1196

Streamlining T-cell engager development

with diverse, fully human CD3-binding antibodies, bispecific engineering technology, and an integrated discovery engine

Lindsay DeVorkin, Juntao (Matt) Mai, Kate Caldwell, Tim M Jacobs, Raffi Tonikian, Karine Herve, Yuri Hwang, Cristina Faralla, Wei Wei, Emma Lathouwers, Rhys S Chappell, Stefan Hannie, Katherine J Lam, Harveer Dhupar, Tran NT Tran, Melissa Cid, Lena M Bolten, Tova Pinsky, Ping Xiang, Courteney Lai, Ahn Lee, Patrick Chan, Jasmine Chin, Sherie Duncan, Aaron Yamniuk, Kush Dalal, **Bryan C Barnhart**

AbCellera

DOWNLOAD POSTER

THE CHALLENGE

CD3 T-cell engagers have the potential to be a cornerstone of immuno-oncology. However, a limited pool of CD3-binding antibodies and technological challenges in engineering bispecifics have hindered their development.

Discovering effective T-cell engagers requires identification of two target-binding arms — a CD3 arm that fine-tunes T cell activation and a tumor arm with high specificity for cancer cells — optimized as a whole to work in concert with each other.

The intensity of T cell activation is a key predictor of safety and efficacy and is, in part, determined by the affinity and epitope of the CD3-binding arm. CD3 binders that strongly activate T cells can trigger dose-limiting toxicities, including cytokine release syndrome. On the other hand, CD3 binders that weakly activate T cells can lack potency. The result is a narrow therapeutic window for T-cell engager development.

THE SOLUTION

Beginning with diverse panels of parental antibodies increases the probability of finding appropriately potent and developable T-cell engagers, reducing the need for downstream engineering to eliminate liabilities and improve functionality.

Our goal was to develop a large and diverse panel of fully human CD3-binding antibodies and partner to discover T-cell engagers with optimized specificities, potencies, biophysical properties, and cytokine release.

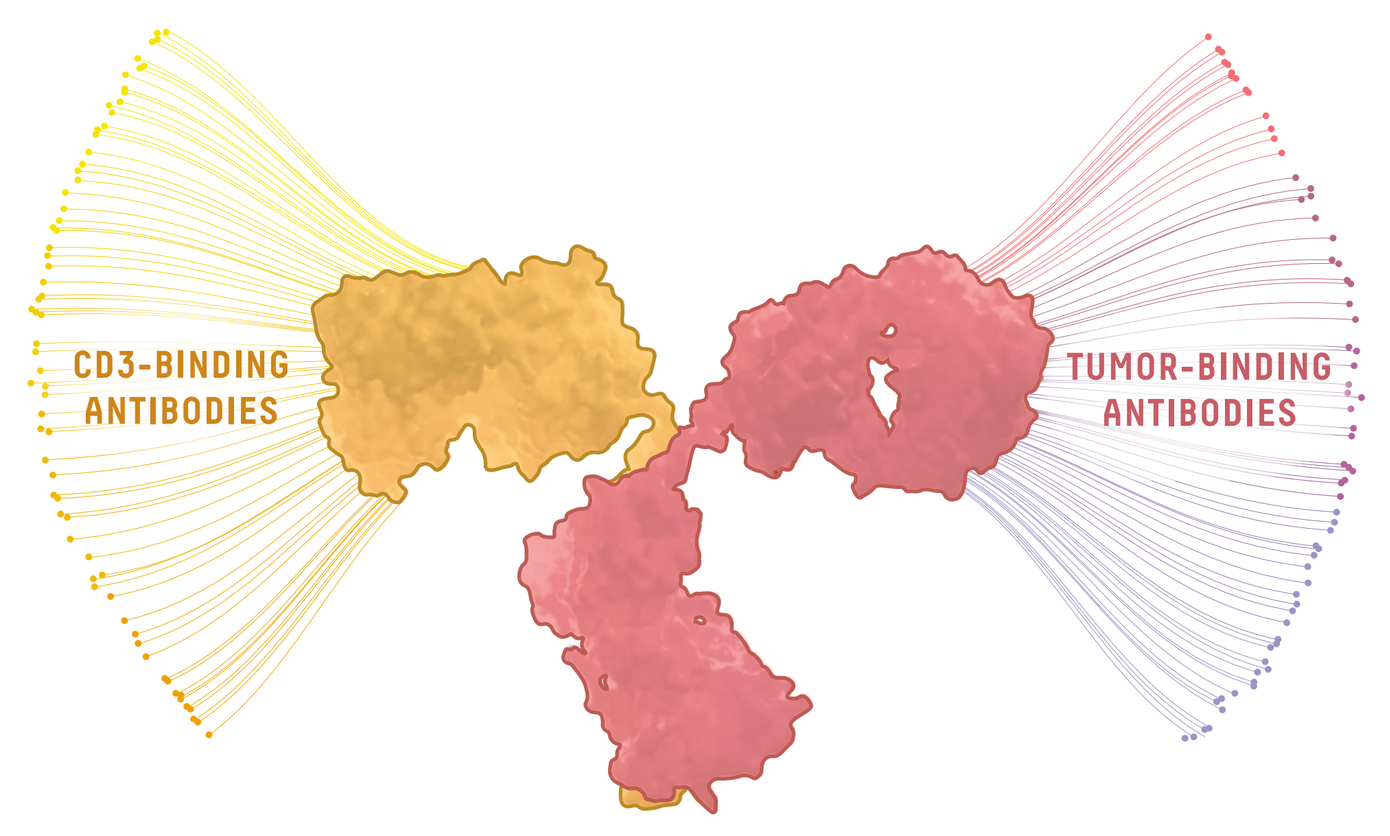
CD3-BINDING ANTIBODIES
TUMOR-BINDING ANTIBODIES

THE OUTCOME

A panel of fully human CD3-binding antibodies that are:

DIVERSE	DEVELOPABLE	VALIDATED
NINE different epitope bins	**FAVORABLE** biophysical properties	**CD3 x EGFR** proof-of-concept T-cell engagers
– A range of CD3 affinities (K_D from ~1 nM - 1 μM) – A broad range of T cell activation potencies (EC_{50} from ~6 - 200 nM) – Multiple epitope bins containing human and cyno cross-reactive binders	– Including high thermal stability and low hydrophobicity, self-association, and polyspecificity	– A broad range of T cell activation potencies (EC_{50} from ~2 pM - 2 nM) – A broad range of tumor cell killing potencies (EC_{50} from ~0.01 - 1 nM) – Potent molecules that achieve >90% cell killing with low cytokine release

All characterization data shown are for the subset of total antibodies that have been expressed and analyzed to date.

DIVERSITY

Hundreds of unique human antibodies with sequence diversity.

Diverse antibody sequences from mature immune responses in humanized mice

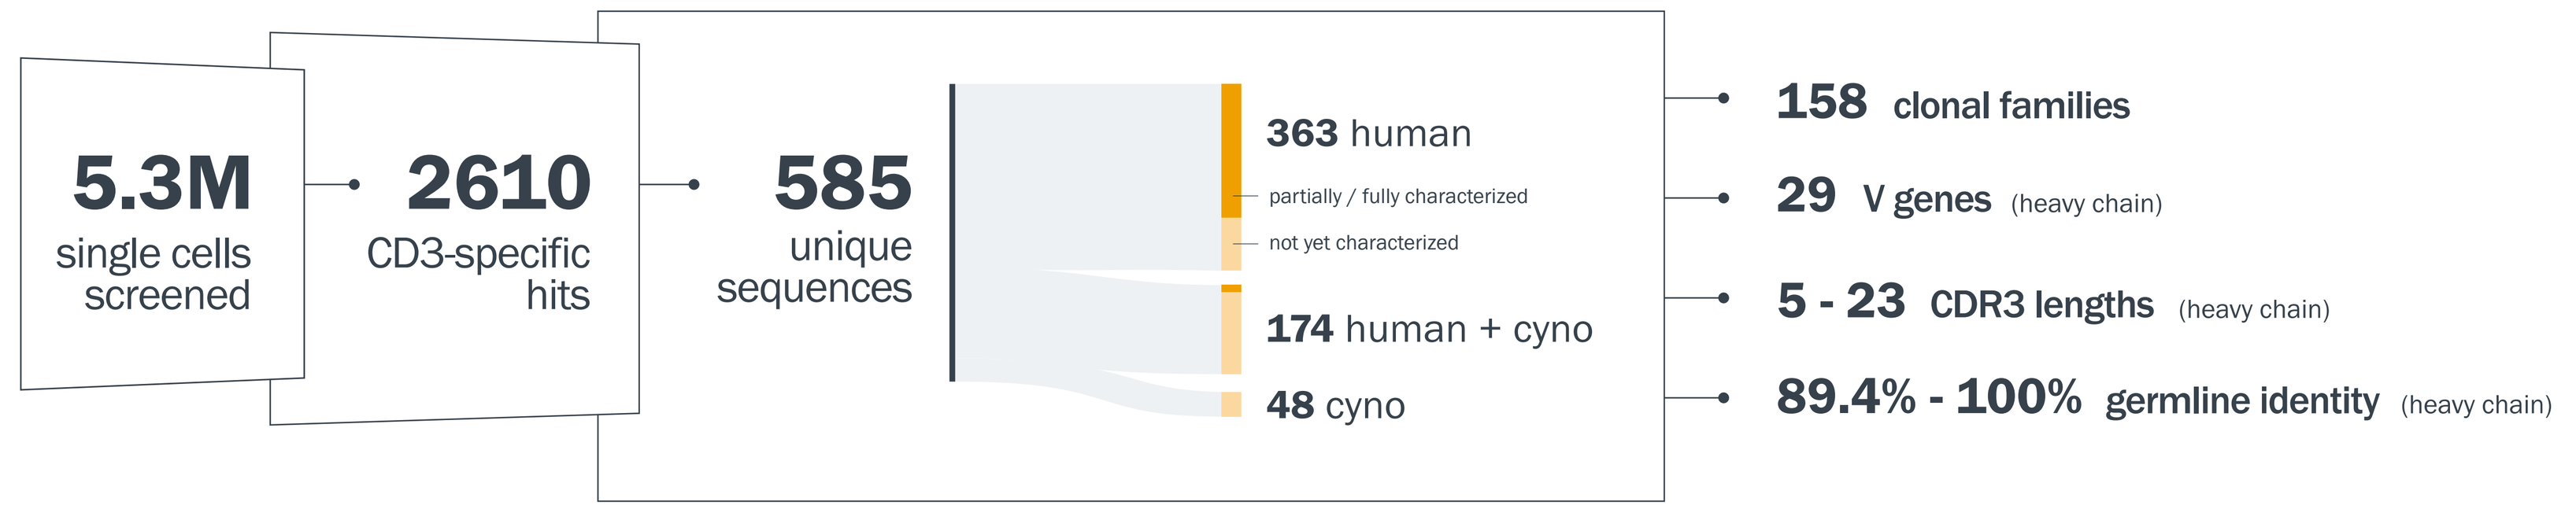
5.3M single cells screened
2610 CD3-specific hits
585 unique sequences
363 human
partially / fully characterized
not yet characterized
174 human + cyno
48 cyno
158 clonal families
29 V genes (heavy chain)
5 - 23 CDR3 lengths (heavy chain)
89.4% - 100% germline identity (heavy chain)

Figure 1. Proprietary immunization strategies using intact human and/or cyno CD3 complexes in humanized mice enriched antibody diversity from the start. CD3-specific antibody sequences had diverse V gene usage, CDR3 lengths, and identity to germline. Antibodies with specificities to human CD3, cyno CD3, and human + cyno CD3 were recovered.

CD3-specific antibodies with binding and functional diversity.

Diverse CD3 affinity KD from ~1 nM - 1 μM

A range of on/off rates Diverse target engagement times

Broad T cell activation potencies EC_{50} from ~6 - 200 nM

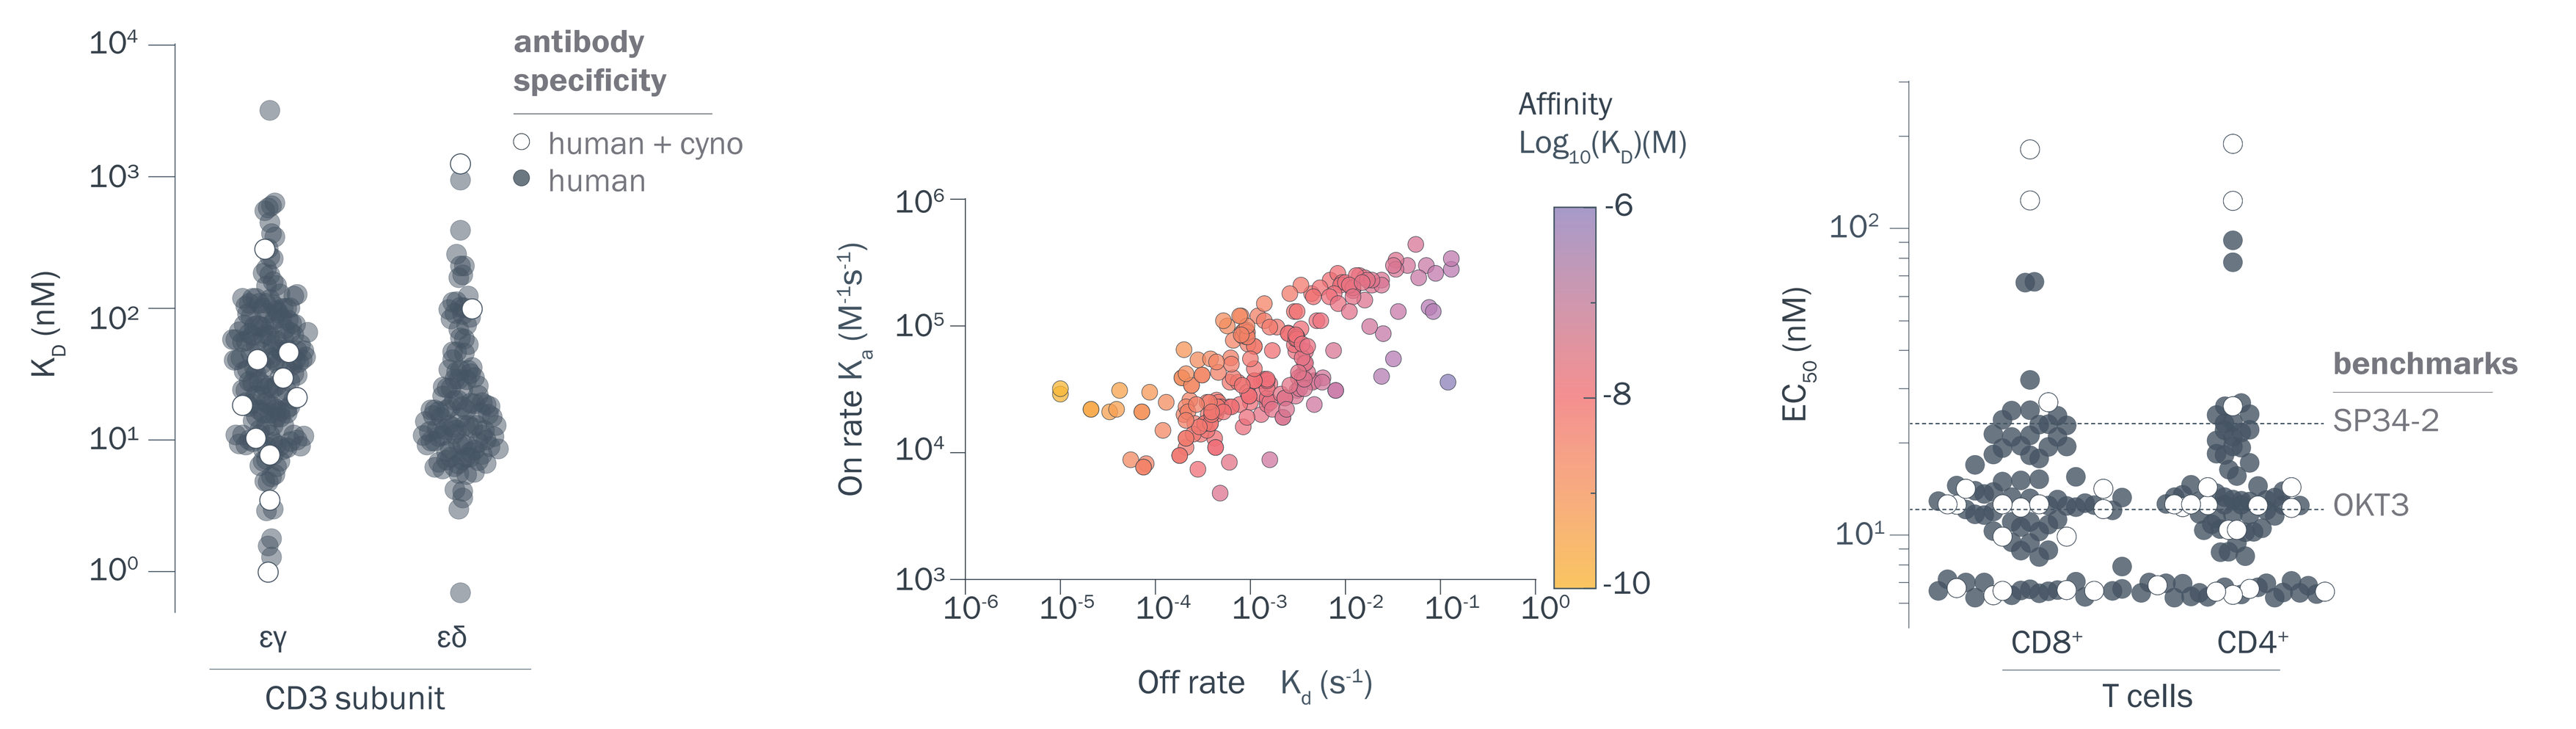
antibody specificity
human + cyno
human
CD3 subunit
Affinity Log10(KD)(M)
On rate Ka (M-1s-1)
Off rate Kd (s-1)
benchmarks
SP34-2
OKT3
CD8+
CD4+
T cells

Figure 2. High-throughput antibody expression and analysis showed diverse affinites, target engagement times, and T cell activation potencies. (A) Surface Plasmon Resonance (SPR)-coupled kinetics analysis of antibody binding affinity to human CD3 εγ and εδ subunits.

(B) SPR-coupled kinetics of antibody binding to the human CD3 εδ subunit.

(C) T cell activation (CD25 and CD69 upregulation) measured by flow cytometry. Naive primary human T cells were incubated with plate-bound anti-CD3 antibodies for 24 hours. Values are the concentration of each antibody needed to induce 50% of the maximal activation of primary human CD4+ and CD8+ T cells.

9 epitope bins, 4 containing cross-reactive binders from the antibodies characterized to date

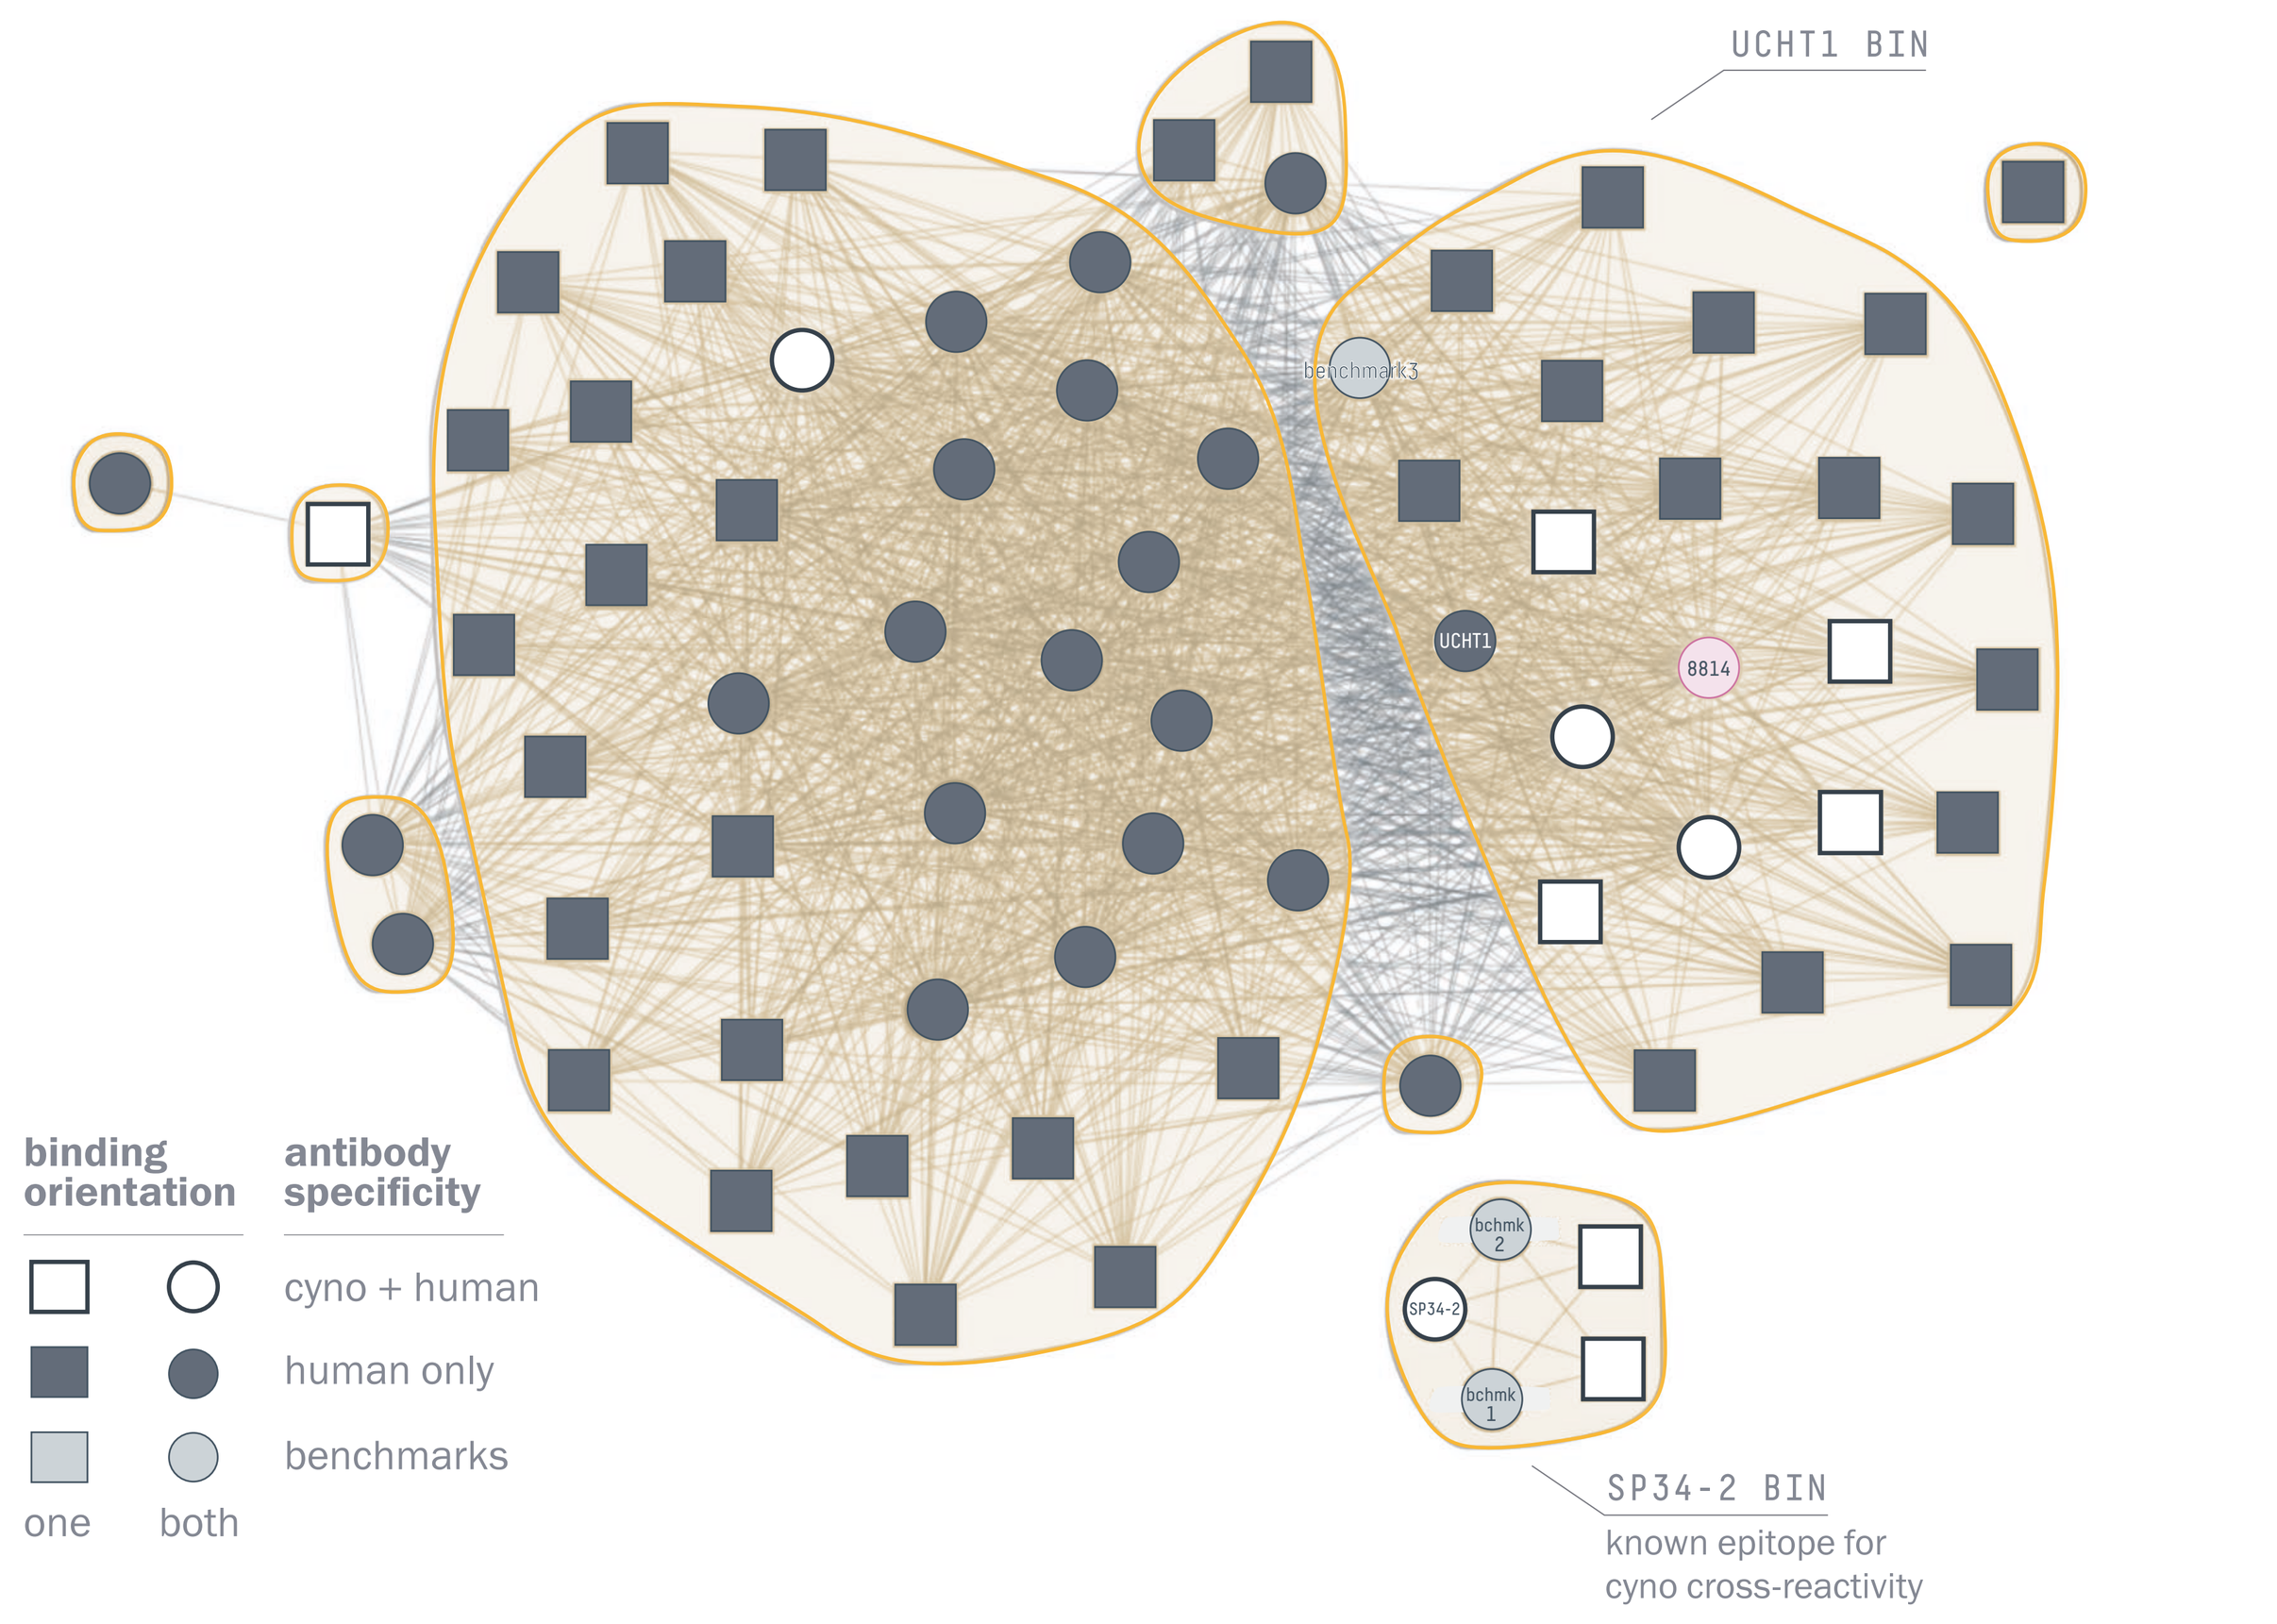
UCHT1 BIN
SP34-2 BIN
known epitope for cyno cross-reactivity
binding orientation
antibody specificity
cyno + human
human only
benchmarks
one
both

Figure 3. High-throughput SPR epitope binning of CD3-binding antibodies revealed nine epitope bins. Antibodies with cross-reactivity to human and cyno CD3 were found in four epitope bins, three of which were distinct from the SP34-2 bin. UCHT1 and SP34-2 are characterized, commercially available CD3-specific antibodies.

MOST CD3 T-CELL ENGAGERS ARE DERIVED FROM SP34-2

Finding novel CD3 binders is hard.

The CD3 complex on T cells is a challenging target. Finding antibodies that bind both human and cyno CD3 adds another layer of complexity due to the relatively low sequence similarity (~81%) between them.

As a result, many drug developers have relied on the limited number of publicly disclosed human + cyno cross-reactive antibodies, with SP34-2 being the most common.

Commonly used CD3 binders have limitations.

SP34 was discovered in 1985 from mice immunized with SDS-denatured protein (1). It binds to a linear epitope on the N-terminus of the CD3 ε protein, leaving most of the conformational epitopes of the complex unexplored. In addition, the antibody is often heavily modified to mitigate liabilities.

Novel, cross-reactive, fully human anti-CD3 antibodies that bind diverse epitopes greatly expand opportunities for development of CD3 T-cell engagers, fine-tuned for function.

DEVELOPABILITY

Scalable antibodies with favorable biophysical properties.

Low hydrophobicity aHIC · **Low self-association** AC-SINS · **Low polyspecificity** BVP-ELISA

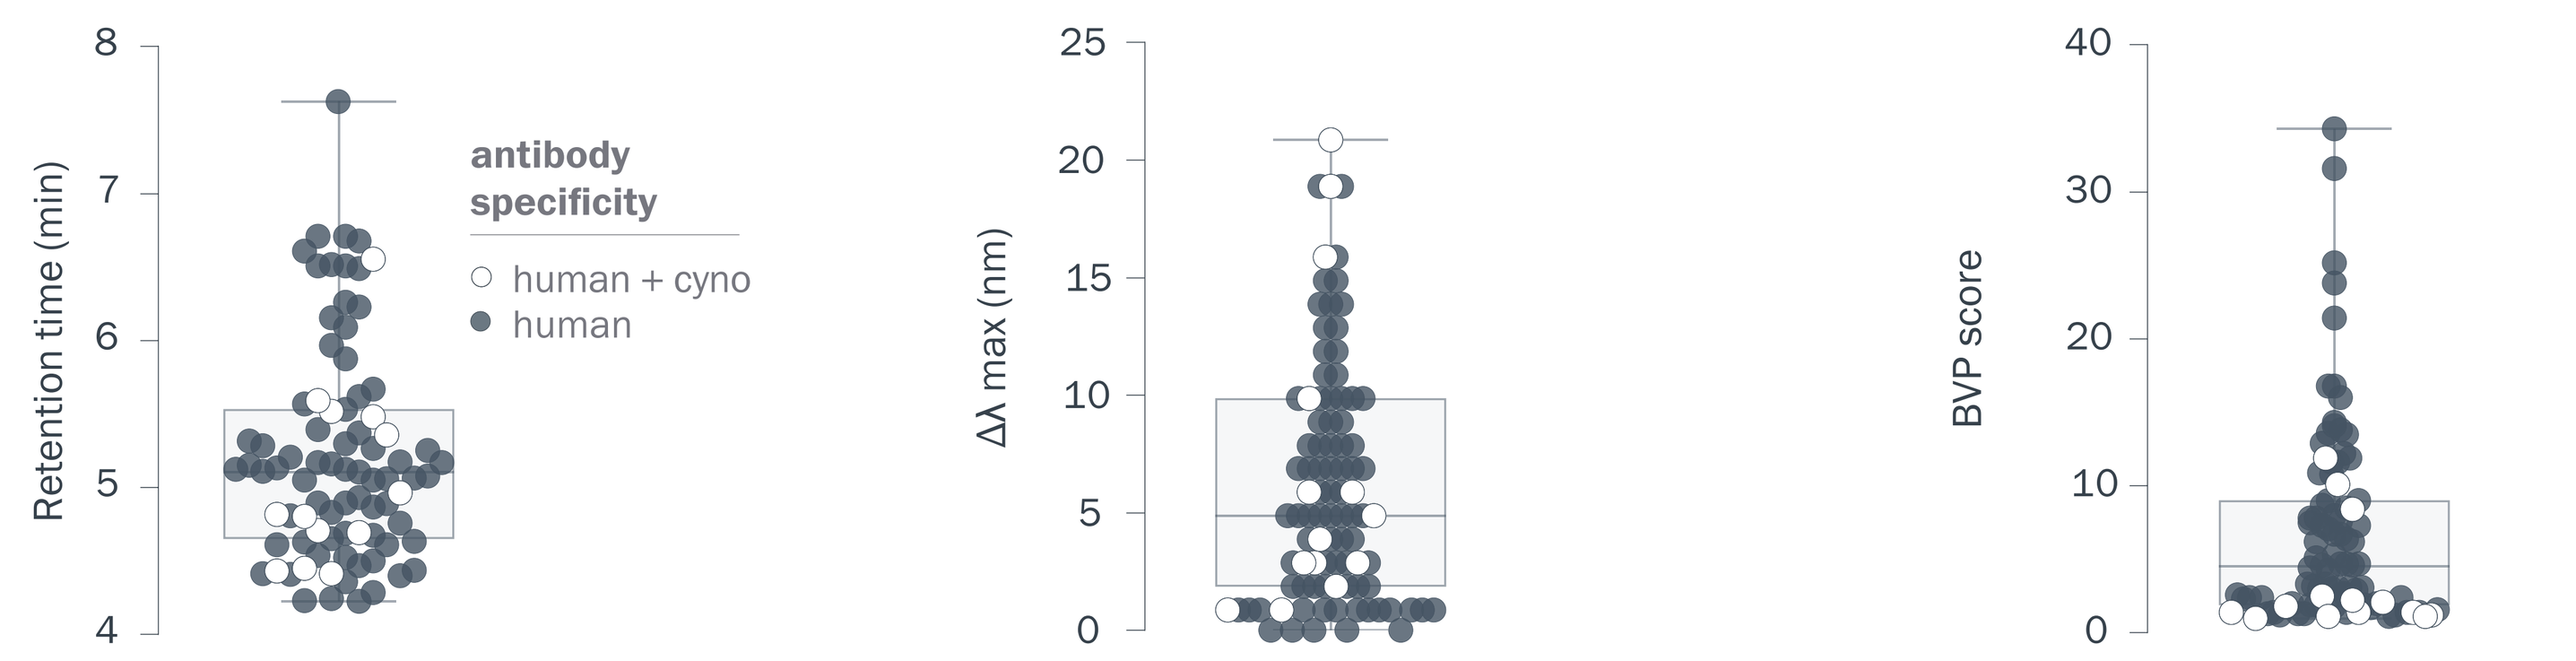
Retention time (min)
Δλ max (nm)
BVP score
antibody specificity
human + cyno
human

Figure 4. Biophysical analysis of CD3-specific antibodies revealed favorable developability properties. (A) Relative surface hydrophobicity measured by analytical Hydrophobic Interaction Chromatography (aHIC). **(B)** Self-association assessed by Affinity-Capture Self-Interaction Nanoparticle Spectroscopy (AC-SINS). **(C)** Polyspecificity assessed by Baculovirus Particle Enzyme-Linked Immunosorbent Assay (BVP-ELISA).

VALIDATION

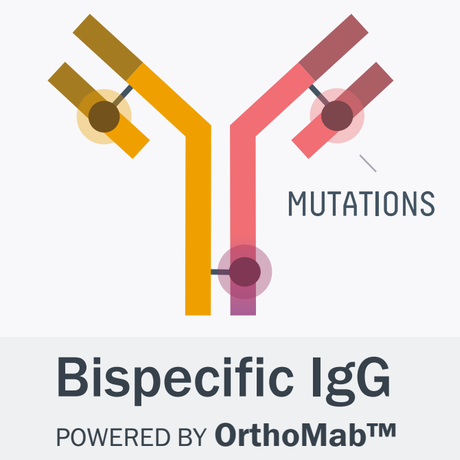
MUTATIONS
Bispecific IgG
POWERED BY OrthoMab™

CD3-specific antibodies validated as proof-of-concept CD3 x EGFR bispecifics using OrthoMab.

Favorable biophysical properties comparable to CD3 parental antibodies

Pairing Intact MS · **Self-association** AC-SINS · **Polyspecificity** BVP-ELISA

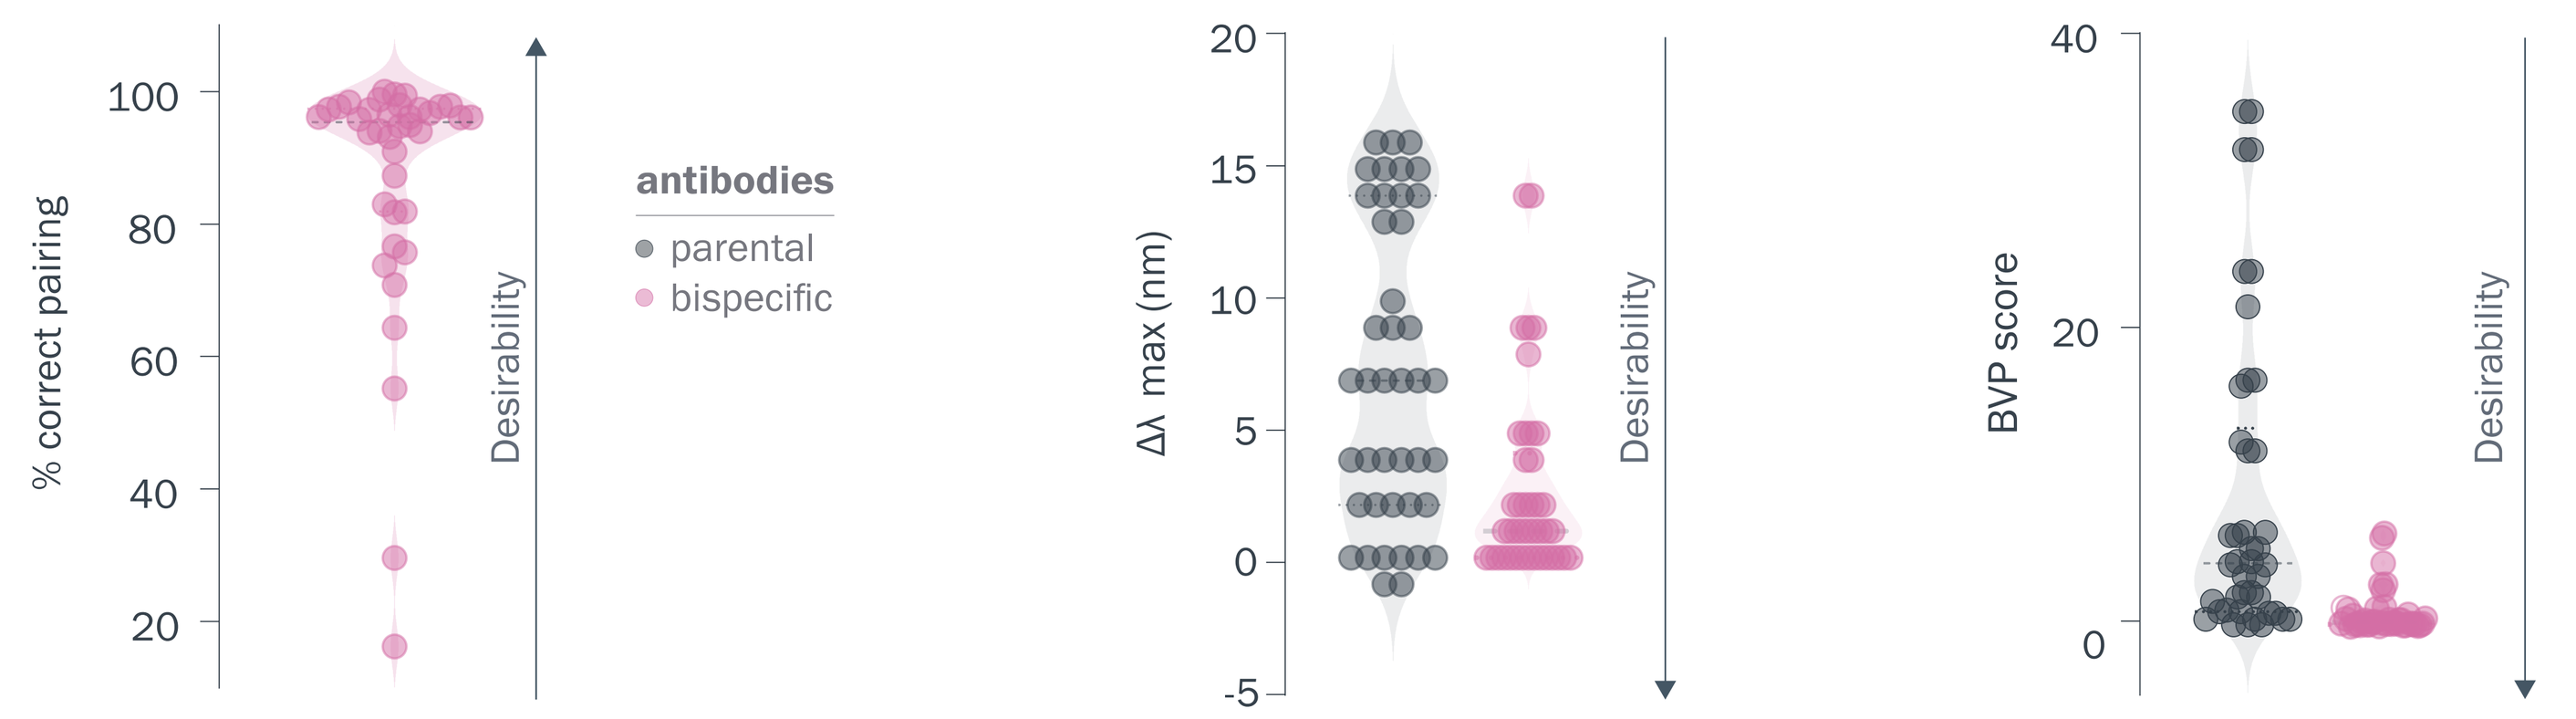
% correct pairing
Desirability
antibodies
parental
bispecific
Δλ max (nm)
BVP score

Figure 5. Biophysical analyses showed developability properties comparable to CD3-specific parental antibodies. The fractional percentage of correctly paired vs. incorrectly paired bispecifics was measured by mass spectrometry (MS). Self-association and polyspecificity were measured by AC-SINS and BVP-ELISA, respectively. Stability (measured by nano differential scanning fluorimetry) and aggregation (measured by aSEC) also remained comparable to CD3-specific parental antibodies (data not shown).

Broad T cell activation potencies EC_{50} from ~2 pM - 2 nM

Broad tumor cell killing potencies EC_{50} from ~0.01 - 1 nM

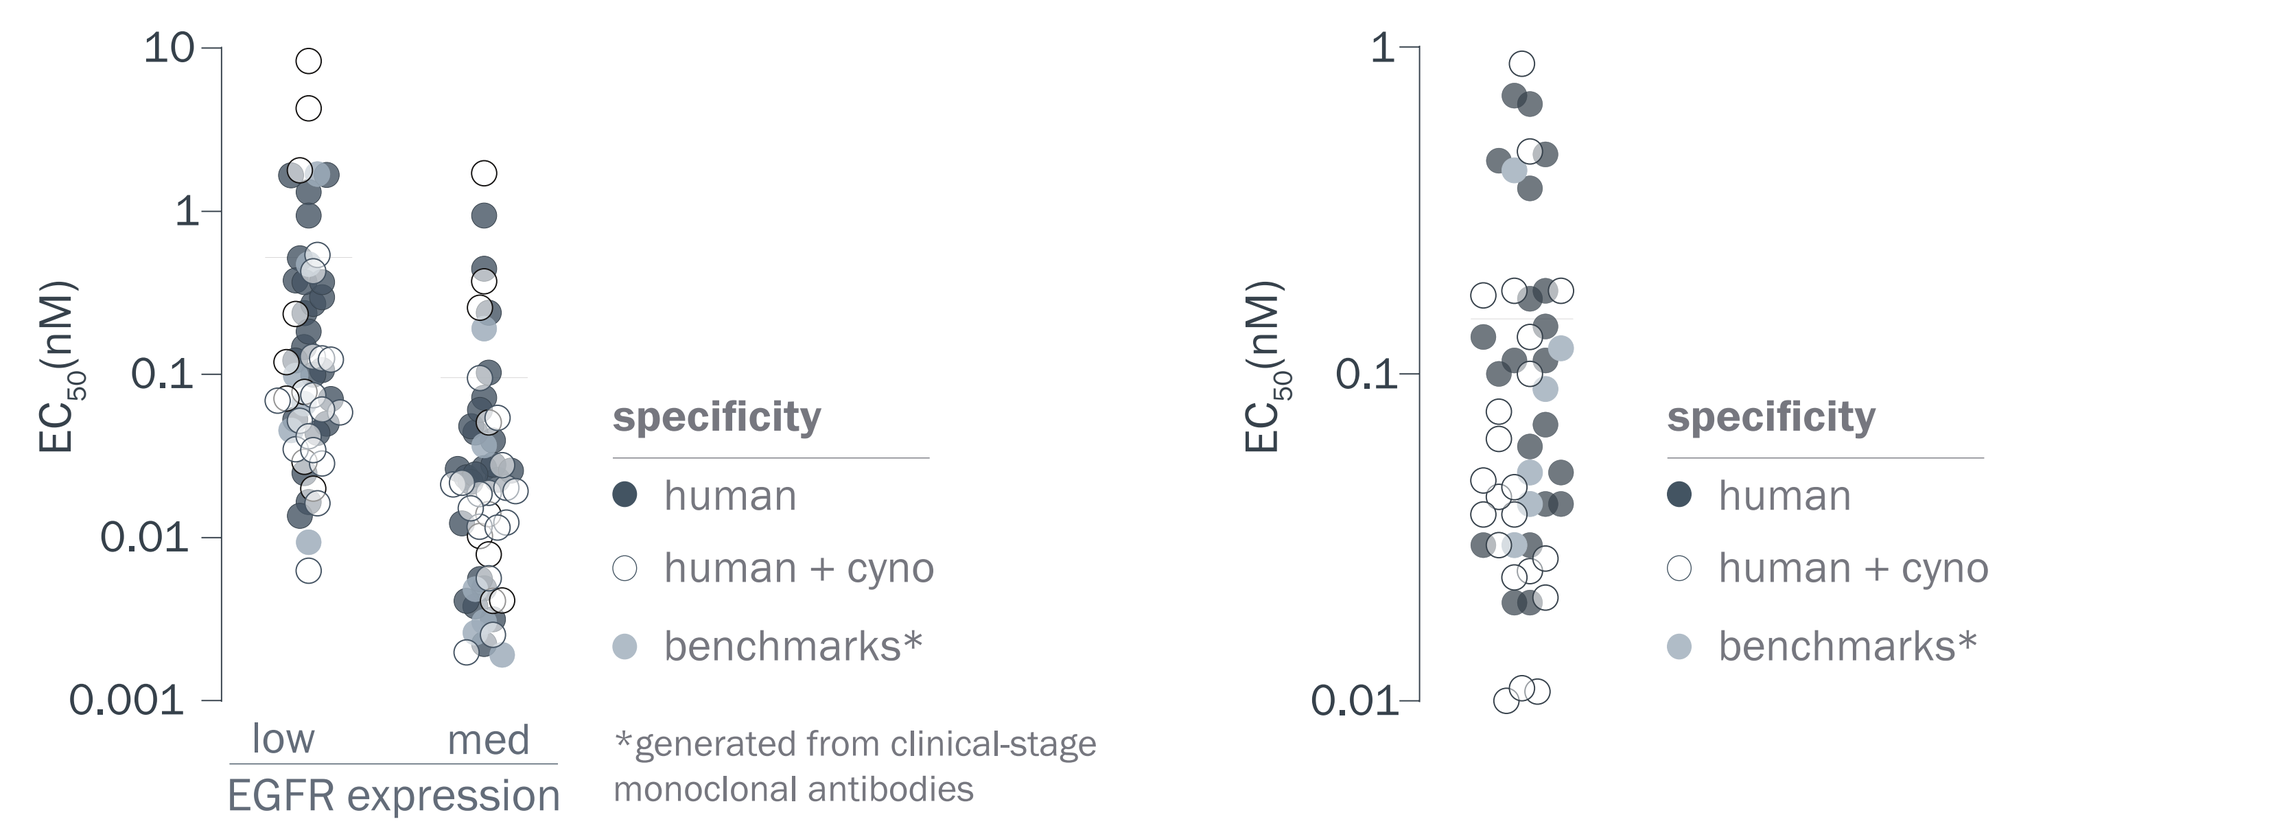
EC50 (nM)
specificity
human
human + cyno
benchmarks*
low
med
EGFR expression
*generated from clinical-stage monoclonal antibodies

Figure 6. Functional analysis of CD3 x EGFR bispecific antibodies showed broad potencies (A) The concentration of each bispecific antibody needed to induce 50% of the maximal activation of NFAT reporter Jurkat T cells. Effector T cells were incubated with target CHO-K1 cells expressing low or medium levels of EGFR for 6 hours at a ratio of 7:1.

(B) The concentration of each bispecific antibody needed to induce 50% of the maximal T cell-mediated killing of HeLa cells. Unactivated human T cells were incubated with target cells at a ratio of 10:1. Cell killing was measured at 48 hours using an xCELLigence assay.

A range of cytokine release Potency [bsAb] 0.02 - 5 nM at 60% | 0.07 - 5 nM at 80% tumor cell killing

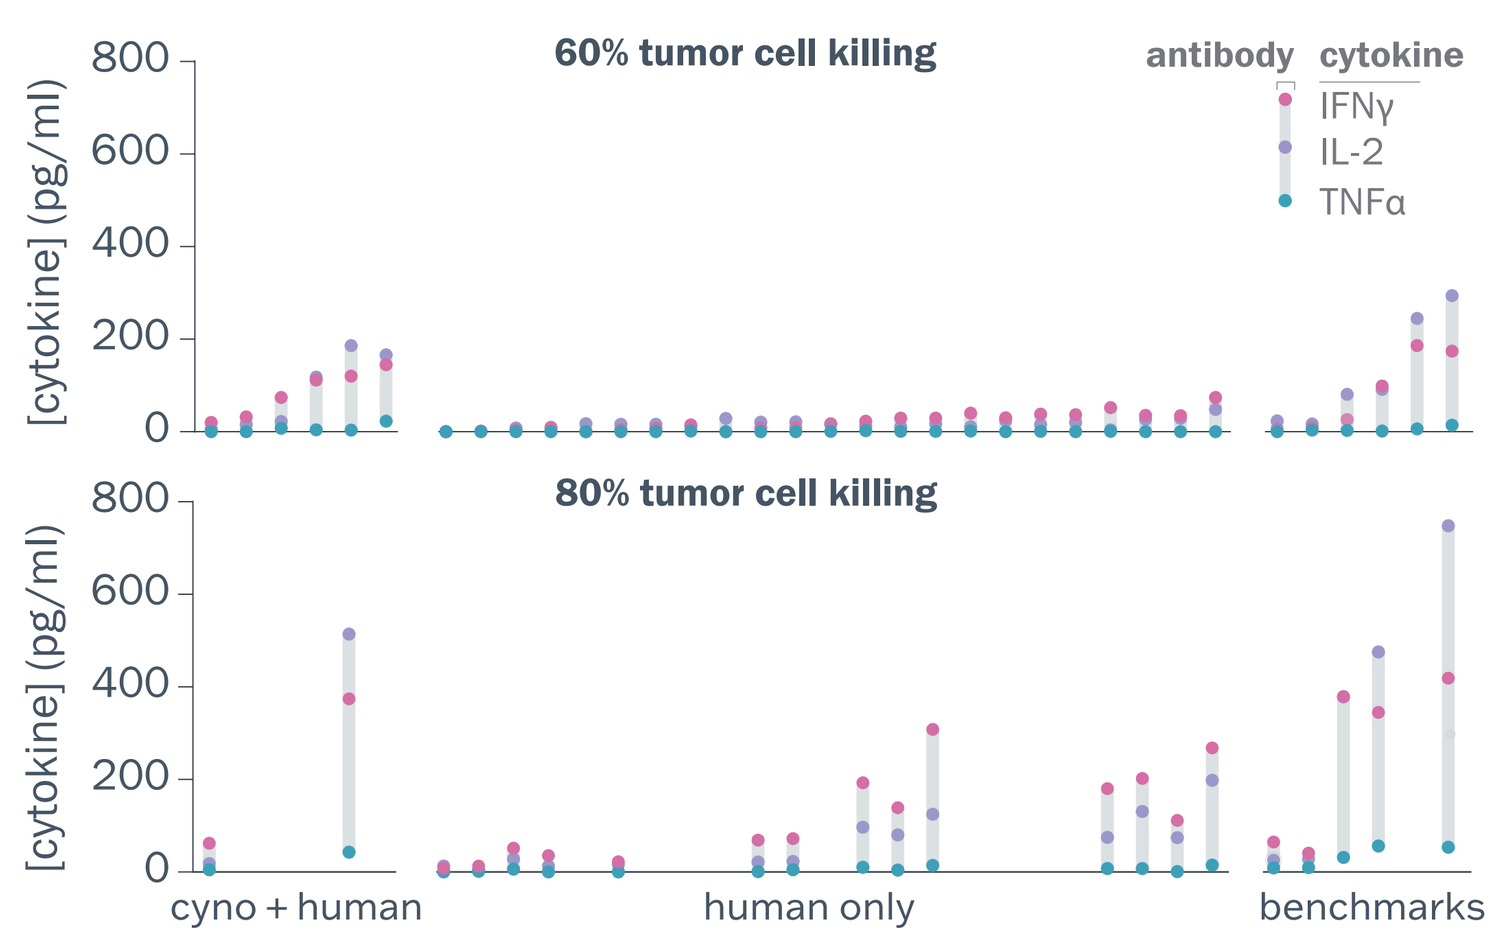
60% tumor cell killing
80% tumor cell killing
antibody
cytokine
IFNγ
IL-2
TNFα
[cytokine] (pg/ml)
cyno + human
human only
benchmarks

Figure 7. Functional analysis revealed potent bispecific antibodies with minimal cytokine release. In the same assay described in Figure 6B, cytokines were measured after 48 hours of incubation. Cytokine concentrations were calculated for each antibody at 60% and 80% HeLa cell killing.

Proof-of-concept CD3 T cell engagers that strike the right balance.

Potent tumor cell killing and low cytokine release

Human-specific CD3 parental antibody · Cross-reactive CD3 parental antibody

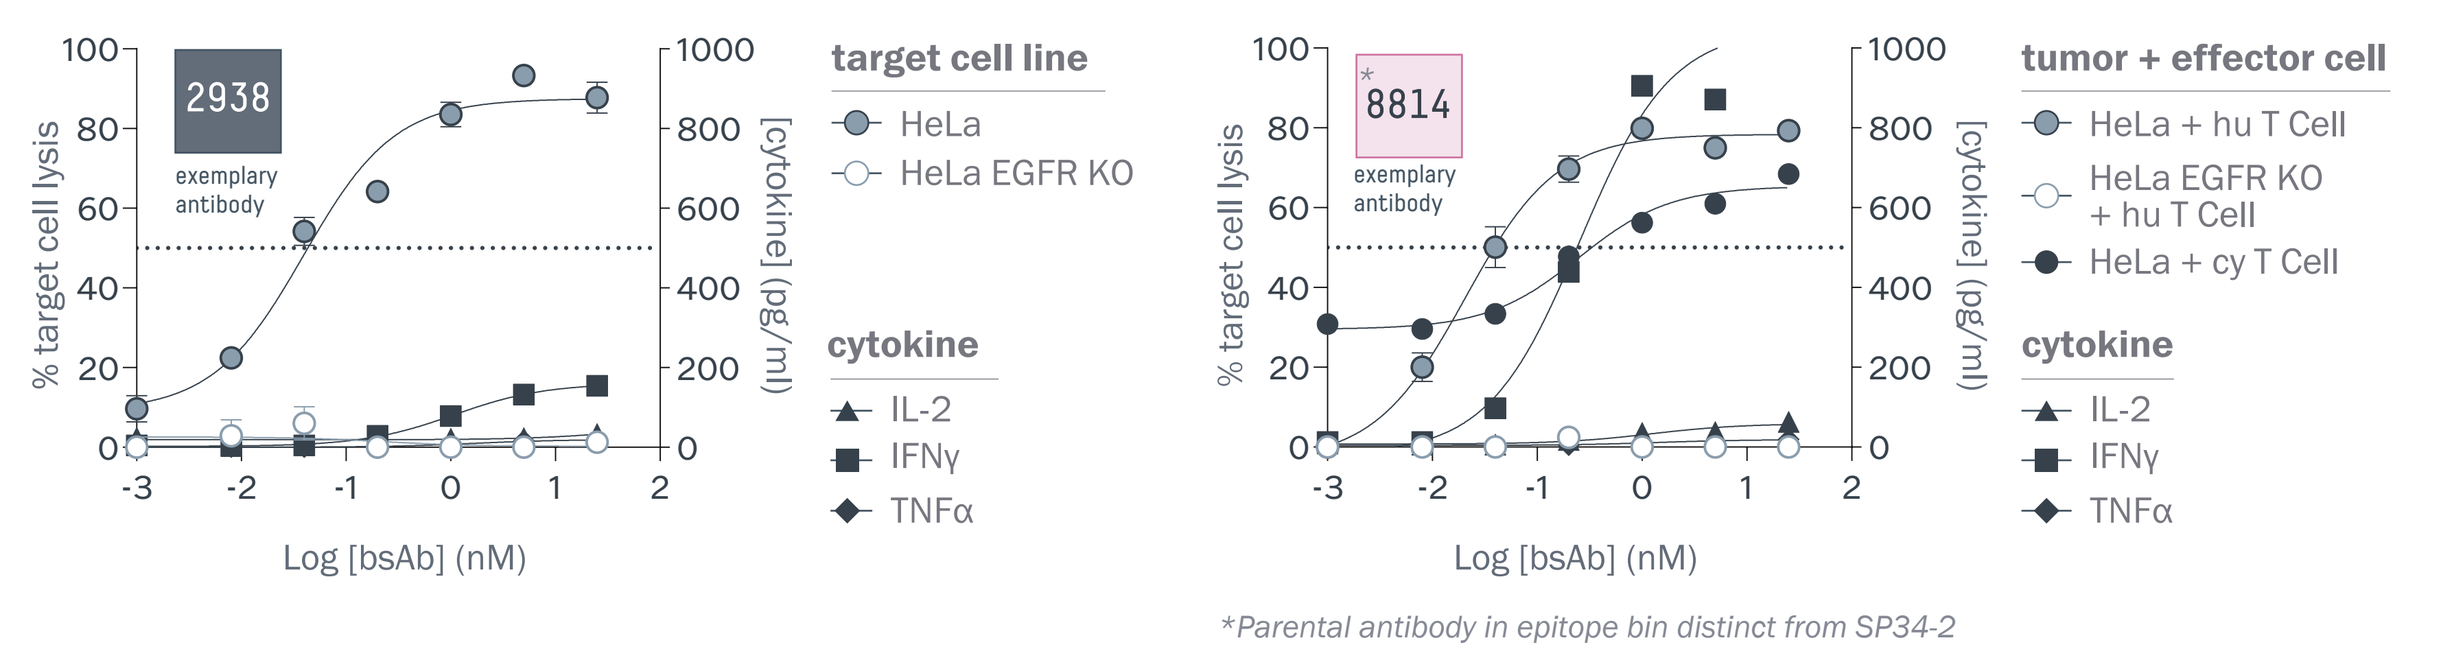
2938
exemplary antibody
8814
% target cell lysis
[cytokine] (pg/ml)
Log [bsAb] (nM)
target cell line
HeLa
HeLa EGFR KO
tumor + effector cell
HeLa + hu T Cell
HeLa EGFR KO + hu T Cell
HeLa + cy T Cell
cytokine
IL-2
IFNγ
TNFα
*Parental antibody in epitope bin distinct from SP34-2

Figure 8. Pairing our CD3 panel with our bispecifics platform identified T-cell engagers that balance potency and cytokine release. Profiles of **(A, left)** a human-specific CD3 x EGFR bispecific antibody and **(B, right)** a human and cyno cross-reactive CD3 x EGFR bispecific antibody that exhibit strong tumor cell killing and minimal cytokine release.

	T cell activation ($EGFR^{MED}$)	Tumor cell killing	Affinity (εδ CD3)
human + cyno 8814	EC_{50} 15.1 pM	EC_{50} 20.7 pM (human T cells) EC_{50} 249 pM (cyno T cells)	K_D 0.99 nM
human 2938	EC_{50} 23.4 pM	EC_{50} 39.5 pM (human T cells)	K_D 351 nM

REFERENCES:

1. S Pessano et al. (1985). The T3/T cell receptor complex: antigenic distinction between the two 20-kd T3 (T3-delta and T3-epsilon) subunits. EMBO J. Feb;4(2):337-44.

COPYRIGHT © ABCELLERA