Poster 590

A first-in-human, phase 1 trial of IMA203 PRAME-directed TCR T-cell therapy with PRAME encoding mRNA-4203 in previously treated, unresectable or metastatic cutaneous melanoma or synovial sarcoma (NCT06946225)

James Smithy[1], Rodabe N. Amaria[2], Adil Daud[3], Adi Diab[4,5], Karam Khaddour[6], Patrick Ott[6], Derrick Tao[3], Alyssa Flynn[7], Lin Guey[7], Brendan Weiss[7], Norbert Hilf[8], M. Alper Kursunel[8], Andrea Mayer-Mokler[8], Liane Preußner[8], Cedrik M. Britten[8], Dejka Araujo[9]

[1]Department of Medicine, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Melanoma Medical Oncology, University of Texas MD Anderson Center, Houston, TX, USA; [3]Department of Medicine (Hematology/Oncology), UCSF Health, San Francisco, CA, USA; [4]Cleveland Clinic, Abu Dhabi, United Arab Emirates; [5]Department of Melanoma, Cleveland Clinic, Cleveland, Ohio, USA; [6]Medical Oncology, Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA; [7]Moderna Inc, Cambridge, MA, USA; [8]Immatics Biotechnologies GmbH, Tuebingen, Germany; [9]Sarcoma Medical Oncology, University of Texas MD Anderson Center, Houston TX, USA.

BACKGROUND

- TCR T-cell therapy has demonstrated promising activity in treatment-refractory advanced cutaneous melanoma (CM) and synovial sarcoma (SS); there remain opportunities to improve outcomes by combining immunotherapy modalities[1]
- PRAME is expressed in more than 50 cancer types; 95% of CM and SS cancers express PRAME[2,3]
- **Anzutresgene autoleucel (anzu-cel; formerly IMA203)** is a PRAME-directed TCR T-cell therapy engineered to recognize intracellular PRAME-derived peptides presented by HLA-A*02:01 on the cell surface and initiate a potent and specific antitumor response[1]
- In a phase 1 trial (NCT03686124), anzu-cel exhibited favorable tolerability and encouraging antitumor activity in heavily pretreated patients with advanced melanoma at RP2D[4]
 - Confirmed ORR was 56% (18/32) and median DOR was 12.1 months
 - Median PFS was 6.1 months and OS was 15.9 months
 - Most frequent TEAE at all dose levels and indications (N=74) were lymphodepletion-related cytopenias (99%)
 - CRS was mostly lower grade (84% G1/2; 11% G3; no ≥G4) and ICANS was infrequent (10% G1/2; 4% G3; no ≥G4)
- mRNA-4203 is an LNP-encapsulated mRNA cell therapy enhancer that encodes the PRAME sequence that when translated by APCs, drives activation and proliferation of anzu-cel TCR T cells in the lymph node before encountering the tumor (**Fig. 1**)

Figure 1. Mechanism of action of anzu-cel and mRNA-4203

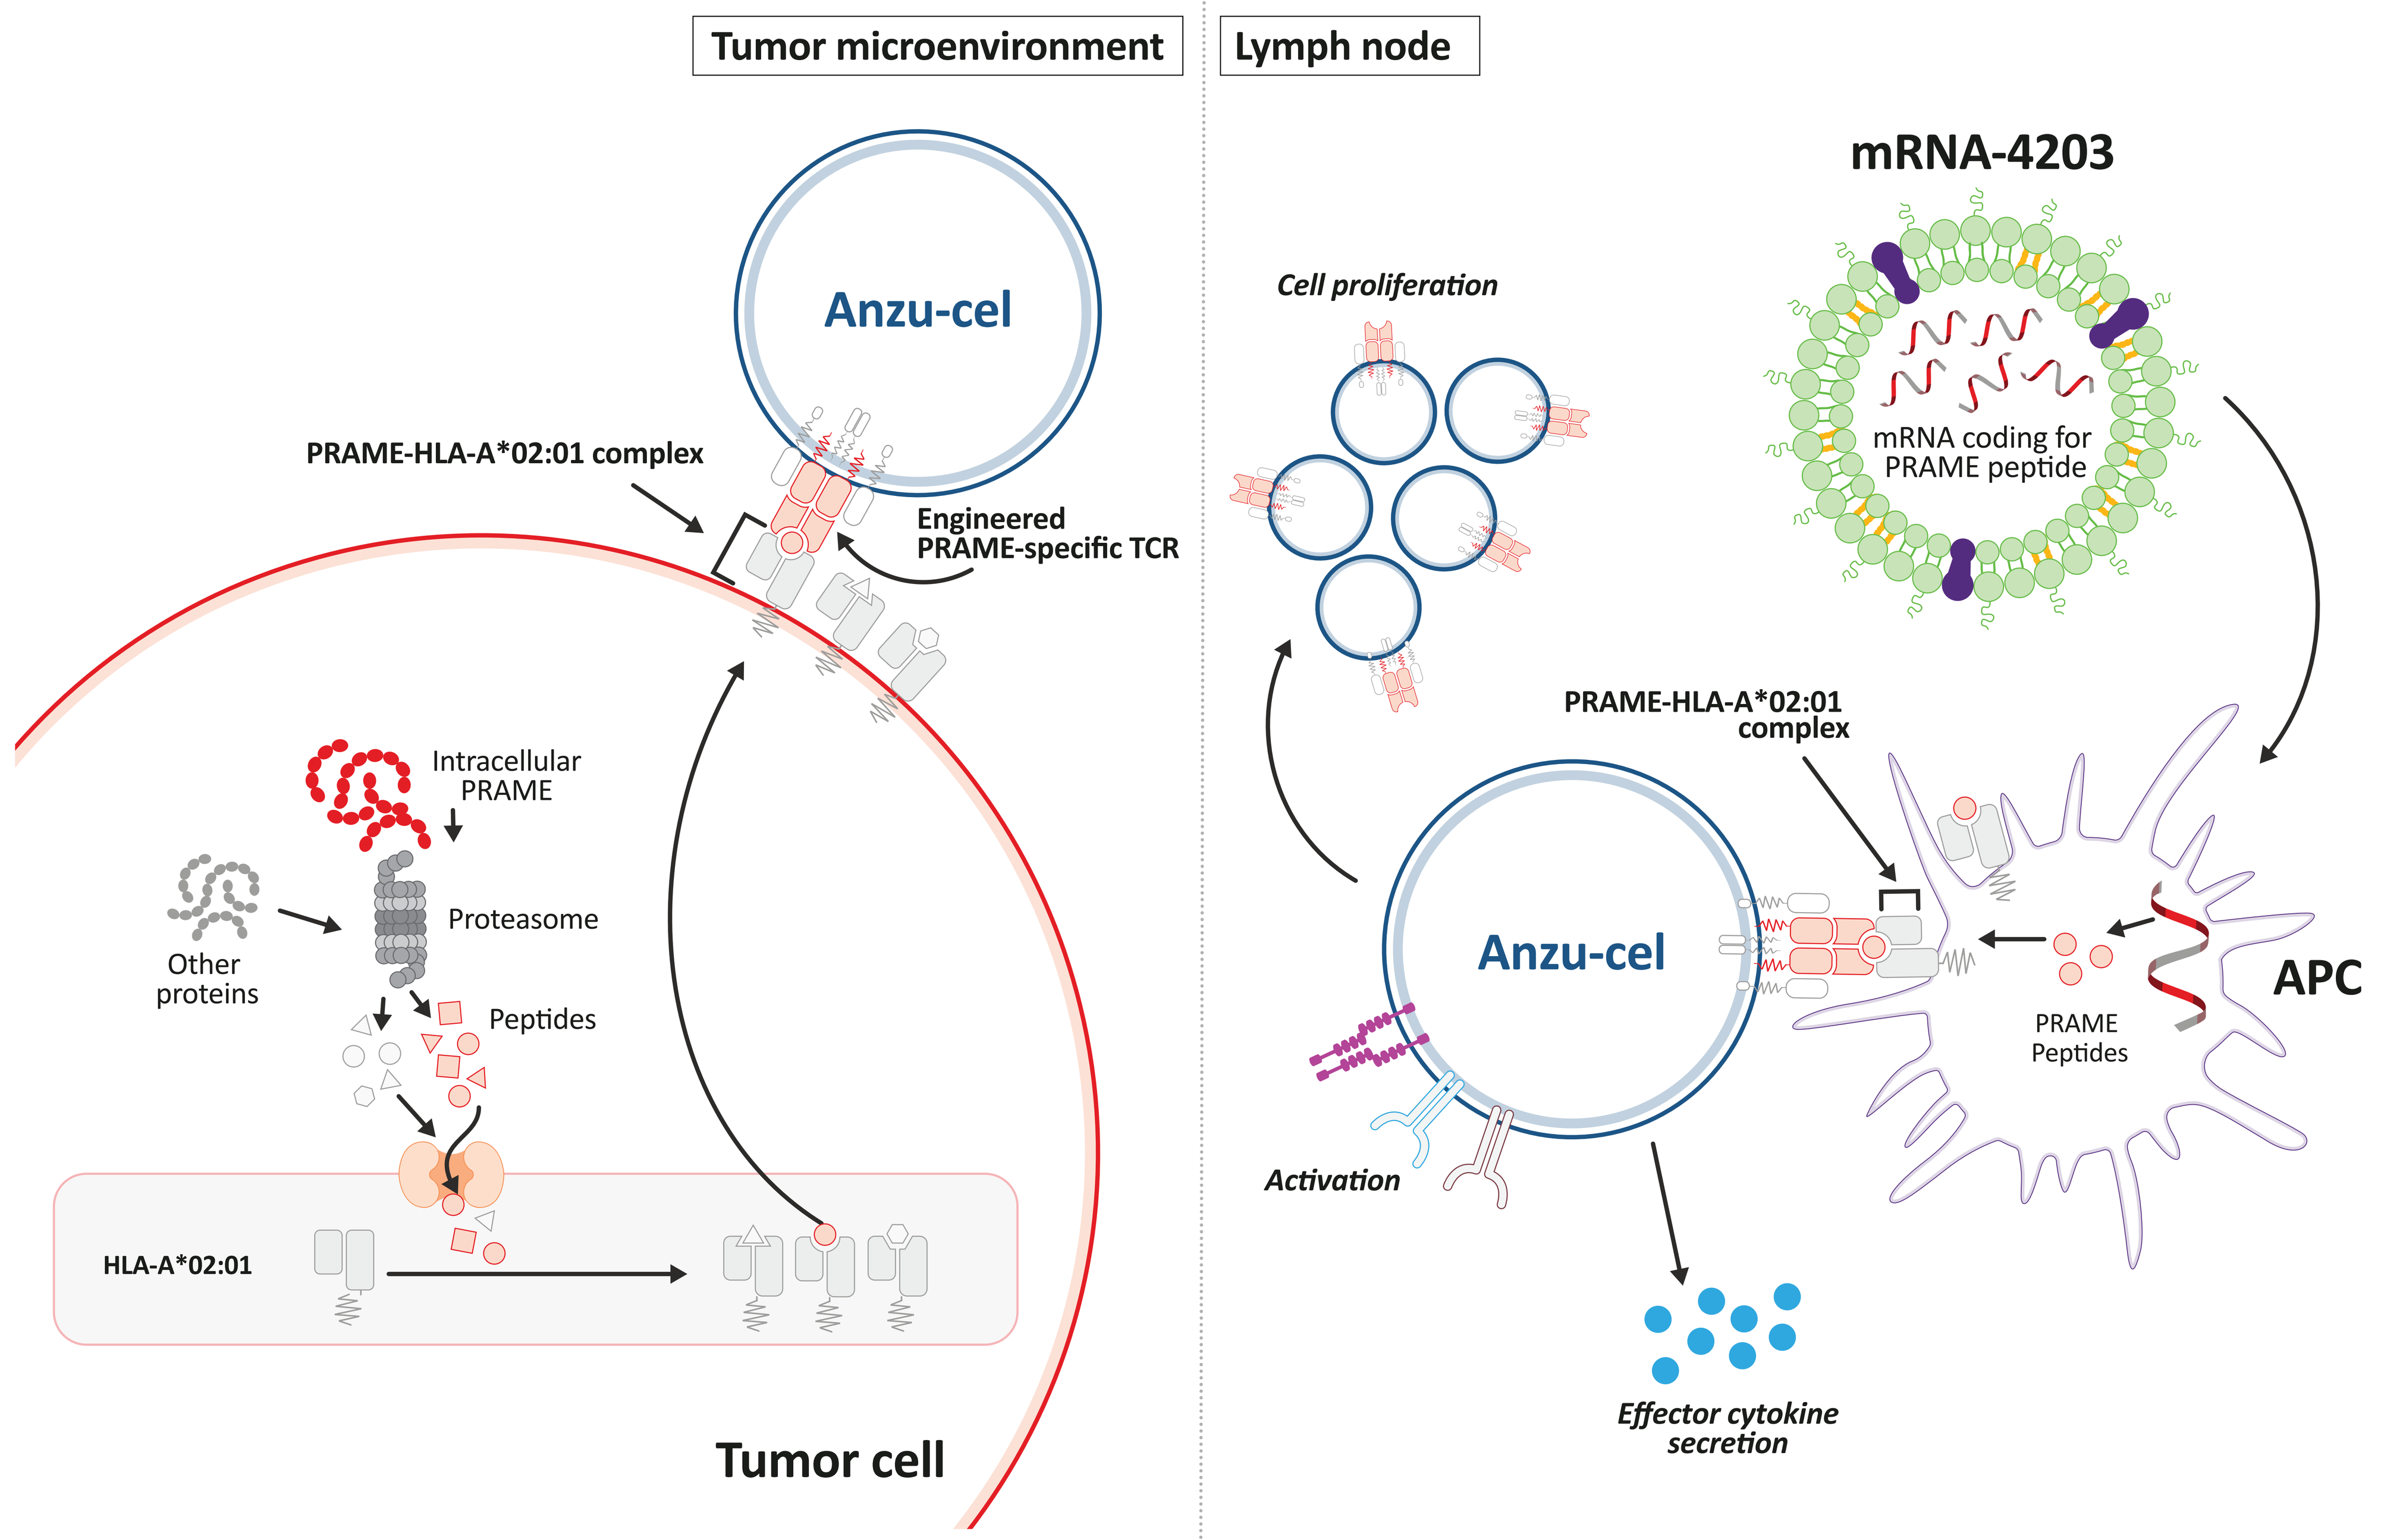

Tumor microenvironment
Lymph node
Anzu-cel
PRAME-HLA-A*02:01 complex
Engineered PRAME-specific TCR
Intracellular PRAME
Other proteins
Proteasome
Peptides
HLA-A*02:01
Tumor cell
mRNA-4203
mRNA coding for PRAME peptide
Cell proliferation
PRAME-HLA-A*02:01 complex
Anzu-cel
APC
PRAME Peptides
Activation
Effector cytokine secretion

Following one-time infusion, anzu-cel targets PRAME (presented on tumor cells across >50 cancer types), resulting in immune activation and antitumor response

Anzu-cel antitumor response is boosted by mRNA-4230 administration on Day 15 and later

METHODS

- IMA203-102 is an ongoing open-label, multicenter, first-in-human phase 1a/b trial (NCT06946225) investigating the safety, tolerability, and antitumor activity of anzu-cel in combination with different doses of mRNA-4203 in patients with CM or SS (**Fig 2**)
- Primary endpoints:
 - DLTs of the combination; safety and tolerability
- Secondary endpoints:
 - ORR, DOR, DCR, PFS, PK

Eligibility Criteria

Key Inclusion Criteria	Key Exclusion Criteria
• Unresectable or metastatic CM or SS after ≥1 line of treatment	• Serious autoimmune disease
• HLA-A*02:01 positive	• Active infections and HIV+
• ECOG PS 0-1	• LDH > 2.0 × ULN
• Measurable disease (RECIST 1.1)	• Active brain metastases
• Life expectancy >5 months	• Other malignancies (exceptions apply) within the last 3 years
• Adequate organ function	• Prior ASCT or solid organ transplantation
• Prospective PRAME testing not required	

Figure 2. Trial Design

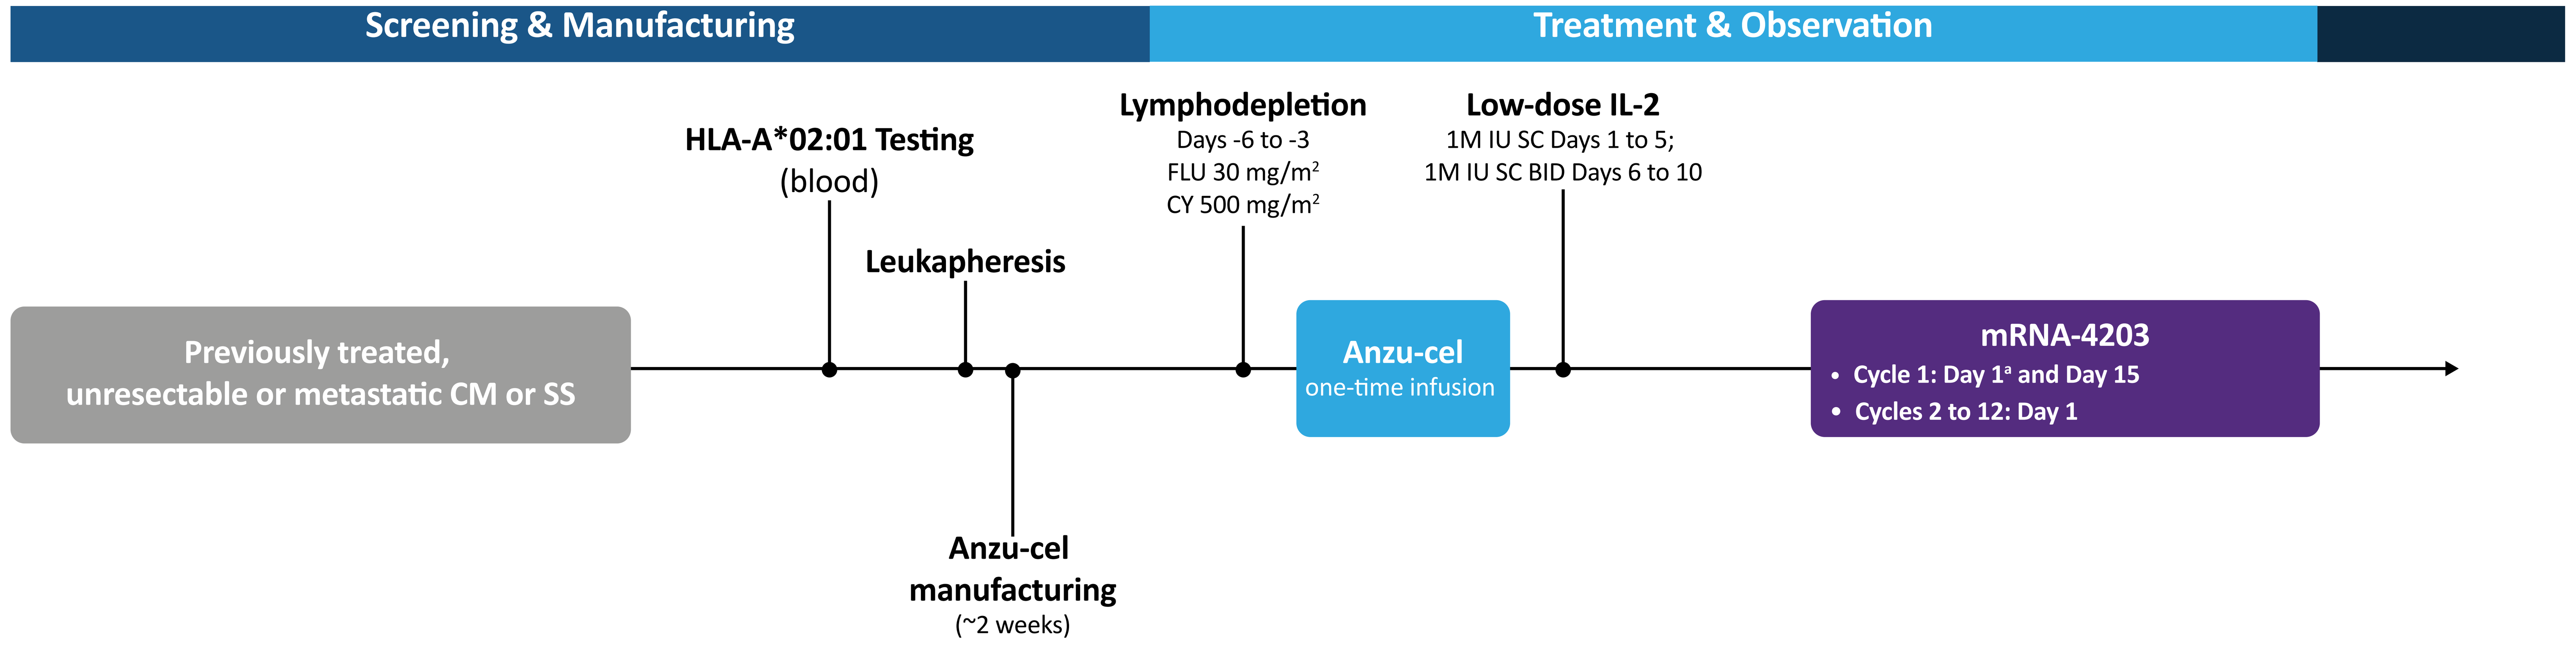

Screening & Manufacturing
Treatment & Observation
HLA-A*02:01 Testing
(blood)
Leukapheresis
Lymphodepletion
Days -6 to -3
FLU 30 mg/m2
CY 500 mg/m2
Low-dose IL-2
1M IU SC Days 1 to 5;
1M IU SC BID Days 6 to 10
Previously treated, unresectable or metastatic CM or SS
Anzu-cel
one-time infusion
mRNA-4203
• Cycle 1: Day 1a and Day 15
• Cycles 2 to 12: Day 1
Anzu-cel manufacturing
(~2 weeks)

[a] Cycles start on the day of the first mRNA-4203 administration. The first mRNA-4203 administration occurs between 15 to 71 days following anzu-cel infusion (once clinical and laboratory criteria are met).

ENROLLMENT

- The trial is expected to enroll approximately 15 patients across 4 sites in the United States

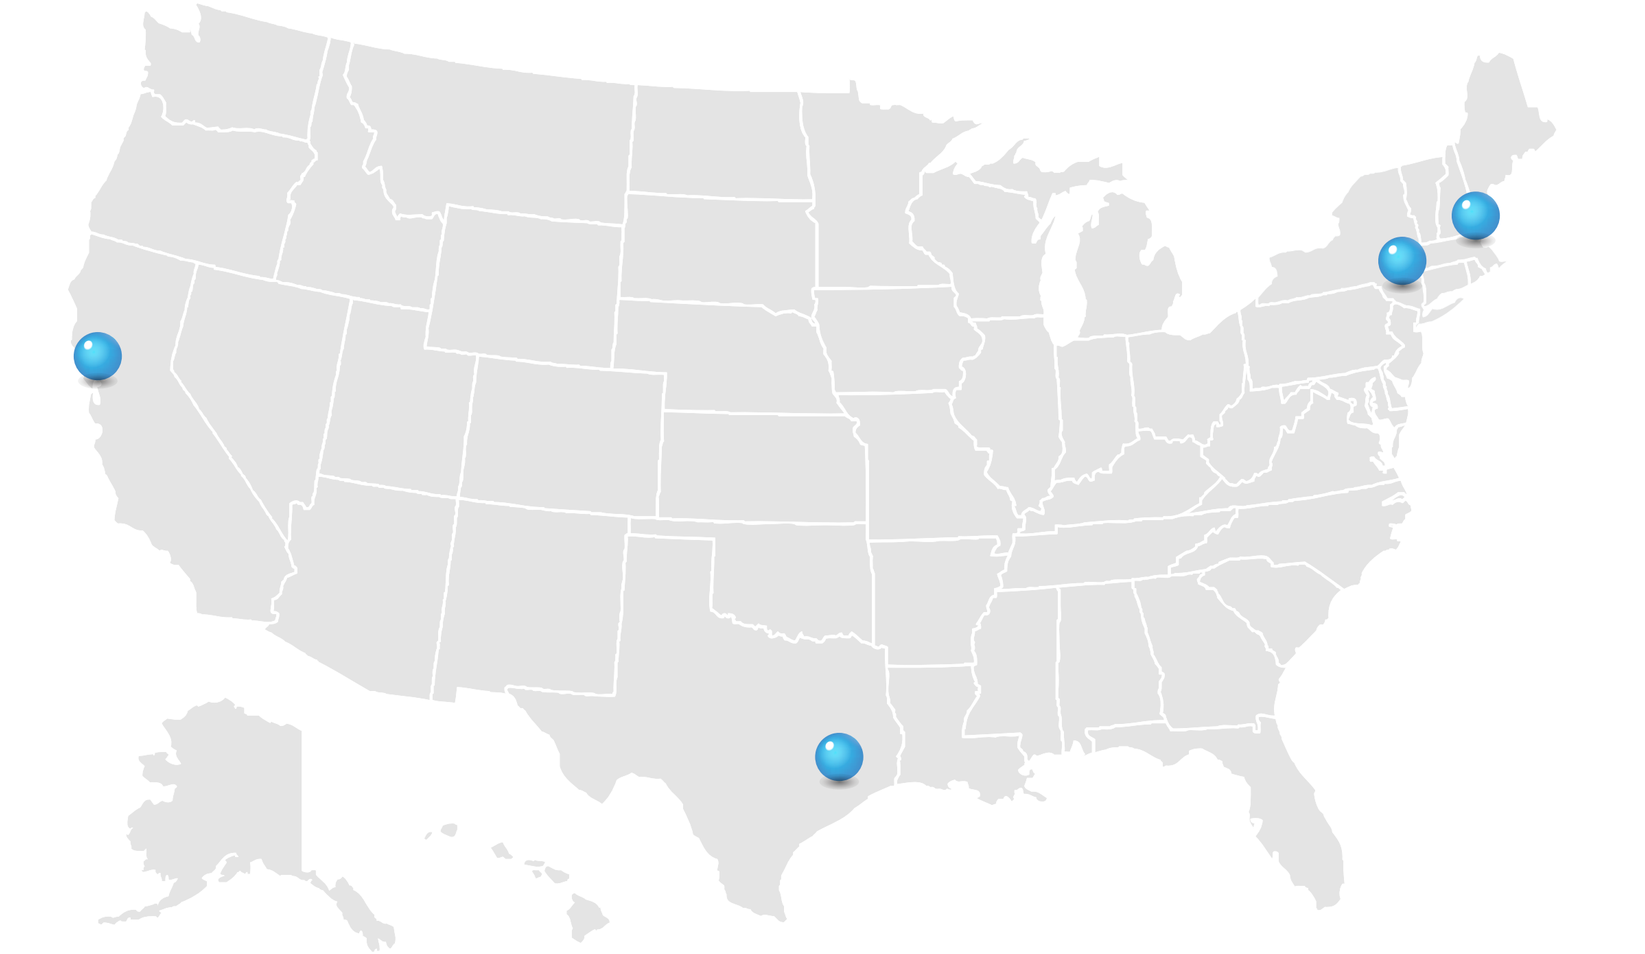

ACKNOWLEDGMENTS

- We thank the patients, their families, and caregivers for their participation in this study
- We thank the study investigators, site personnel, and independent data monitoring committee

ABBREVIATIONS: APC, antigen presenting cell; ASCT, autologous stem cell transplant; BID, twice-daily; CRS, cytokine release syndrome; CY, cyclophosphamide; DCR, disease control rate; DLT, dose-limiting toxicity; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; FLU, fludarabine; G; grade; HIV, human immunodeficiency virus; HLA, human leukocyte antigen; ICANS, immune effector cell-associated neurotoxicity syndrome; IHC, immunohistochemistry; IL-2, interleukin-2; LDH, lactate dehydrogenase; LNP, lipid nanoparticle; mRNA, messenger ribonucleic acid; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; PK, pharmacokinetics; PRAME, preferentially expressed antigen in melanoma; RECIST, Response Evaluation Criteria in Solid Tumors; RP2D, recommended phase 2 dose; SC, subcutaneous; TEAE, treatment-emergent adverse event; TCR, T-cell receptor; ULN, upper limit of normal.

REFERENCES:

1. Wermke M, et al. *Nat Med*. 2025; 31:2365-2374.
2. Data on file; PRAME target prevalence is based on a proprietary mass spec-guided expression threshold applied to RNAseq and/or IHC data.
3. Field MG, et al. *Clin Cancer Res*. 2016;22:1234-1242.
4. Wermke M, et al. Presented at: American Society of Clinical *Oncology* (ASCO) 2025. Abstr 2508.

Please contact medinfo@immatics.com for additional information

FPO

Presented at the Society for Immunotherapy of Cancer (SITC) 40th Anniversary Annual Meeting • National Harbor, MD • November 5-9, 2025