Soluble TL1A/Anti-TL1A Immune Complexes Induce *De Novo* Production of TL1A, Restore FcγR Binding and Induce Anti-drug Antibodies That May Cross-Bind Approved IBD Antibodies

SHATTUCK LABS

PP1330

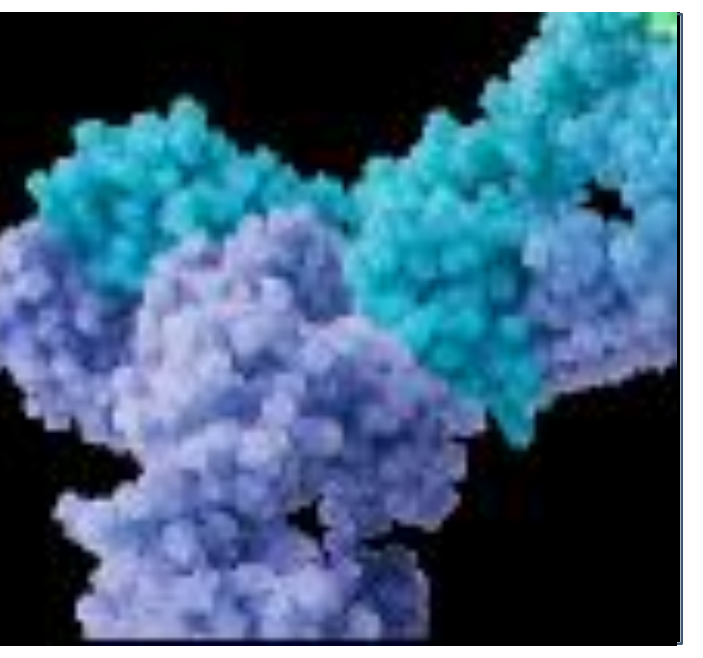

Harunobu Kato, Noah Murr, Karen Lenz, Mahmud Hussain, Anne Y. Lai, Arpita Patel, Izabela Webster, Alexandra Hahn, Nate Oien, Ronita Shukla, Taylor H. Schreiber and Suresh de Silva

***Shattuck Labs, Inc.* Austin, TX & Durham, NC**

1. Background

- Neutralization and blockade of soluble and membrane forms of TL1A, respectively, by monoclonal antibodies (mAb) have shown promising clinical responses in IBD patients (tulisokibart, afimkibart and duvakitug)
- Binding of anti-TL1A antibodies to soluble, trimeric TL1A (sTL1A) in blood can create immune complexes (IC)
- IC formation contributes to high rates of anti-drug antibody (ADA) response for anti-TL1A antibodies in 48-100% patients in Phase 1 clinical trials
- ICs between anti-TL1A antibodies and TL1A may catalyze a positive feedback loop of increased TL1A production, because TL1A is known to be induced by ICs. Accumulation of ICs could further increase the risk of immunogenicity and broaden the clonality of the anti-TL1A mAb ADA response.
- A majority of the ADA response to anti-TL1A mAbs has been described as 'non-neutralizing', indicating that the ADA bind the framework regions of the anti-TL1A antibodies instead of the paratopic/CDR regions
- Non-neutralizing ADA can lead to loss of exposure of anti-TL1A antibody therapies and reduced efficacy in patients
- The heavy chain framework regions of anti-TL1A mAbs shares 95-97% sequence identity to Risankizumab (Risa) and Vedolizumab (Vedo), suggesting ADA to TL1A blocking antibodies are likely to cross-bind Risa and Vedo

	HC Framework (% identity)		
Tulisokibart			
Afimkibart	96		
Duvakitug	96	96	
Vedolizumab	95	95	96
Risankizumab	95	96	97
Mirikizumab	90	90	90
Guselkumab	91	90	93
	Tulisokibart	Afimkibart	Duvakitug

	LC Framework (% identity)		
Tulisokibart			
Afimkibart	97		
Duvakitug	49	48	
Vedolizumab	87	87	46
Risankizumab	84	85	46
Mirikizumab	87	88	48
Guselkumab	49	48	97
	Tulisokibart	Afimkibart	Duvakitug

2. Study objectives

- Evaluate whether sTL1A/anti-TL1A ICs lead to de novo production of TL1A
- Evaluate the relative immunogenicity of sTL1A, anti-TL1A mAbs, or sTL1A/anti-TL1A ICs in pre-clinical models
- Evaluate whether ADA's against sTL1A/anti-TL1A ICs can cross-bind and increase the clearance of other therapeutic antibodies that share high degree of sequence identity in the framework region.

3. *In vitro* workflow

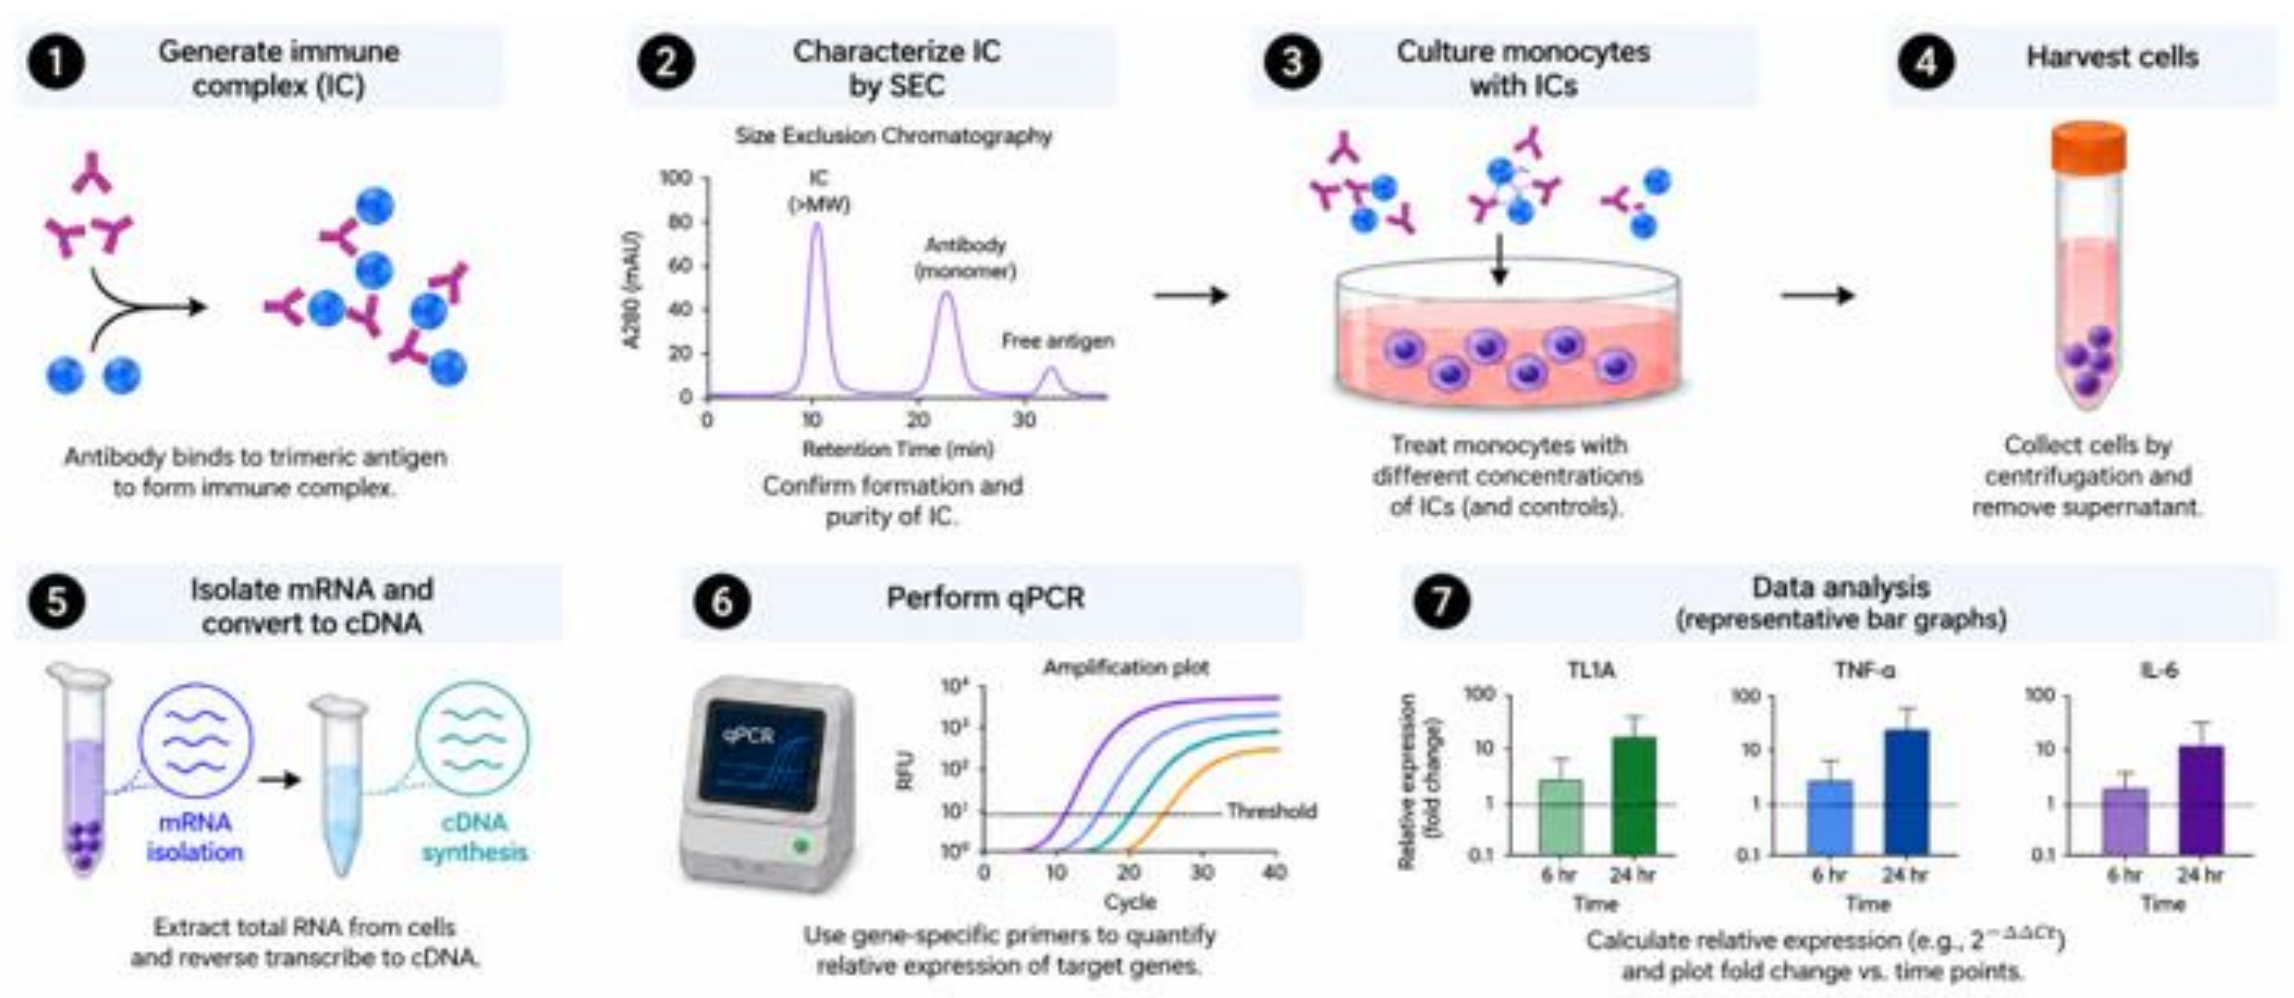

1 Generate immune complex (IC)
Antibody binds to trimeric antigen to form immune complex.
2 Characterize IC by SEC
Size Exclusion Chromatography
Confirm formation and purity of IC.
3 Culture monocytes with ICs
Treat monocytes with different concentrations of ICs (and controls).
4 Harvest cells
Collect cells by centrifugation and remove supernatant.
5 Isolate mRNA and convert to cDNA
Extract total RNA from cells and reverse transcribe to cDNA.
6 Perform qPCR
Use gene-specific primers to quantify relative expression of target genes.
7 Data analysis (representative bar graphs)
Calculate relative expression (e.g., 2^-ΔΔCt) and plot fold change vs. time points.

4. Immunocomplexes (IC) trigger TL1A production in antigen presenting cells

A. Human IgG-based ICs can induce TL1A production in human monocytes

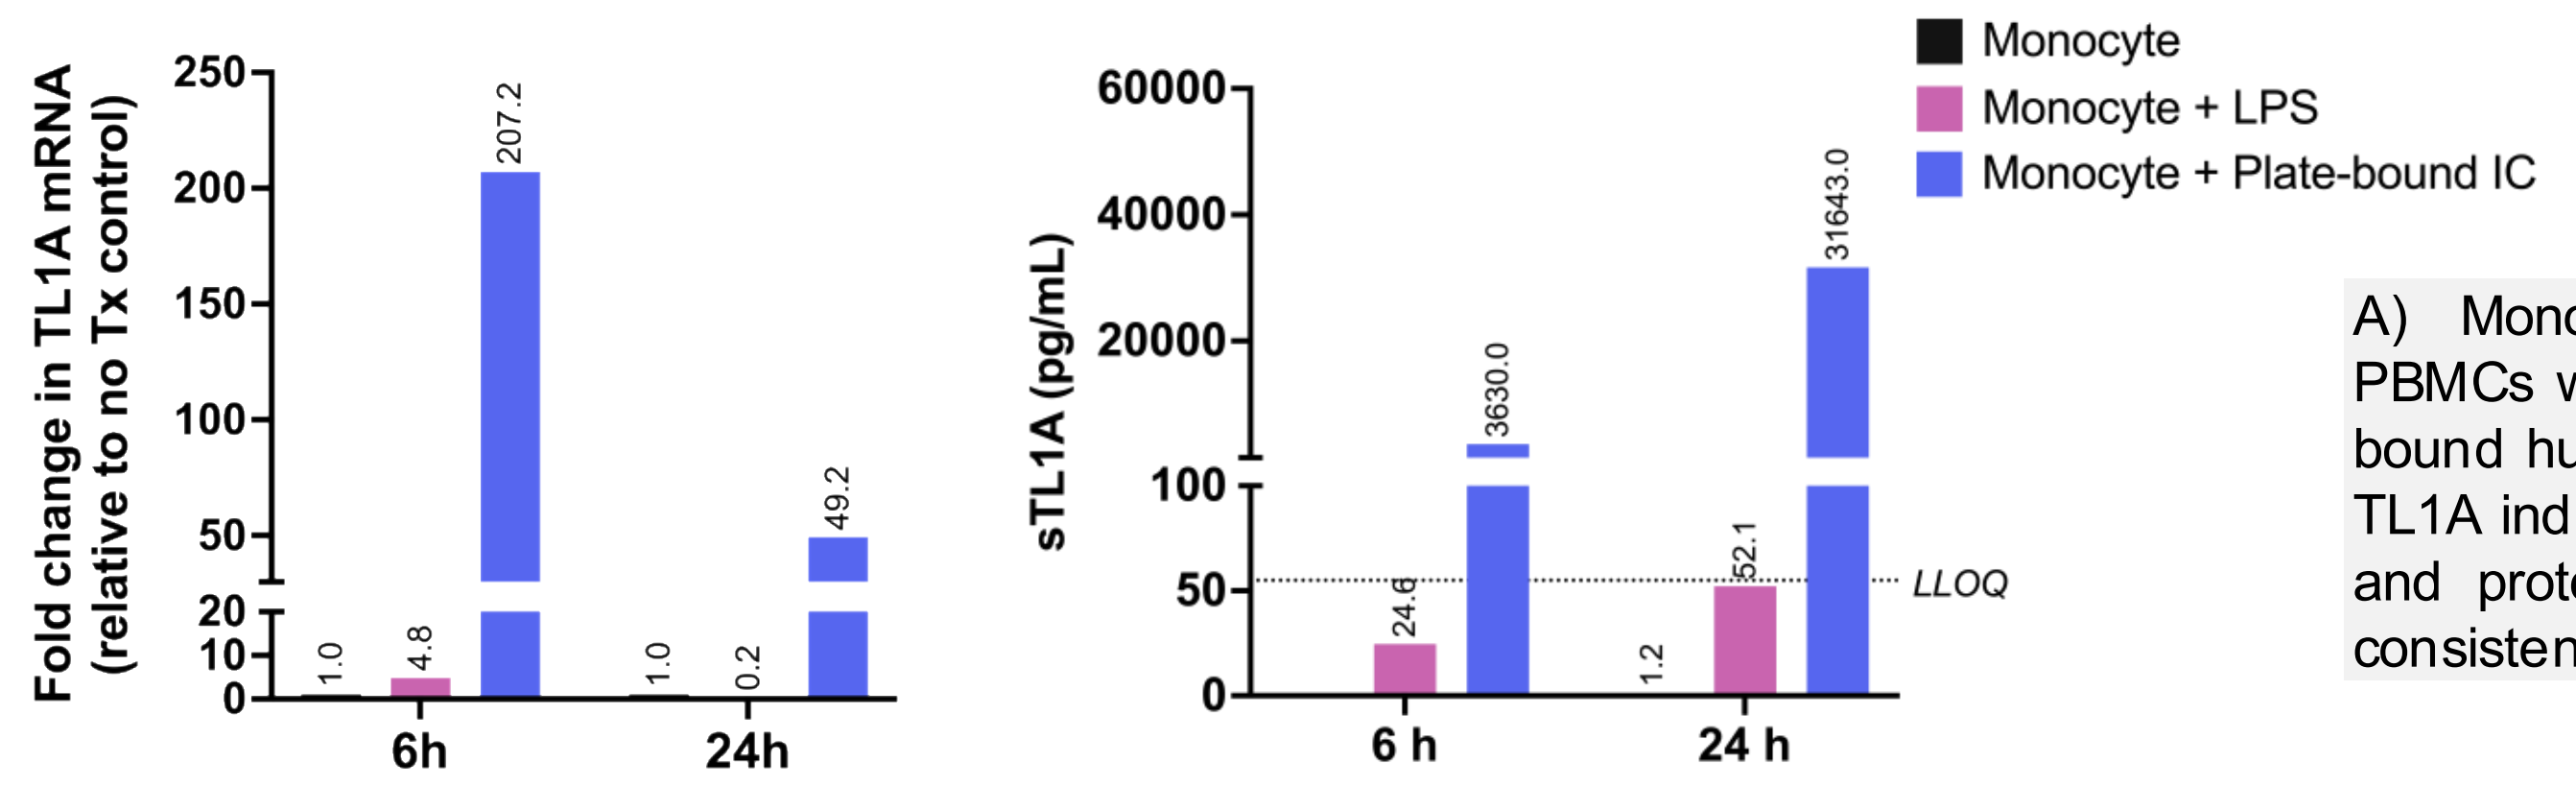

Fold change in TL1A mRNA (relative to no Tx control)
207.2
49.2
6h
24h
sTL1A (pg/mL)
3630.0
31643.0
LLOQ
6 h
24 h
Monocyte
Monocyte + LPS
Monocyte + Plate-bound IC

A) Monocytes isolated from human PBMCs were activated with either plate-bound human IgG IC or LPS to confirm TL1A induction at the mRNA (qRT-PCR) and protein (MSD) level. This data is consistent with previous publications[1-3].

B. TL1A/Tulisokibart ICs induce TL1A expression in monocytes similar to TNFα/Adalimumab ICs

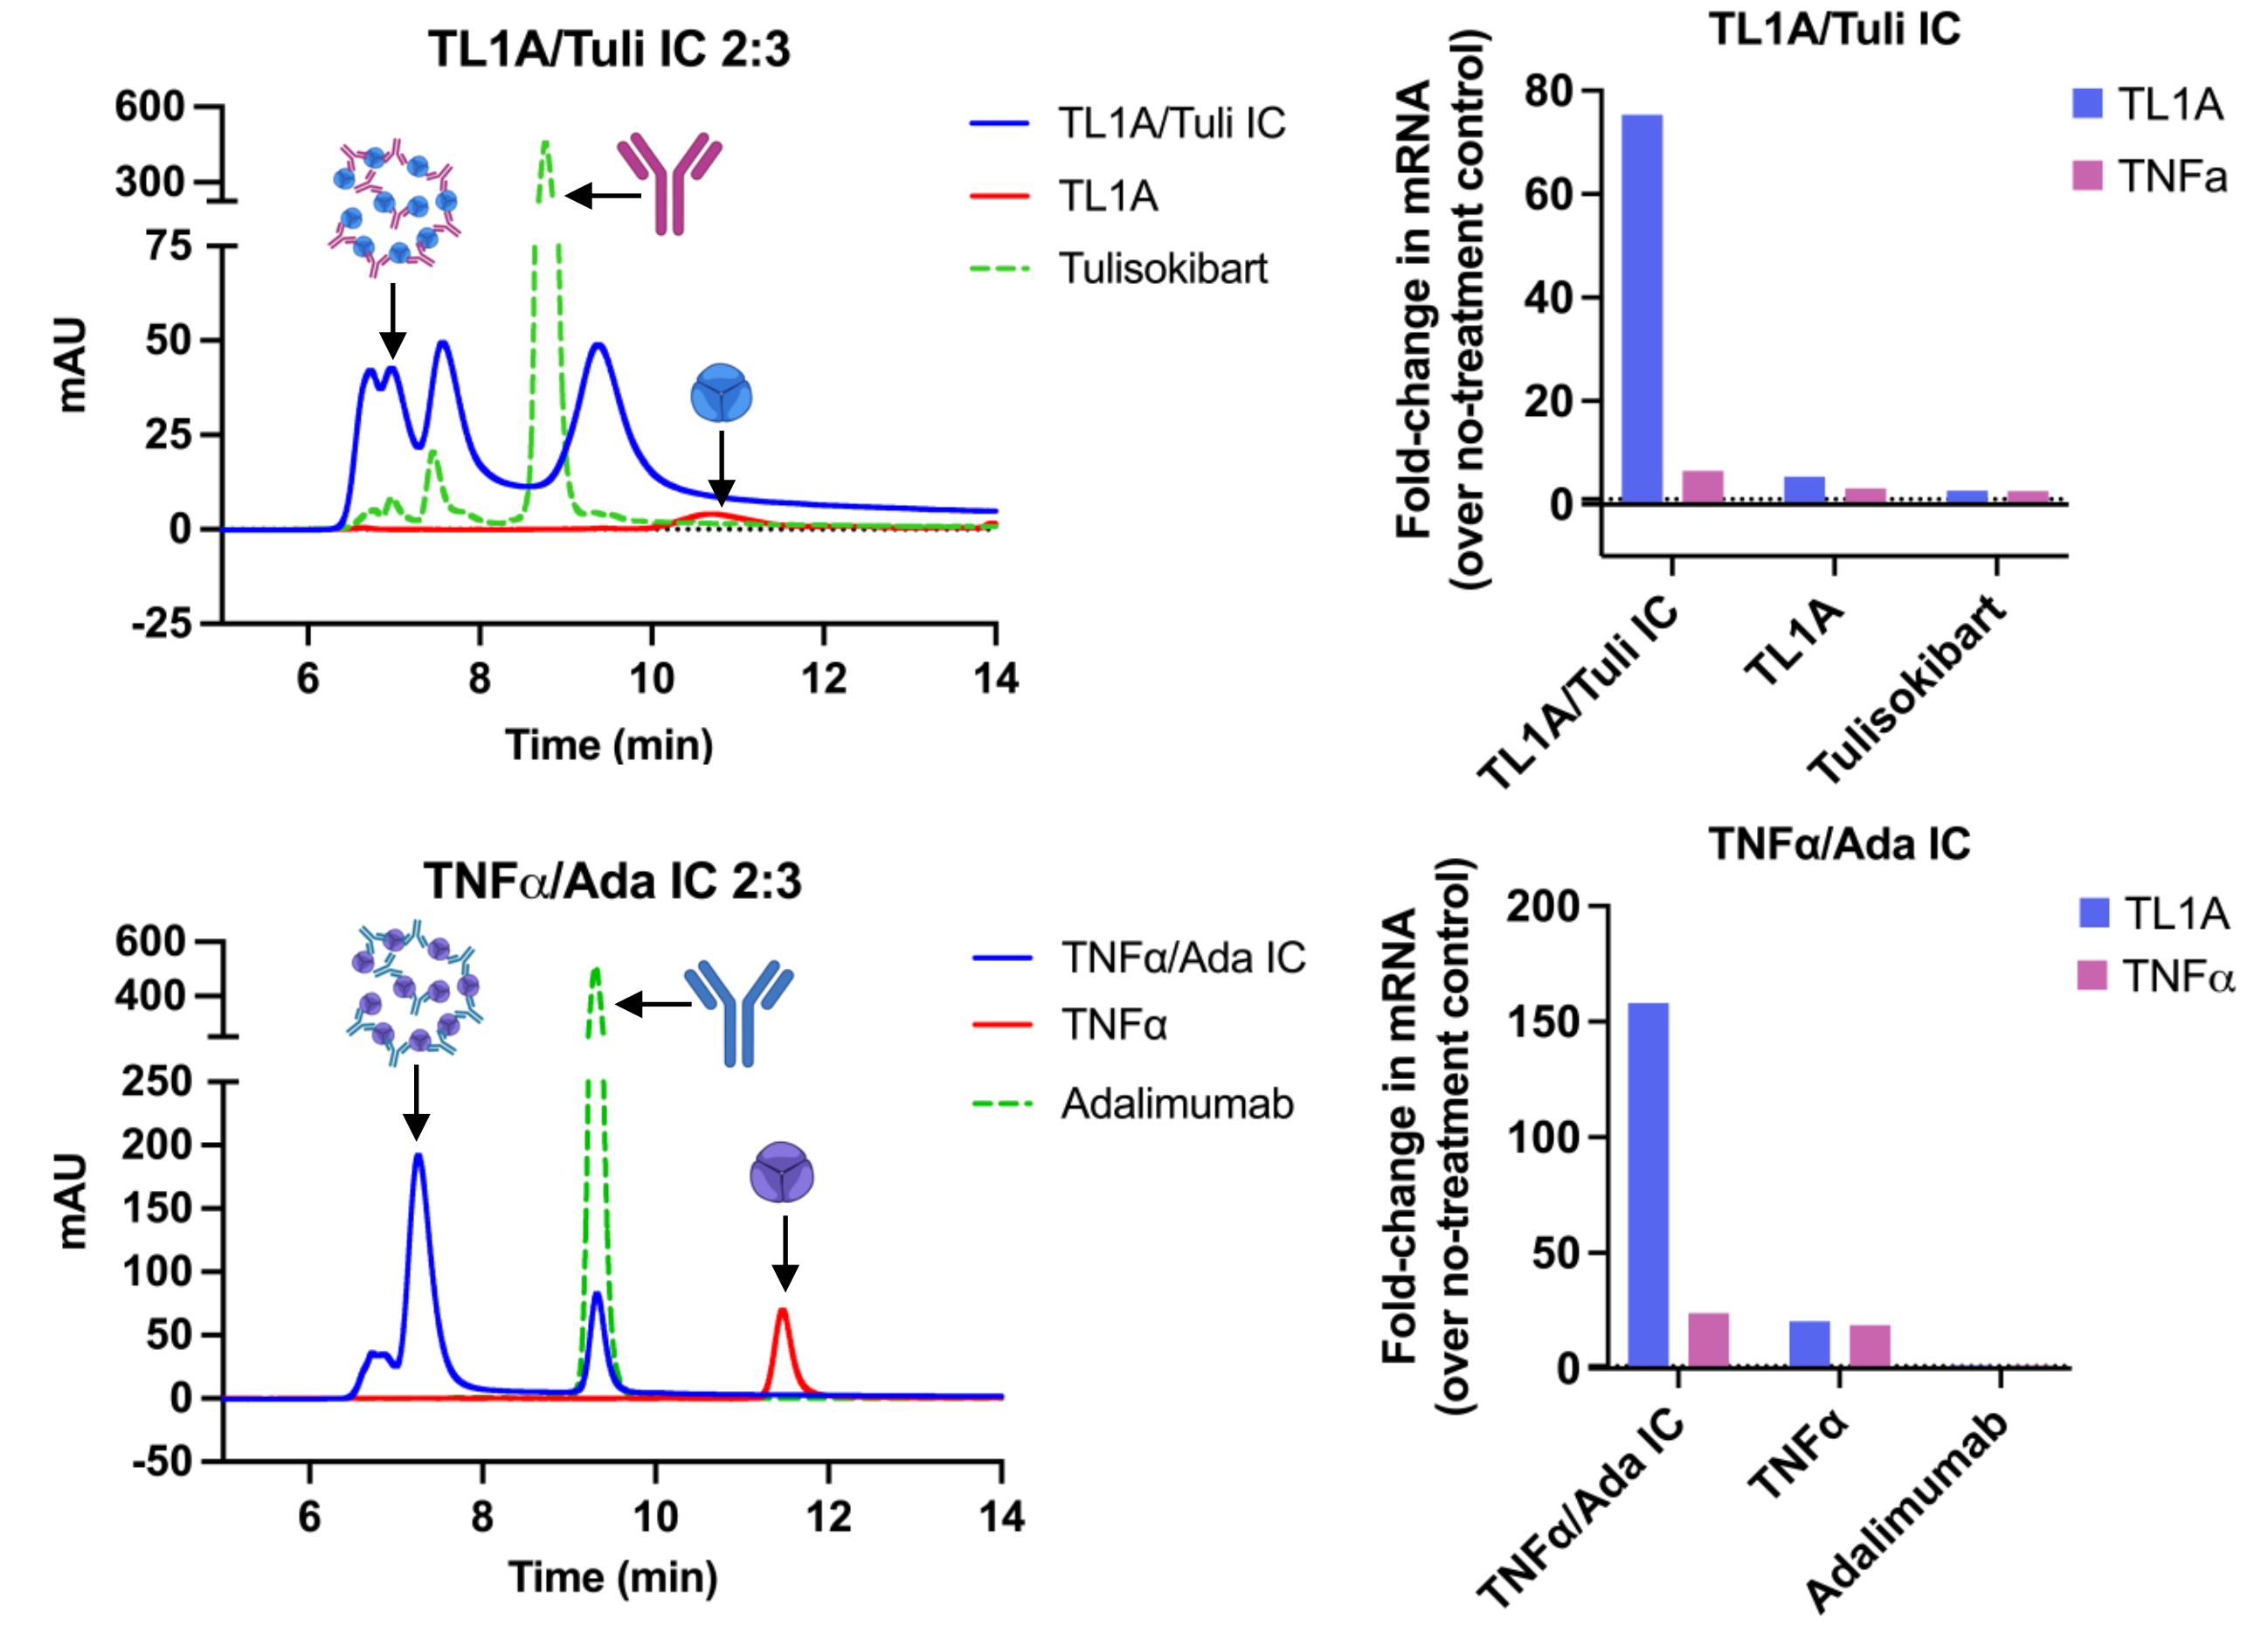

TL1A/Tuli IC 2:3
TL1A/Tuli IC
TL1A
Tulisokibart
TNFα/Ada IC 2:3
TNFα/Ada IC
TNFα
Adalimumab
mAU
Time (min)
Fold-change in mRNA (over no-treatment control)
TL1A
TNFa

- TL1A protein and Tulisokibart (Tuli; Top) or TNFα protein and adalimumab (Ada; Bottom) sequence equivalent antibodies were mixed at a 2:3 molar ratio overnight at 4ºC to generate IC and profiled by size exclusion chromatography (SEC) – left panels.
- Human Monocytes were isolated from healthy donor PBMCs and incubated with either TL1A/Tuli IC or TNFα/Adalimumab ICs for 6 hours and the level of TL1A and TNFα mRNA was measured using qRT-PCR - right panels. Beta-2-microglobulin mRNA levels were used as a house keeping gene control for normalization. Fold changes in mRNA levels following treatment with ICs were calculated using the ΔΔCT method.

5. Crosslinked and non-crosslinked TL1A/Tulisokibart ICs restore Fcγ receptor binding

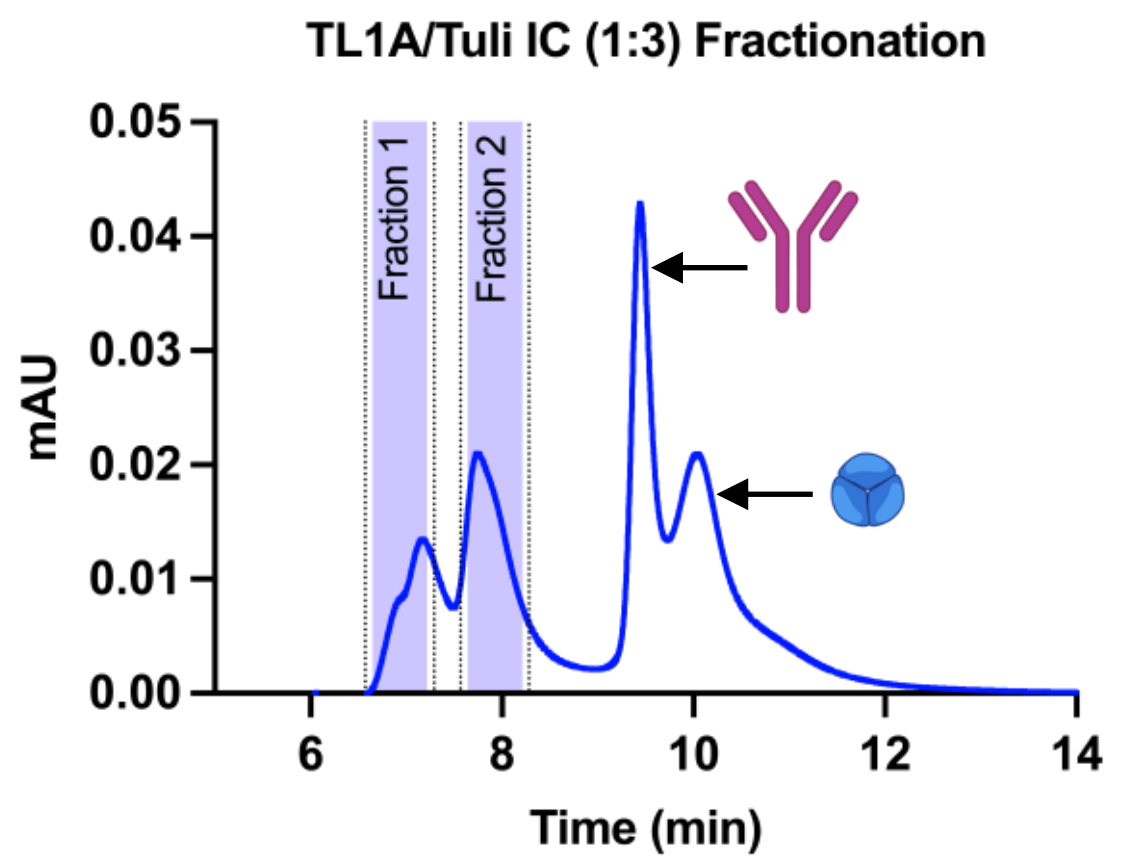

TL1A/Tuli IC (1:3) Fractionation
Fraction 1
Fraction 2
mAU
Time (min)

- TL1A/Tuli IC were prepared, generating non-cross linked species (1 TL1A trimer + 3 Tuli, Fraction 2) and cross-linked (2+ TL1A trimer + 4+ Tuli, Fraction 1) species.

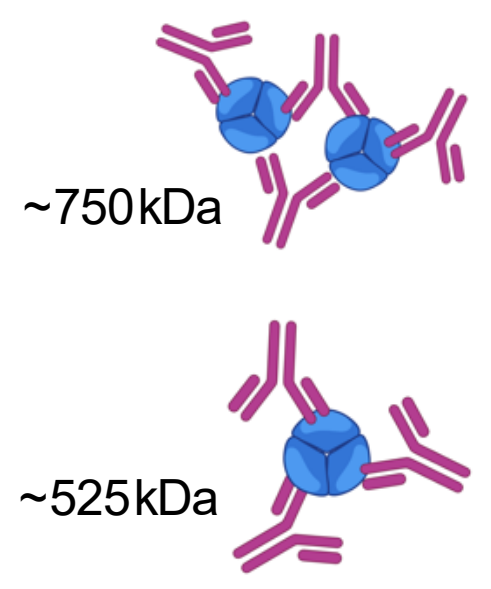

~750kDa
~525kDa

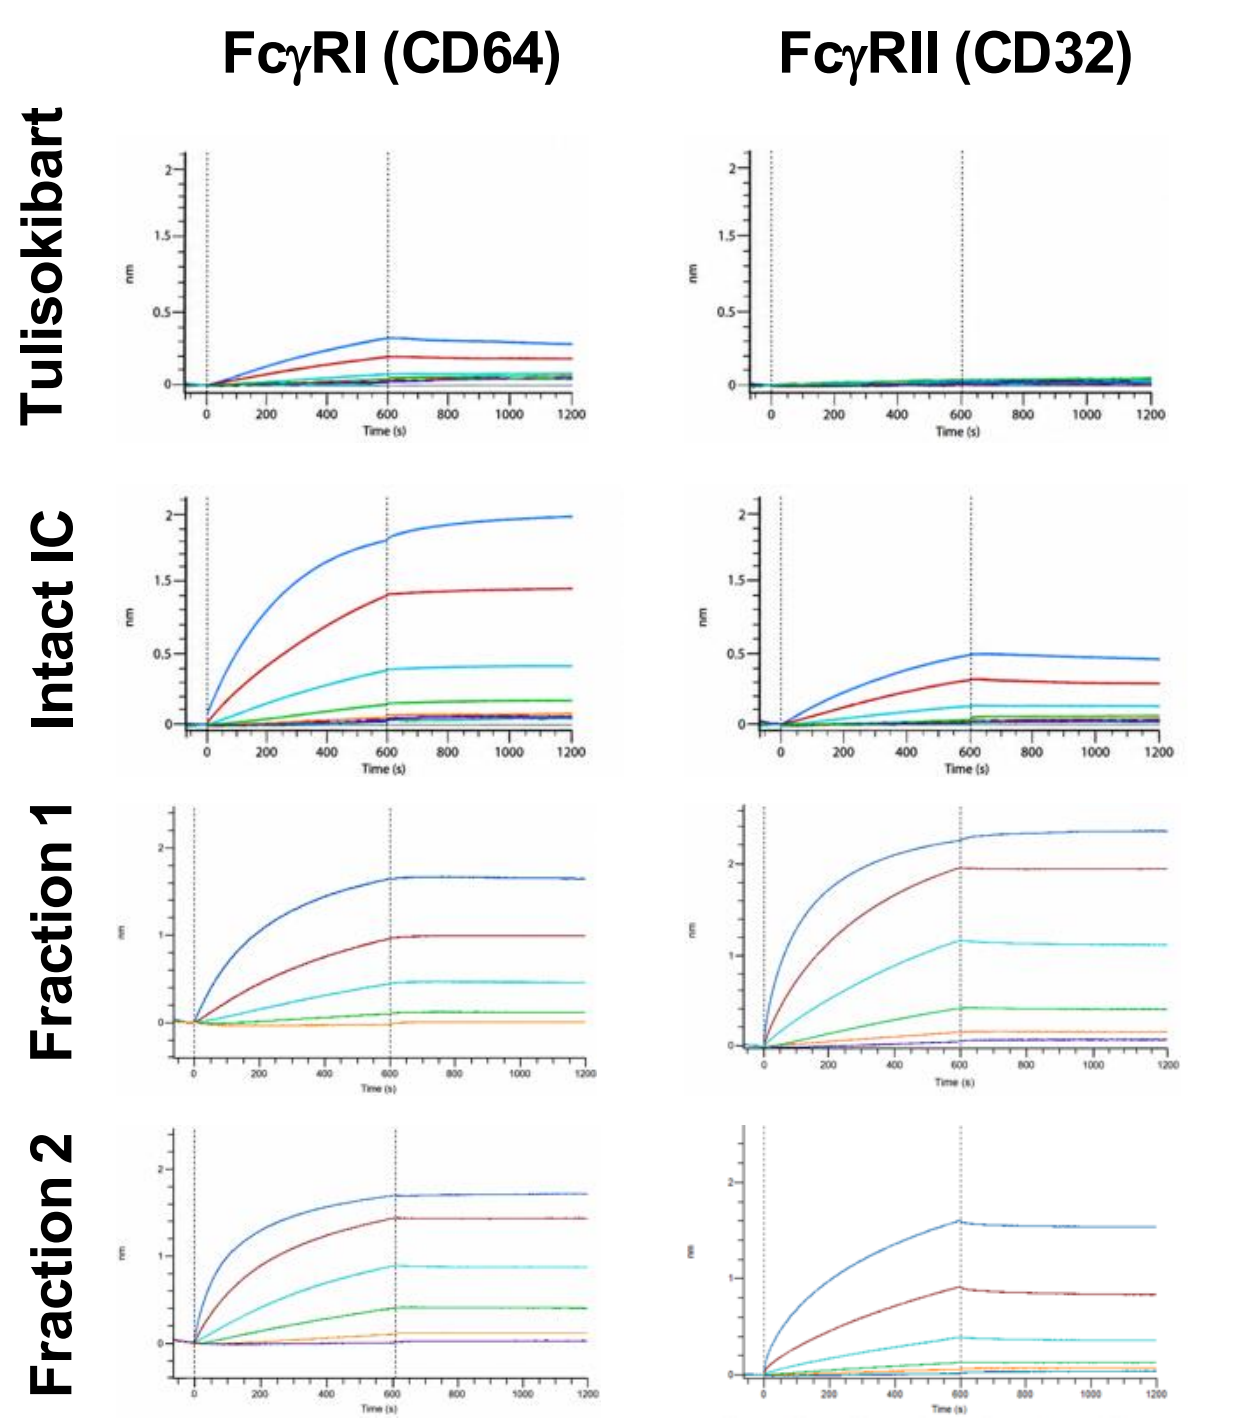

FcγRI (CD64)
FcγRII (CD32)
Tulisokibart
Intact IC
Fraction 1
Fraction 2

- Peak fractions were isolated by SEC and tested for FcγRI (CD64) and FcγRII (CD32) binding by BLI.
- Tulisokibart was used as a control to show the lack of FcγRI and RII due to Fc silencing mutations in the Fc domain (LALA + G237A)

6. TL1A/Tulisokibart IC prime for accelerated clearance of Risankizumab and Vedolizumab

A) *In vivo* study design in C57BL/6 mice with TL1A/Tuli IC

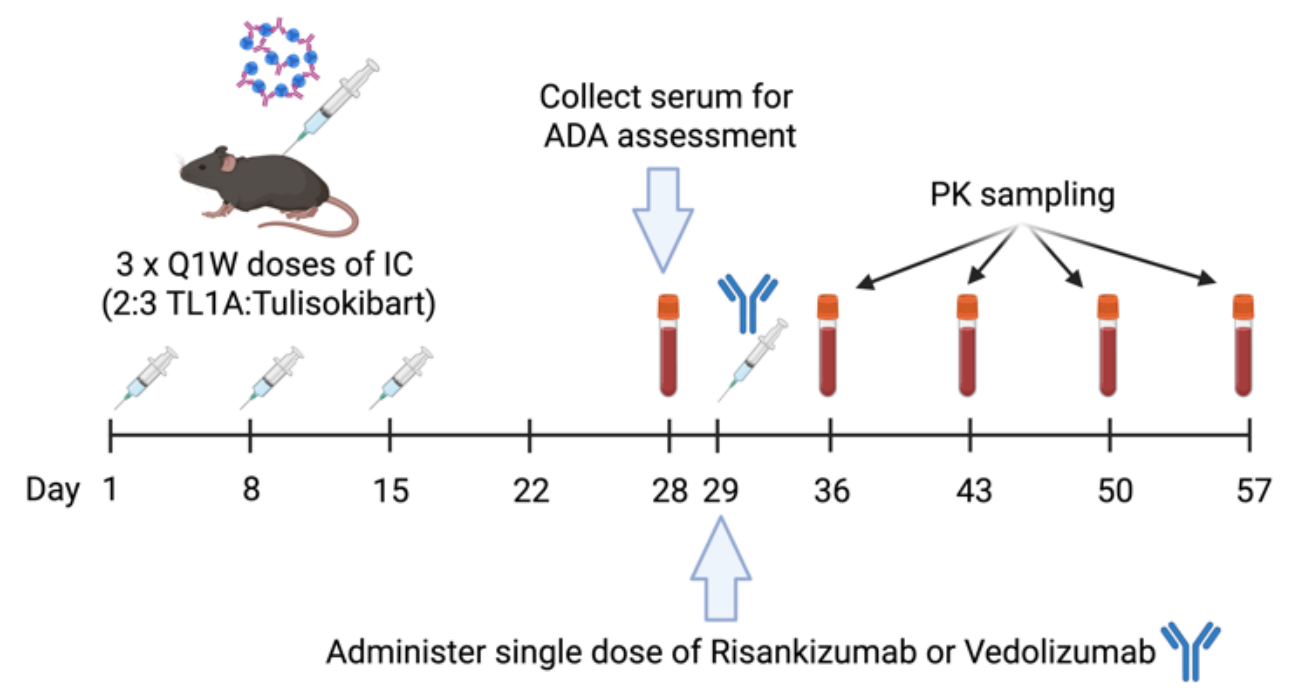

3 x Q1W doses of IC (2:3 TL1A:Tulisokibart)
Collect serum for ADA assessment
PK sampling
Day 1 8 15 22 28 29 36 43 50 57
Administer single dose of Risankizumab or Vedolizumab

B) Anti-IC ADA in mice primes for boosted immunogenicity to Risa and Vedo

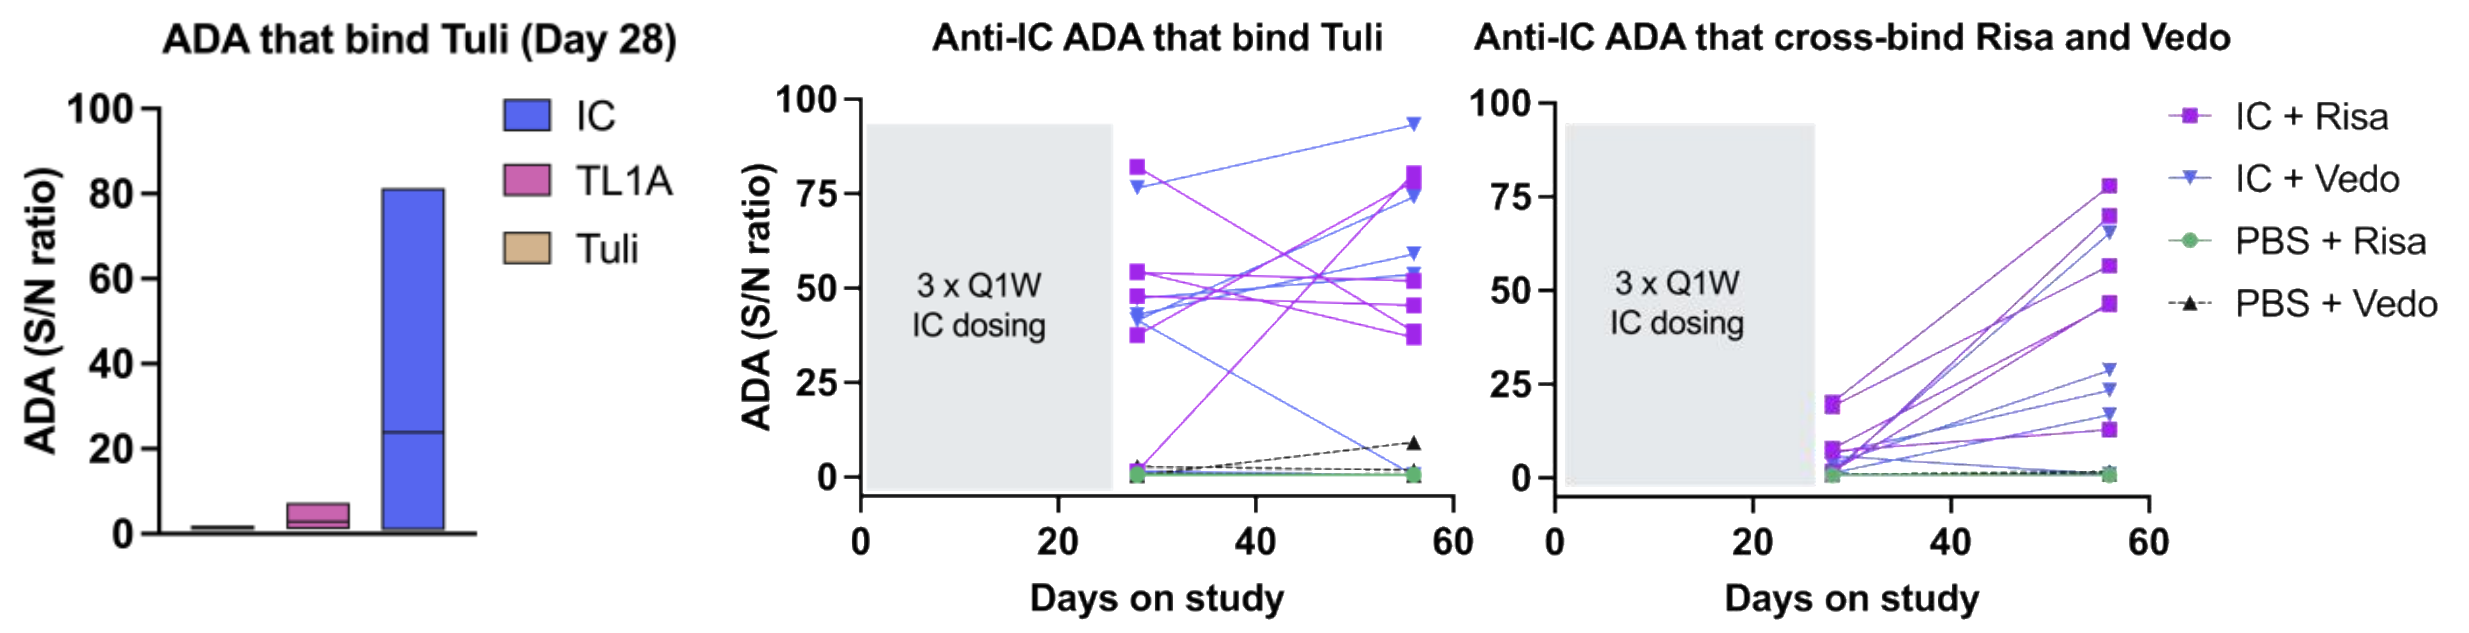

ADA that bind Tuli (Day 28)
Anti-IC ADA that bind Tuli
Anti-IC ADA that cross-bind Risa and Vedo
ADA (S/N ratio)
IC
TL1A
Tuli
3 x Q1W IC dosing
Days on study
IC + Risa
IC + Vedo
PBS + Risa
PBS + Vedo

C) Non-neutralizing anti-Tulisokibart ADA accelerate clearance of Risa and Vedo in mice

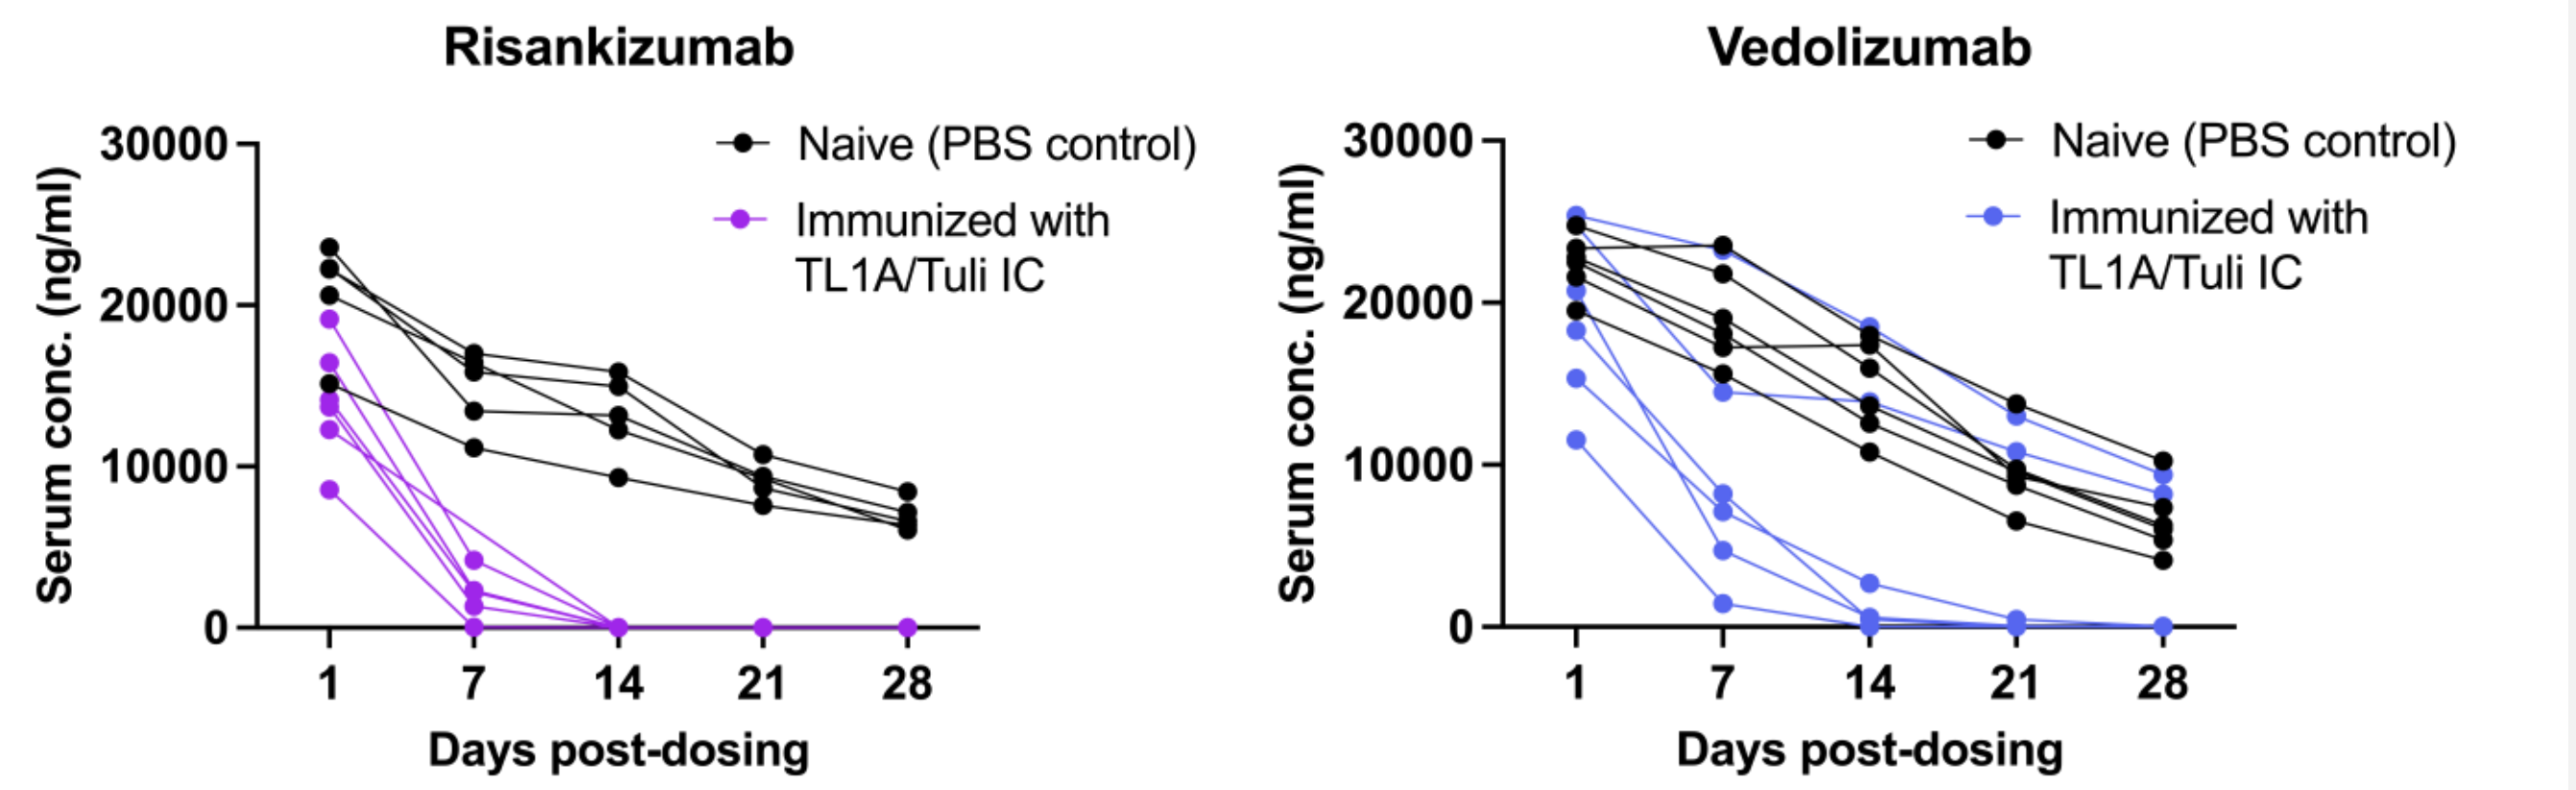

Risankizumab
Vedolizumab
Serum conc. (ng/ml)
Naive (PBS control)
Immunized with TL1A/Tuli IC
Days post-dosing

A) Schematic of the *in vivo* study design. B) ADA in mouse serum that bind Tuli was assessed using an MSD assay with Tuli as the capture antibody and a sulfo-tagged anti-mouse Fc antibody as the detector (left). The same serum was tested for cross-binding to Risa and Vedo in similar MSD assays with Risa and Vedo as the capture antibodies. ADA is represented as a signal-to-noise (S/N) ratio. C) Risa and Vedo concentrations were measured in the serum using separate MSD pharmacokinetic assays.

D) ICs with Afimkibart (Afim) and Duvakitug (Duva) are immunogenic, cross-bind Risa and increase clearance

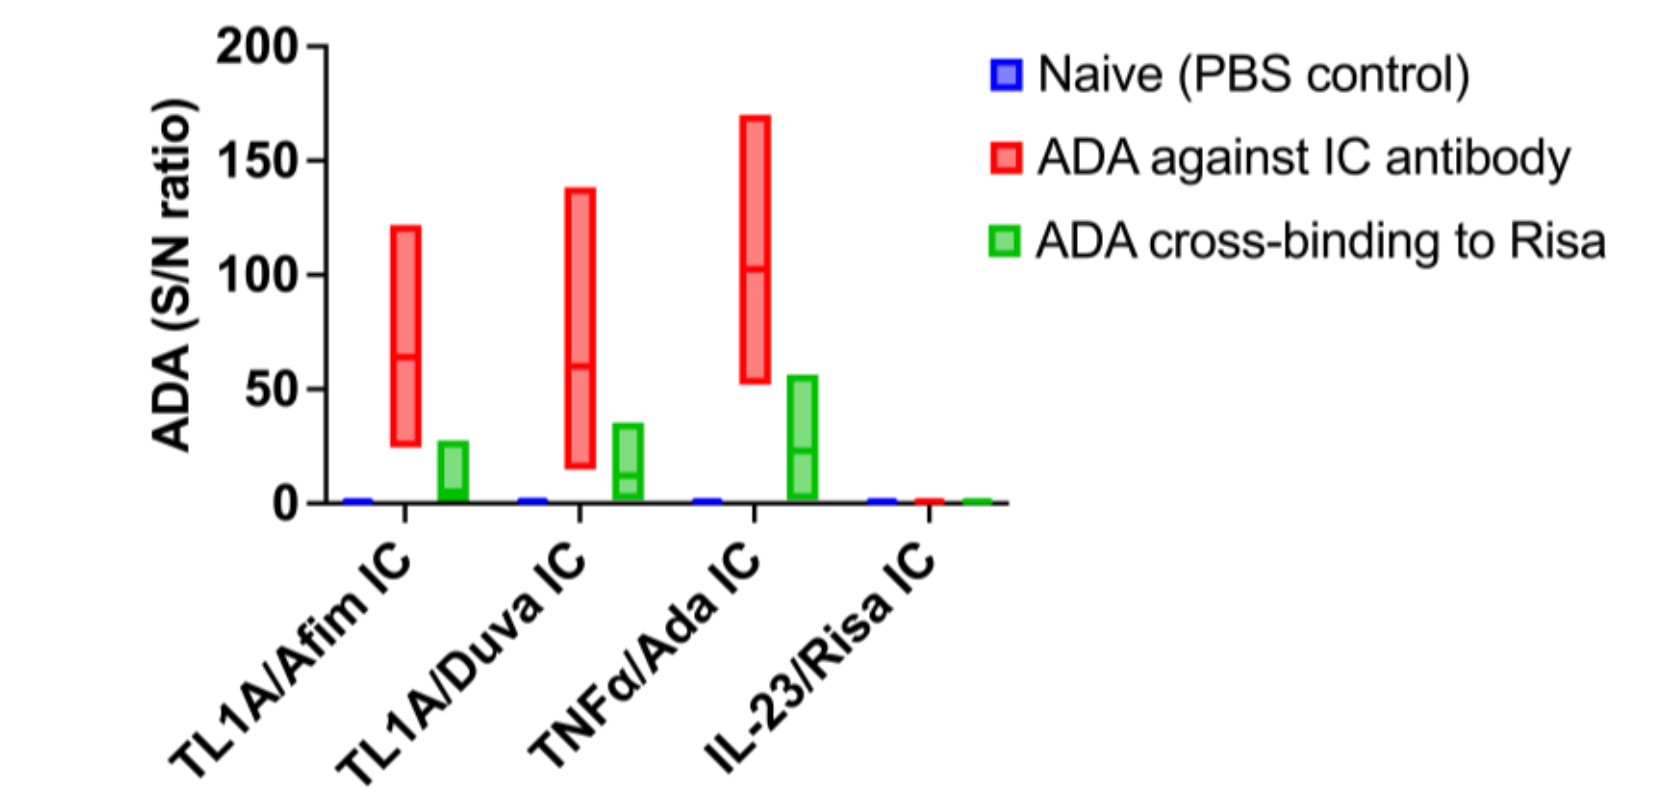

ADA (S/N ratio)
Naive (PBS control)
ADA against IC antibody
ADA cross-binding to Risa
TL1A/Afim IC
TL1A/Duva IC
TNFα/Ada IC
IL-23/Risa IC

E) IC lacking the Fc domain of Tuli induce lower rate of ADA confirming the potential role of Fc receptors in the induction of ADA

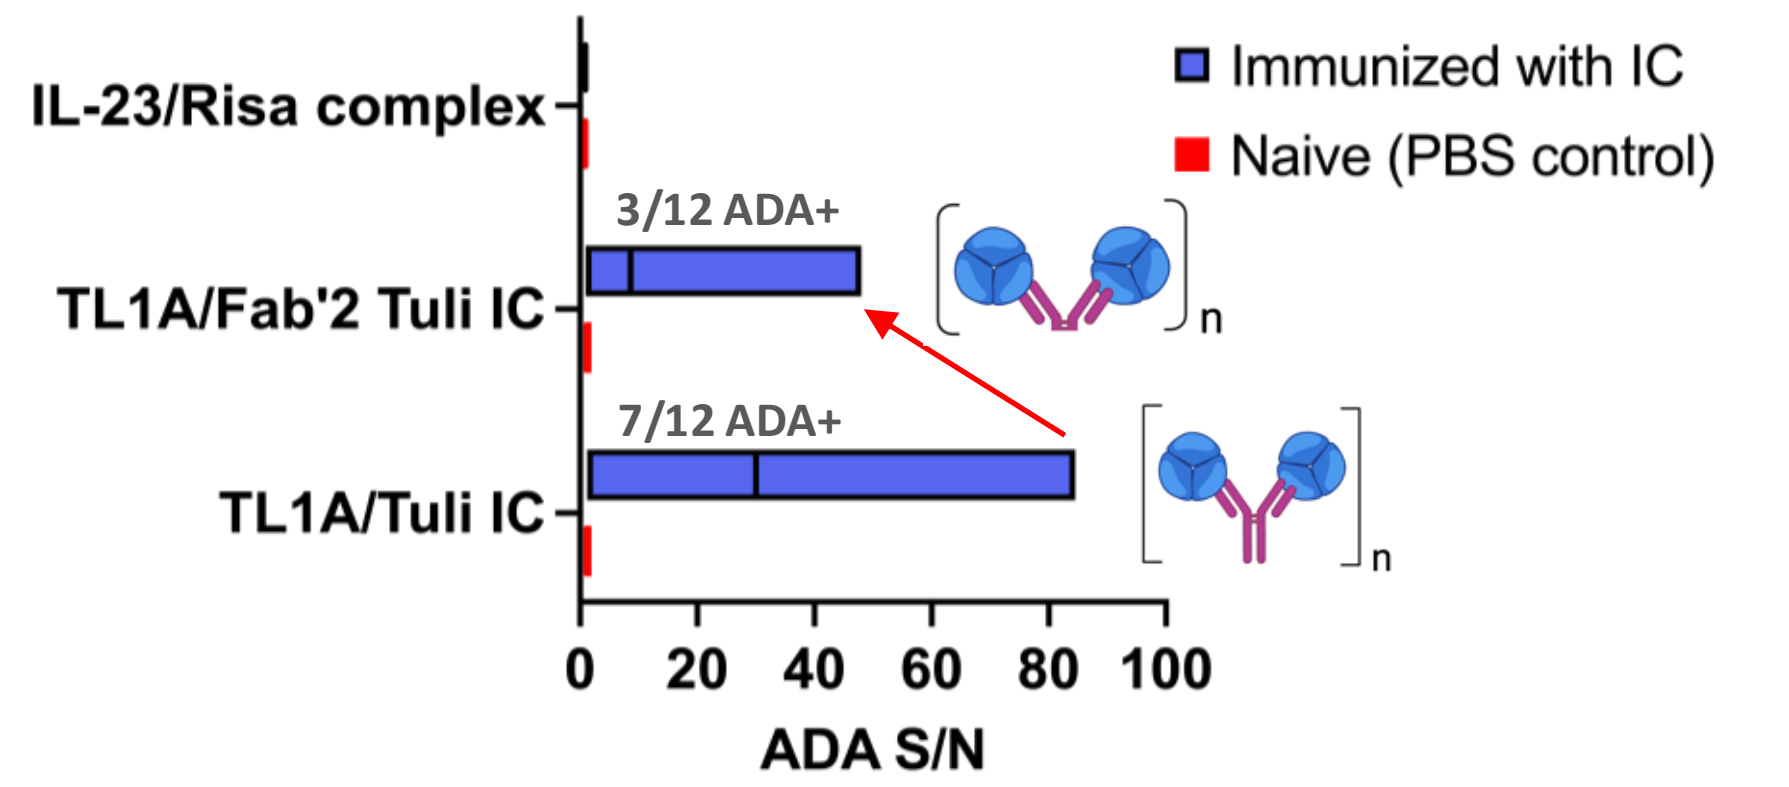

IL-23/Risa complex
TL1A/Fab'2 Tuli IC
TL1A/Tuli IC
3/12 ADA+
7/12 ADA+
Immunized with IC
Naive (PBS control)
ADA S/N

D) ICs were generated with TL1A and Afimkibart or Duvakitug and injected in C57BL/6 mice as described in A. TNFα/Adalimumab and IL-23/Risankizumab complexes were used as positive and negative controls, respectively. ADA that bind the constituent antibody in each IC was measured on day 28 using specific ADA assays and are represented as signal-to-noise ratios. Accelerated clearance of Risa and Vedo was observed in Afim and Duva IC-treated animals (data not shown) E) Tulisokibart was digested with pepsin and the Fab'2 fragments were purified using affinity chromatography. ICs were generated by mixing Fab'2 with TL1A (2:3) to create ICs and tested in mice (3 x Q1W dosing) to compare ADA rates compared to the TL1A/Tuli (whole antibody) IC. ADA was measured on day 28 using an MSD assay with Tuli as the capture and a sulfo-tagged anti-Fc detection antibody.

7. Conclusions

- TL1A/anti-TL1A ICs are highly immunogenic, restore FcγR binding, and are rapidly internalized by human monocytes.
- ADA generated against Tulisokibart, Afimkibart and Duvakitug ICs cross-bind IBD antibodies Risankizumab and Vedolizumab, which share >90% framework sequence identity.
- Cross-binding ADA have the potential to accelerate clearance of other antibodies with high sequence identity, including Risankizumab and Vedolizumab.
- Further evaluation of this phenomenon is warranted using serum samples from patients previously treated with anti-TL1A antibodies

References: [1]Cassatella MA., *J Immunol* 2015; [2]Meylan F., *Immunity* 2008; [3]Kroenke MA., *Front in Immunol* 2021

Conflict of declaration – all authors are employees and stock option holders of Shattuck Labs Inc.