1291

Profiling bispecific T-cell engagers: Strategies for enhancing potency while minimizing cytokine release

AUTHORS

Peter Bergqvist*, Amy Burke*, Ryan Blackler, Patrick Farber, Gesa Volkers, Nathalie Blamey, Creagh Briercliffe, Kate Caldwell, Stefania Carrara, Lauren Clifford, Melissa Cid, Cindy-Lee Crichlow, Harveer Dhupar, Jared Dutra, Cristina Faralla, Jessica Fernandes Scortecci, Kate Gibson, Marian Haustein, Lucas Kraft, Katherine Lam, Ahn Lee, Lucas Maddalena, Matt Mai, John Marwick, Esther Odekunle, Patrick Rowe, Britany Rufenach, Oscar Urtatiz, Elena Vigano, Riley Walsh, Wei Wei, Shirley Zhi, Kelly Bullock, Sophie Cullen, Sherie Duncan, Ester Falconer, Kevin Heyries, Michael Kennedy, Ingrid Knarston, Nicole Lee, Grace Leung, Kathleen Lisaingo, Stephanie K. Masterman, Amanda Moreira, Marta Szabat, Katherine Vousden, Aaron Yamniuk, Christopher Williamson, Bryan C. Barnhart, Allison Goodman, and **Lindsay DeVorkin**.

*co-first author

Presenter: Lindsay DeVorkin

AUTHOR AFFILIATION

AbCellera, Vancouver, Canada

DOWNLOAD POSTER

REFERENCES

1. Sung H et al. (2020) Global cancer statistics 2020: GLOBOCAN estimates of incidence and mortality worldwide for 36 cancers in 185 countries. *CA Cancer J Clin*, 71:209–49.
2. Lim K et al. (2024) Clinical Pharmacology Strategies for Bispecific Antibody Development: Learnings from FDA-Approved Bispecific Antibodies in Oncology, *Clin Pharmacol Ther*, 116(2):315-327.
3. Ahn M et al. (2023) Tarlatamab for Patients with Previously Treated Small-Cell Lung Cancer, *NEJM*, 389:2063-207.
4. Hassel et al. (2023) Three-Year Overall Survival with Tebentafusp in Metastatic Uveal Melanoma, *NEJM*, 389:2256-2266.
5. Murphy AJ et al. (2019). Patent Application Publication, Pub. No: US 2019/0389951 A1.

BACKGROUND

T-cell engagers for difficult-to-treat cancers

Solid tumors, which account for more than 90% of all cancers,[1] remain challenging indications with high unmet need. Solid tumor treatments have been underrepresented despite a recent wave of targeted immunotherapies, with seven of the nine approved T-cell engagers (TCEs) treating hematological cancers.[2] However, recent FDA approvals highlight the potential of TCEs as a drug class for patients with difficult-to-treat cancers.[3,4]

To date, limitations in efficacy have hindered TCE development — a challenge that is amplified within immunosuppressive solid tumor microenvironments. To advance TCEs for solid tumor indications, it is critical to obtain clinical efficacy, maintain a manageable safety profile, and avoid induction of excessive cytokine release. Doing so requires optimization of multiple TCE parameters, including T-cell activation, persistence, and cytokine release, which can be achieved using design strategies that are tailored to target and indication.

AIM

Tailored TCE design strategies for diverse targets and indications

To address these challenges, we developed a TCE platform comprising novel T-cell engaging antibodies (CD3 and γδ), costimulatory CD28- and 4-1BB-binding antibodies, multispecific engineering technology, and a high-throughput process for identifying molecules with desired profiles. To generate optimal TCEs, we engineer large panels of bispecifics using diverse CD3- and tumor associated antigen (TAA)-binding arms. We vary TCE parameters that impact function, such as binding affinities, geometries, and epitopes, for both CD3 and TAAs. We then apply a suite of high-throughput assessments to identify TCEs with desired properties.

Here, we present strategies to build optimized TCEs for solid tumor targets (Figure 1) and application of our platform to two internal TCE programs.

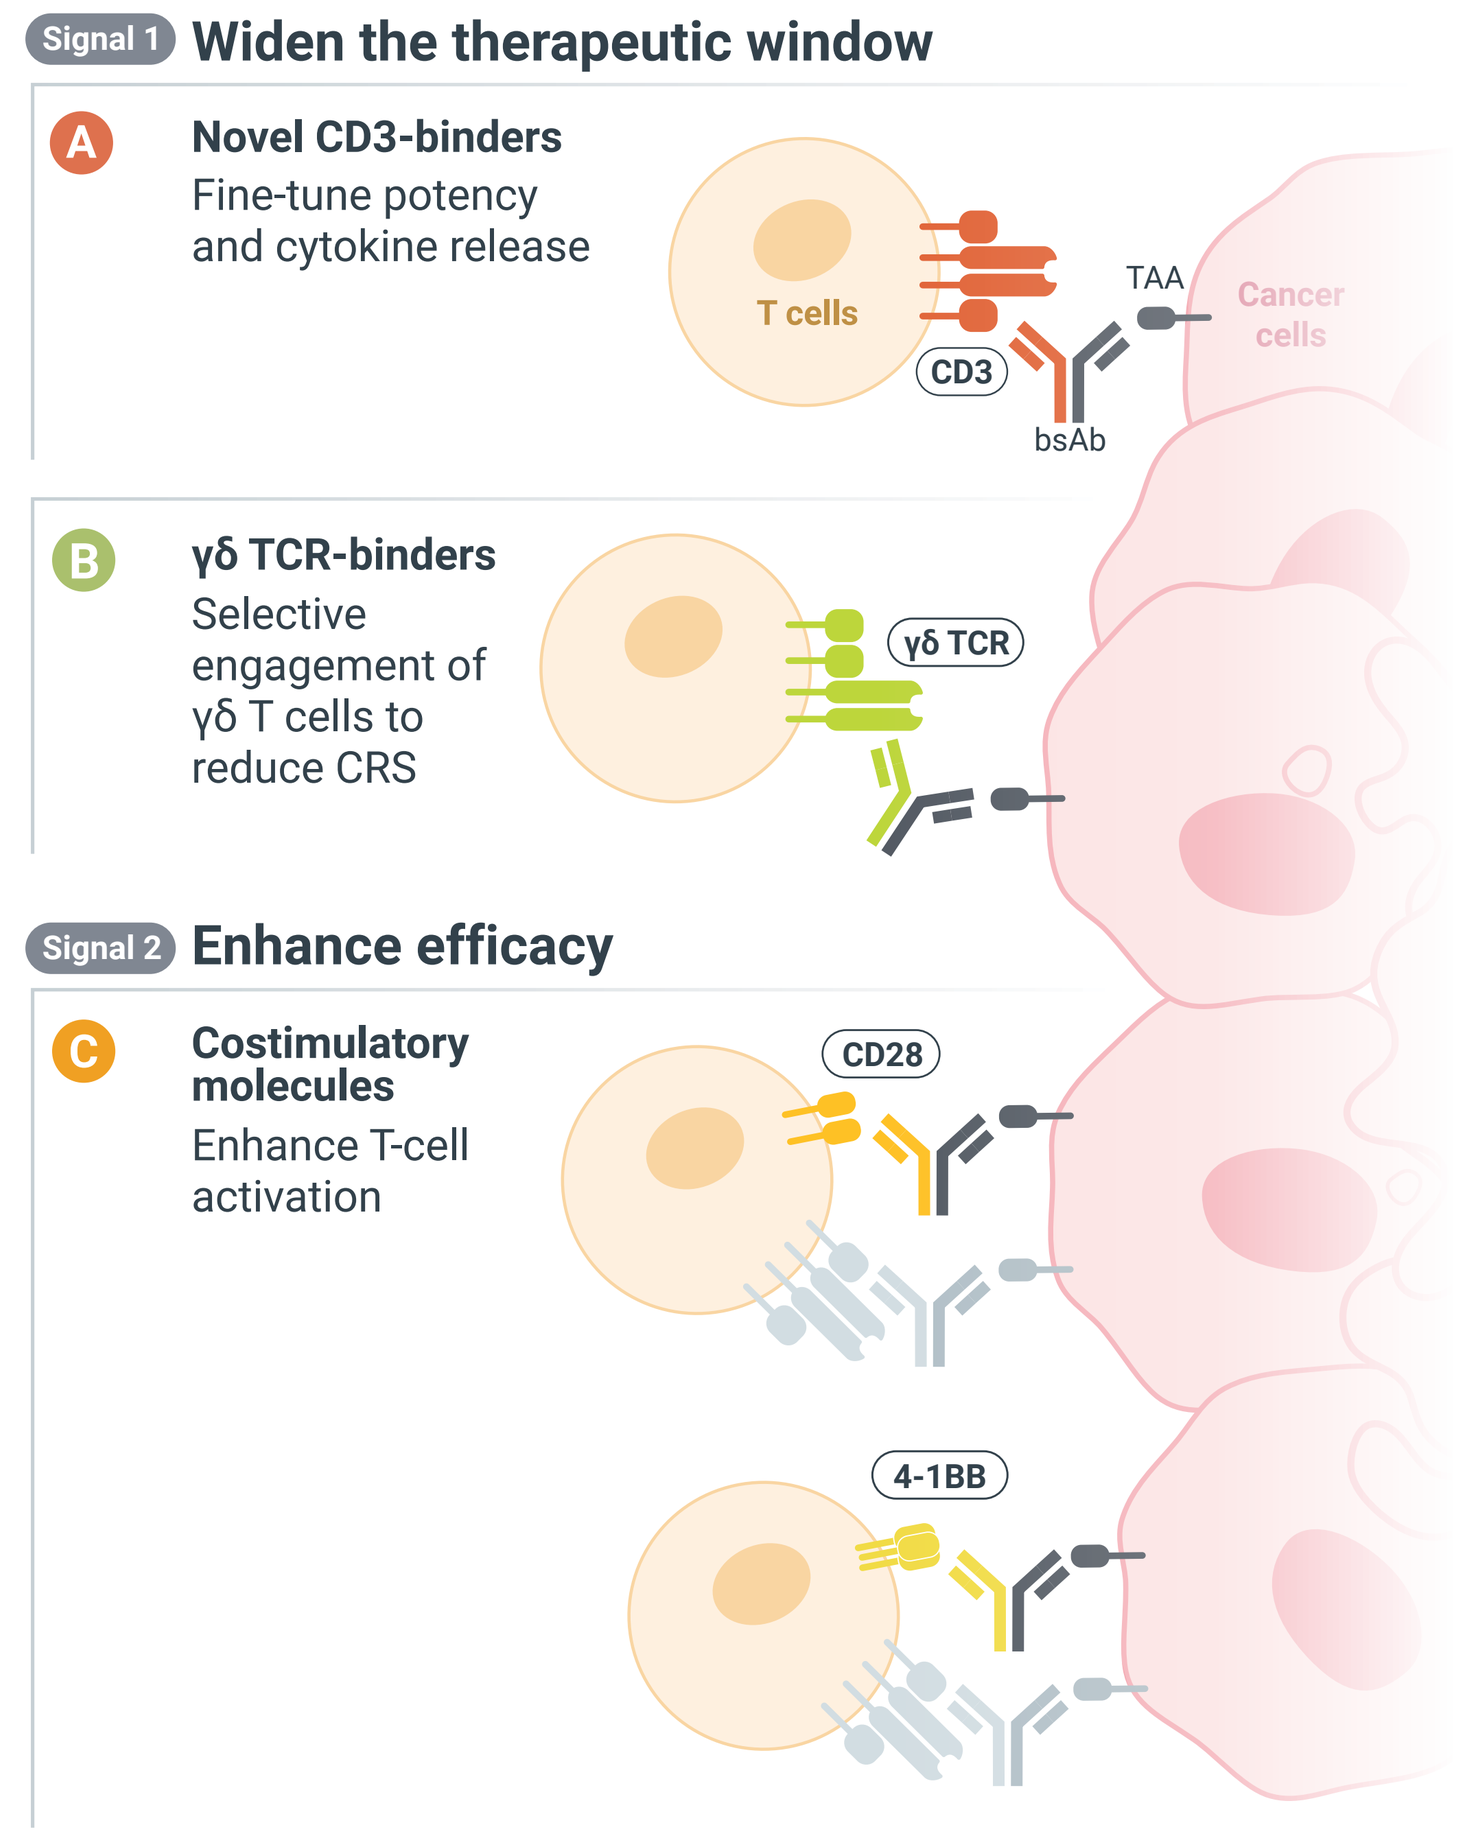

Signal 1 Widen the therapeutic window
A Novel CD3-binders
Fine-tune potency and cytokine release
T cells
CD3
bsAb
TAA
Cancer cells
B γδ TCR-binders
Selective engagement of γδ T cells to reduce CRS
γδ TCR
Signal 2 Enhance efficacy
C Costimulatory molecules
Enhance T-cell activation
CD28
4-1BB

Figure 1. Multiple strategies for building optimized TCEs. **(A)** Novel, fully human CD3-binding antibodies with diverse binding and functional properties to generate TCEs that achieve high potency with optimal cytokine release. **(B)** γδ TCR-binding antibodies may be used to generate TCEs that selectively recruit γδ T cells to target tumor cells, potentially reducing the risk of cytokine release syndrome (CRS). **(C)** CD28- and 4-1BB-binding antibodies to enhance anti-tumor activity of T-cell activating therapies while reducing T-cell exhaustion.

Novel CD3-binding antibodies to drive potent tumor-cell killing and optimal cytokine release

CD3 T-cell engagers with diverse functional profiles for two solid tumor targets

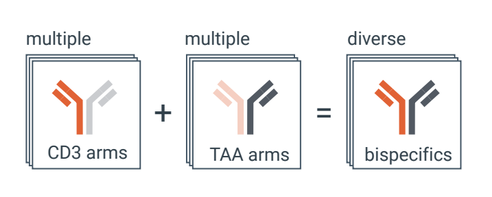

multiple CD3 arms + multiple TAA arms = diverse bispecifics

Tumor cell-killing and cytokine release

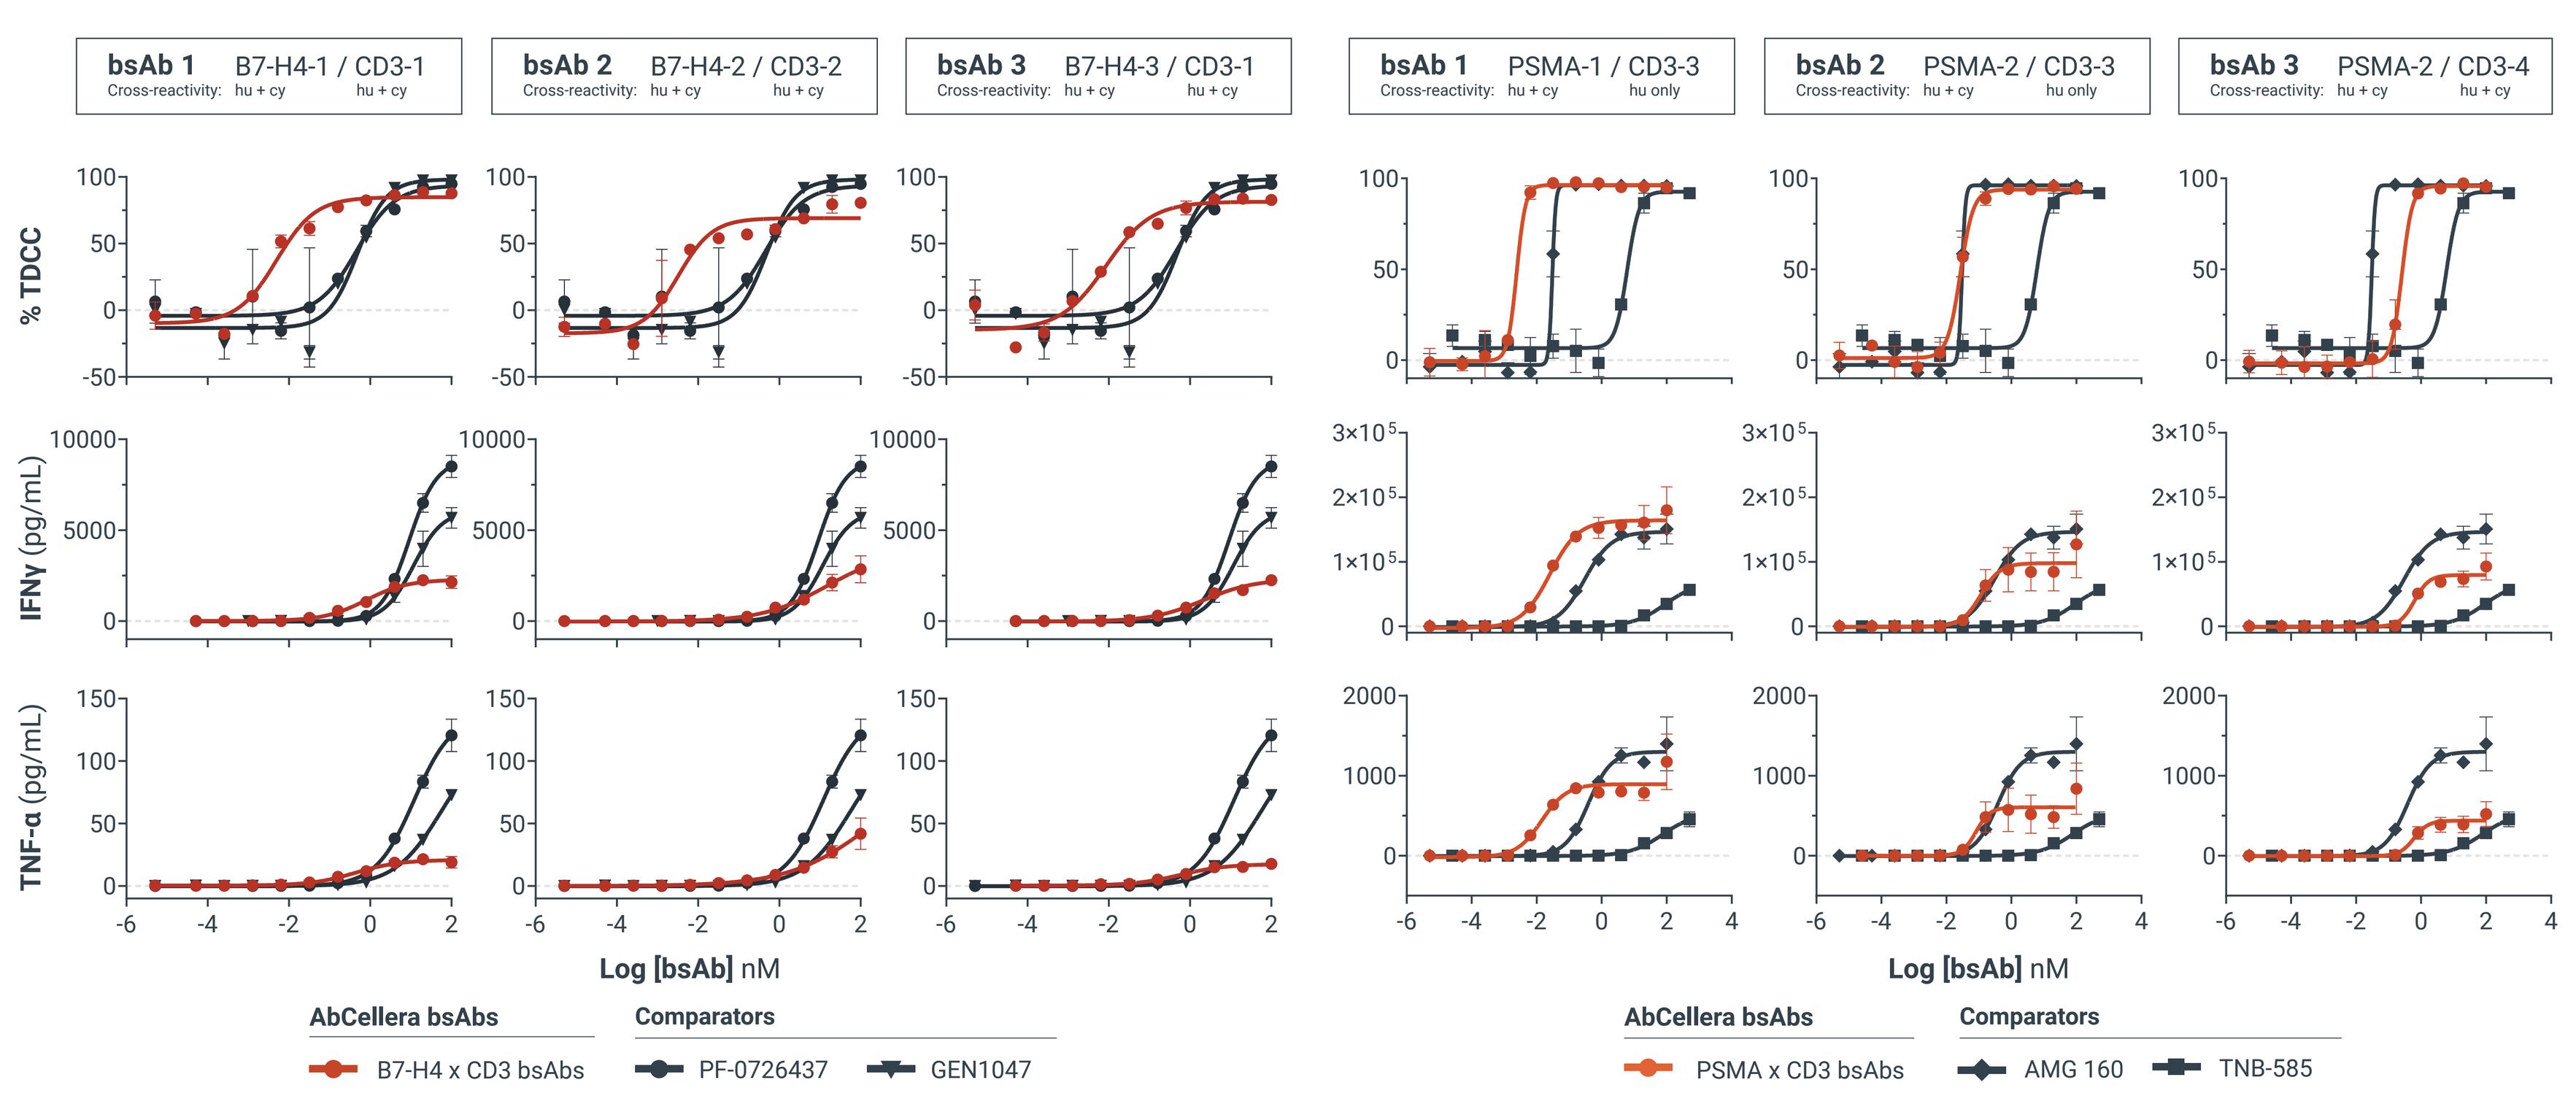

A B7-H4 x CD3 pan-T : HCC1954 cells (8:1)
B PSMA x CD3 pan-T : C4-2 cells (5:1)
bsAb 1 B7-H4-1 / CD3-1
bsAb 2 B7-H4-2 / CD3-2
bsAb 3 B7-H4-3 / CD3-1
bsAb 1 PSMA-1 / CD3-3
bsAb 2 PSMA-2 / CD3-3
bsAb 3 PSMA-2 / CD3-4
% TDCC
IFNγ (pg/mL)
TNF-α (pg/mL)
Log [bsAb] nM
AbCellera bsAbs
B7-H4 x CD3 bsAbs
PSMA x CD3 bsAbs
Comparators
PF-0726437
GEN1047
AMG 160
TNB-585

Figure 2. B7-H4- and PSMA-targeted TCEs with unique CD3- and TAA-binding arms show differentiation from clinical benchmarks. **(A, B)** Function was assessed with a T-cell-dependent cellular cytotoxicity (TDCC) assay using human pan-T cells incubated with target cells for 72 hours.

TAA arm bound to PSMA

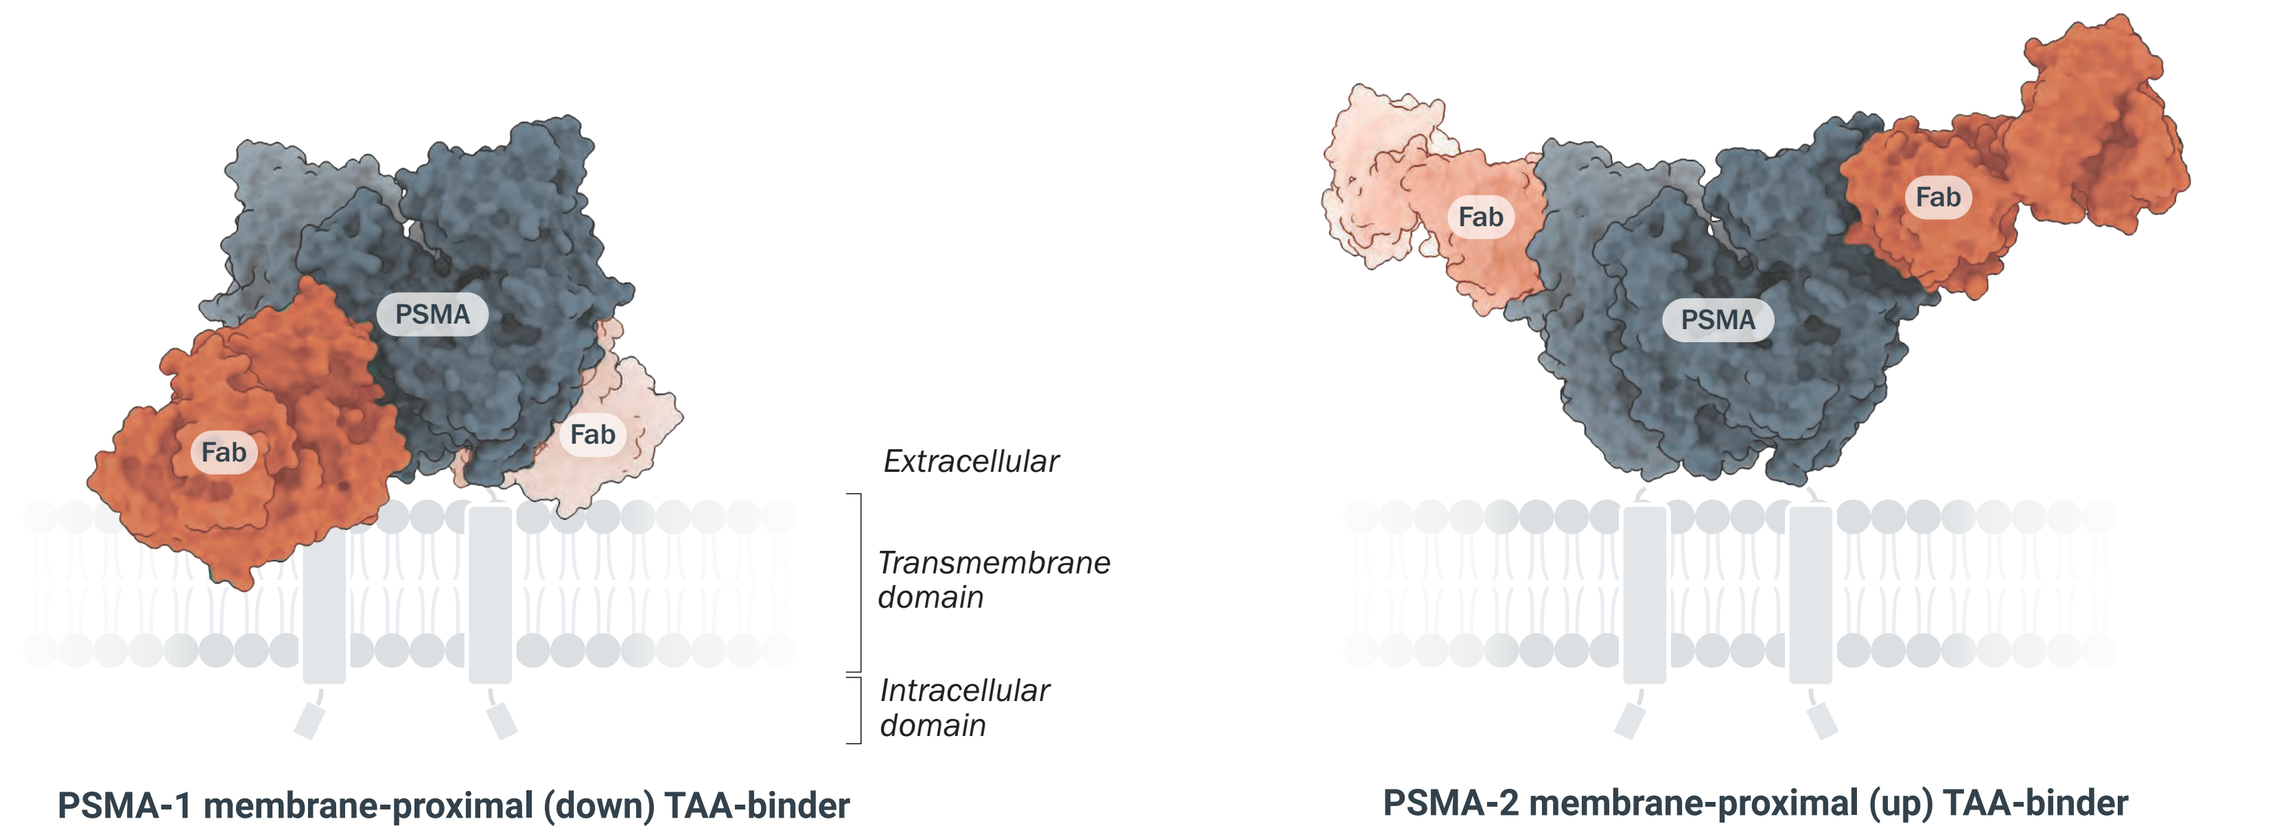

C Cryo-EM structure of Fab domains of PSMA x CD3 bsAbs 1-3
PSMA
Fab
Extracellular
Transmembrane domain
Intracellular domain
PSMA-1 membrane-proximal (down) TAA-binder
PSMA-2 membrane-proximal (up) TAA-binder

Figure 2. (C) PSMA-binding arms that yield TCEs with different functional profiles show membrane-proximal binding in distinct orientations. Antibody-antigen complex structures were generated using a size-exclusion chromatography-purified complex and cryo-electron microscopy.

Molecules to selectively recruit γδ T cells to tumor targets

Diverse TCR γδ1- and γδ2-binding antibodies for TCE development

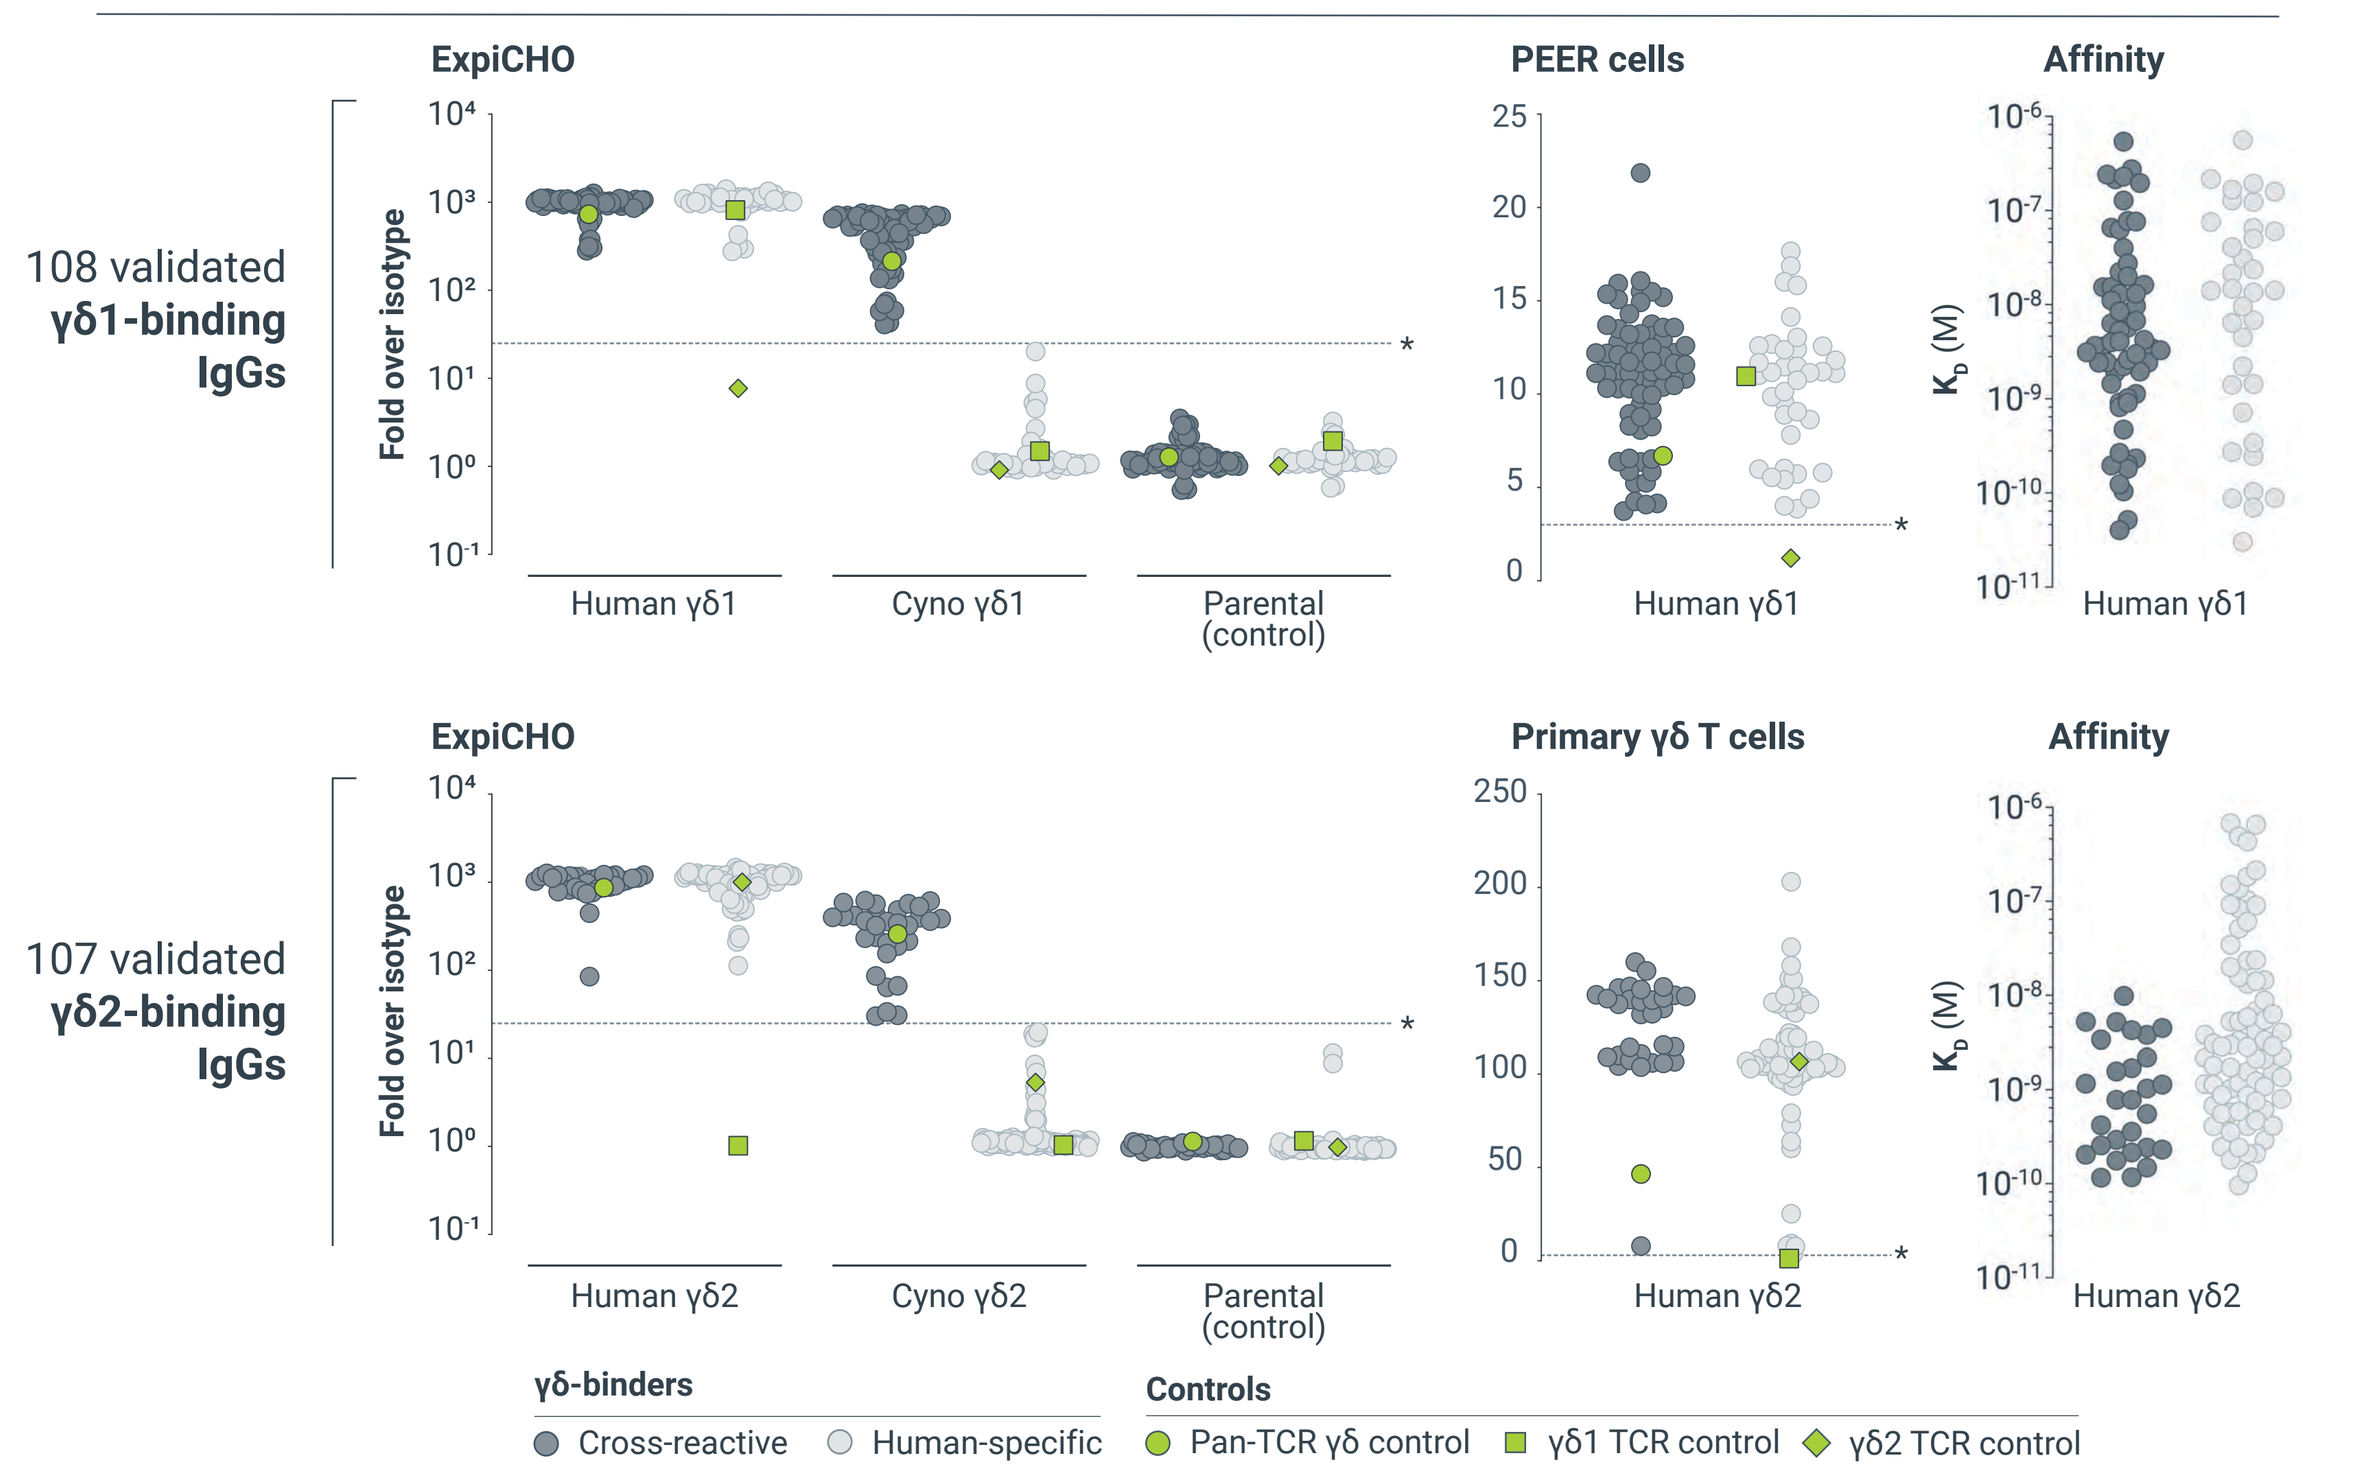

A Hundreds of human/cyno cross-reactive binders
108 validated γδ1-binding IgGs
107 validated γδ2-binding IgGs
ExpiCHO
PEER cells
Primary γδ T cells
Affinity
Fold over isotype
Human γδ1
Cyno γδ1
Human γδ2
Cyno γδ2
Parental (control)
γδ-binders
Cross-reactive
Human-specific
Controls
Pan-TCR γδ control
γδ1 TCR control
γδ2 TCR control

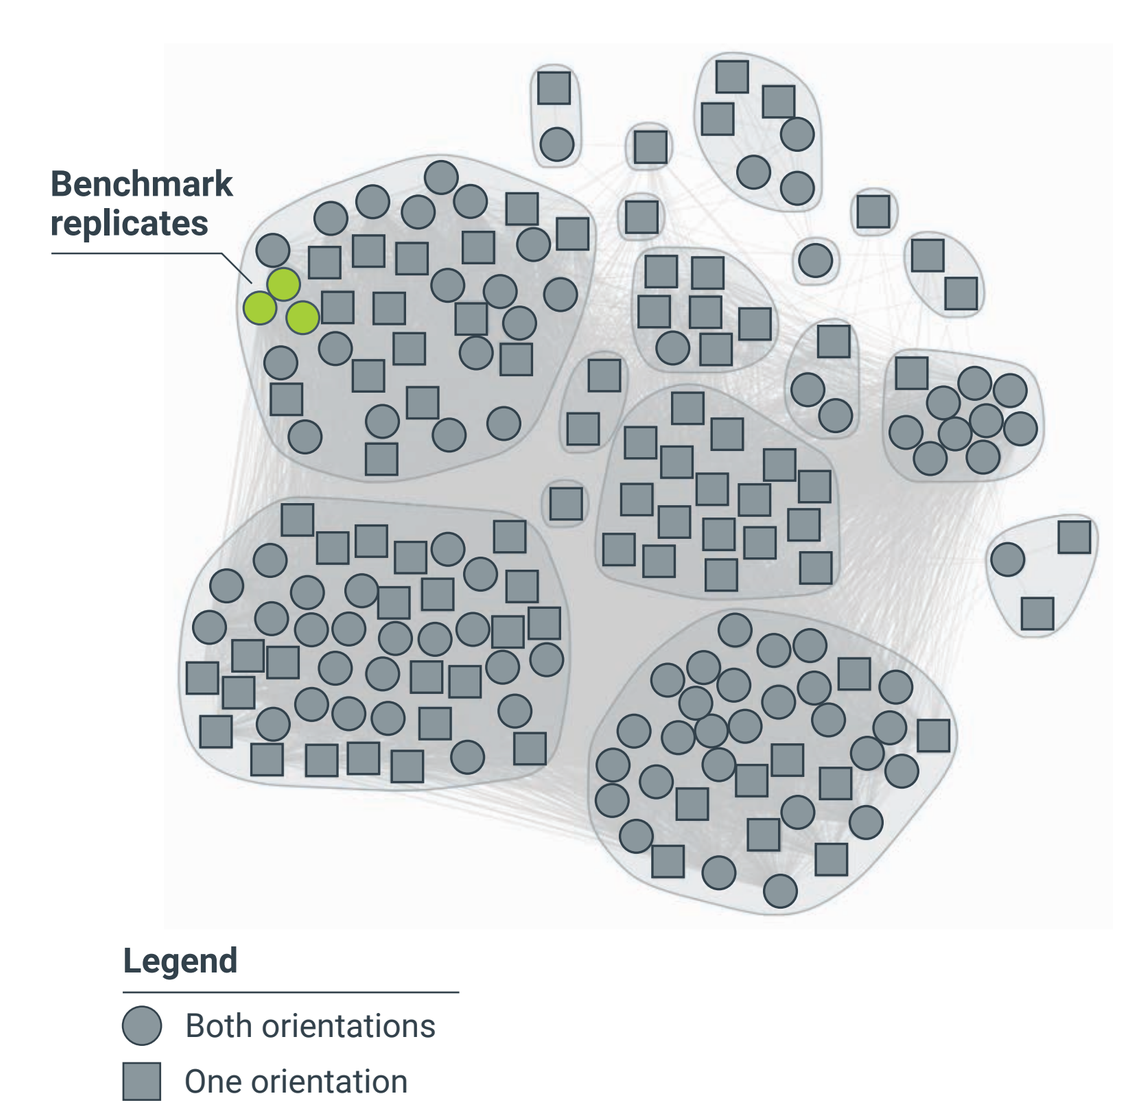

B Broad γδ2 epitopic diversity
17 epitope communities
Benchmark replicates
Legend
Both orientations
One orientation

Figure 3. IgG γδ1/2 antibodies with human and cyno cross-reactivity. **(A)** 184 human-specific and cyno cross-reactive γδ1/2 antibodies were assessed for binding. 71 γδ1 and 31 γδ2 antibodies were human/cyno cross-reactive, with no binding to parental ExpiCHO cells. All γδ1/2 antibodies showed binding to cell lines endogenously expressing γδ1 (PEER) and γδ2 (primary γδ T cells). Antibodies showed a range of affinities by SPR. Preliminary analytical and biophysical characterization showed favorable developability properties (data not shown). **(B)** γδ2-binding antibodies cluster in 17 different epitope communities as determined by ward.D2 hierarchical clustering based on competition events. γδ1-binding antibodies clustered into nine communities (data not shown).

*Dotted line indicates positive binding threshold.

Costimulatory molecules to enhance anti-tumor activity

CD28 monoclonal antibodies that activate T cells without superagonism

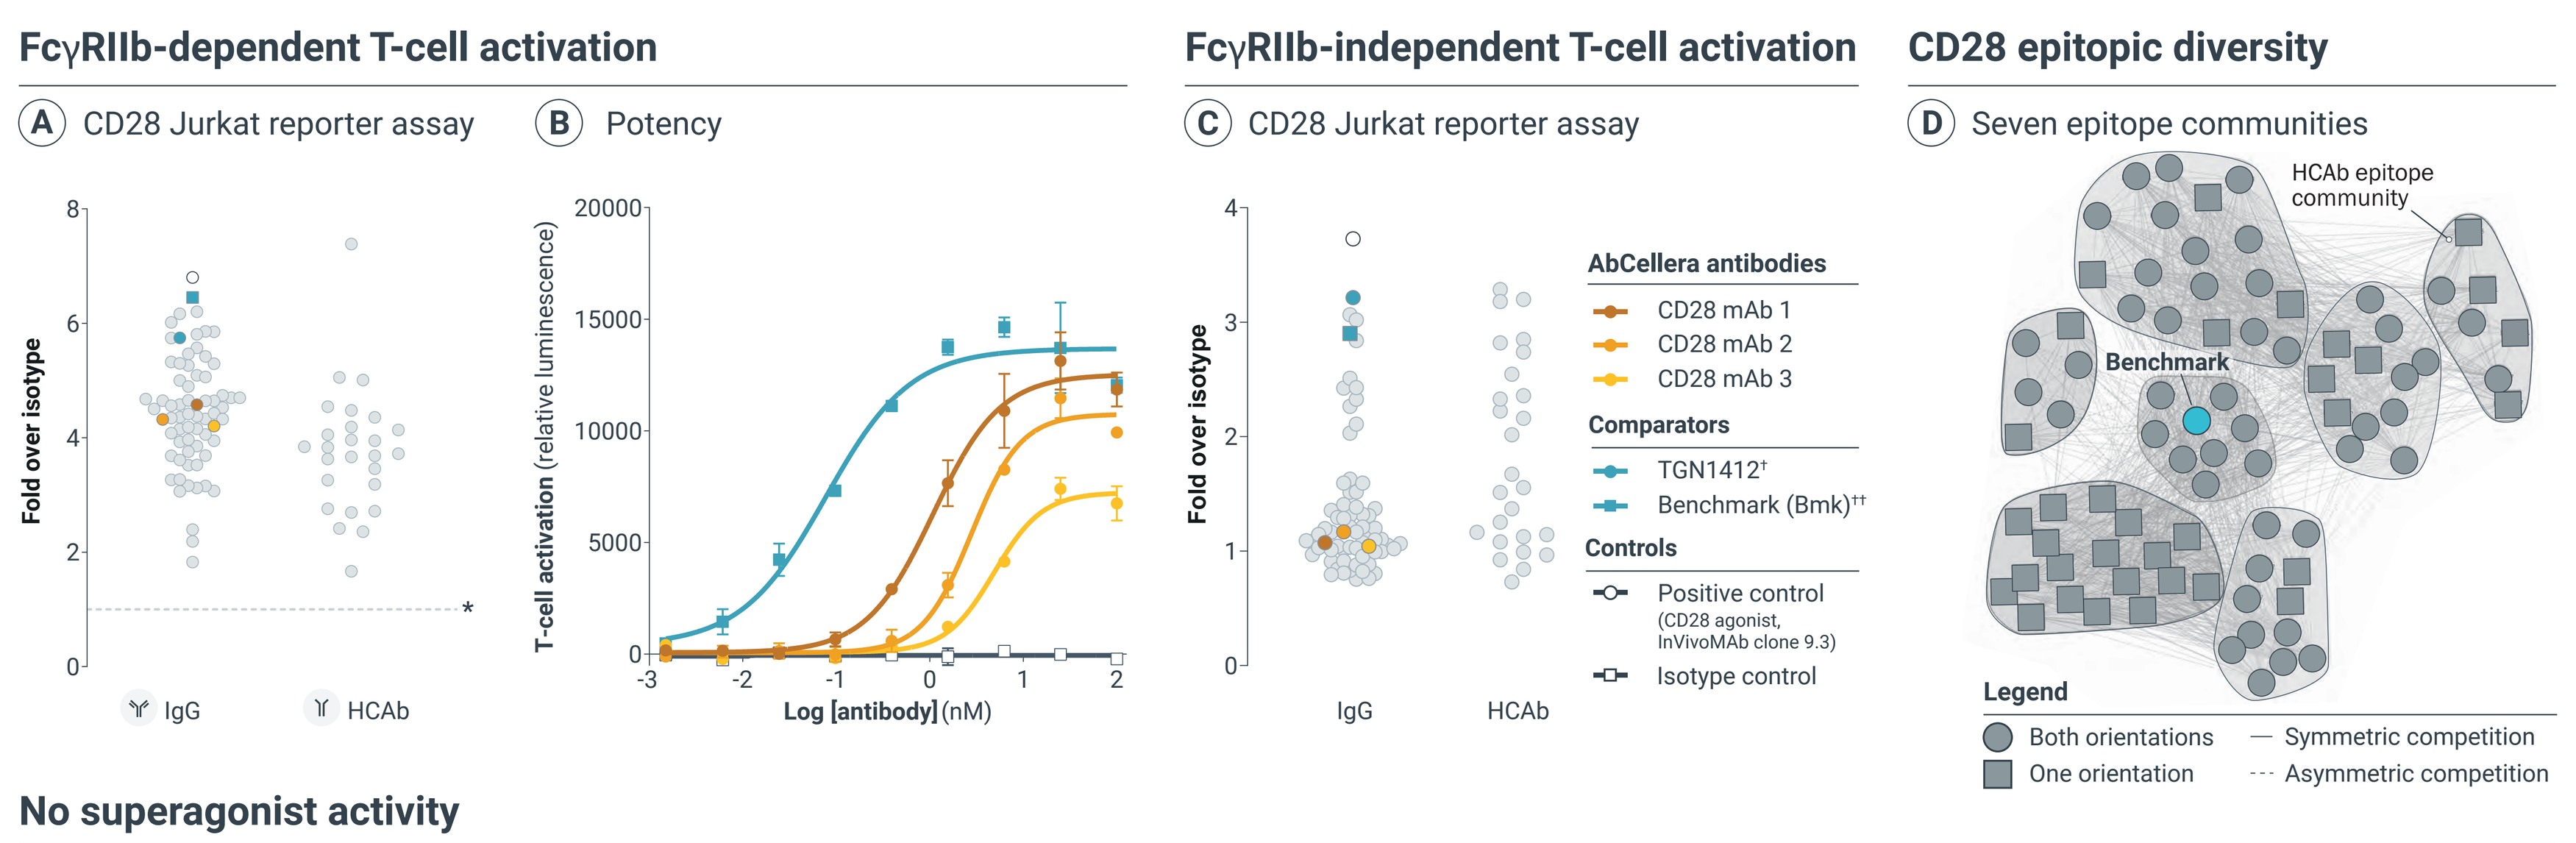

FcγRIIb-dependent T-cell activation
A CD28 Jurkat reporter assay
B Potency
FcγRIIb-independent T-cell activation
C CD28 Jurkat reporter assay
CD28 epitopic diversity
D Seven epitope communities
HCAb epitope community
Benchmark
Fold over isotype
T-cell activation (relative luminescence)
Log [antibody] (nM)
IgG
HCAb
AbCellera antibodies
CD28 mAb 1
CD28 mAb 2
CD28 mAb 3
Comparators
TGN1412†
Benchmark (Bmk)††
Controls
Positive control (CD28 agonist, InVivoMAb clone 9.3)
Isotype control
Legend
Both orientations
One orientation
Symmetric competition
Asymmetric competition

Figure 4. CD28-binding antibodies are conditional agonists with diverse functional activities and epitopes. **(A)** Antibodies (formatted as IgG1 Fc bivalents) were incubated with effector cells (endogenously expressing TCR, CD3, and CD28) and CHO-K1 cells (expressing FcγRIIb and a TCR-engaging protein) to assess FcγRIIb-dependent activation. Example antibodies with diverse functional responses are highlighted. **(B)** Titration series for example antibodies 1-3 are shown. **(C)** FcγRIIb-independent agonist activities were similarly assessed using CHO-K1 cells expressing only a TCR-engaging protein. **(D)** The majority of antibodies competed with the benchmark for binding to CD28, with the exception of molecules in the HCAb epitope community. Antibodies showed diverse binding avidities (870 pM - 6.40 μM), measured using the CD28 extracellular domain by SPR (data not shown).

*Fold over isotype ≥ 1 considered positive for binding.
†TGN1412: Theralizumab is a known CD28 superagonist molecule.
††A bivalent monoclonal CD28 benchmark antibody was generated using CD28 seqeuences derived from REGN5678 patent literature.

No superagonist activity

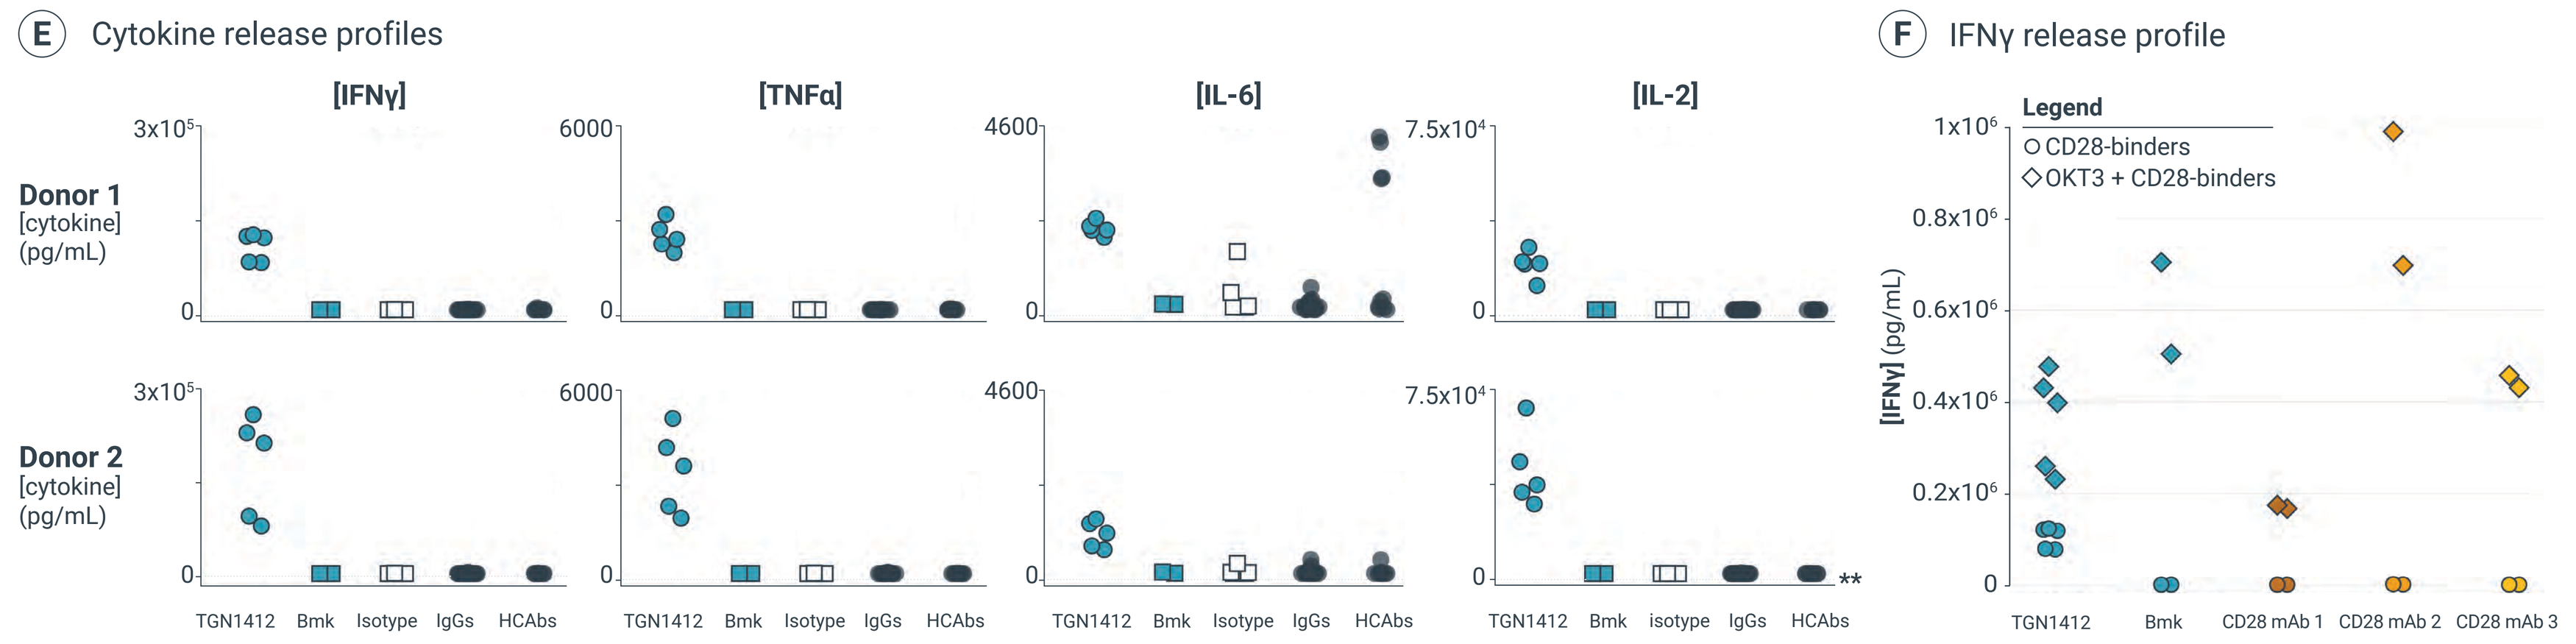

E Cytokine release profiles
[IFNγ]
[TNFα]
[IL-6]
[IL-2]
Donor 1 [cytokine] (pg/mL)
Donor 2 [cytokine] (pg/mL)
TGN1412
Bmk
Isotype
IgGs
HCAbs
F IFNγ release profile
Legend
CD28-binders
OKT3 + CD28-binders
[IFNγ] (pg/mL)
CD28 mAb 1
CD28 mAb 2
CD28 mAb 3

Figure 4. (E) PBMCs from five donors were cultured with wet-coated antibodies as previously described[5] and cytokine profiles of two representative donors are shown. **(F)** In a PoC study for MHC-independent costimulation, PBMCs were cultured with plate-bound CD28- and soluble CD3-binding antibodies. A single PBMC donor and representative cytokine is shown.

**Values below the lower limit of detection are shown at y=0.

Functional 4-1BB-binding antibodies, including ligand-blocking and non-blocking binders

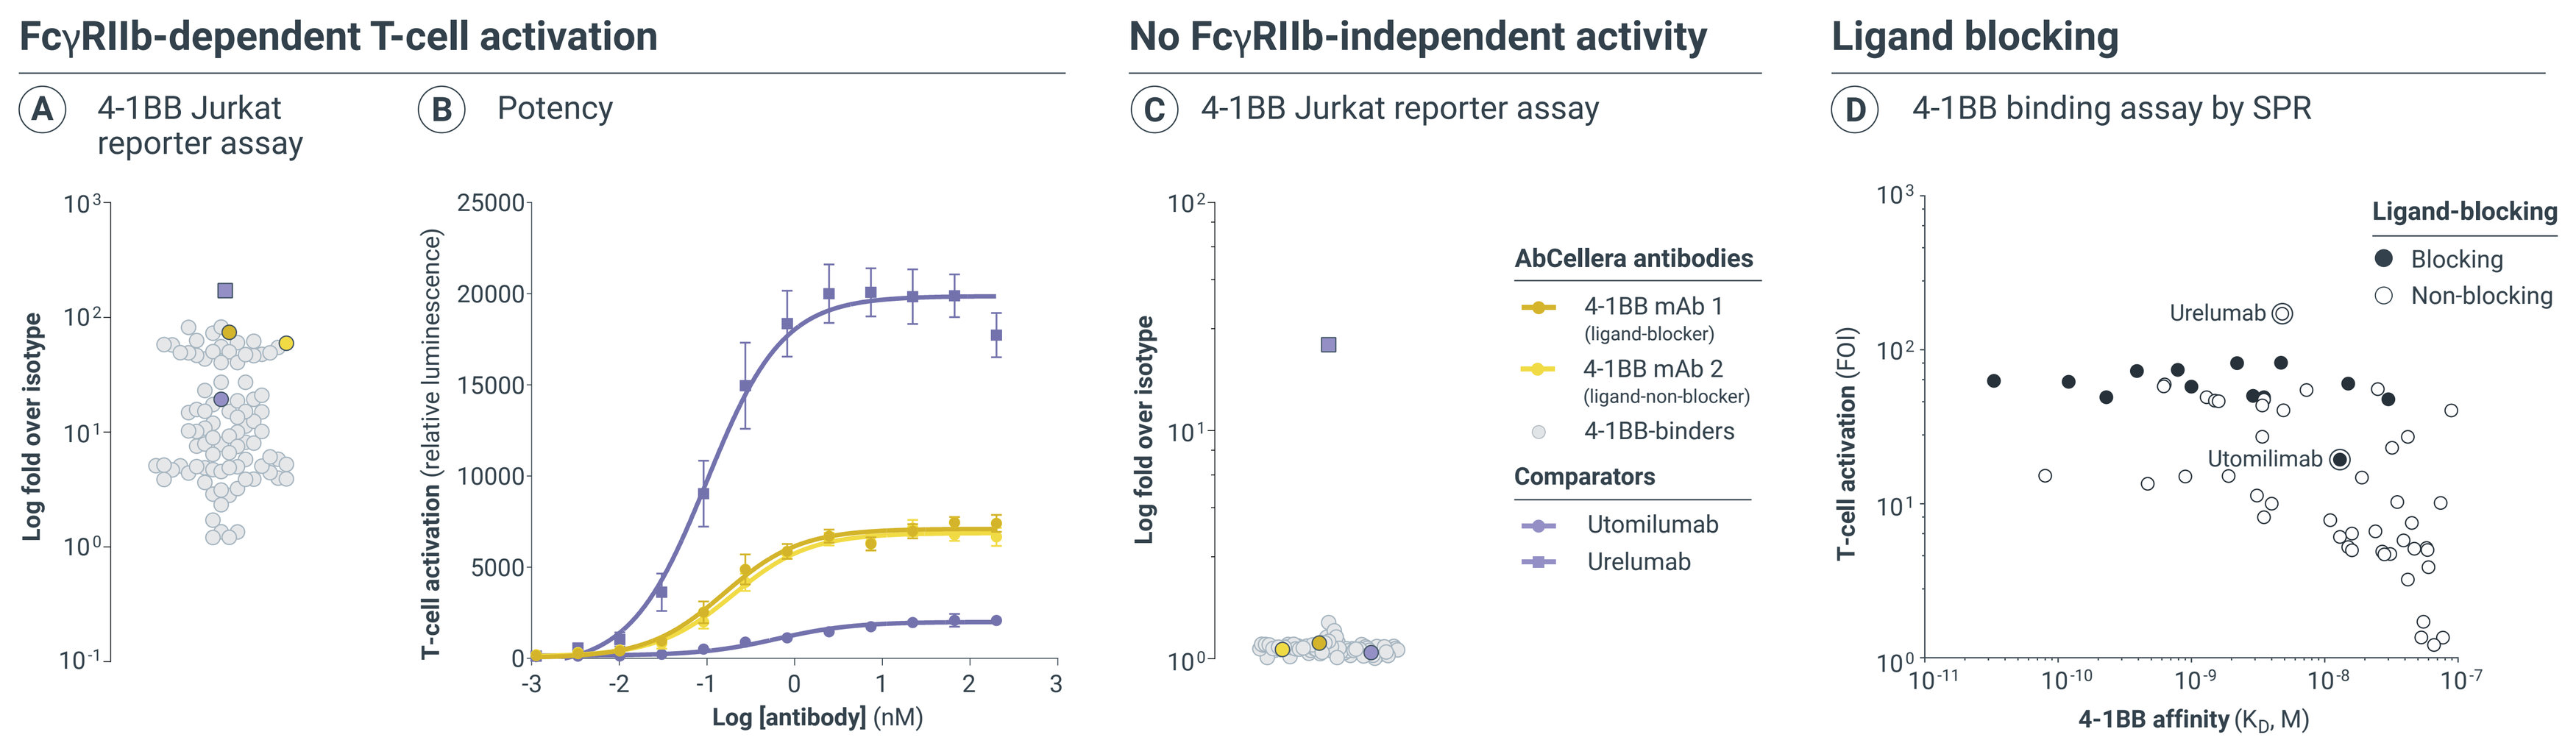

FcγRIIb-dependent T-cell activation
A 4-1BB Jurkat reporter assay
B Potency
No FcγRIIb-independent activity
C 4-1BB Jurkat reporter assay
Ligand blocking
D 4-1BB binding assay by SPR
Log fold over isotype
T-cell activation (relative luminescence)
Log [antibody] (nM)
AbCellera antibodies
4-1BB mAb 1 (ligand-blocker)
4-1BB mAb 2 (ligand-non-blocker)
4-1BB-binders
Comparators
Utomilumab
Urelumab
Ligand-blocking
Blocking
Non-blocking
T-cell activation (FOI)
4-1BB affinity (K_D, M)

Figure 5. Diverse 4-1BB ligand-blockers and non-blockers. **(A)** Antibodies (formatted as IgG1 Fc bivalents) activated reporter cells in a FcγRIIb-dependent manner with a range of potencies. **(B)** Example titration series for three 4-1BB- binding antibodies are shown. **(C)** No FcγRIIb-independent activation of reporter cells was observed. **(D)** Range of affinities and FcγRIIb-dependent activation of Jurkat T cells observed among 4-1BBL-blocking (12) and non-blocking (44) antibodies.

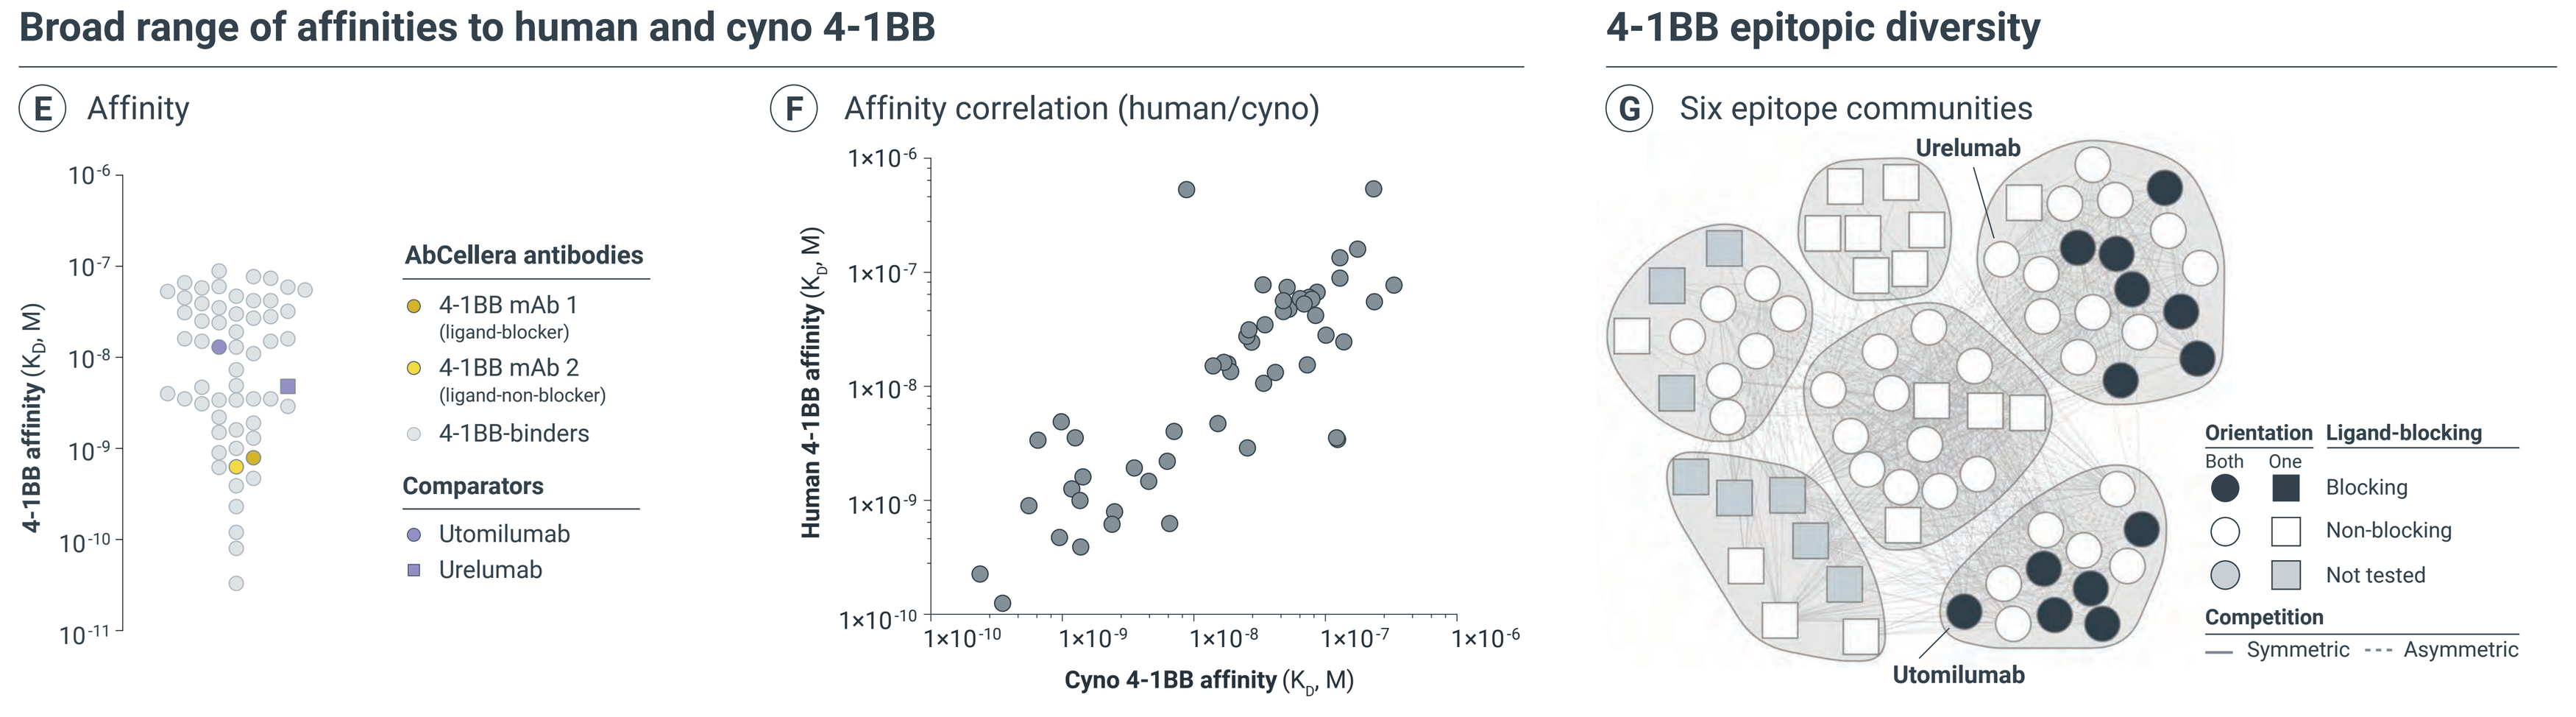

Broad range of affinities to human and cyno 4-1BB
E Affinity
F Affinity correlation (human/cyno)
4-1BB epitopic diversity
G Six epitope communities
4-1BB affinity (K_D, M)
Human 4-1BB affinity (K_D, M)
Cyno 4-1BB affinity (K_D, M)
Urelumab
Utomilumab
Orientation Both One
Ligand-blocking
Blocking
Non-blocking
Not tested
Competition
Symmetric
Asymmetric

Figure 5. (E) 65 antibodies showed human and cyno cross-reactivity **(F)** with similar affinities across the two species homologs. **(G)** Epitope community analysis revealed high epitope diversity, with blocking antibodies clustering into the same communities.

A comprehensive toolkit to tailor design of T-cell engagers for difficult-to-treat indications

STRATEGIES

Widen the therapeutic window	Enhance efficacy
☐ Novel CD3-binders for optimized T-cell engagement ☐ Diverse γδ TCR-binding antibodies ☐ High-throughput TAA-binder discovery ☐ Diverse formats, including IgG and HCAb	☐ CD28 and 4-1BB costimulation ☐ Multispecific engineering

These strategies can be used to fine-tune TCE parameters known to impact clinical efficacy and safety, including affinity, epitope diversity/binding, T-cell activation, and persistence.

PROGRAMS

AbCellera-led T-cell engager programs

Oncology
PSMA, B7-H4, and undisclosed targets

Autoimmune
CD19

We are leveraging the breadth of this platform to design TCEs with optimized combinations of TAA-, CD3-, and costimulatory-binders tailored to tumor target, to ultimately advance programs for difficult-to-treat indications.

Next steps in our TCE programs include:

- Additional preclinical functional characterization including *in vitro* assays and *in vivo* efficacy studies
- Moving CD28- and 4-1BB-binders into monovalent formats for further testing
- Engineering and assessment of novel bi- and multispecific costimulatory TCEs

COPYRIGHT © ABCELLERA