Soluplus® Maintains the Supersaturation of Carbamazepine from Amorphous Solid Dispersions

K. Tsinman[1], O. Tsinman[1], N. Langley[2], and S. Ali[2]

[1] 1Pion Inc., 10 Cook Street, Billerica, MA 01821, USA

[2] BASF, 500 White Plains Road, Tarrytown, NY 10591

ktsinman@pion-inc.com

BASF

The Chemical Company

Introduction

Drug solubilization has drawn attention in recent years because large numbers of NCEs often fail in development due to their poor solubility and bioavailability[1]. To overcome these challenges in bringing new drugs to the market, novel solubilizers for poorly soluble APIs are expected (1) to have better solubilization capacity than known materials, (2) to have unparalleled safety and toxicological properties, and (3) to reduce time and cost spent on the drug development process.

It is also important to have the ability to screen API-solubilizer combinations early in drug development using small quantities of material and media.

This study was aimed at understanding the supersaturation ability of a novel solubilizer Soluplus® with Carbamazepine as a model drug by using API-sparing real time con-centration monitoring with µDISS Profiler™.

Materials and Methods

Carbamazepine (CBZ, Figure 1), Soluplus (Figure 2) and a solid dispersion of Carbamazepine with Soluplus (CBZ-Soluplus HME, 15% CBZ load prepared through hot melt extrusion) were obtained from BASF Corporation.

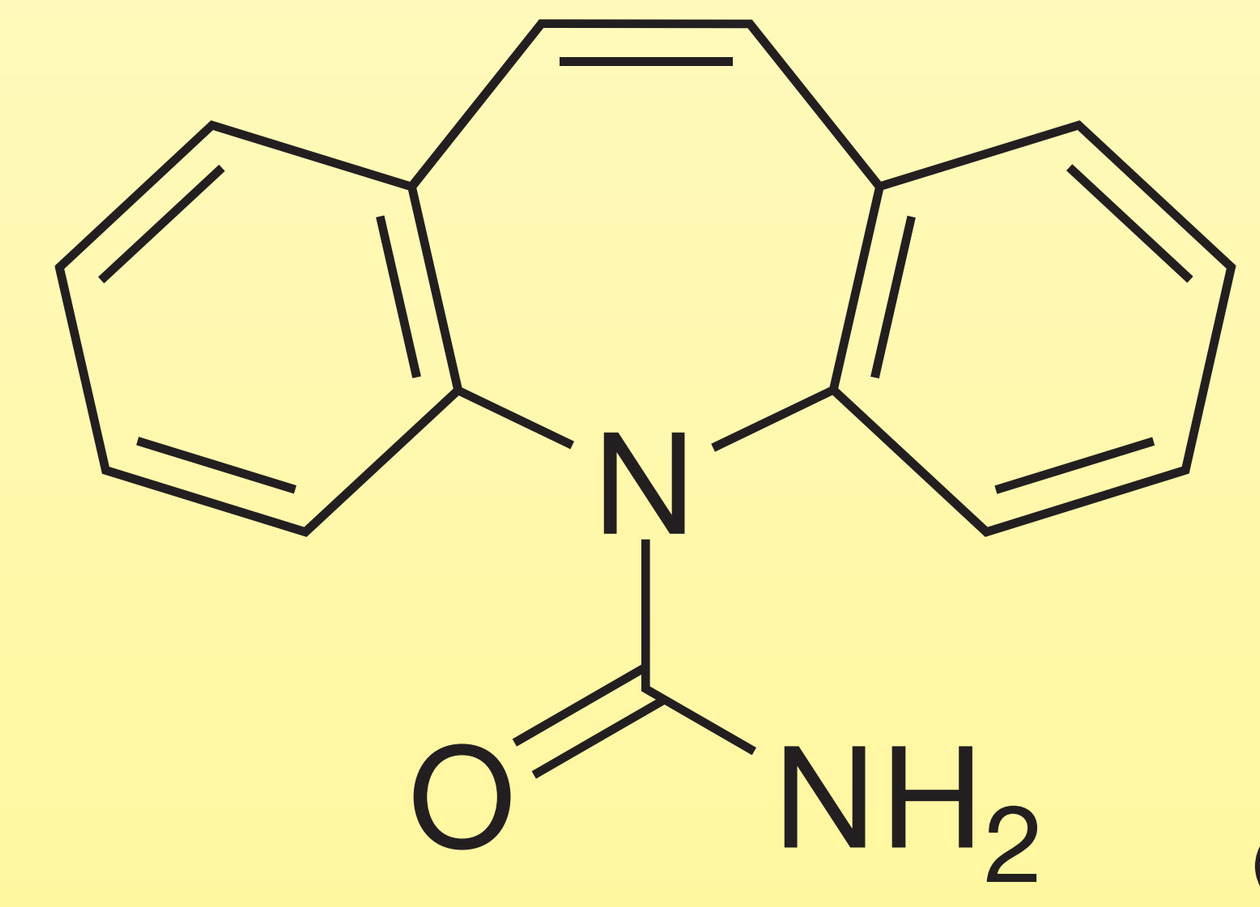

(1)

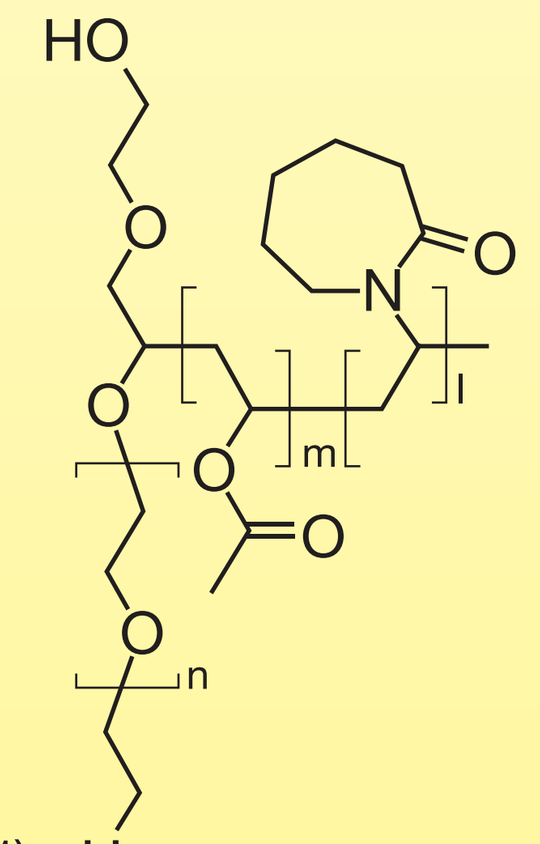

(2)

Figure 1: Structure of Carbamazepine, a model drug used in this study. Figure 2. Structure of Soluplus.

Figure 2 illustrates the structure of Soluplus, a graft copolymer comprising polyethylene glycol, polyvinyl acetate, and polyvinylcaprolactam. Table 1 lists its composition and physicochemical properties[2].

Property	Value
PEG 6000/polyvinlycaprolactan/polyvinly acetate	13/57/30
Appearance	White-yellowish, granular
Solubility	Freely soluble in water and organic solvents
Glass transition temperature	70°C
Lower critical solution temperature (LCST)	37°C
Molecular weight	118,000
K value	31-41
Hydrophilic-lipophilic balance	14
Critical micelle concentration (CMC)	8 mg/L
Particle size, Granules (D_{50})	340 microns
Solutions	
pH 1.2 (0.1 HCl)	70-170 nm (23°C or 37°C)
H 7.0 (PBS)	70-100 nm (23°C) 100 nm at 37°C and 0.01% concentration 130 nm at 37°C and 0.1% concentration 350 nm at 37°C and 1.0% concentration 3,500 nm at 37°C and 10% concentration

Table 1. Physicochemical properties of Soluplus.

Simulated Gastric Fluid (SGF, pH 1.2 HCl/KCl buffer) was prepared according to USP Vol. 35 protocol.

Fast state simulated intestinal fluid (FaSSIF) was prepared from SIF powder following the protocol obtained from Phares (www.ephares.com).

Dissolution behavior and solubility enhancement in SGF and FaSSIF were assessed by the µDISS Profiler (Pion Inc., Figure 3). Experiments were conducted at ambient temperature using 5.7 to 7.8 mg of CBZ or ~ 50 mg of CBZ-Soluplus extrudate powder in 20 mL of media. Stirring speed was set to 200 RPM in all 8 vessels of the miniaturized dissolution bath.

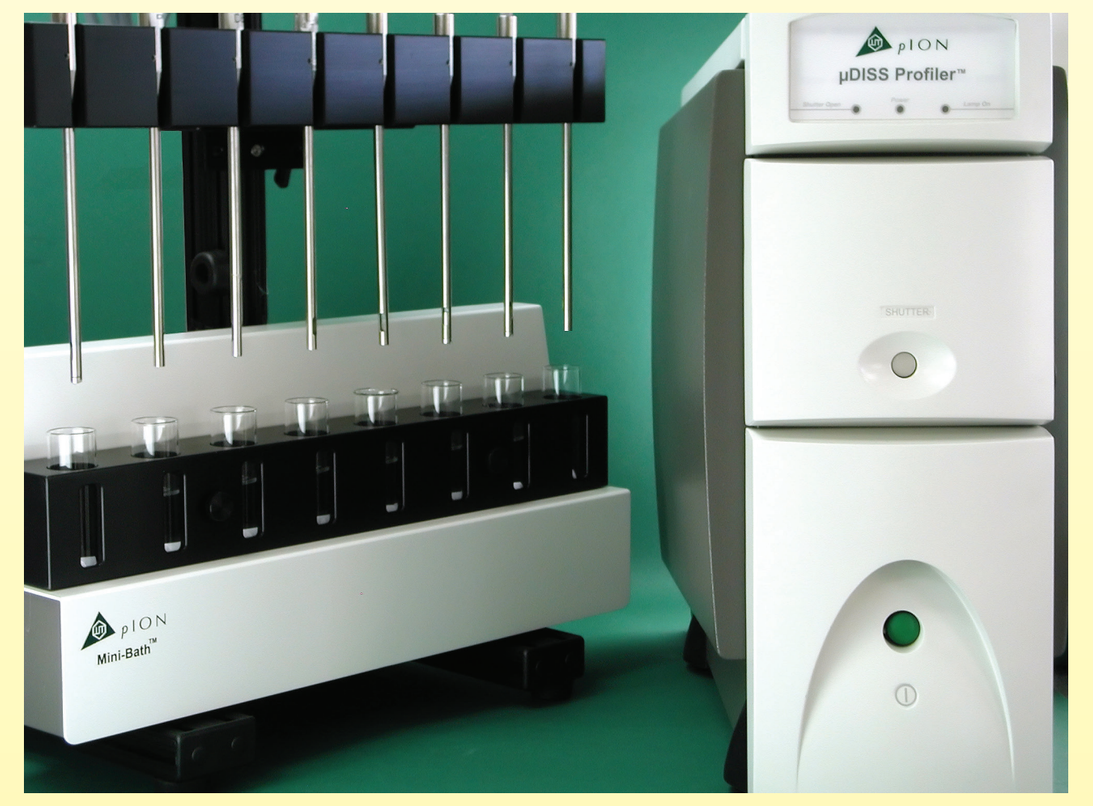
pION
µDISS Profiler™
pION
Mini-Bath™

Figure 3. The µDISS Profiler monitors concentration in real time in 8 temperature and stirring controlled channels using only 1 – 20 mL of dissolution media.

Results and Discussion

Dissolution of CBZ and CBZ-Soluplus HME in SGF and FaSSIF

CBZ is known for forming multiple polymorphic (or pseudo-polymorphic, e.g. hydrates/dehydrates) forms[3]. High energy amorphous CBZ is expected to eventually convert into one of the more stable crystalline forms. Figure 4 shows dissolution/precipitation profile of CBZ in simulated gastric fluid (SGF, pH 1.2). The initial amorphous form exceeds equilibrium solubility, but it starts precipitating within 30 min to 1 h equilibrat-ing at about 145 ± 2 µg/mL solubility level.

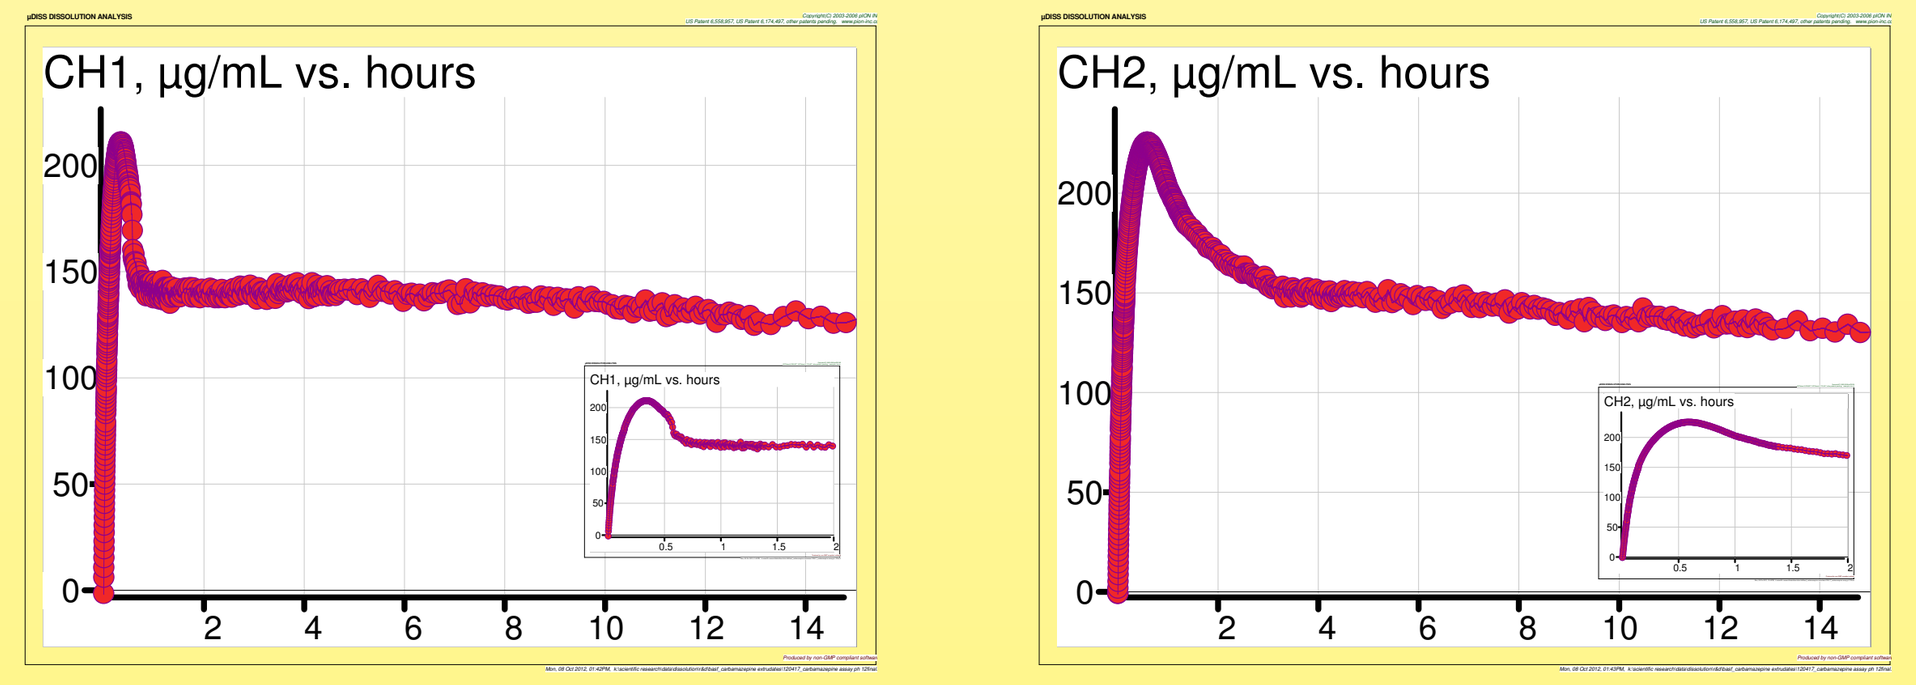
CH1, µg/mL vs. hours
CH2, µg/mL vs. hours

Figure 4. Dissolution/precipitation profile (µg/mL versus hours) for Carbamazapine powder in SGF. Inserts magnify first 2 hours of the experiment.

However, the supersaturation reached by dissolving amorphous CBZ, can be extended with the use of Soluplus as a co-polymer for the hot melt extrusion. Figure 5 illustrates that in the case of CBZ-Soluplus HME where the concentration of CBZ exceeded 300 µg/mL and was quasi-stable at this level for the duration of the assay, 16+ hours.

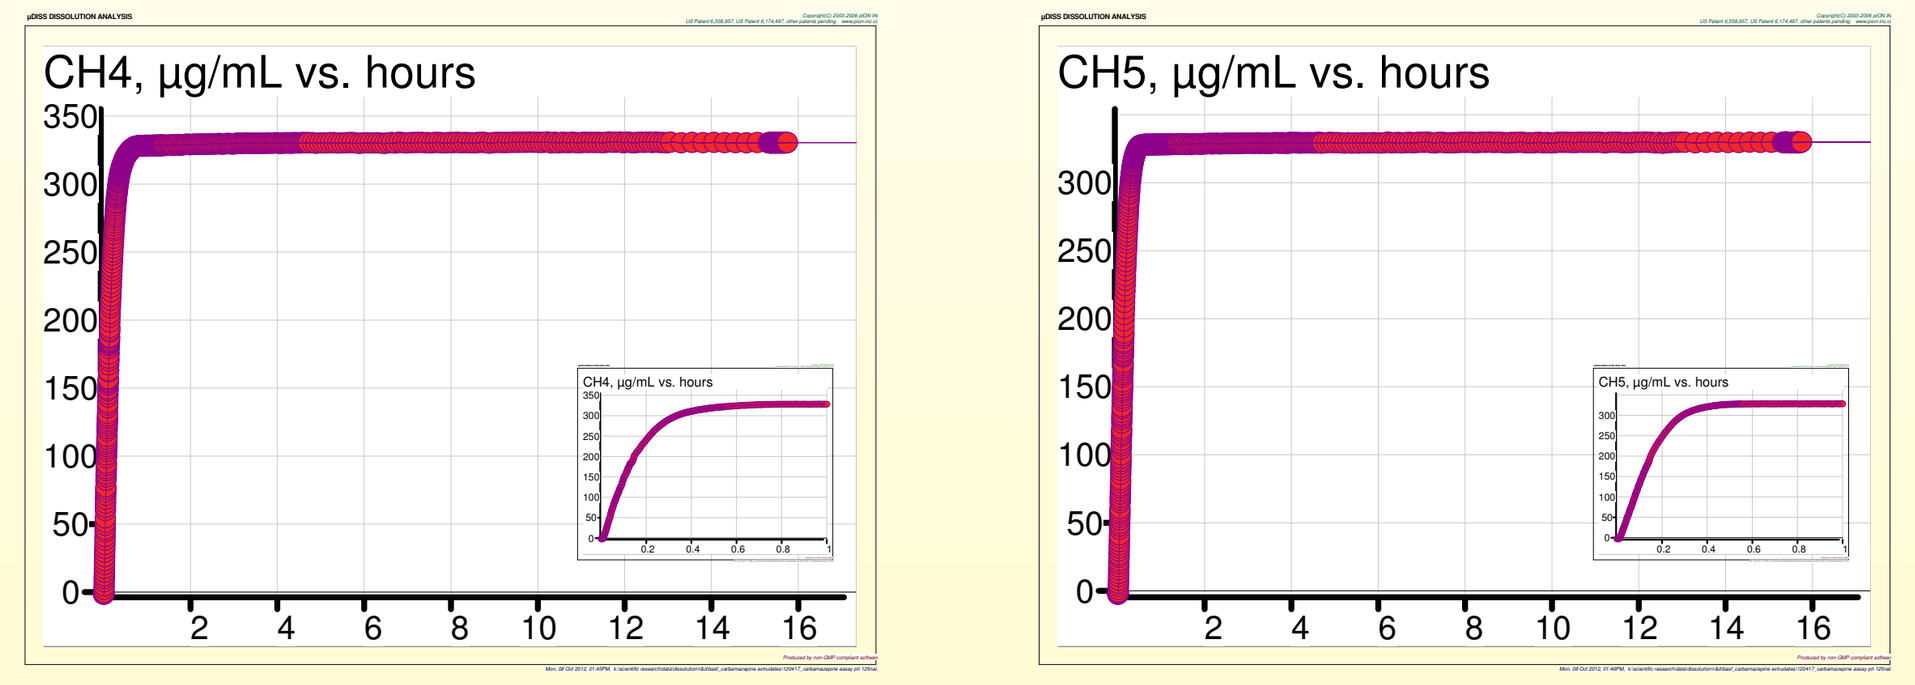
CH4, µg/mL vs. hours
CH5, µg/mL vs. hours

Figure 5. Dissolution profile (µg/mL versus hours) for CBZ-Soluplus hot melt extrudate powder in SGF. Inserts magnify first hour of the experiment where carbamazepine reached quasi-equilibrium.

Analysis of the dissolution curves on Figure 4 and Figure 5 implemented in the µDISS Profiler Command Software allowed estimation of additional characteristics parame-ters of the dissolving powders[4] (Table 2).

Parameter	SGF Medium CBZ (initial 25 min)	SGF Medium CBZ (final)	SGF Medium CBZ-Soluplus	FaSSIF Medium CBZ (initial 25 min)	FaSSIF Medium CBZ (final)	FaSSIF Medium CBZ-Soluplus
Solubility, µg/mL	220 ± 8	145 ± 2	322* ± 10	229 ± 1	166 ± 4	342* ± 11
IDR5, µg cm-2 min-1	27.8 ± 0.3	—	40.7 ± 1.2	30.3 ± 0.3	—	43.1 ± 1.4
IDR5, µg cm-2 min-1	1.26 ± 0.12	—	1.01 ± 0.10	1.06 ± 0.05	—	0.75 ± 0.06
IDR5, µg cm-2 min-1	18.4 ± 1.5	—	22.9 ± 2.5	21.7 ± 1.1	—	30.9 ± 2.4
Effective Diffusion Layer Thickness, µm	11.3 ± 0.6	—	14.4 ± 1.5	12.5 ± 0.4	—	15.0 ± 0.6

Table 2. Dissolution related parameters (average from 3 replicates) estimated from the regression analysis of the corresponding dissolution curves.

Initial solubility, effective area of the powder, and particle size of CBZ were estimated based on the first 25 minutes of the dissolution curve. The final solubility of CBZ was assessed after at least 16 hours of continuous monitoring.

* Concentration of CBZ dissolved in CBZ-Soluplus solid solution was within 80% to 95% of the available CBZ amount. Solubility of this CBZ formulation could be even higher.

Spherical particle radius values in Table 2 refer to a hypothetical monodisperse population of spherical particles that would produce the same initial dissolution rate as was measured experimentally. The Wang-Flanagan[4,6] model was used for the initial effective diffusion layer thickness estimation.

Figure 6 shows the first 160 minutes of CBZ dissolution in FaSSIF at 25°C. Similarly to SGF (Figure 4), CBZ re-precipitates after about 30 minutes reducing its concentration from a maximum level of around 230 µg/mL to 166 ± 4 µg/mL.

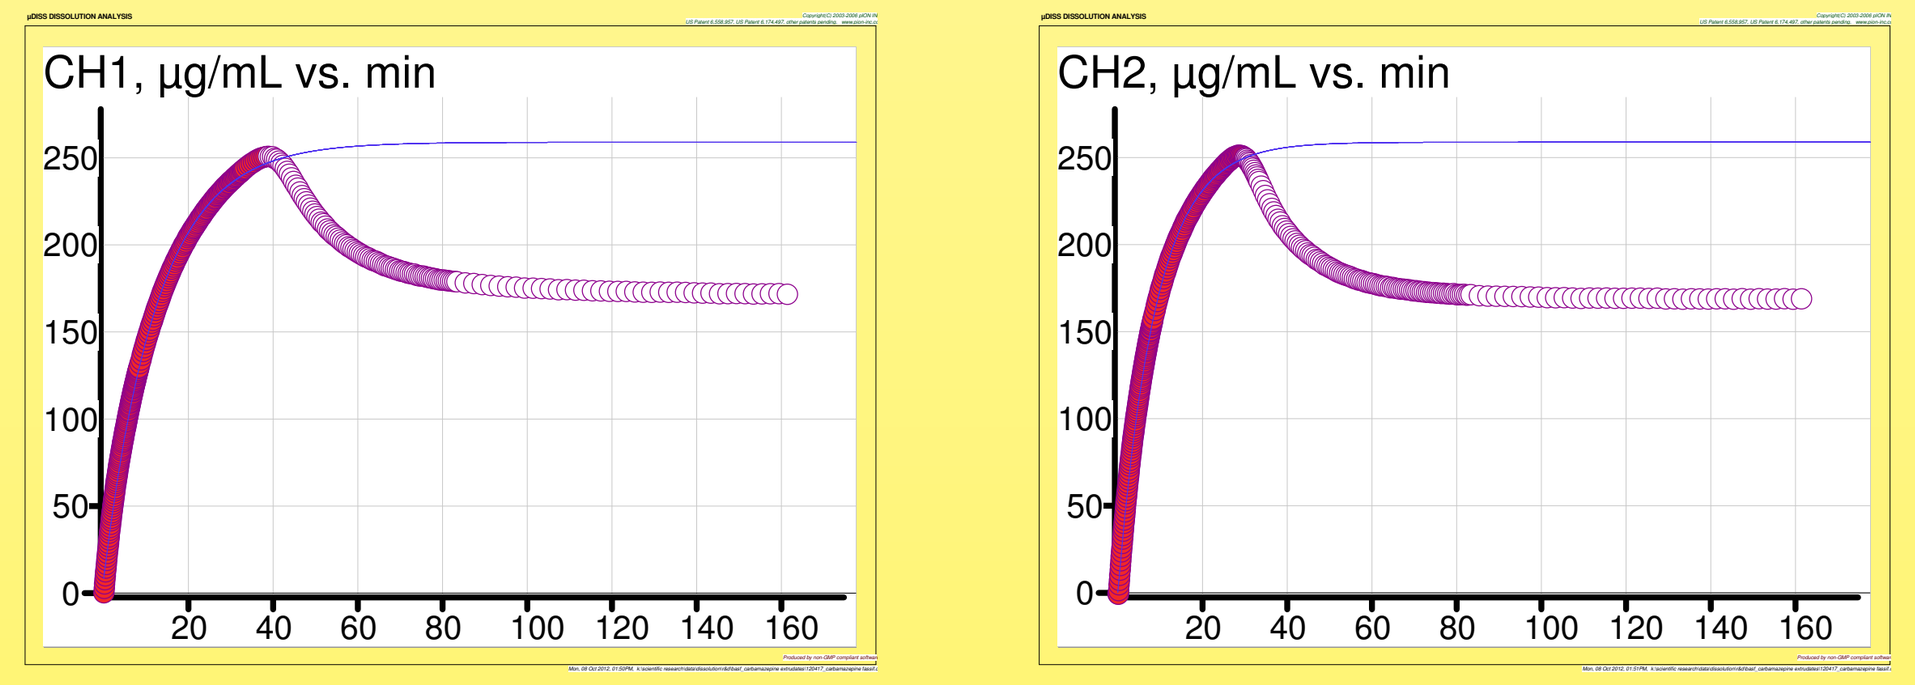
CH1, µg/mL vs. min
CH2, µg/mL vs. min

Figure 6. Examples of two replicates showing dissolution/precipitation profile (µg/mL versus minutes) for Carbamazapine powder in FaSSIF. Filled points indicate the portion of the dissolution curve that was used for model fitting (blue line) that produced parameters listed in the Table 2.

Unlike pure CBZ, the CBZ-Soluplus extrudate sustained elevated solubility in FaSSIF (supersaturated state) over 24 hours of incubation. Figure 7 shows the initial hours of dissolution where a quasi-equilibrium solubility of at least 300 µg/mL was reached within first the 60 minutes of the experiment.

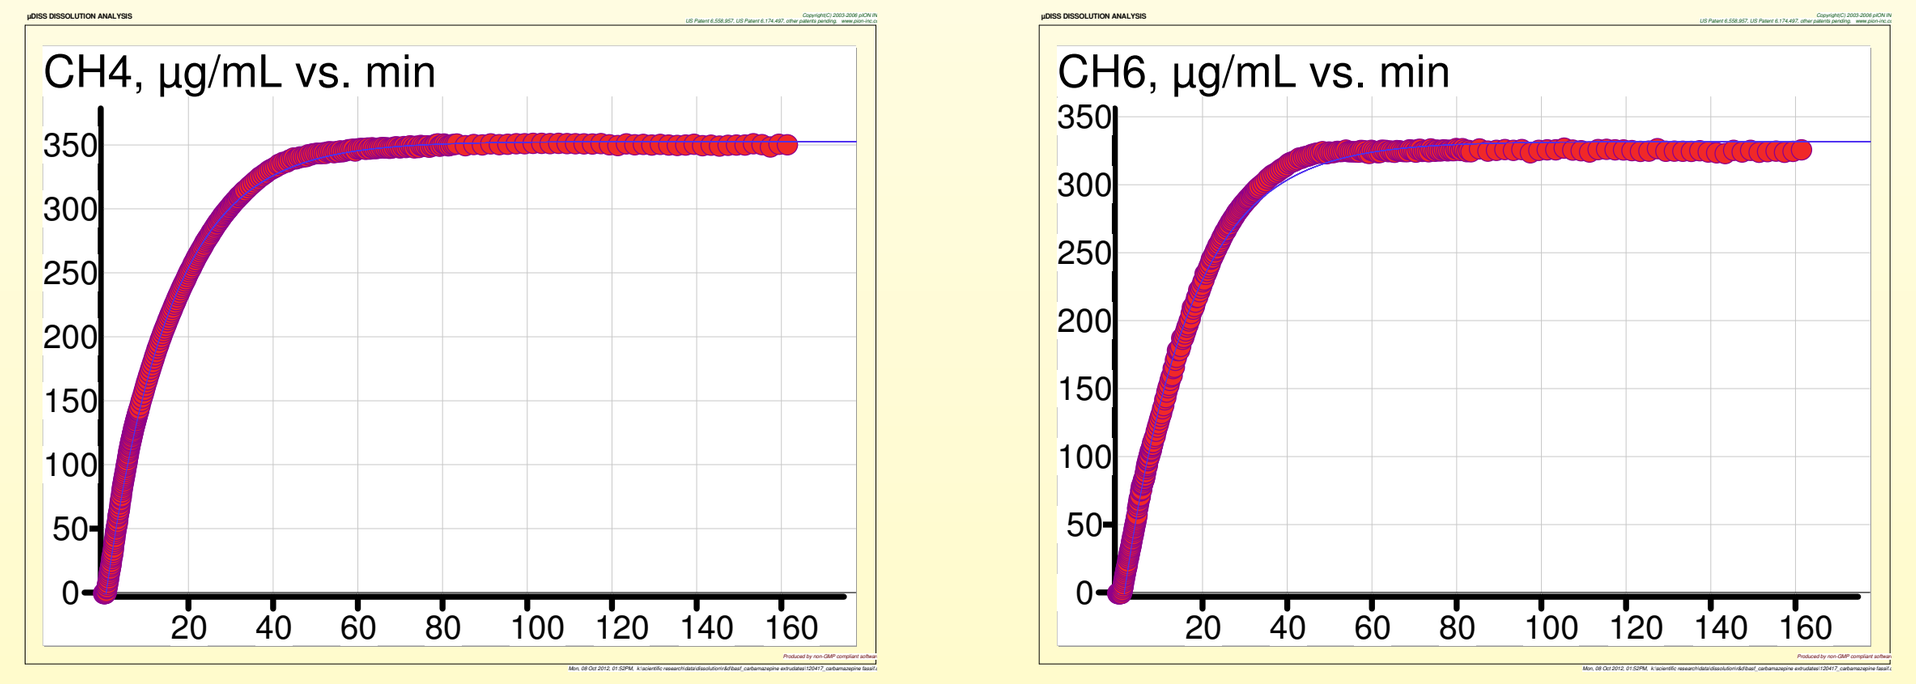
CH4, µg/mL vs. min
CH6, µg/mL vs. min

Figure 7. Examples of two replicates showing dissolution profile (µg/mL versus minutes) for CBZ-Soluplus powder in FaSSIF. The maximum reached concentration of CBZ was maintained over 24 hours of continuous monitoring.

Conclusions

CBZ-Soluplus hot melt extrudates maintained quasi-stable supersaturation of CBZ in both of the studied media while pure CBZ re-precipitated within 1 hour of reaching its maximum kinetic solubility.

Prolonged supersaturation of CBZ hot melt extrudates with Soluplus indicated a potential for higher bioavailability of CBZ from such formulations.

The µDISS Profiler provides a fast and effective way of selecting an appropriate solubilizer for enhancing the bioavailability of poorly soluble compounds.

Its ability to monitor the concentration of dissolving/precipitating compounds *in situ* in small volumes makes the µDISS Profiler the tool of choice for studying kinetic phenomena in a variety of dissolution media.

References

1. Mehta M. AAPS/FDA Workshop on Bio-pharmaceutics Classification Systems. September 2002, 25-27.
2. Hardung H., Djuric D., and Ali S. Combining HME & Solubilization: Soluplus® - The Solid Solution. *Drug Delivery Tech.*, **2010**, 10 (3), 20-27.
3. Grzeiak A.L., Lang M., Kim K., and Matzger A.J. Comparison of the four anhydrious polymorphs of carbamazepine and the crystal structure of form I. *J. Pharm. Sci.*, **2003**, 92, 2260-2271.
4. Avdeef A., Tsinman K., Tsinman O., Sun N., and Voloboy D. Miniaturization of Powder Dissolution Measurement and Estimation of Particle Size. *Chem. Biodev.* **2009**, 6 (11), 1796-1811.
5. Tsinman K., Avdeef A., Tsinman O., and Voloboy D. Powder Dissolution Method for Estimating Rotating Disk Intrinsic Dissolution Rates of Low Solubility Drugs. *Pharm. Res.*, **2009**, 26 (9), 2093-2100.
6. Wang J., Flanagan D.R., Genaral Solution for Diffusion-Controlled Dissolution of Spherical Particles. 1. Theory. *J. Pharm. Sci.*, **1999**, 88, 731-738.

Pion

AAPS Annual Meeting, Chicago, 2012