing regimens.
- Zumilokibart was generally well tolerated, with a safety profile consistent with other therapies targeting the IL-13 or IL-4/IL-13 pathway.

Collectively, these results support continued evaluation of zumilokibart as a novel therapy for AD, including:
- Dose optimization in APEX Part B (Week 16 readout expected Q2 2026)
- Initiation of phase 3 program (expected 2H 2026)

ABBREVIATIONS

AD, atopic dermatitis; BSA, body surface area; CI, confidence interval; CFBL, change from baseline; EASI, Eczema Area and Severity Index; EASI-75, achieving at least 75% reduction from baseline in EASI; EASI-90, achieving at least 90% reduction from baseline in EASI; EASI-100, achieving 100% reduction from baseline in EASI; DLQI, Dermatology Life Quality Index; Fc, fragment crystallizable; FcRn, neonatal Fc receptor; IL, interleukin; I-NRS, Itch-Numeric Rating Scale; LS, least squares; mITT, modified intent-to-treat; Q12W, every 12 weeks; Q24W, every 24 weeks; vIGA-AD, Validated Investigator Global Assessment for AD; W, week; YTE, triple-amino acid modification in the Fc region that extends antibody half-life.

REFERENCES

1. Silverberg JI, et al. *J Eur Acad Dermatol Venereol.* 2024;38:2139-2148. **2.** Gorelick J, et al. *Am J Clin Dermatol.* 2025;26:199-211. **3.** Miron Y, et al. *Transl Clin Immunol.* 2022;150:690-700. **4.** Zhu E, et al. *J Allergy Clin Immunol Glob.* 2025;4:100545. **5.** Lim XQ, et al. *Clin Transl Sci.* 2025;18:e70456. **6.** Guttman-Yassky E, et al. Presented at the European Academy of Dermatology and Venerology (EADV) Congress 2025; Paris, France. **7.** Guttman-Yassky E, et al. Presented at the American Academy of Dermatology (AAD) Congress 2026; Denver, CO, USA.

ACKNOWLEDGMENTS

Apogee Therapeutics, Inc. sponsored the study reported in this poster. Medical writing and editorial support for this poster were provided by Kate Smigiel of Apogee Therapeutics, Inc., and Nathan Yardley and Heidi Reinholdt, both of Helios Global Group, on behalf of Apogee Therapeutics, Inc.

DISCLOSURES

EG-Y is an employee of Mount Sinai and has received research grants (paid to the institution) from and/or is a consultant for: AbbVie, Aclaris Therapeutics, Almirall, Alumis, Amgen, AnaptysBio, Apogee Therapeutics, Apollo Therapeutics, Arcutis, Artax Biopharma, Astria, Boeringer Ingelheim, BMS, Celldex, Centrexion Therapeutics, Connect Biopharm, Concerto Biosciences, Coty, DBV, Dualitas Therapeutics, Eli Lilly, Enveda Biosciences, Escient Pharmaceuticals, Galderma, Gate Bio, GSK, GSK Immunology, Incyte, Inmagene, Janssen Biotech, Jasper Therapeutics, Kymera Therapeutics, Kyowa Kirin, LEO Pharma, Matchpoint Therapeutics, Merck, Nektar Therapeutics, NUMAB Therapeutics, Obsidio, OTSUKA, Pfizer, Pharmaxis, Proteologix, Q32 Bio, RAPT, RayThera, Regeneron, Ribon Therapeutics, Sanofi, SATO, Schrödinger, Sitryx, Sun Pharma, Takeda, Teva, TRex Bio, UCB, VRG Therapeutics, and Xencor. **AB** has served as a speaker (received honoraria) for Almirall, Eli Lilly, LEO Pharma, Sanofi, and UCB; has served as a scientific adviser (received honoraria) for AbbVie, Almirall, Alumis, Amgen, Anaptysbio, Apogee Therapeutics, Arcutis, Eli Lilly, Incyte, Janssen, Khanda Therapeutics, LEO Pharma, Novartis, Oruka, Pfizer, Regeneron, Sanofi, Sun Pharma, Takeda, and UCB; and owns stock in Lipidio and Oruka. **MG** has served as an investigator, speaker and/or advisor for AbbVie, Acelyrin, Alumis, Amgen, Akros, Arcutis, Aristea, AnaptysBio, Apogee Therapeutics, Bausch Health, BMS, Boehringer Ingelheim, Dermira, Dermavant, Eli Lilly, Galderma, GSK, Incyte, Inmagene, JAMP Pharma, Janssen, LEO Pharma, L'Oreal, MedImmune, Meiji, Moonlake, Nektar, Nimbus, Novartis, Organon, Oruka, Pfizer, Q32 Bio, Regeneron, Sanofi Genzyme, Sun Pharma, Tarsus, Takeda, UCB, Union, Ventyx, and Vyne. **KK** has received consulting fees, honoraria, grant support, and/or lecture fees from AbbVie, Amgen, Apogee Therapeutics, Eli Lilly, Kyowa Kirin, Japan Tobacco, LEO Pharma, Maruho, Mitsubishi Tanabe, Ono Pharmaceutical, Pfizer, Procter & Gamble, Sanofi, Taiho, and Torii Pharmaceutical. **JIS** has received honoraria as a consultant and/or advisory board member for AbbVie, Aldena, Amgen, AObiome, Apollo, Arcutis, Arena, Attovia, Boehringer Ingelheim, BMS, Castle Biosciences, Connect Biopharma, Corevitas, Dermavant, Eli Lilly, FIDE, Formation Bio, Galderma, GSK, Immunocore, Incyte, Inmagene, Invea, LEO Pharma, Merck, Nektar, Novartis, Pfizer, RAPT, Recludix, Regeneron, Sandoz, Sanofi Genzyme, Shaperon, TARGET-RWE, Teva, Triveni, UCB, Union, and UpToDate; speaker for AbbVie, Arcutis, Dermavant, Eli Lilly, Galderma, LEO Pharma, Pfizer, Regeneron, and Sanofi Genzyme; institution received grants from Galderma, Incyte, and Pfizer. **SW**, **OC**, **JX**, **CW**, **AK**, and **KN** are employees of Apogee Therapeutics, Inc. and hold company stock/stock options. **CLD** is an employee of Apogee Therapeutics, Inc. and holds company stock/stock options; serves on the Board of Directors of Oruka Therapeutics, Inc.; and holds stock/stock options in Oruka, Spyre, and Junevity.

Presented at the Revolutionizing Atopic Dermatitis (RAD) Annual Meeting, Nashville, TN, USA, June 17 – 19, 2026

For questions, please contact MedAffairs@apogeetherapeutics.com

©Apogee Therapeutics, Inc.

Supplement to: Zumilokibart (APG777) provides robust, progressive improvement in the signs and symptoms of atopic dermatitis: 52-week results from the phase 2 APEX Part A study

Emma Guttman-Yassky[1], Andrew Blauvelt[2], Melinda Gooderham[3], Kenji Kabashima[4], Susanna Wen[5], Olivia Choi[5], Jiahong Xu[5], Cindy Wu[5], Amol Kamboj[5], Carl L. Dambkowski[5], Kristine Nograles[5], Jonathan I. Silverberg[6]

[1]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [2]University of Maryland School of Medicine, Baltimore, MD, USA; [3]SKiN Centre for Dermatology, Peterborough, Ontario, Canada and Queen's University, Kingston, Ontario, Canada; [4]Kyoto University Graduate School of Medicine, Kyoto, Japan; [5]Apogee Therapeutics, Inc., Waltham, MA, USA; [6]George Washington University School of Medicine and Health Sciences, Washington, DC, USA

SUPPLEMENTAL RESULTS

Participant disposition

- In total, 109 (88.6%) participants completed the induction period and rolled over into maintenance (N=72 zumilokibart induction arm, N=37 placebo induction arm).
- Among all patients who entered the maintenance period and received ≥1 dose of zumilokibart (N=107), 88.8% completed the maintenance period (**Supplemental Figure 1**).
 - The most common reason for discontinuation was withdrawal by participant (n=4, Q12W arm; n=2, Q24W arm; n=1, crossover arm); 1 participant discontinued due to an AE (crossover arm).

Supplemental Figure 1: Participant disposition

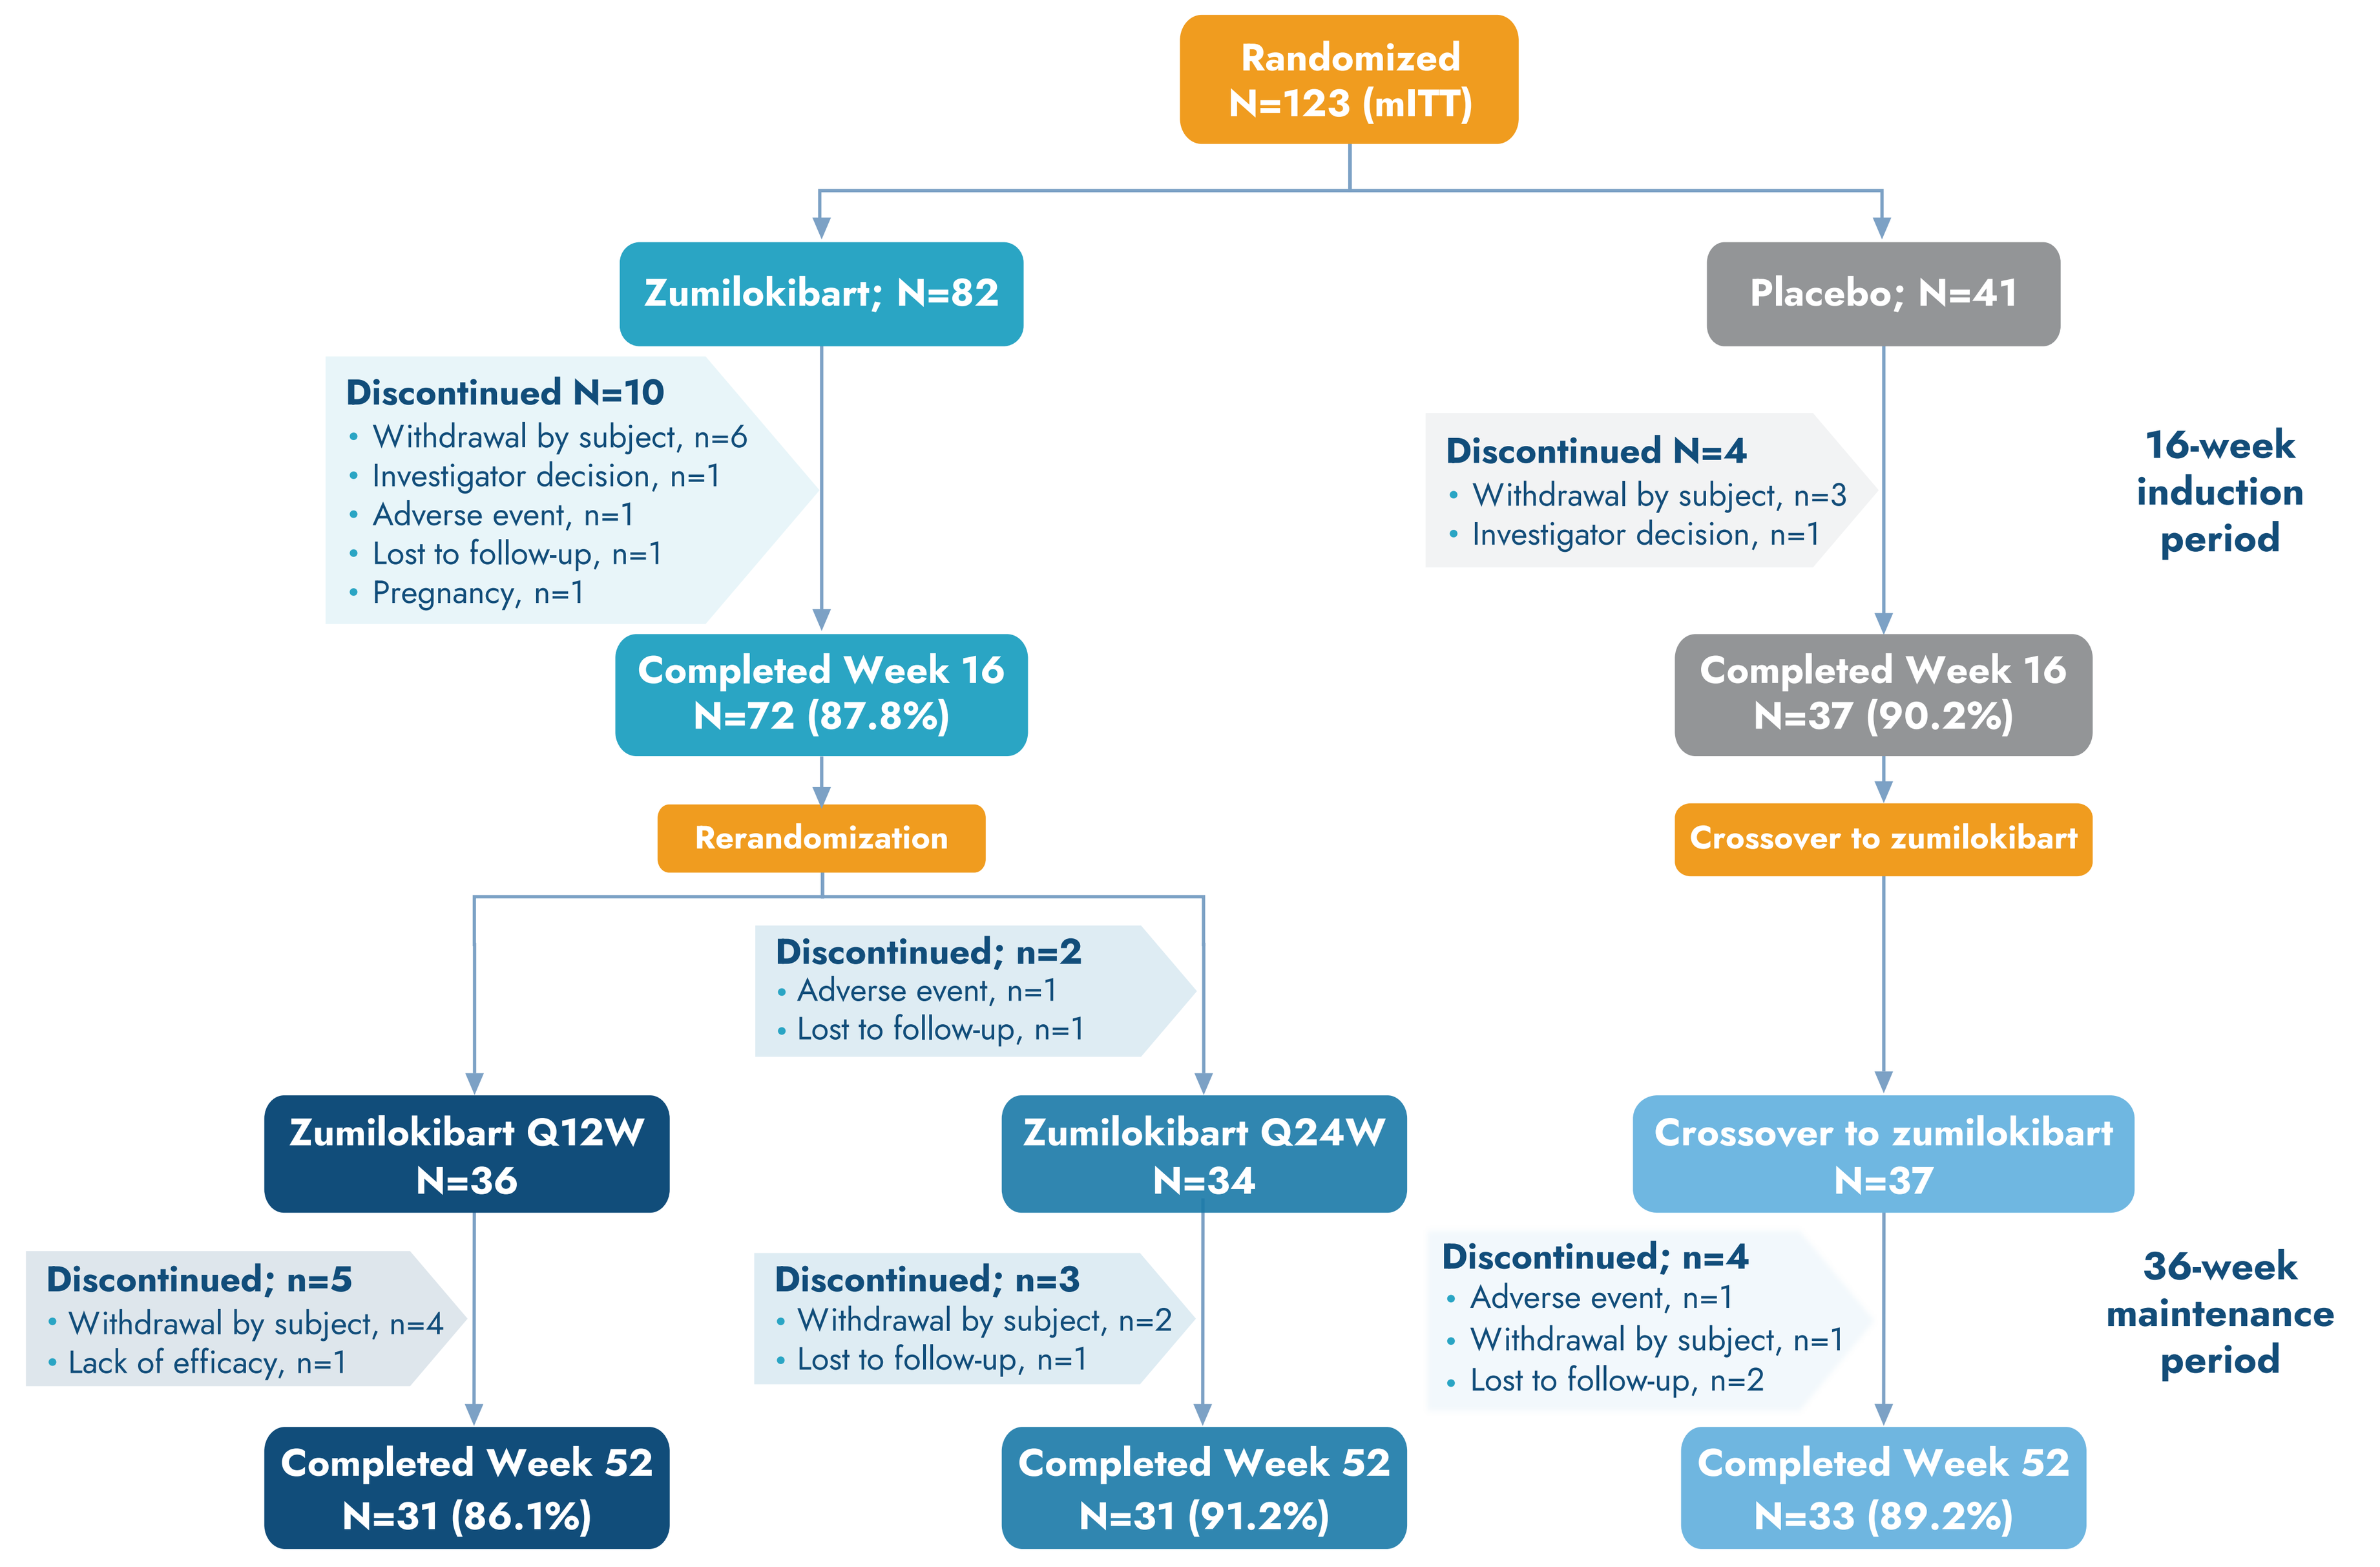
Randomized
N=123 (mITT)
Zumilokibart; N=82
Placebo; N=41
Discontinued N=10
• Withdrawal by subject, n=6
• Investigator decision, n=1
• Adverse event, n=1
• Lost to follow-up, n=1
• Pregnancy, n=1
Discontinued N=4
• Withdrawal by subject, n=3
• Investigator decision, n=1
16-week induction period
Completed Week 16
N=72 (87.8%)
Completed Week 16
N=37 (90.2%)
Rerandomization
Crossover to zumilokibart
Discontinued; n=2
• Adverse event, n=1
• Lost to follow-up, n=1
Zumilokibart Q12W
N=36
Zumilokibart Q24W
N=34
Crossover to zumilokibart
N=37
Discontinued; n=5
• Withdrawal by subject, n=4
• Lack of efficacy, n=1
Discontinued; n=3
• Withdrawal by subject, n=2
• Lost to follow-up, n=1
Discontinued; n=4
• Adverse event, n=1
• Withdrawal by subject, n=1
• Lost to follow-up, n=2
36-week maintenance period
Completed Week 52
N=31 (86.1%)
Completed Week 52
N=31 (91.2%)
Completed Week 52
N=33 (89.2%)

mITT is defined as all randomized participants who received at least 1 dose of study drug during the induction period.

Baseline characteristics

- Baseline demographics and disease characteristics were generally comparable between groups (**Table 1**).
- The maintenance arms were also well balanced following re-randomization at Week 16 (**Supplemental Table 1**).

Supplemental Table 1: Select baseline characteristics

	Study Baseline			
	Participants randomized and who received ≥1 dose of study drug in induction		Participants randomized to zumilokibart in induction and who received ≥1 dose of zumilokibart in maintenance	
	Placebo (N=41)	Zumilokibart (N=82)	Zumilokibart Q12W (N=36)	Zumilokibart Q24W (N=34)
Age, mean ± SD, years	36.0 ± 13.7	38.7 ± 15.6	34.6 ± 12.9	43.4 ± 17.6
Female, n (%)	19 (46.3)	41 (50.0)	21 (58.3)	17 (50.0)
Body mass index, mean ± SD, kg/m²	27.7 ± 6.7	30.0 ± 8.4	28.9 ± 6.9	30.8 ± 10.4
Race, n (%)				
White	30 (73.2)	54 (65.9)	20 (55.6)	24 (70.6)
Black	6 (14.6)	13 (15.9)	8 (22.2)	3 (8.8)
Asian	4 (9.8)	12 (14.6)	6 (16.7)	6 (17.6)
Other[a]	1 (2.4)	3 (3.7)	2 (5.6)	1 (2.9)
Previous use of systemic treatment, n (%)	11 (26.8)	35 (42.7)	13 (36.1)	14 (41.2)
EASI, mean ± SD	25.3 ± 10.8	25.2 ± 10.8	25.8 ± 10.6	24.2 ± 9.8
vIGA-AD score=4, n (%)	14 (34.1)	27 (32.9)	11 (30.6)	11 (32.4)
BSA affected, mean ± SD, %	33.2 ± 22.6	37.2 ± 22.3	38.2 ± 24.7	35.6 ± 17.3
I-NRS score, mean ± SD	6.7 ± 1.9	6.4 ± 2.1	6.4 ± 1.9	6.5 ± 2.2
DLQI, mean ± SD	13.3 ± 7.1	14.3 ± 7.0	13.5 ± 5.7	14.3 ± 7.1

[a]Other included American Indian or Alaska Native, Native Hawaiian or Pacific Islander, other, and unknown.

vIGA-AD 0/1 responders

- At Week 52, 71.9% and 51.6% of participants in the Q12W and Q24W arms, respectively, achieved clear/almost clear skin with a vIGA-AD 0/1 response, showing further improvement of response during the maintenance period (**Supplemental Figure 2**).

Supplemental Figure 2: Improvement in vIGA-AD 0/1 response rates with zumilokibart over 52 weeks of treatment

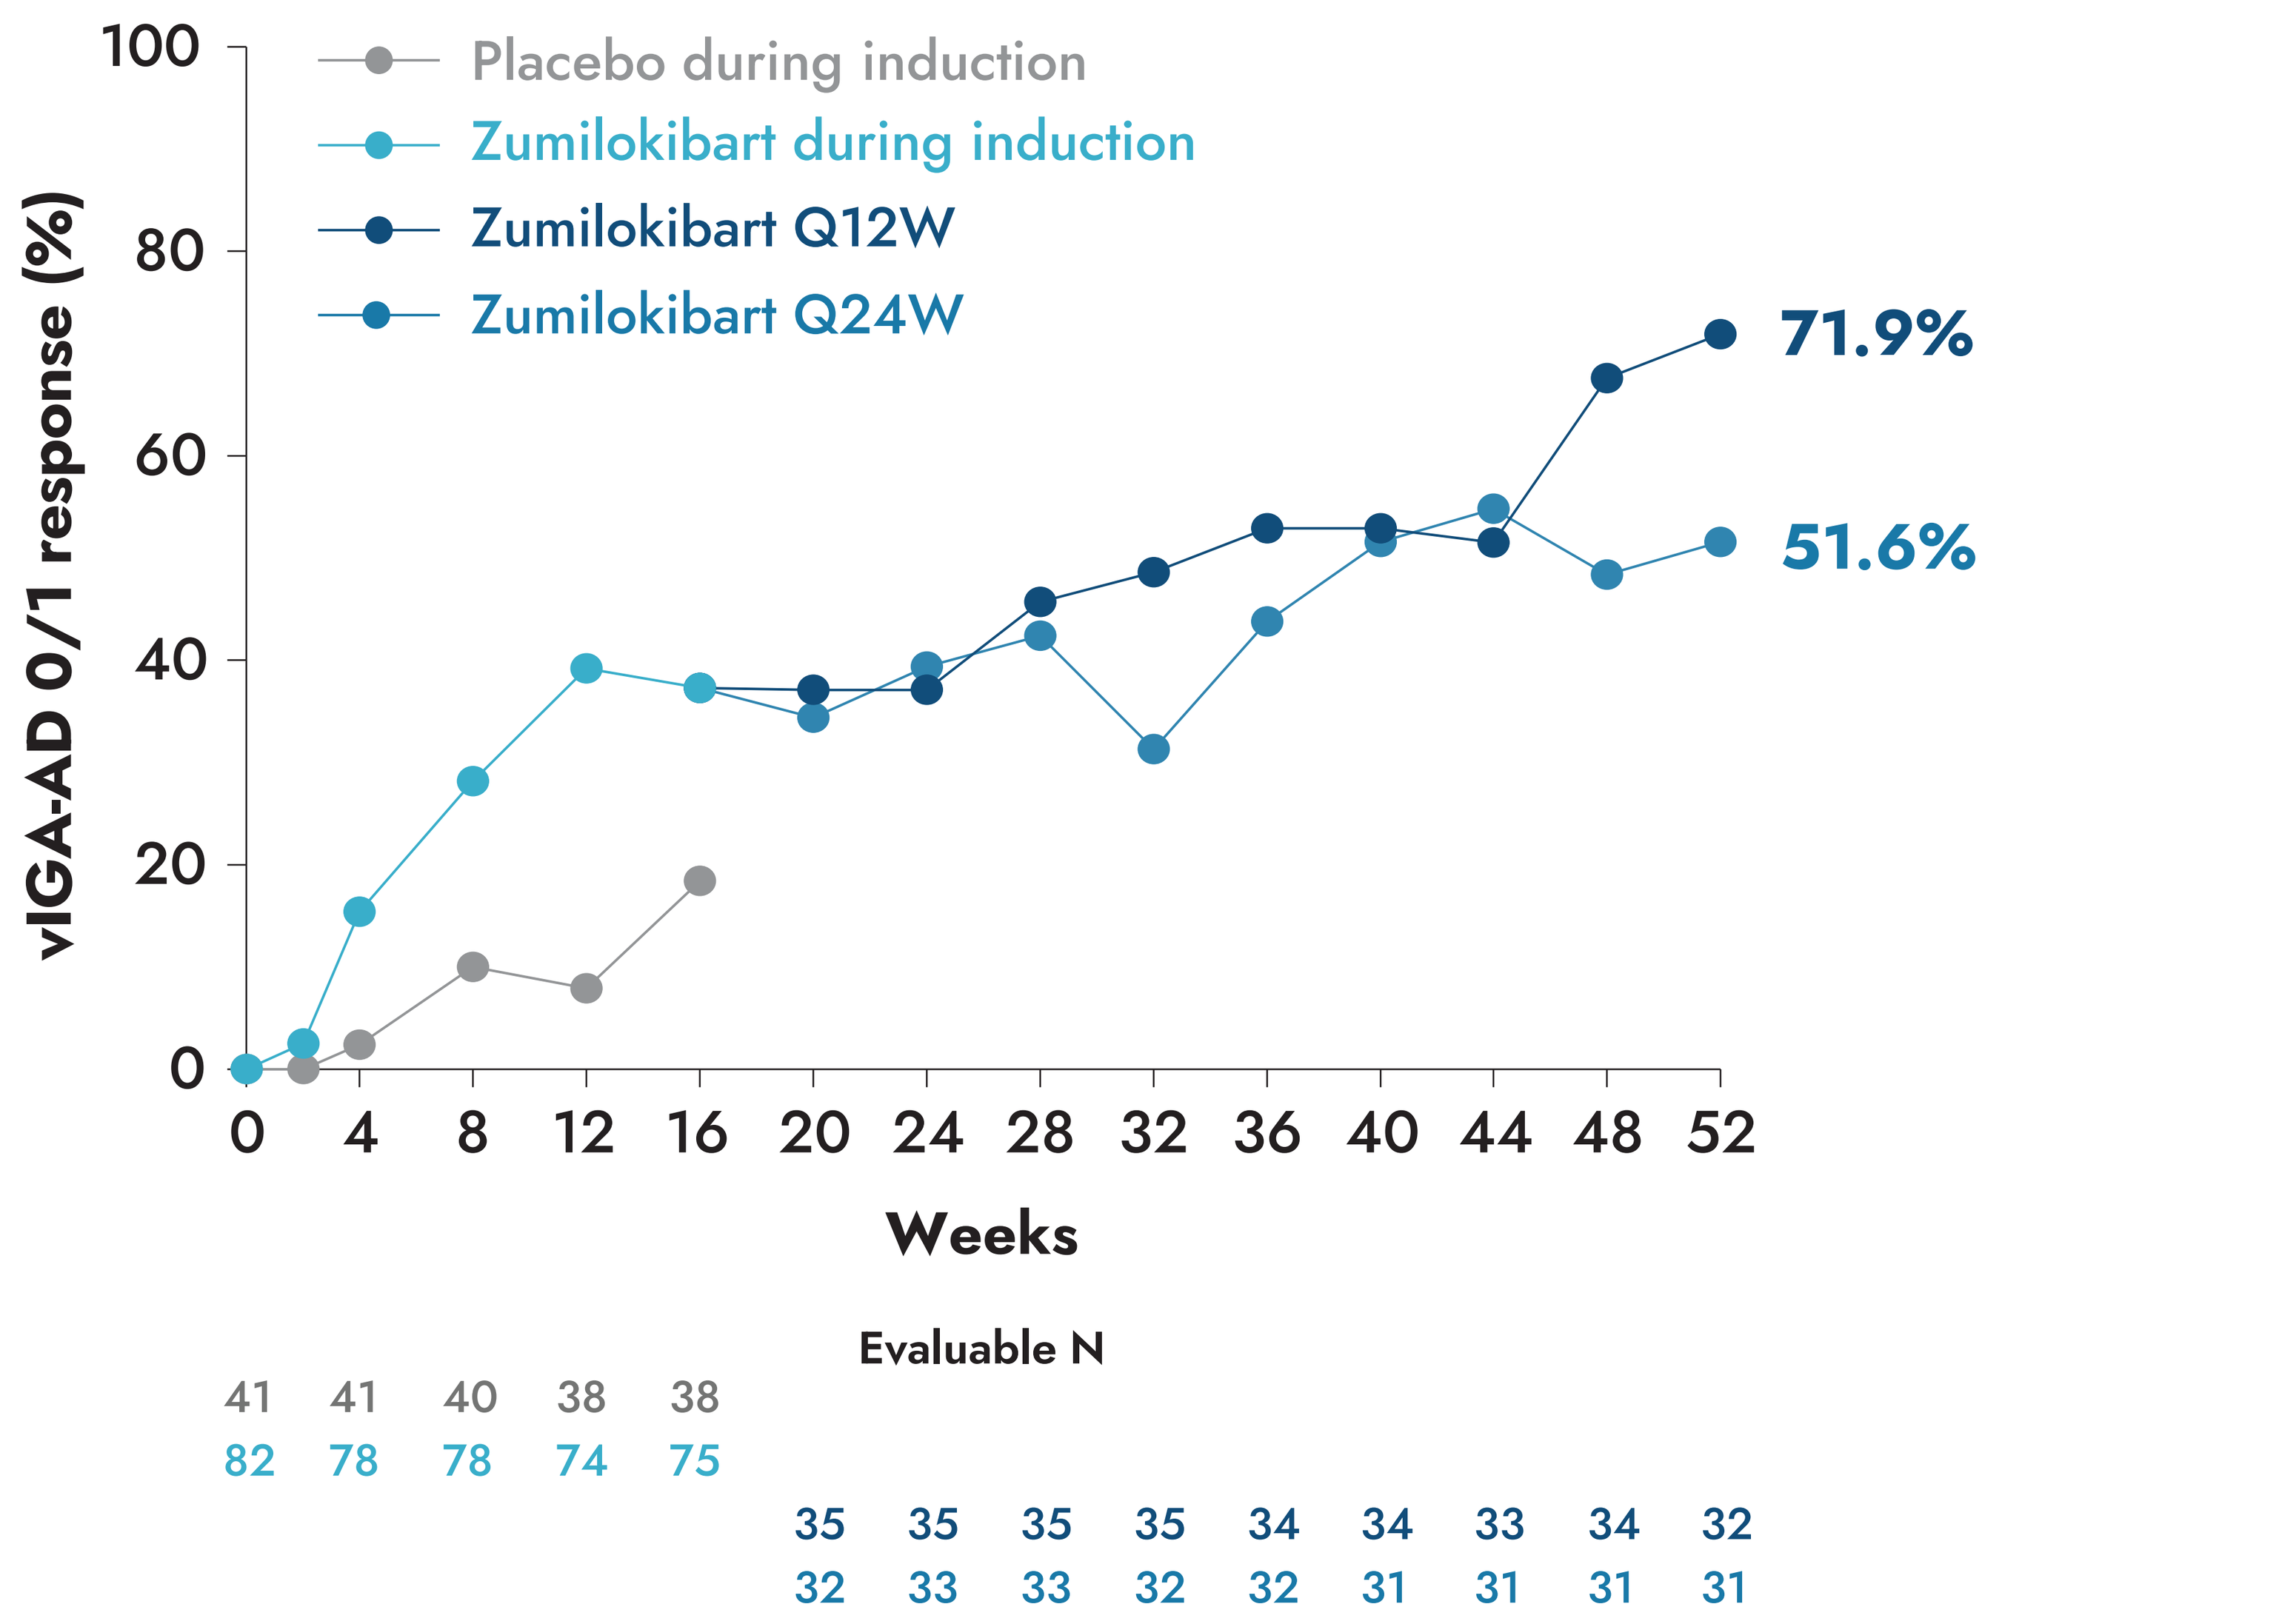
vIGA-AD 0/1 response (%)
100
80
60
40
20
0
Placebo during induction
Zumilokibart during induction
Zumilokibart Q12W
Zumilokibart Q24W
71.9%
51.6%
0 4 8 12 16 20 24 28 32 36 40 44 48 52
Weeks
Evaluable N
41 41 40 38 38
82 78 78 74 75
35 35 35 35 34 34 33 34 32
32 33 33 32 32 31 31 31 31

For induction, N = number of participants randomized and received ≥1 dose of study drug in induction.
For maintenance, N = number randomized to zumilokibart in induction and received ≥1 dose of study drug in maintenance.
A vIGA-AD 0/1 response was defined as achieving a vIGA-AD score of 0 or 1 with a ≥2-point reduction from baseline.
Efficacy data were evaluated as-observed without imputation for missing data or rescue medication use.

ABBREVIATIONS
AE, adverse event; BSA, body surface area; EASI, Eczema Area and Severity Index; DLQI, Dermatology Life Quality Index; I-NRS, Itch-Numeric Rating Scale; mITT, modified intent-to-treat; Q12W, every 12 weeks; Q24W, every 24 weeks; SD, standard deviation; vIGA-AD, Validated Investigator Global Assessment for Atopic Dermatitis.

Presented at the Revolutionizing Atopic Dermatitis (RAD) Annual Meeting, Nashville, TN, USA, June 17 – 19, 2026
For questions, please contact MedAffairs@apogeetherapeutics.com

©Apogee Therapeutics, Inc.