1367

A rational approach for selecting CD3-binding antibodies for T-cell engager development

AUTHORS

Valentine de Puyraimond†, Patrick Rowe†, Juntao (Matt) Mai†, Phillipe Pouliot, Kate Caldwell, Lauren Clifford, Allie Goodman, Patrick Farber, Harveer Dhupar, Davide Tortora, Peter Bergqvist, Janice Reimer, Karine Herve, Vivian Li, Irene Yu, John Marwick, Stefania Carrarra, Nathalie Blamey, Melissa Cid, Cindy-Lee Crichlow, Lena M. Bolten, Tova Pinsky, Gesa Volkers, Jessica Fernandes Scortecci, Girija Bodhankar, Caitlyn De Jong, Sohpie Cullen, Ping Xiang, Courtenay Lai, Ahn Lee, Gabrielle Conaghan, Antonios Samiotakis, Stefan Hannie, Rhys Chappell, Emma Lathouwers, Lauren Chong, Kate Gibson, Stephanie K. Masterman, Kirstin Brown, Kelly Bullock, Raffi Tonikian, Lindsay DeVorkin, Sherie Duncan, Aaron Yamniuk, Kush Dalal, Tim M. Jacobs, **Bryan C. Barnhart***

† *these authors contributed equally* * *presenter*

AUTHOR AFFILIATION

AbCellera, Vancouver, Canada

DOWNLOAD POSTER

OUR PLATFORM

Diverse CD3-binding antibodies for TCE development

Effective CD3 T-cell engagers (TCEs) must balance potency with potential toxicities by maximizing tumor-cell killing while fine-tuning cytokine release. However, most TCEs in clinical development are derived from a small number of CD3-binding antibodies, such as SP34-2[1], limiting the number of antibodies available for optimal immunological synapse formation.

To address these barriers, AbCellera developed a complete TCE platform: a discovery engine to identify diverse tumor-binding antibodies; a portfolio of fully human, CD3-binding antibodies that are distinct from SP34-2; a clinically validated multispecific engineering platform; and a high-throughput strategy to rapidly identify TCE candidates[2-4].

THE CHALLENGE

CD3-binding antibodies result in diverse TCE functional profiles

Previously, we demonstrated that identical CD3-binders result in variable function when paired with different tumor-binding arms[4]. However, due to the limited number of CD3-binding antibodies that have been assessed in the field, the full extent to which CD3-binding parameters impact TCE function is not well understood.

To address this, we designed and engineered our largest panel of TCEs to date. We paired hundreds of CD3-binders with a single tumor-binding paratope and assessed bispecifics in high-throughput functional assays. We performed a clustering analysis to identify sets of CD3-binders that result in similar function as bispecifics, and developed a process that leverages these functional clusters to streamline selection of CD3-binders for different tumor targets.

OUR APPROACH

A streamlined approach for identifying the optimal CD3-binder for any tumor target

We integrated large antibody datasets to gain insights into parameters that impact TCE function. We combined these with our previous data demonstrating the impact of the tumor-binding arm on TCE function and developed a strategy to rationally select tumor- and CD3-binding antibodies for each target. We leveraged these data to select TCEs for further assessment for multiple programs, two of which are shown here.

Multiple CD3-binding parameters impact TCE function

CD3-binding kinetics impact tumor-cell killing and cytokine release

(A) We engineered a panel of TCEs to assess the impact of the CD3-binding arm on function

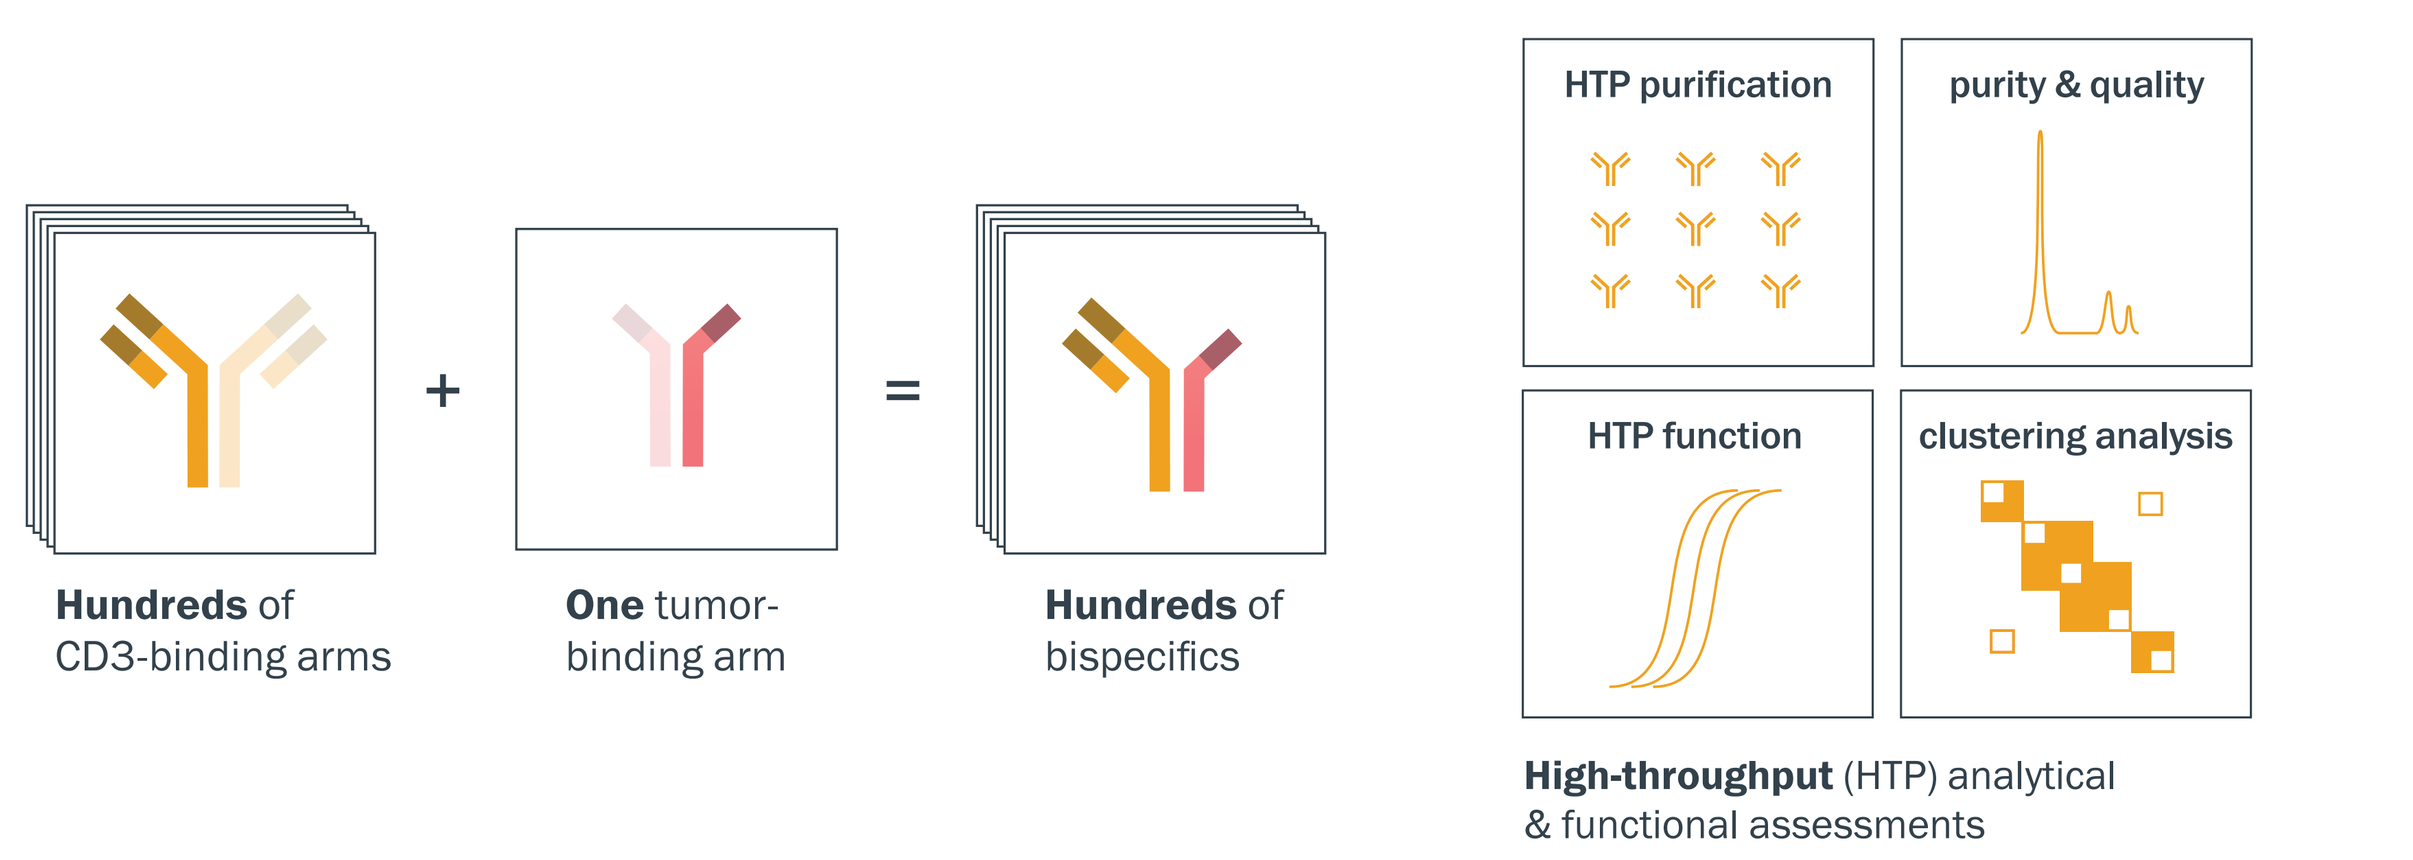

Hundreds of CD3-binding arms
One tumor-binding arm
Hundreds of bispecifics
HTP purification
purity & quality
HTP function
clustering analysis
High-throughput (HTP) analytical & functional assessments

(B) We compared CD3-binding kinetics to TCE function

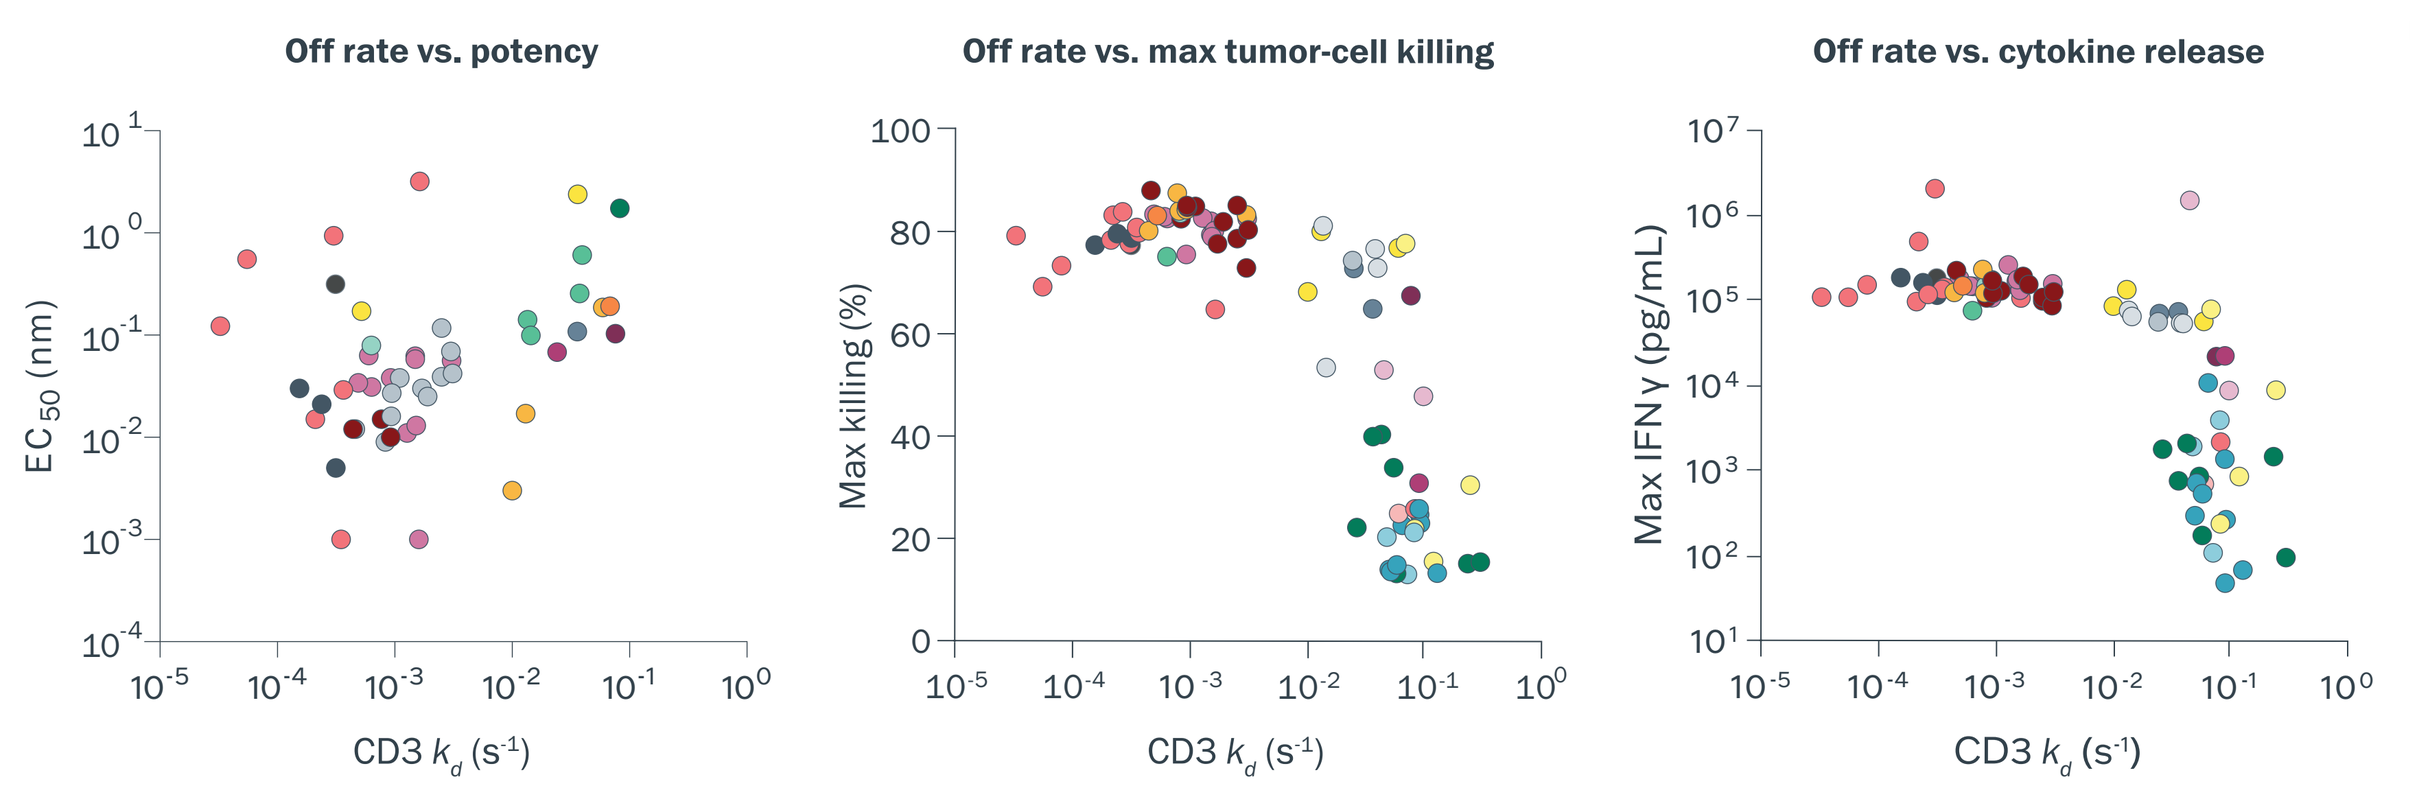

Colored by CD3-binding antibody clonal family
Off rate vs. potency
Off rate vs. max tumor-cell killing
Off rate vs. cytokine release
EC50 (nm)
Max killing (%)
Max IFNγ (pg/mL)
CD3 kd (s-1)

Figure 1. Hundreds of bispecifics were engineered to assess the impact of the CD3-binding arm on TCE function. (A) AbCellera's CD3-binding antibodies[2-4] were paired with a single tumor-binding arm. The resulting bispecifics were expressed and analyzed using high-throughput functional assays. **(B)** Binding kinetics for the human CD3 εδ subunit were compared to TCE potency, tumor-cell killing, and cytokine release. Because off rate was found to be the primary driver of affinity, and release of TNFα and IL-2 was consistent with that of IFNγ, these parameters are shown. CD3-binding antibody clonal family (defined as antibodies sharing the same V gene, J gene, CDR3 length, and animal origin for both heavy and light chains) is overlaid using color. Off rate was found to be a significant contributor to cytokine release and tumor-cell killing, but was not sufficient to fully describe these properties, particularly at faster off rates. In contrast, potency was not tightly correlated with either off rate or clonal family.

Binding kinetics alone cannot predict TCE function

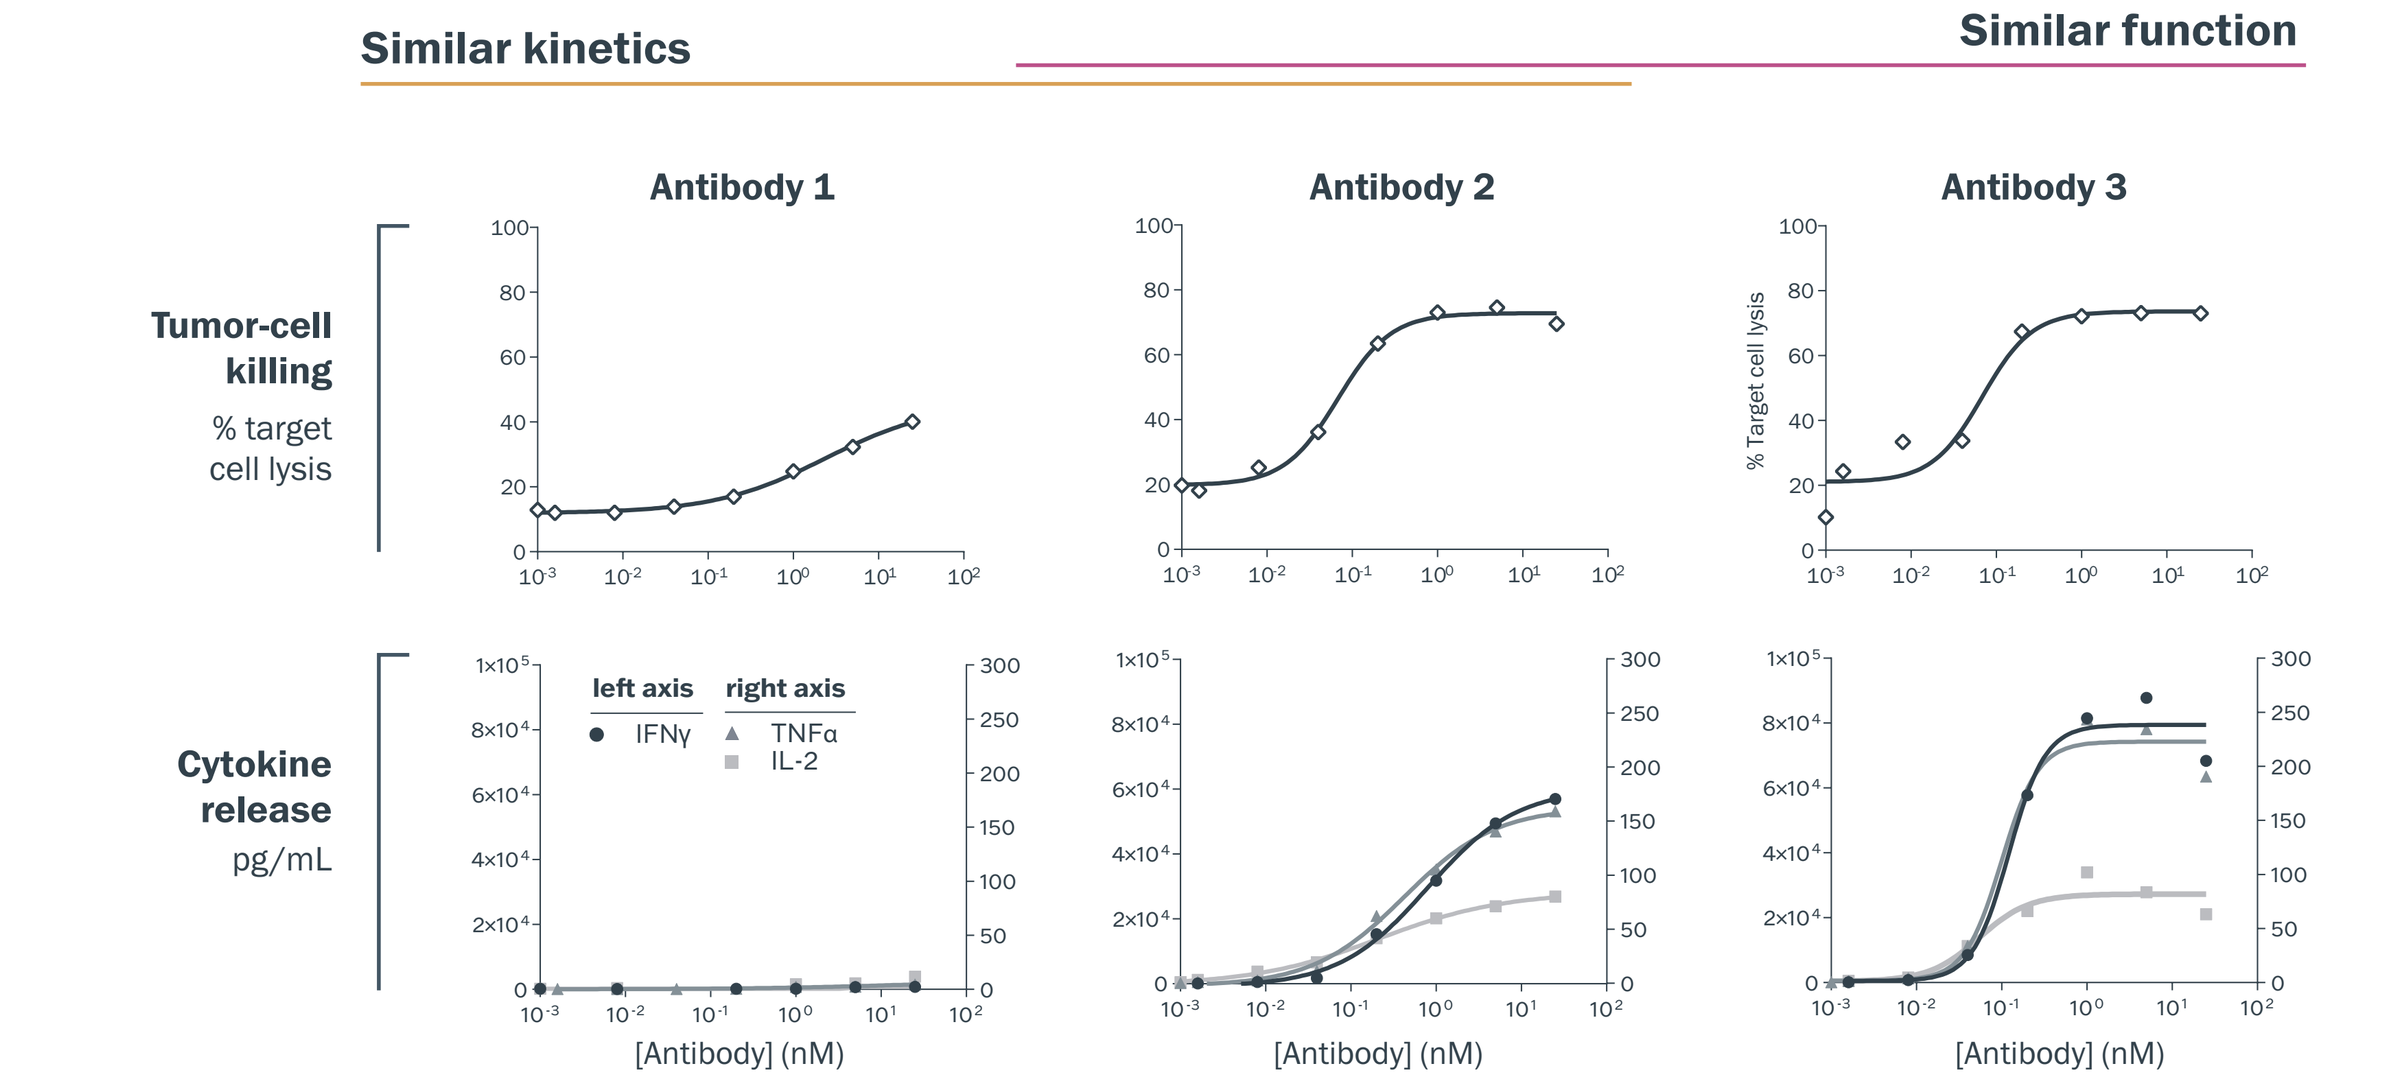

Similar kinetics
Similar function
Antibody 1
Antibody 2
Antibody 3
Tumor-cell killing
% target cell lysis
Cytokine release
pg/mL
left axis
IFNγ
right axis
TNFα
IL-2
[Antibody] (nM)

	Antibody 1	Antibody 2	Antibody 3
CD3-binding properties			
Affinity (nM)	320	600	38
Off rate (s^{-1})	4×10^{-2}	2×10^{-2}	3×10^{-3}
Subunit	epsilon	epsilon	epsilon
TCE properties			
Max killing (%)	40	75	73
Max IFNγ (pg/mL)	772	5.7×10^4	8.6×10^4
Max TNFα (pg/mL)	5	160	243
Max IL-2 (pg/mL)	12	81	102
Functional cluster*	2	3	3

Figure 2. Diverse functional profiles were observed despite similar kinetic parameters. Tumor-cell killing and cytokine release curves for three example TCEs are shown. The CD3-binding properties and functional readouts for each are summarized in the table below. Antibodies 1 and 2 have similar binding kinetics but display distinct functional profiles, while antibodies 2 and 3 have similar functional profiles but different binding kinetics. All three antibodies are from different clonal families.

*Functional clustering is described in Figure 3.

Select functionally diverse CD3-binding antibodies

AbCellera's CD3-binding antibodies can be clustered into 19 groups based on function

(A) We normalized and categorized functional data

(B) Functional clustering analysis

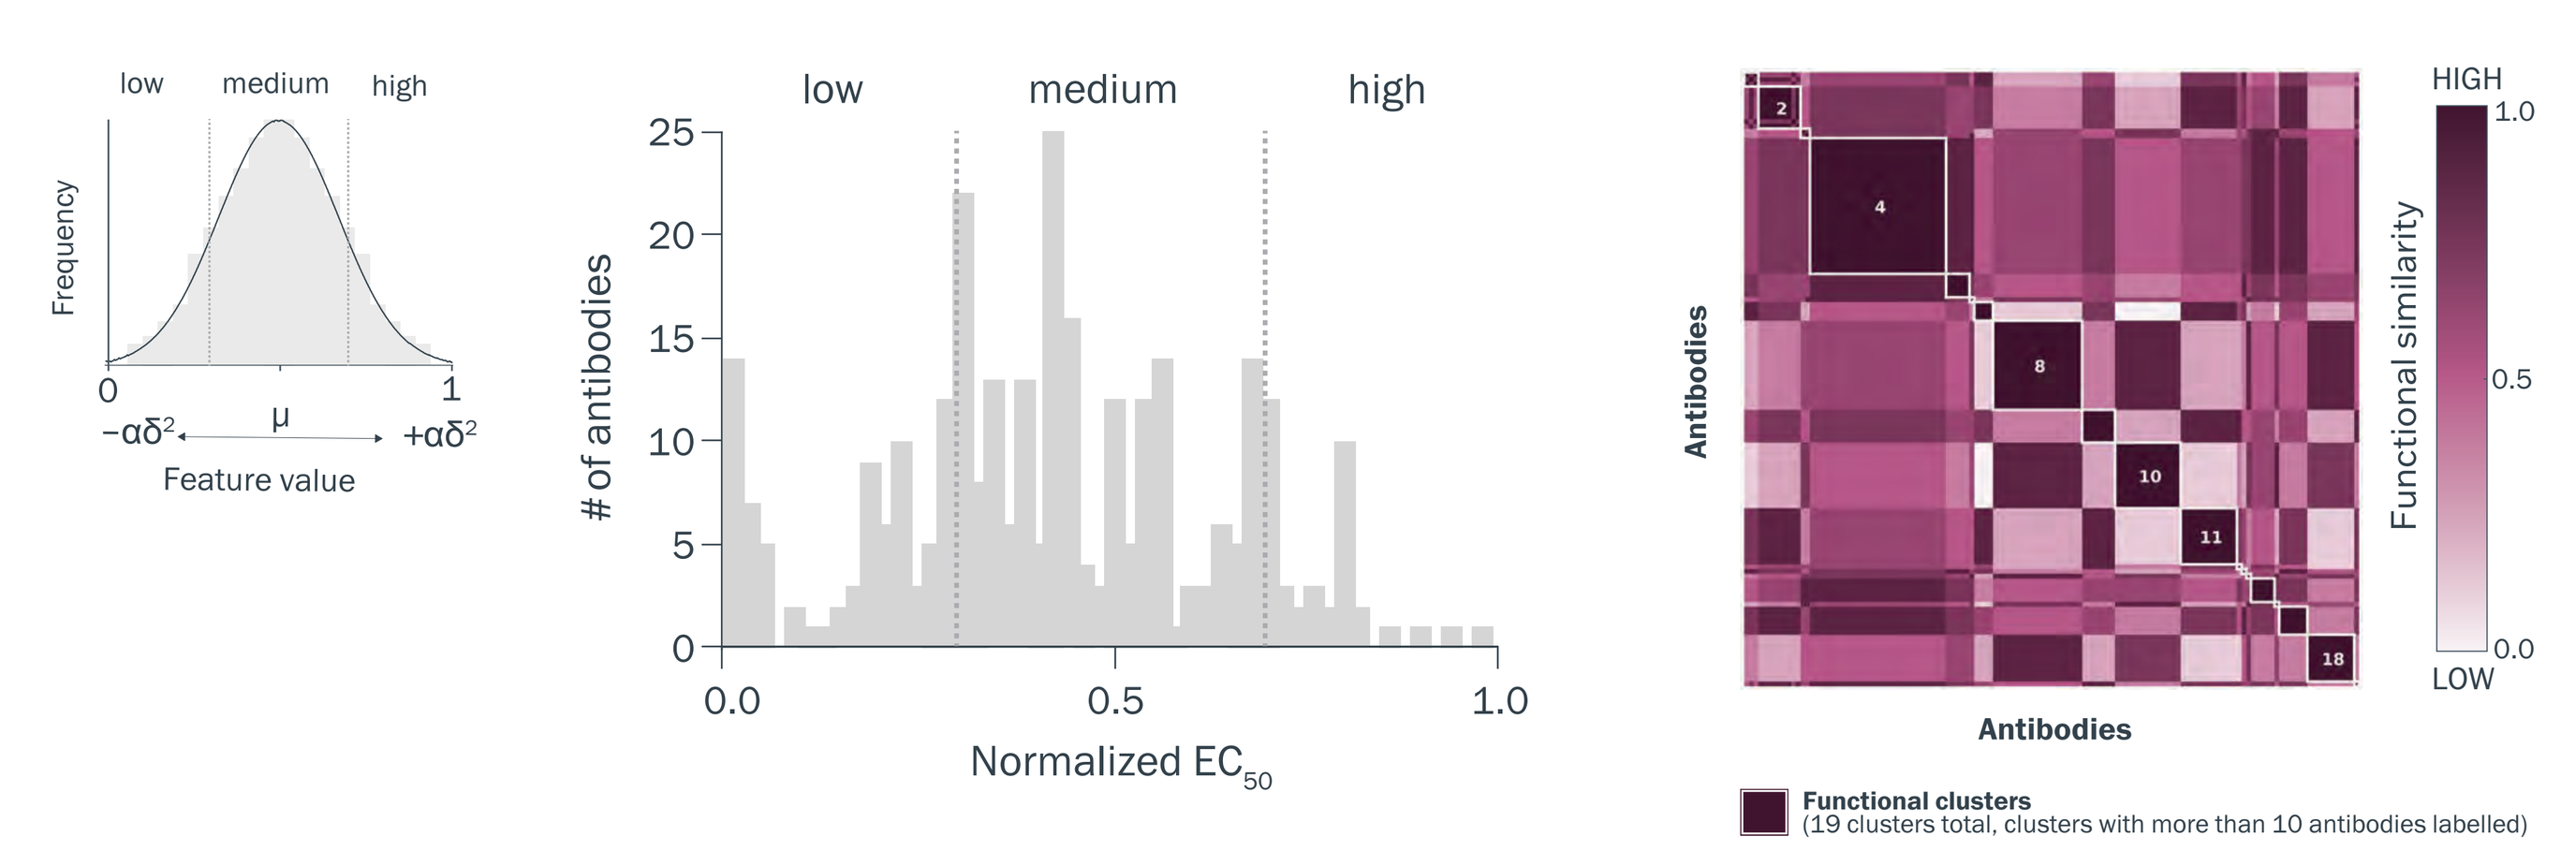

low
medium
high
Frequency
Feature value
of antibodies
Normalized EC50
Antibodies
Functional similarity
HIGH
LOW
Functional clusters
(19 clusters total, clusters with more than 10 antibodies labelled)

Figure 3. CD3-binding antibodies were clustered based on TCE functional profiles. (A) Tumor-cell killing and cytokine release data were normalized to range (0, 1) and antibodies were categorized as high, medium, or low based on their distance from the mean. Example normalized EC_{50} data is shown. **(B)** Antibodies were then clustered based on functional profiles as shown in the white boxes along the diagonal of the heatmap. Example antibodies can be seen in Figure 2, where antibodies 1 and 2 are in different clusters and antibodies 2 and 3 are in the same cluster.

Functional clusters streamline selection of CD3-binders with relevant diversity

(A) **Functional clusters**
CD3-binders with **distinct functional profiles** are paired with diverse tumor-binding arms.

(B) **Screening panel**
Hundreds of bispecifics are analyzed by **high-throughput** functional and biophysical assays.

(C) **Lead panel**
Dozens of lead panel TCEs are selected for **detailed** lead assessment and optimization.

(D) **Development candidates**
A few development candidates are selected for **IND-enabling** studies.

(E) **Clinical candidate**
One optimal TCE is selected for clinical trials.

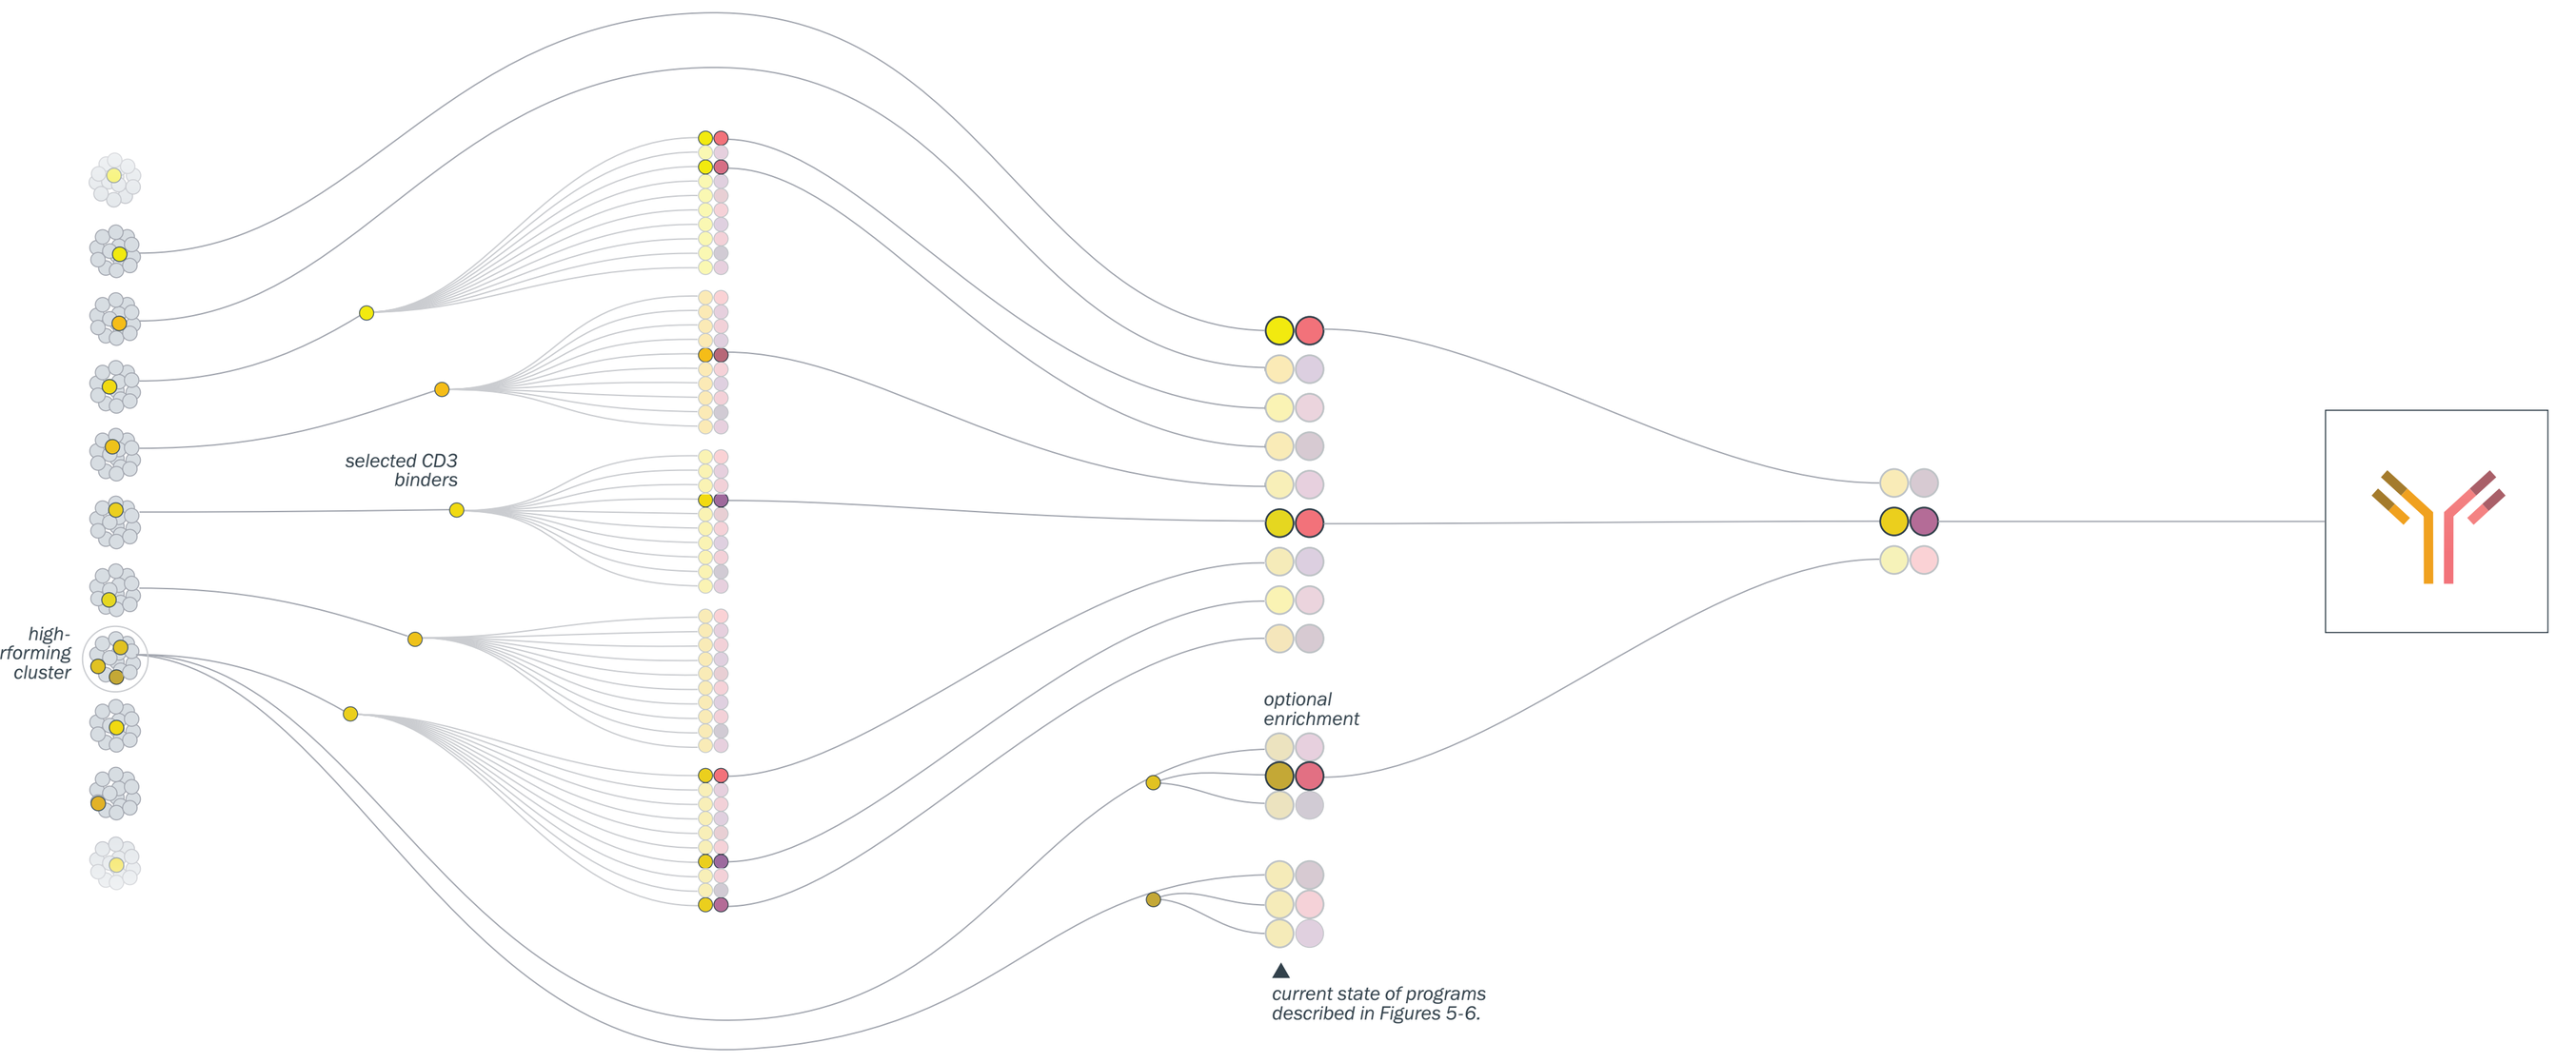

high-performing cluster
selected CD3 binders
optional enrichment
current state of programs described in Figures 5-6.

Figure 4. Functional clusters are used to select diverse CD3-binding antibodies for different tumor targets. As shown previously, the same CD3-binder can display different function depending on the tumor-binding arm it is paired with[4]. As a result, functional profiles for different tumor targets cannot be accurately predicted. To address this challenge, we developed a streamlined, function-first approach to identify optimal CD3- and tumor-binding pairs for each target.

(A) We maximize functional diversity upfront by pairing CD3-binders from distinct clusters with diverse tumor-binding arms, prioritizing desired properties such as cyno cross-reactivity and subunit specificities where needed.

(B) We engineer and analyze hundreds of bispecifics using high-throughput functional and biophysical assessments.

(C) We select dozens of lead panel antibodies based on function and developability. If needed, we can enrich the lead panel with CD3-binding arms from top-performing clusters.

(D) We analyze selected antibodies with robust functional, biophysical, analytical, stability, and structural assessments, and select ~1-3 development candidates for IND-enabling studies.

(E) Based on these studies, a final clinical candidate is selected for GMP manufacturing.

Identify the optimal pair of antibodies for each tumor target

Functionally diverse CD3-binders were used to engineer TCEs for two tumor targets

(A) AbCellera TCE program 1: PSMA x CD3

300+ CD3-binders » **19** functional clusters » **10** CD3-parentals

400+ PSMA-binders » **92** expressed & characterized » **9** PSMA-parentals

180+ bispecifics engineered

(B) AbCellera TCE program 2: MAGE-A4 x CD3

300+ CD3-binders » **19** functional clusters » **19** CD3-parentals

200 MAGE-A4-binders » **45** expressed & characterized » **6** MAGE-A4-parentals

200+ bispecifics engineered

Figure 5. (A) CD3-binding antibodies selected for our PSMA x CD3 program were prioritized based on cyno cross-reactivity. In addition to having functional diversity, antibodies had different subunit specificities and affinities ranging from 1 nM to 12 µM. We selected nine developable PSMA-binding antibodies from five different epitope bins with affinities ranging from 46 pM to 31 nM.

(B) Cyno cross-reactivity was not required for our MAGE-A4 x CD3 program, enabling selection from a broader range of functional profiles. The 19 selected CD3-binders ranged in affinity from 1 nM to 3 µM. We discovered developable antibodies with high specificity for a MAGE-A4 peptide-MHC[5] and selected six for bispecific engineering.

High-throughput assessment guided selection of TCEs for further assessment

(A) We selected nine PSMA x CD3 TCEs with functional profiles that span the range of clinical molecules

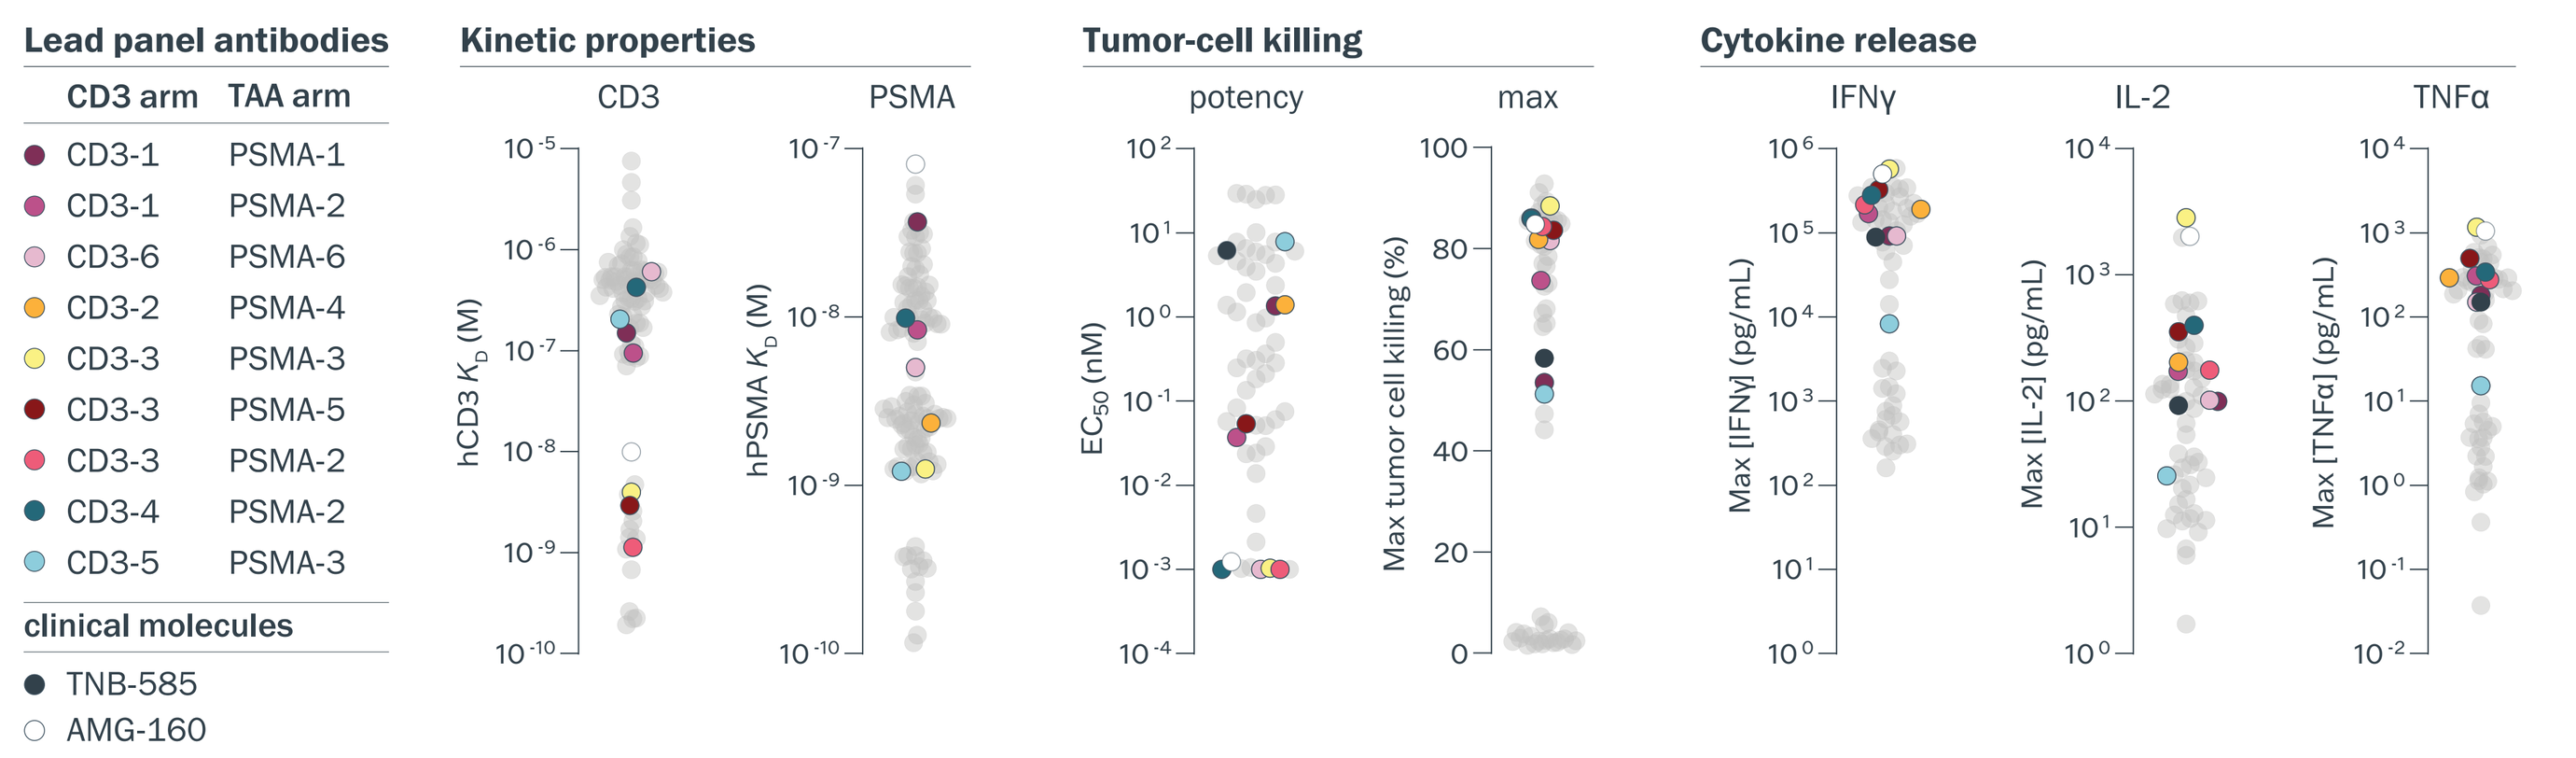

Lead panel antibodies
CD3 arm TAA arm
CD3-1 PSMA-1
CD3-1 PSMA-2
CD3-6 PSMA-6
CD3-2 PSMA-4
CD3-3 PSMA-3
CD3-3 PSMA-5
CD3-3 PSMA-2
CD3-4 PSMA-2
CD3-5 PSMA-3
clinical molecules
TNB-585
AMG-160
Kinetic properties
CD3
PSMA
hCD3 KD (M)
hPSMA KD (M)
Tumor-cell killing
potency
max
EC50 (nM)
Max tumor cell killing (%)
Cytokine release
IFNγ
IL-2
TNFα
Max [IFNγ] (pg/mL)
Max [IL-2] (pg/mL)
Max [TNFα] (pg/mL)

(B) We selected 12 MAGE-A4 x CD3 TCEs with desired functional profiles and peptide-MHC specificity

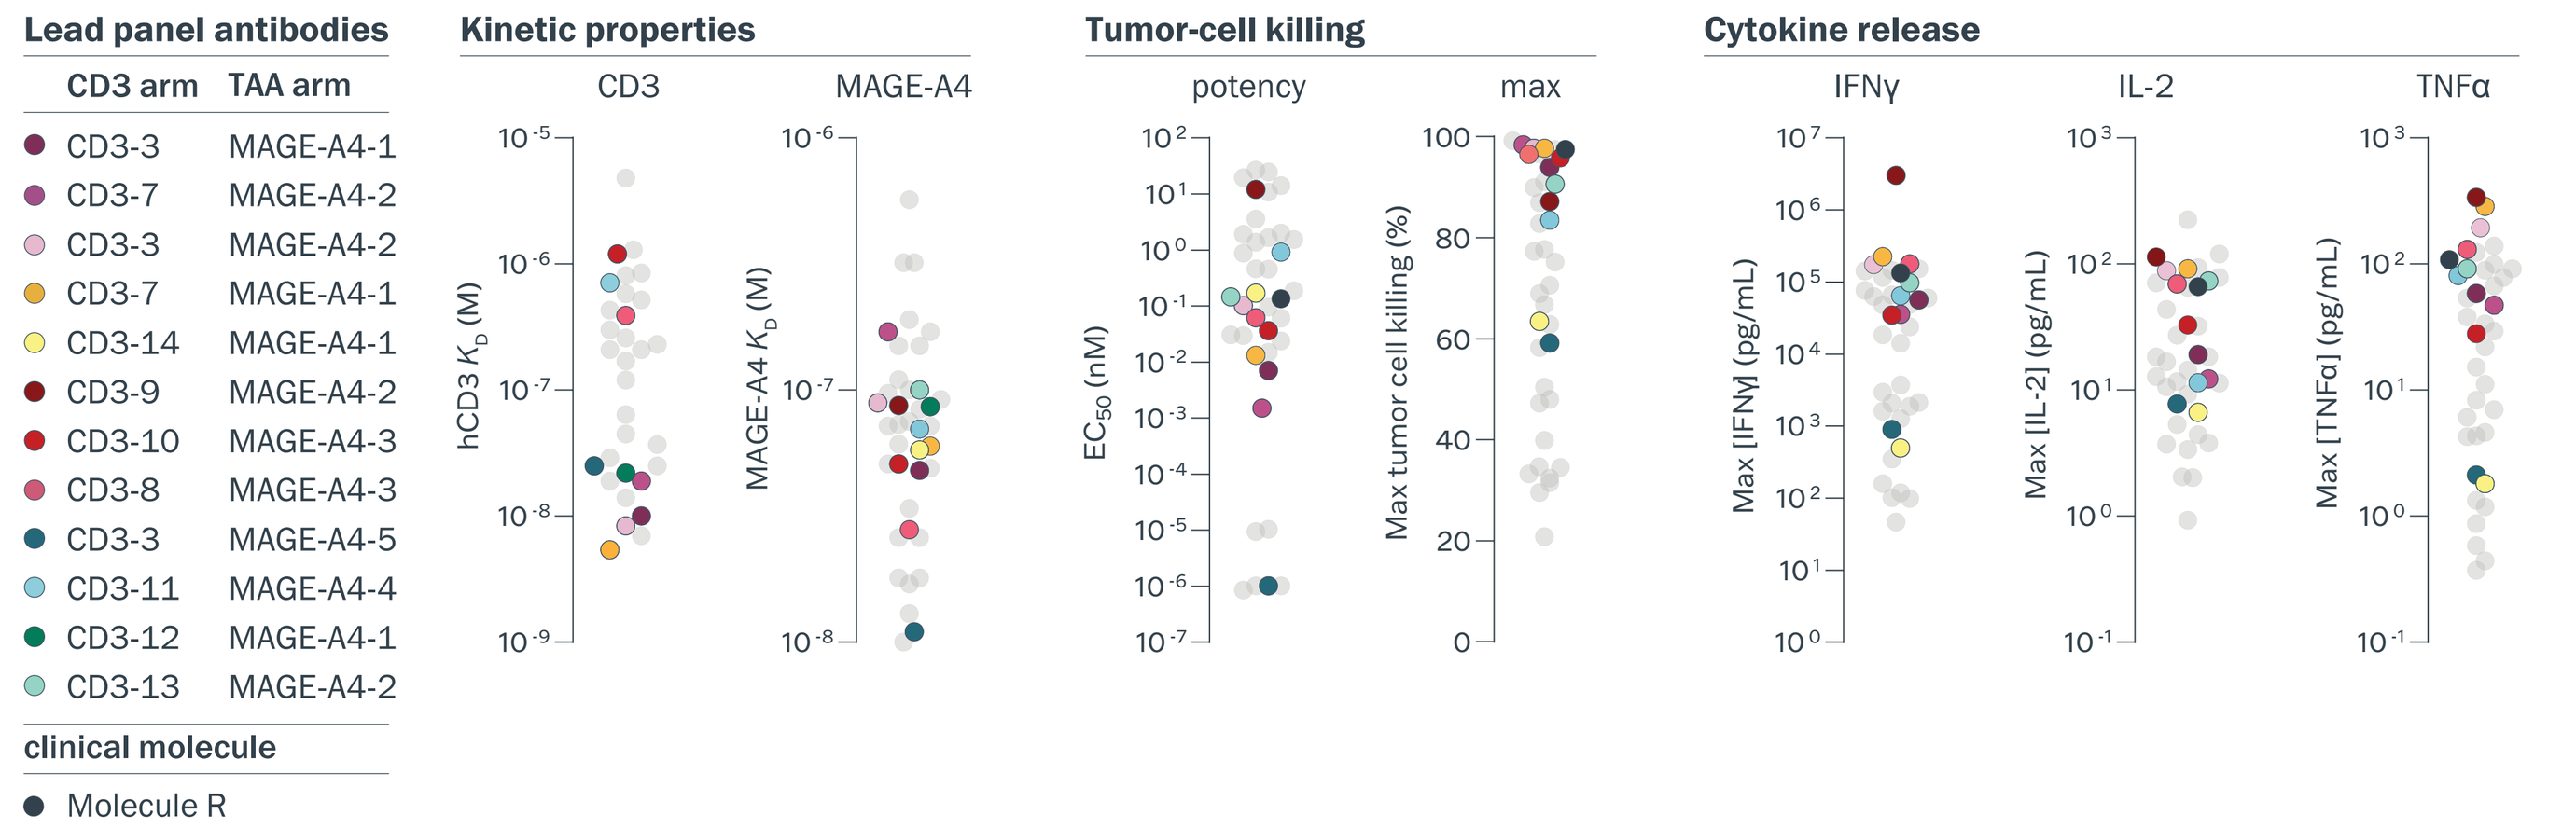

Lead panel antibodies
CD3 arm TAA arm
CD3-3 MAGE-A4-1
CD3-7 MAGE-A4-2
CD3-3 MAGE-A4-2
CD3-7 MAGE-A4-1
CD3-14 MAGE-A4-1
CD3-9 MAGE-A4-2
CD3-10 MAGE-A4-3
CD3-8 MAGE-A4-3
CD3-3 MAGE-A4-5
CD3-11 MAGE-A4-4
CD3-12 MAGE-A4-1
CD3-13 MAGE-A4-2
clinical molecule
Molecule R
Kinetic properties
CD3
MAGE-A4
hCD3 KD (M)
MAGE-A4 KD (M)
Tumor-cell killing
potency
max
EC50 (nM)
Max tumor cell killing (%)
Cytokine release
IFNγ
IL-2
TNFα
Max [IFNγ] (pg/mL)
Max [IL-2] (pg/mL)
Max [TNFα] (pg/mL)

Figure 6. High-throughput assessment of TCEs enabled selection of molecules for further assessment. (A) 180+ PSMA x CD3 TCEs were assessed and compared to two clinical-stage molecules (TNB-585[6] and AMG-160[7]) in a high-throughput assay using unactivated human T cells incubated with PSMA-expressing LNCaP cells at a ratio of 10:1 for 48 hours. Nine selected antibodies are highlighted. **(B)** 200+ 1x1 MAGE-A4 x CD3 T-cell engagers were assessed and compared to Molecule R[8] (057D03 paired with V9 in a 2x1 bispecific format) in a high-throughput assay using unactivated human T cells incubated with wild-type or MAGE-A4 knockout A375 cells at a ratio of 10:1 for 48 hours. Twelve selected antibodies are highlighted.

See more data on our MAGE-A4 x CD3 program

POSTER #1395

REFERENCES

1. Pessano, S. *et al.* (1985). *The EMBO Journal*. 4(2):337-44.
2. DeVorkin, L. *et al.* (2022). *Cancer Research.* 82(12_supplement):312.
3. DeVorkin, L. *et al.* (2022). *Journal for ImmunoTherapy of Cancer.* 10(Issue Suppl 2):1196.
4. Mai, J. *et al.* (2023). *Cancer Research.* 83(7_supplement):1886.
5. Tortora, D. *et al.* (2023). *Cancer Research.* 83(7_supplement):1891.
6. Dang, K. *et al.* (2021). *Journal for ImmunoTherapy of Cancer.* 9:e002488.
7. Deegen, P. *et al.* (2021). *Clinical Cancer Research.* 27(10):2928-2937.
8. Weinzierl, T. *et al.* (2021). *Antibodies binding to HLA-2/MAGE-A4* (International Publication No. WO/2021/122875). World Intellectual Property Organization.

COPYRIGHT © ABCELLERA